The role of SHOX gene in Idiopathic Short Stature: an Italian multicenter study

R. Minari*[1], A. Vottero[1], S. Azzolini[2], D. Barbaro[3], G. Bindi[4], M. Bozzola[5], C. Burrai[6], G. M. Cardinale[7], D. Cioffi[8], M. Cisternino[9], M. S. Coccioli[10], M. Delvecchio[11], E. Fabbrizi[12], M. Ferrari[13], F. Gallarotti[14], F. Gallo[15], L. Ghizzoni[16], M. C. Maggio[17], B. Mainetti[18], R. Montinaro[19], G. Municchi[20], A. Panariello[21], M. Parpagnoli[22], L. Perrone[23], M. Petraroli[24], G. Radetti[25], A. F. Radicioni[26], A. Rossodivita[27], M. C. Salerno[28], S. Savasta[9], S. Seminara[22], L. Tafi[29], M. Tomat[30], A. Tummolo[31], M. Wasniewska[32], L. Iughetti[33] and S. Bernasconi[1].

[1]University of Parma, Department of Clinical and Experimental Medicine, Parma, Italy; [2]University Hospital of Padua, Pediatric Endocrinology and Adolescence Unit at Pediatric Department, Padua, Italy; [3]ASL 6, Section of Endocrinology, Livorno, Italy; [4] Pistoia Hospital, Unit of Pediatrics, Pistoia, Italy; [5]San Matteo Hospital, Unit of Auxology, Pavia, Italy; [6]University Hospital of Sassari, Endocrinology Unit, Sassari, Italy; [7]Ferrari Hospital, Unit of Pediatrics, Casarano (LE), Italy; [8]Santobono Pausilipon Hospital, Auxology and Endocrinology Units, Naples, Italy; [9]San Matteo Hospital, Unit of Pediatrics, Pavia, Italy; [10]Camberlingo Hospital, Unit of Pediatrics, Francavilla Fontana (BR), Italy; [11]IRCCS Casa Sollievo della Sofferenza, Pediatrics Unit, San Giovanni Rotondo (FG), Italy; [12]Civili Hospital, Unit of Pediatrics, Fermo, Italy; [13]AUSL 1, Department of Pediatrics, Massa-Carrara, Italy; [14]"S. Croce e Carle" Hospital, Department of Pediatrics, Cuneo, Italy; [15]Perrino Hospital, Unit of Pediatrics, Brindisi, Italy; [16]University of Turin, Department of Internal Medicine, Division of Endocrinology and Metabolism, Turin, Italy; [17]"G.Di Cristina" Hospital, Unit of Pediatrics, Palermo, Italy; [18]AUSL, Unit of Pediatrics, Forlì, Italy; [19]"S. Caterina Novella" Hospital, Unit of Pediatrics, Galatina (LE), Italy; [20]University of Siena, Pediatric Department, Siena, Italy; [21]San Salvatore Hospital, Department of Pediatrics, Pesaro, Italy; [22]Meyer Children Hospital, Department of Pediatrics, Florence, Italy; [23]Second University of Naples, Pediatric 2 Unit Department "Donna, Bambino e Chirurgia Generale e Specialistica", Naples, Italy; [24]Santa Maria Nuova Hospital, Unit of Pediatrics, Reggio Emilia, Italy; [25]Regional Hospital, Department of Pediatrics, Bolzano, Italy; [26]Sapienza University, Department of Experimental Medicine, Rome, Italy; [27]UCSC, Department of Pediatrics, Rome, Italy; [28]University of Naples "Federico II", Department of Pediatrics, Naples, Italy; [29]AUSL 8, Department of Pediatrics, Arezzo, Italy; [30]University Hospital of Udine, Unit of Pediatrics, Udine, Italy; [31]Children Hospital "Giovanni XXIII", Clinical Genetics and Metabolic Diseases Unit, Bari, Italy; [32]University of Messina, Department of Pediatrics, Messina, Italy; [33]University of Modena and Reggio Emilia, Department of Pediatrics, Modena, Italy.

P1-D1-147

BACKGROUND

- The short stature homeobox-containing (SHOX) gene, located in the telomeric pseudoautosomal region 1 (PAR1) on the short arm of both sex chromosomes, is important for linear growth.
- AIM: to evaluate the presence of SHOX gene deletions/point mutations in children with short stature in order to understand the role of SHOX gene in Idiopathic Short Stature (ISS) and estimate its frequency.

