Regulation of bone growth via ligand-specific activation of estrogen receptor alpha

Maryam Iravani[1], Marie Lagerquist[2], Claes Ohlsson[2] and Lars Sävendahl[1]

Disclosure statement: The project was funded partly by an unrestricted grant from Pfizer.

Conclusions

- **Selective ERα agonist (PPT) or estradiol (E2) treatments decrease tibia and femur longitudinal growth and the height of the tibia growth plate cartilage in female mice.**
- **Our data show that the estrogenic effects on bone growth and growth plate maturation are mainly mediated via ERα.**

Aim

The aim of the study was to evaluate the effects of estradiol (E2) and selective ERα (PPT), ERβ (DPN) and GPER-1 (G1) agonists on bone growth and growth plate maturation.

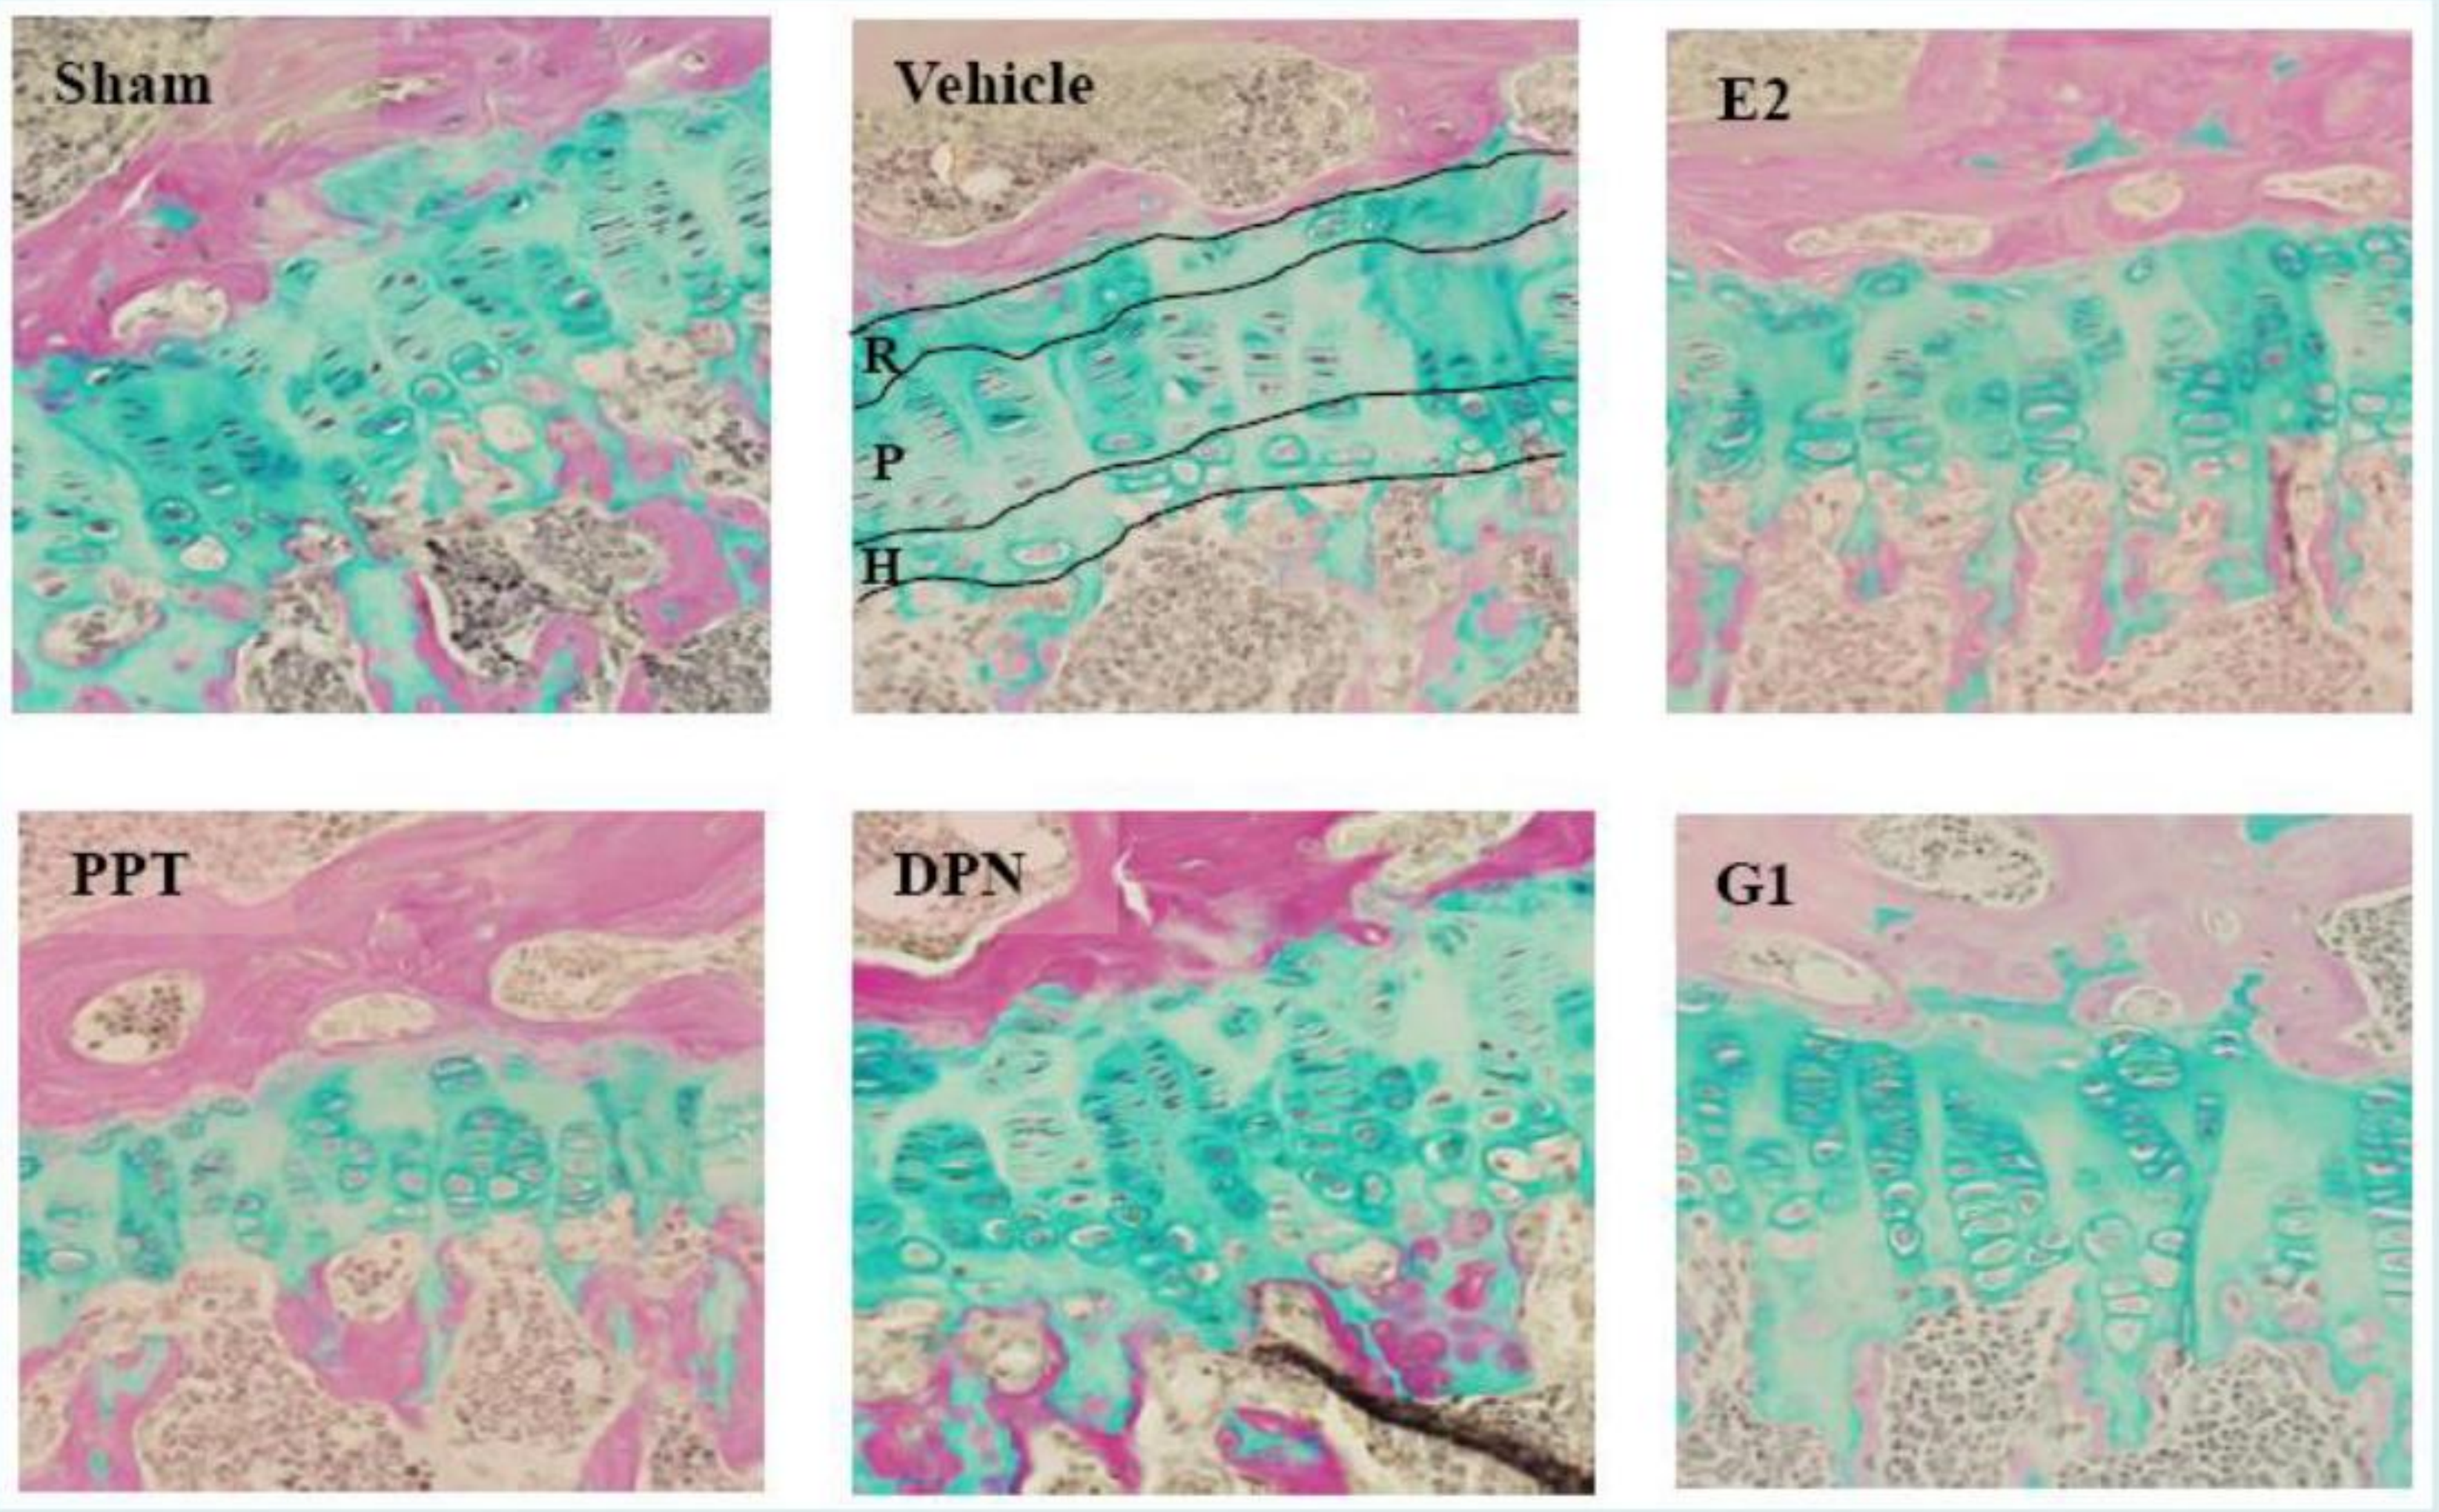
Sham
Vehicle
R
P
H
E2
PPT
DPN
G1

Figure 1. Effects of E2 and selective ERα (PPT), ERβ (DPN) and GPER-1 (G1) agonists on tibia growth plate length and different chondrocytes zones. R=resting zone, P=proliferative zone and H=hypertrophic zone.

Introduction

Estrogens are well known to promote bone maturation and at high doses to induce growth plate closure and thereby stop further growth. High-dose estrogen treatment has therefore been used to limit growth in extremely tall girls. However, recent data suggest that this treatment may have severe side effects, including increased risk of cancer and reduced fertility.

Results

- E2 and PPT treated mice had shorter tibia and femur when compared to vehicle treated controls while those animals treated with DPN and G1 had similar bone lengths as controls (Fig. 1 and 2A).
- The histological evaluation of PCNA staining of tibia growth plate cartilage showed lower proliferation in E2 or PPT treated OVX mice compared to OVX vehicle (Fig. 2B).
- Growth plate height and proliferative zone height were reduced in animals treated with E2 or PPT but not in those treated with DPN or G1 (Fig. 1 and 3).

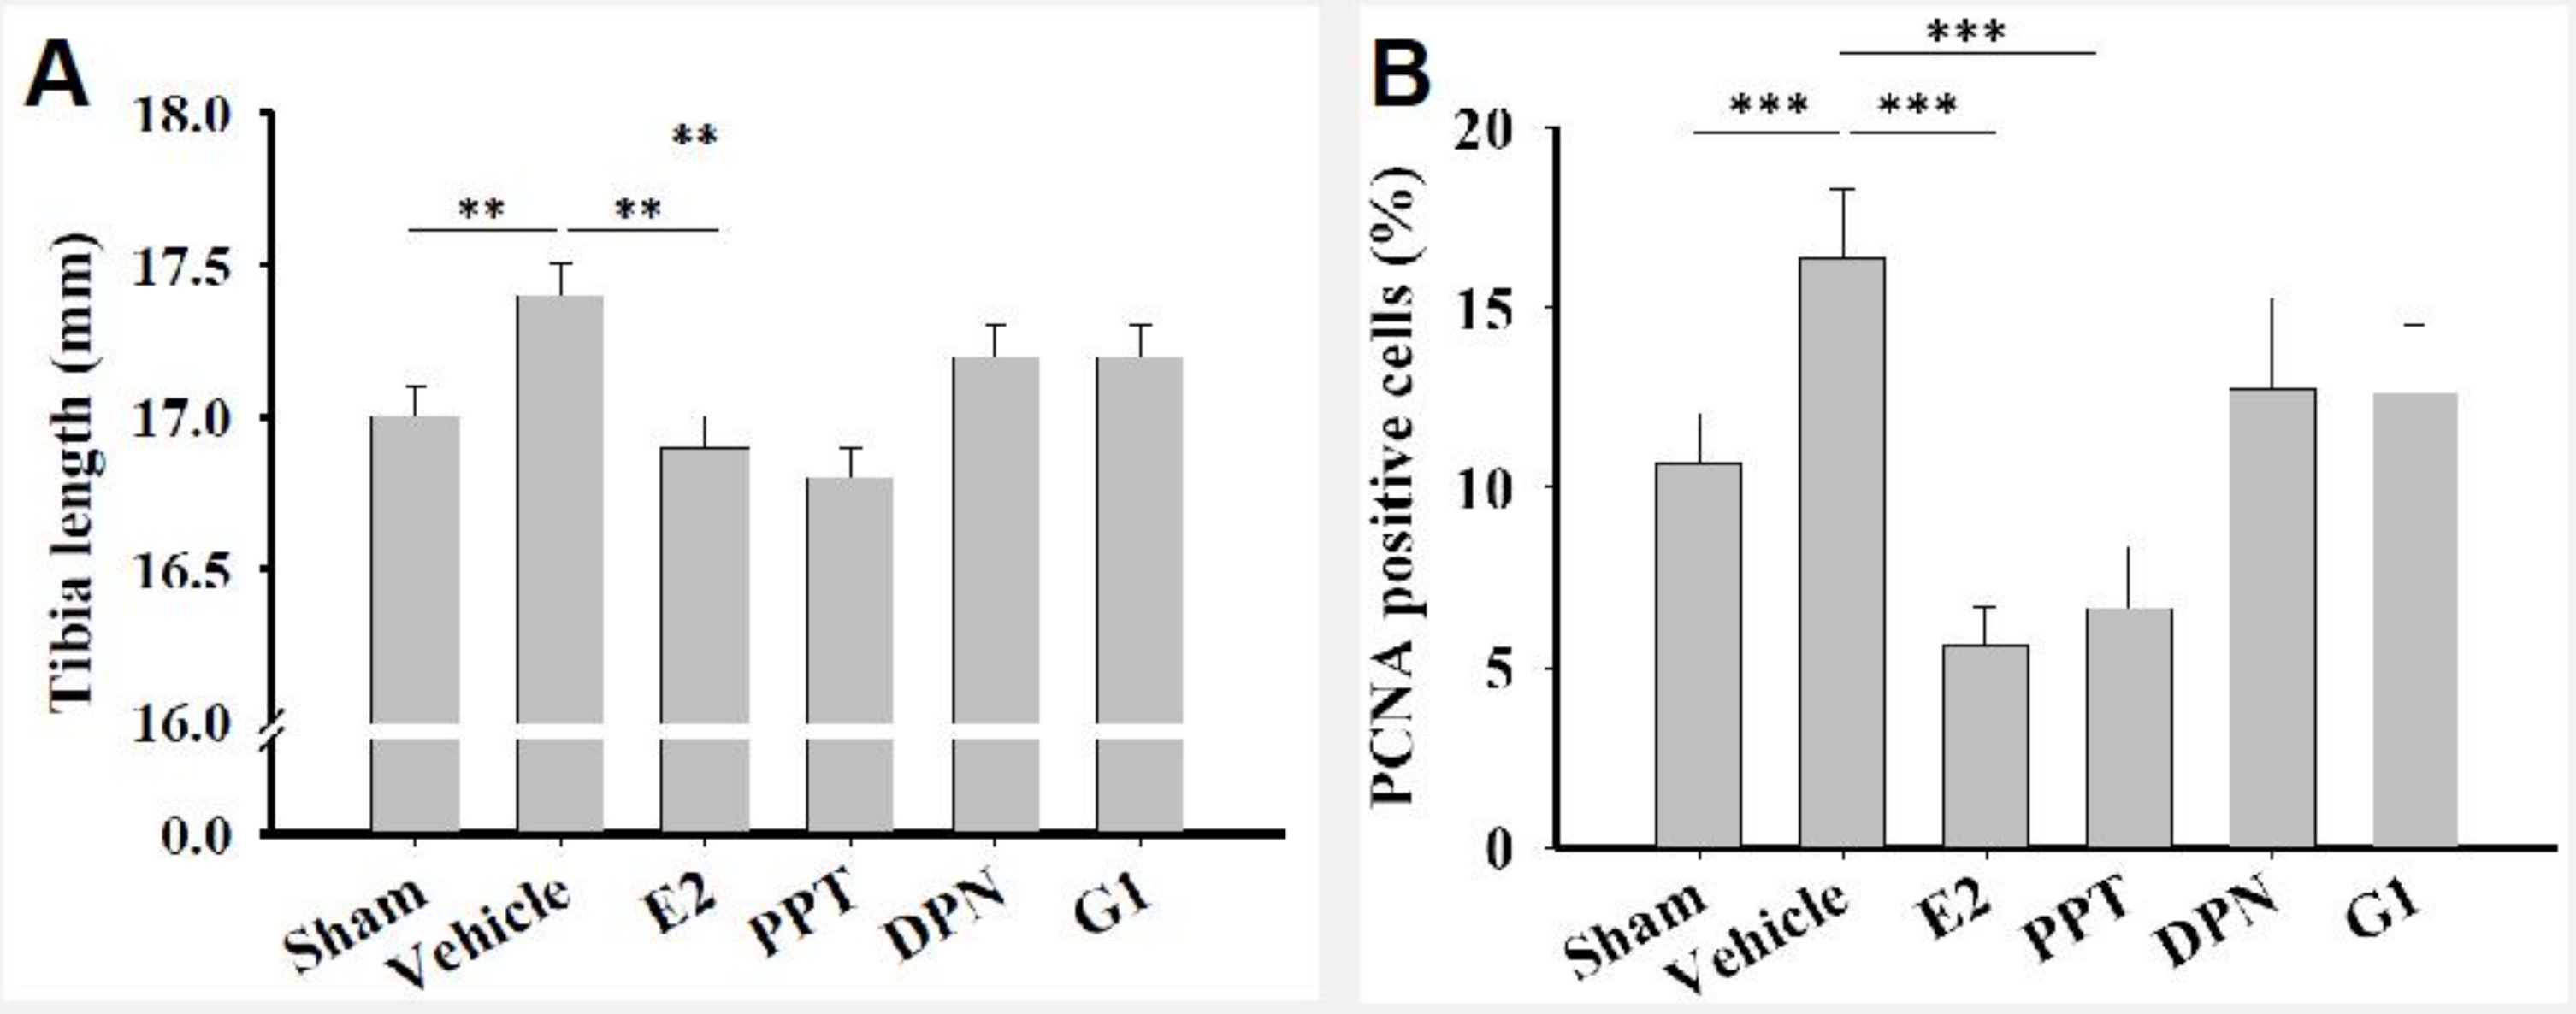
A
Tibia length (mm)
18.0
17.5
17.0
16.5
16.0
0.0
**
**
**
Sham
Vehicle
E2
PPT
DPN
G1
B
PCNA positive cells (%)
20
15
10
5
0