MATERIALS AND METHODS

This study, supported by the Eli Lilly Italia and approved by the Italian Society for Pediatric Endocrinology and Diabetes (ISPED), is a multicenter study involving several Italian Pediatric Endocrinology Units. Out of a total number of 152 blood samples received, 68 were from patients with ISS.

Figure 1. Patients' classification by phenotype

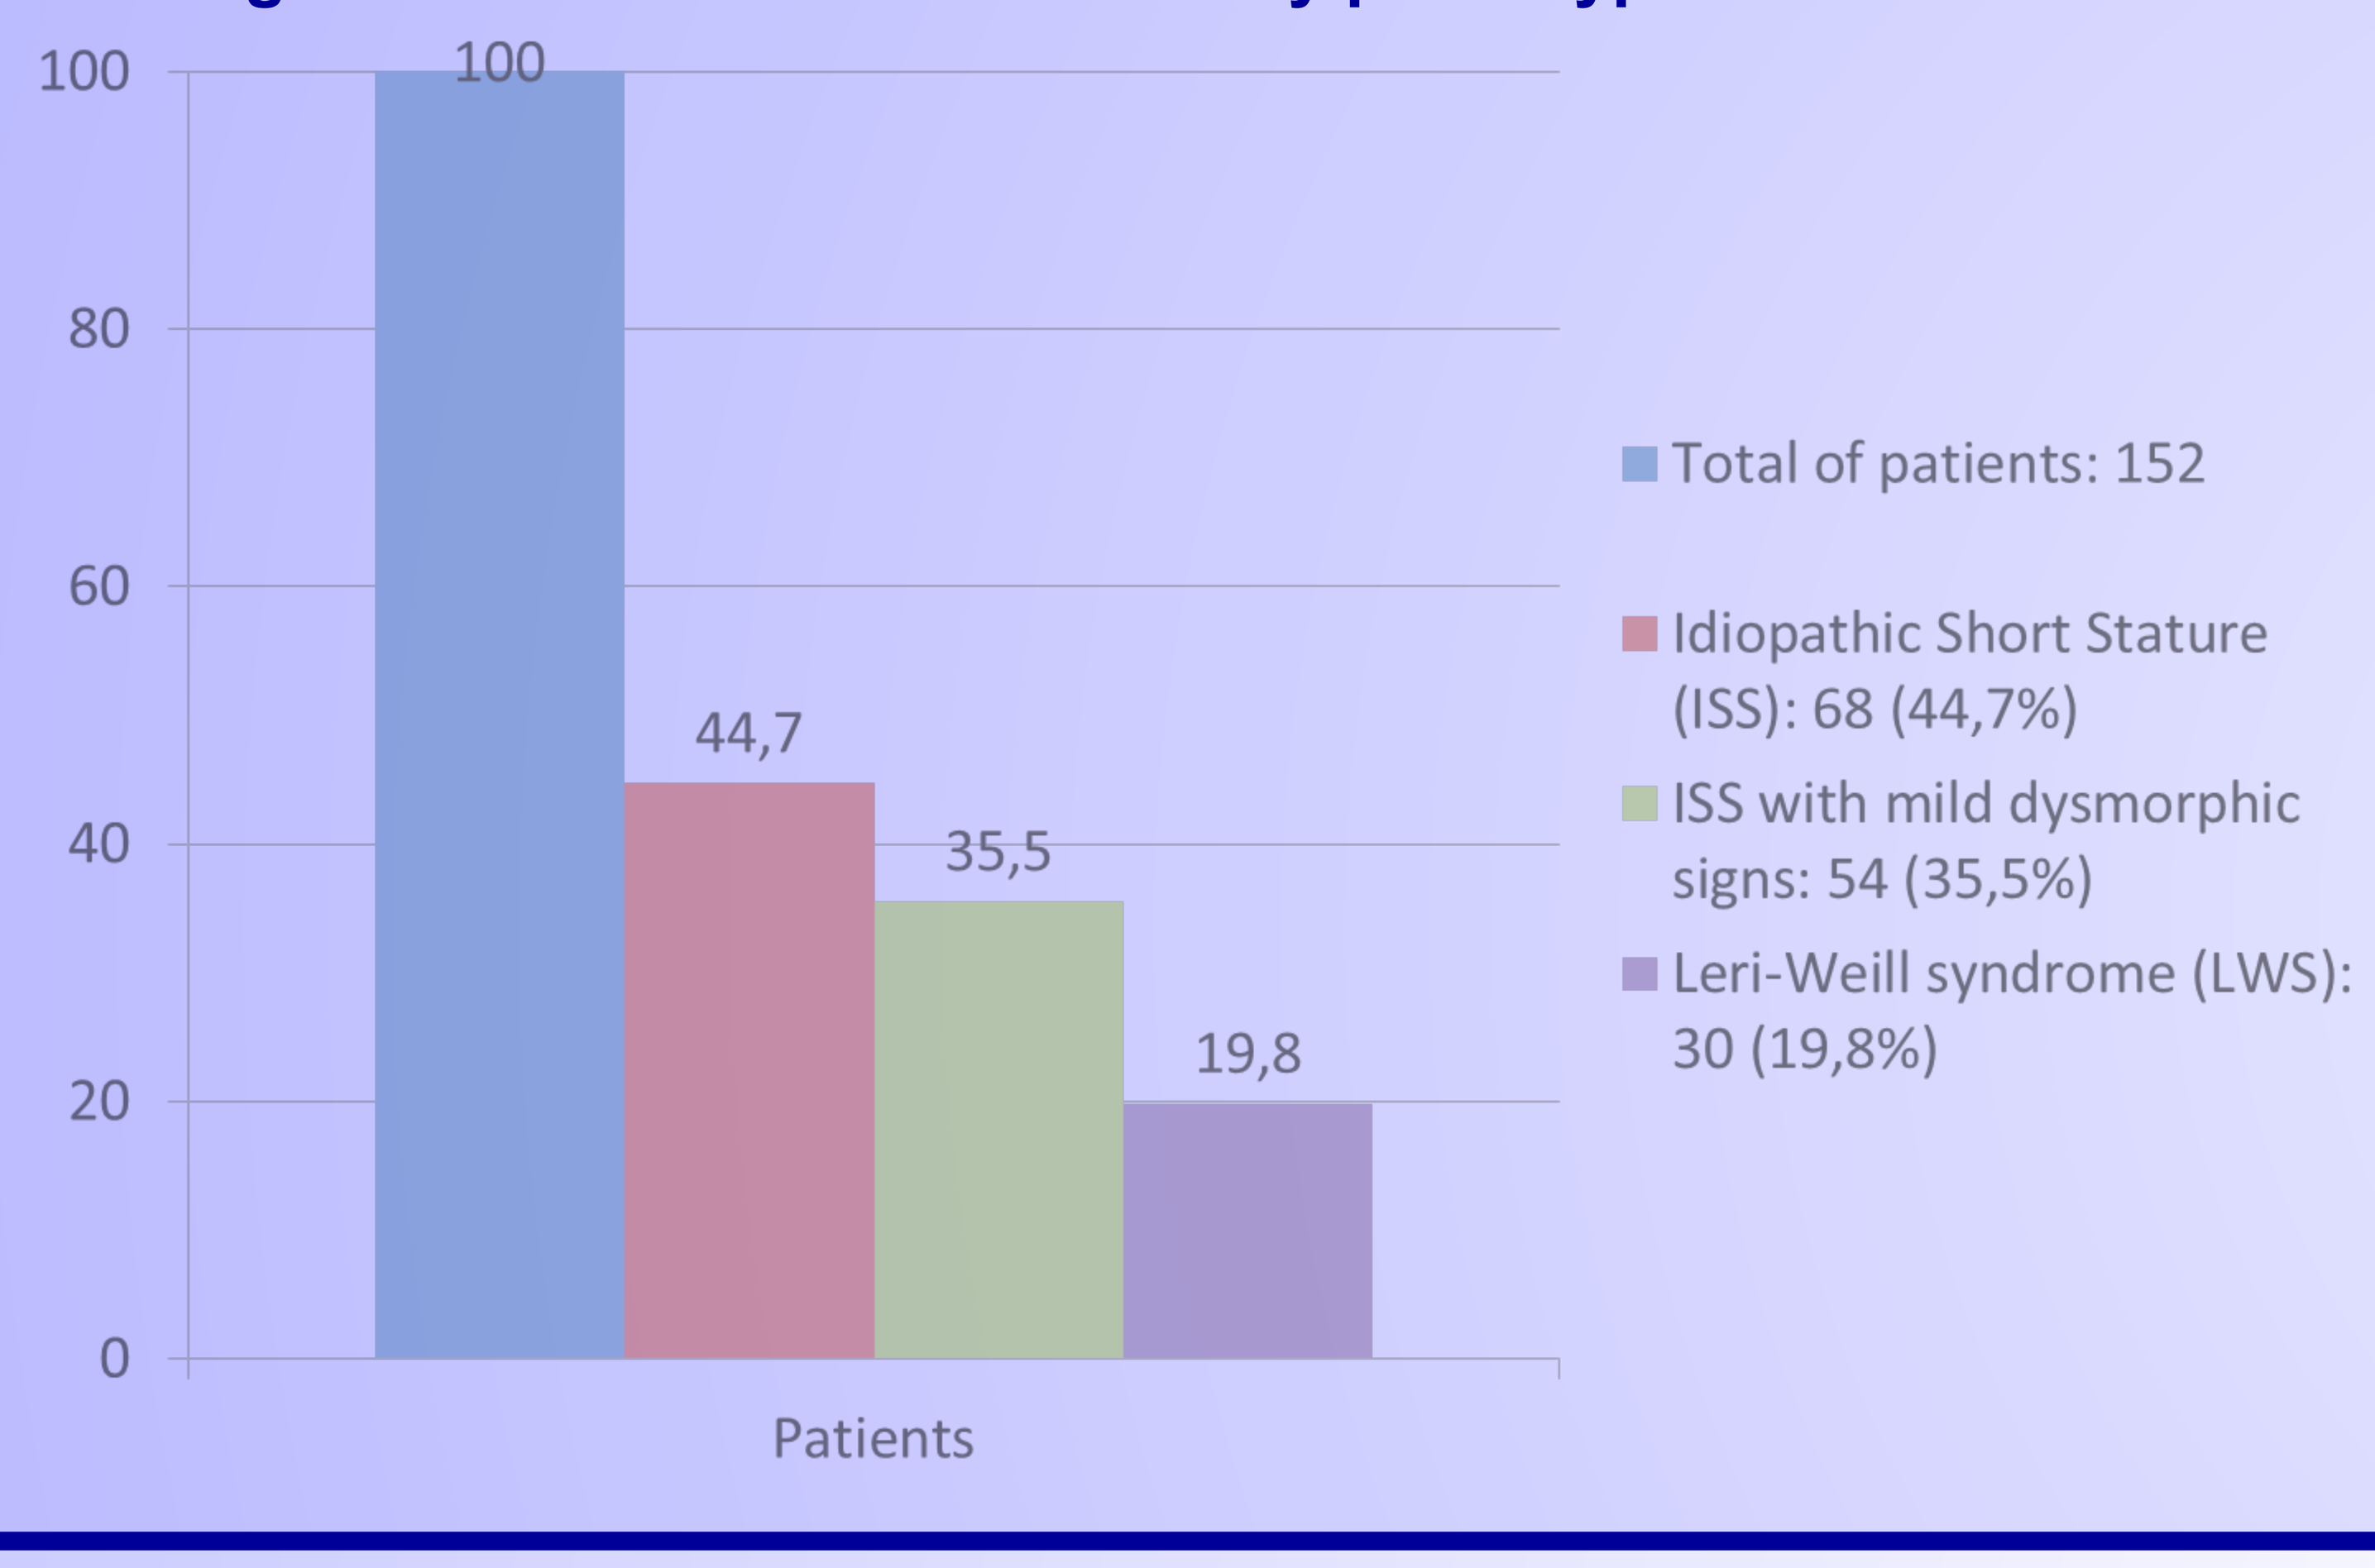

100
80
60
40
20
0
100
44,7
35,5
19,8
Patients
Total of patients: 152
Idiopathic Short Stature (ISS): 68 (44,7%)
ISS with mild dysmorphic signs: 54 (35,5%)
Leri-Weill syndrome (LWS): 30 (19,8%)

Table 1. Clinical data of the study population

	ISS	ISS with mild dysmorphic signs	LWS
Chronological age, years	9,83 ± 4,34	9,07 ± 3,11	9,06 ± 5,39
Sex, male/female	30/38	23/31	11/19
Height, cm	124,07 ± 28,75	119,71 ± 16,64	114,85 ± 23,65
Height, SDS	-2,25 ± 0,83	-2,30 ± 0,88	-2,15 ± 1,00
Span, cm	121,19 ± 23,30	117,36 ± 17,40	113,87 ± 21,25
Sitting height/Height	0,54 ± 0,03	0,54 ± 0,06	0,56 ± 0,03
BMI, kg/m2	18,17 ± 3,78	16,64 ± 2,60	17,79 ± 4,26
BMI, SDS	-0,31 ± 1,24	-0,52 ± 1,24	-0,41 ± 1,61
Ogive palat	0/68	37/54	12/30
Brachimetacarpia 4th finger	0/68	14/54	5/30
Cubitus Valgus	0/68	27/54	15/30
Mesomelia	0/68	0/54	8/30
Madelung deformity	0/68	0/54	19/30

Genomic DNA was extracted and used for Multiplex Ligation-dependent Probe Amplification (MLPA) and sequencing analysis. MLPA was performed in the Pediatric Laboratory of Parma, using the SALSA MLPA P018-F1 SHOX probemix kit, analyzing both the coding region and the enhancer of the SHOX gene. Deletions and duplications were confirmed with Copy Number Assay (Applied Biosystems) in Real Time PCR.

RESULTS

Table 2. Phenotype and genotype characteristics of patients with mutations

Case	Gender	Age, years	Genotype	Phenotype	Height (SDS)
1	F	10,4	Complex caryotype	Short stature	-2,42
2	M	13	SHOX deletion	Short stature, mesomelia	-1.24
3	F	8,2	SHOX deletion	Short stature, mesomelia	-2,35
4	F	11,6	LOH CRLF2 gene	Short stature, minor skeletal alteration	-2,4
5	F	7,5	LOH CRLF2 gene	Short stature, minor skeletal alteration	-1,76
6	F	8	SHOX deletion	Short stature	-3,19
7	M	11,9	Gain CRLF2	Short stature	-1,7
8	M	7,7	Gain SHOX	Short stature, minor skeletal alteration	-2,16
9	M	12,6	46, X, der(Y)	Short stature, minor skeletal alteration	-1,98
10	M	12,8	Gain CRLF2	Short stature, Madelung deformity	-3,14
11	F	6,4	46,X i (Xq10)	Short stature	-3,84
12	M	11,3	SHOX deletion	Short stature, Madelung deformity	-1,17
13	F	0,3	SHOX deletion	Short stature	-2,08
14	M	7,3	Gain CRLF2	Short stature	-2,66
15	F	2,9	Mutation	Short stature	-1,3
16	M	8,8	Gain CRLF2	Short stature, minor skeletal alteration	-1,94
17	F	15	Gain CRLF2	Short stature	-1,65
18	F	7	LOH CNE8 and 9	Short stature, minor skeletal alteration	-2,98
19	M	10,5	LOH CNE9	Short stature, Madelung deformity	-1,3
20	M	9,9	SHOX deletion	Short stature, Madelung deformity	-2,52
21	F	12	SHOX deletion	Short stature, Madelung deformity	0,72
22	F	5,9	LOH SHOX area	Short stature	-2,92
23	F	4,1	SHOX deletion	Short stature, Madelung deformity	-2,05
24	F	11,9	Mutation	Short stature, minor skeletal alteration	-2,13
25	M	10,2	SHOX deletion	Short stature	-1,49
26	F	9,5	LOH CNE8 and 9	Short stature, minor skeletal alteration	-2,22
27	F	7,9	SHOX deletion	Short stature	-3,21
28	M	14,9	Mutation	Short stature	-1,04
29	M	6,2	Dup CNE8 and 9	Short stature, minor skeletal alteration	-2,22
30	M	3,4	SHOX deletion	Short stature	-1,97
31	F	2,3	Mutation	Short stature	-2,37
32	F	12,6	Mutation	Short stature, Madelung deformity	-1,4

Figure 2. Genotype/phenotype correlation and frequency

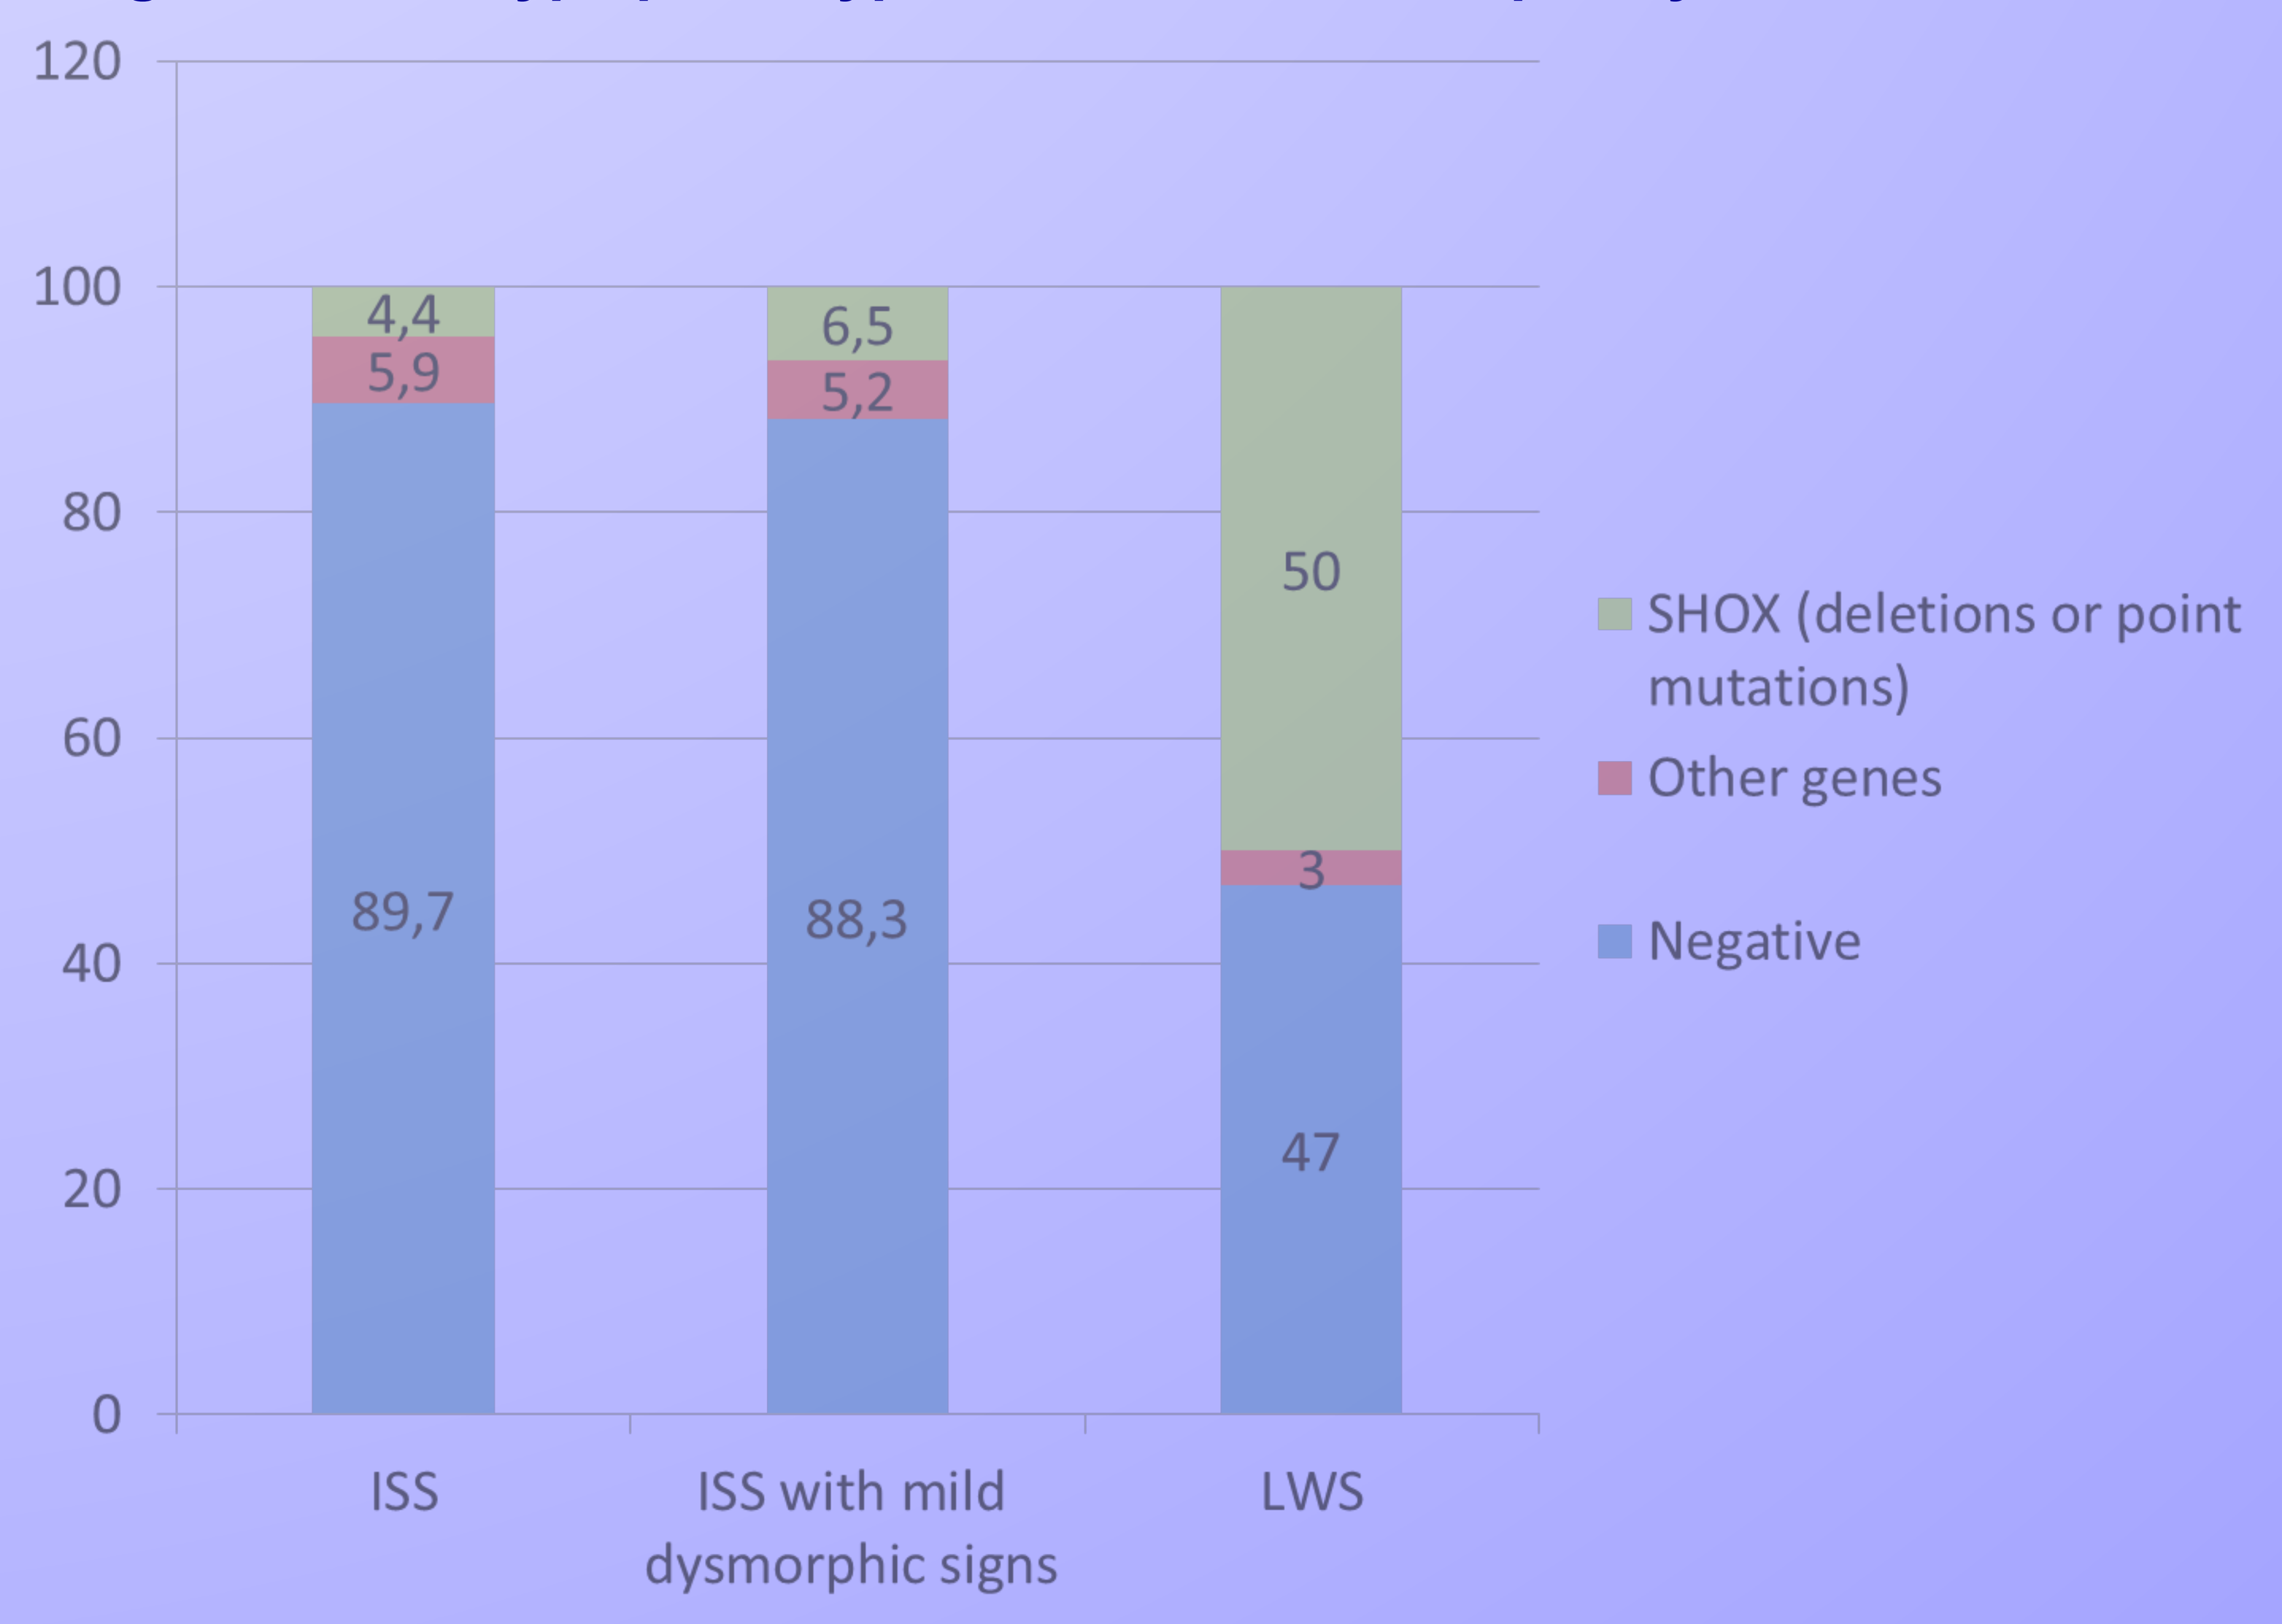

120
100
80
60
40
20
0
4,4
5,9
89,7
6,5
5,2
88,3
50
3
47
ISS
ISS with mild dysmorphic signs
LWS
SHOX (deletions or point mutations)
Other genes
Negative

CONCLUSIONS

If we exclude the patients with Leri-Weill syndrome who presented a SHOX gene deletion in 50%, in our cohort of patients with ISS the incidence of SHOX gene alterations was very low (4,4%), suggesting that the presence of mesomelia, minor skeletal abnormalities, and eventually subtle radiographic signs are essential for requiring genetic analysis properly.