Figure 2. Effects of E2 and selective ER agonists on tibia length and chrondrocyte proliferation (A) and on proliferation (B). Values are means ± SEM. **p<0.01and ***p<0.001.

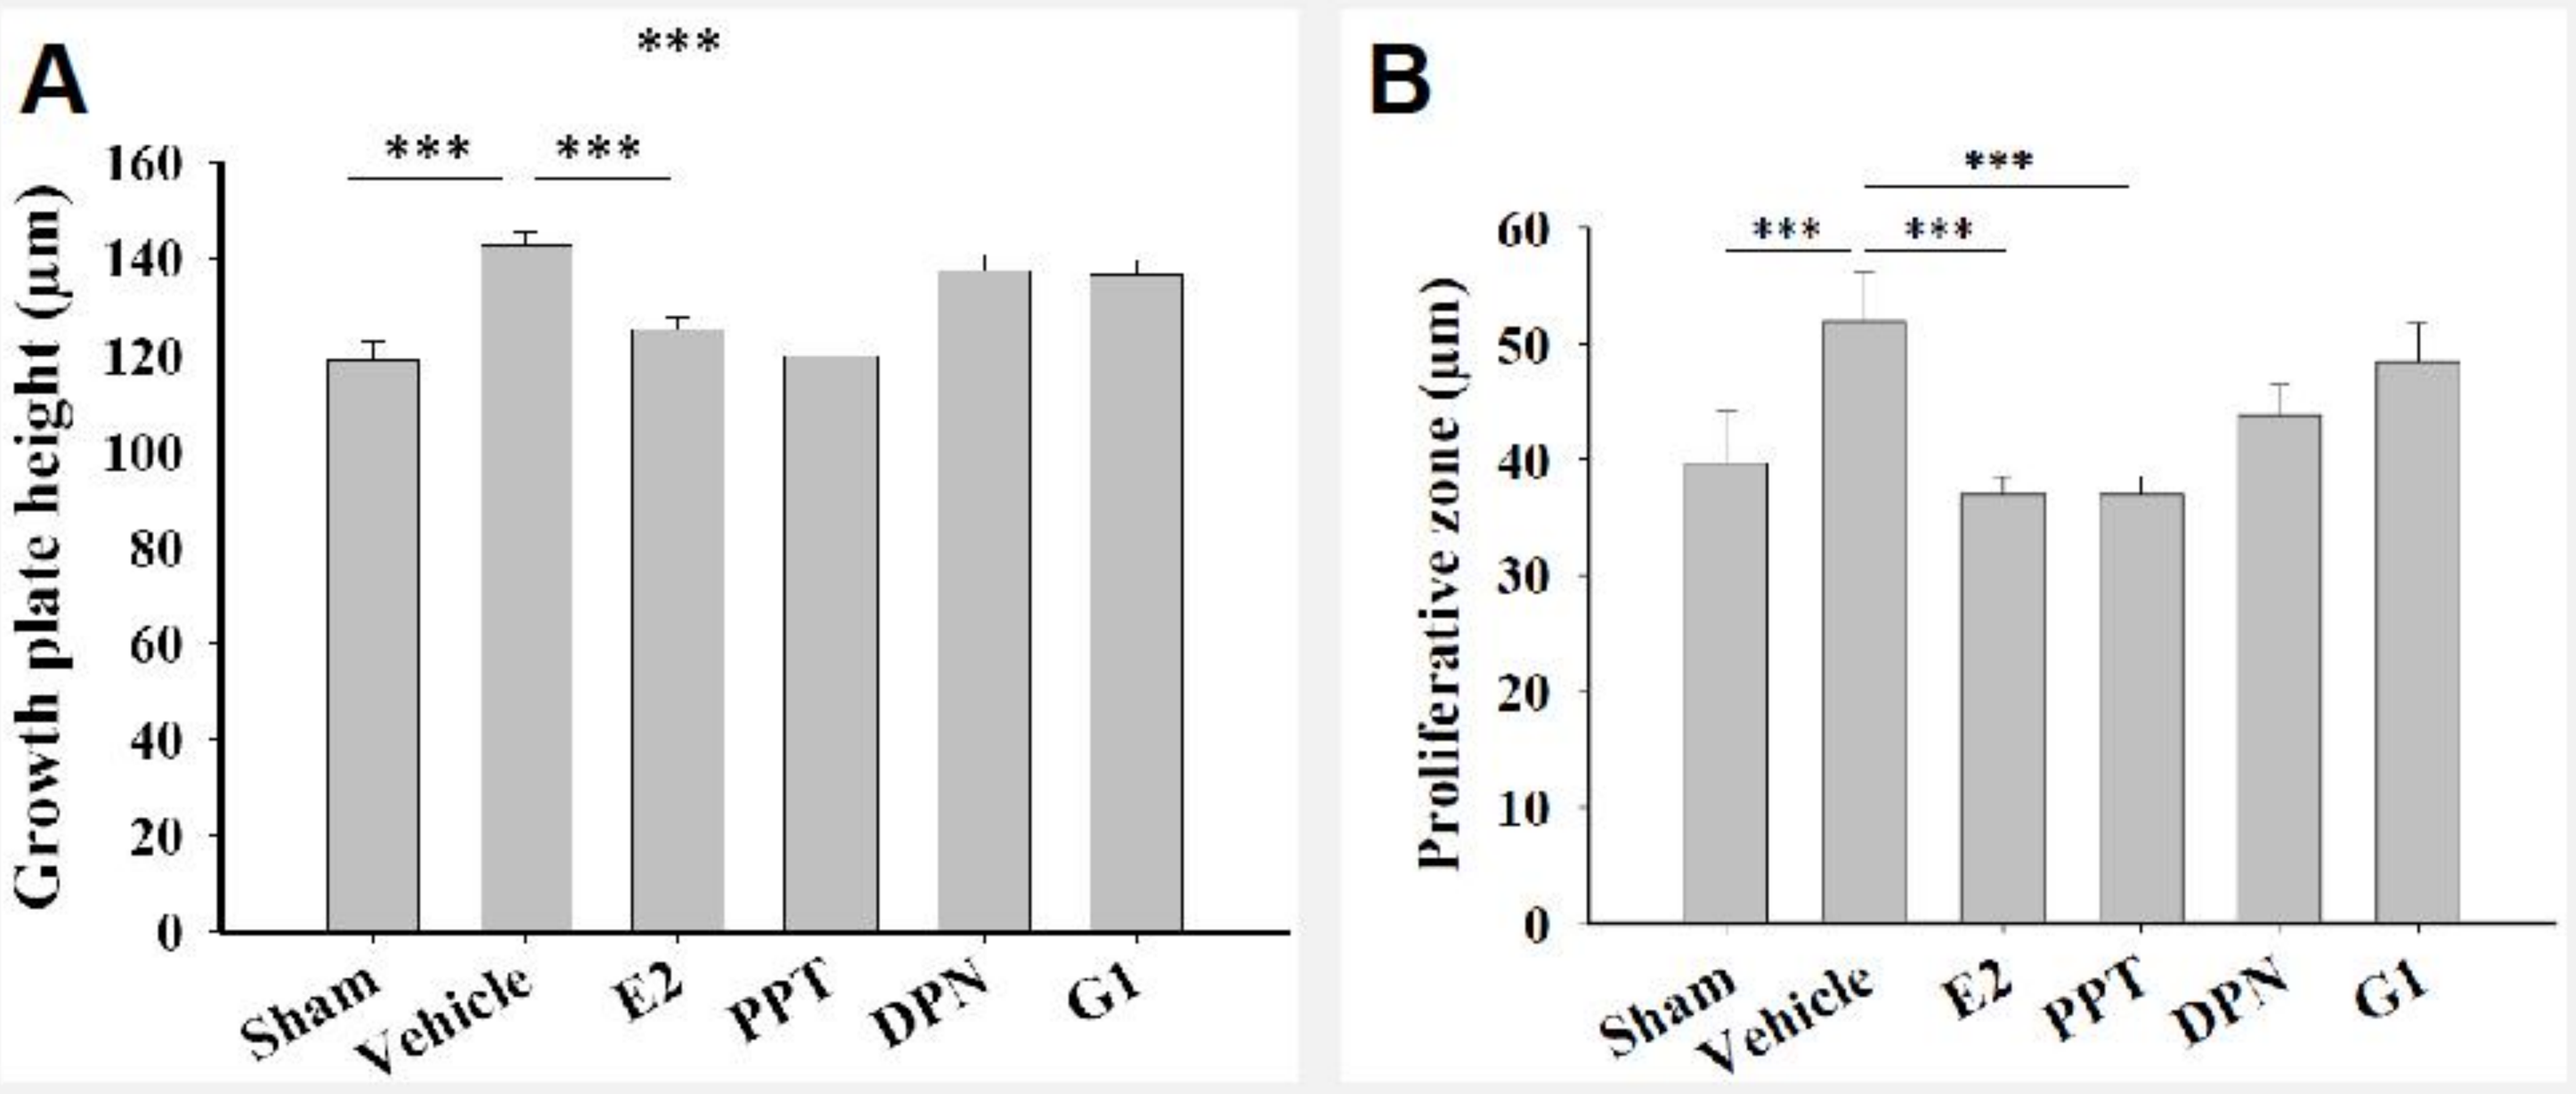
A
Growth plate height (µm)
160
140
120
100
80
60
40
20
0

Sham
Vehicle
E2
PPT
DPN
G1
B
Proliferative zone (µm)
60
50
40
30
20
10
0

Figure 3. Effects of E2 and selective ER agonists on tibia growth plate height (A) and proliferative zone height (B). Values are means ± SEM. ***p<0.001.

Materials and Methods

Twelve-week old ovariectomized female C57BL/6 mice were injected 5 days per week for 4 weeks with E2 (1 µg), PPT (175 µg), DPN (105 µg) or G1 (5 µg) per mouse. Tibia and femur lengths were measured after sacrifice and growth plate morphology was analyzed.

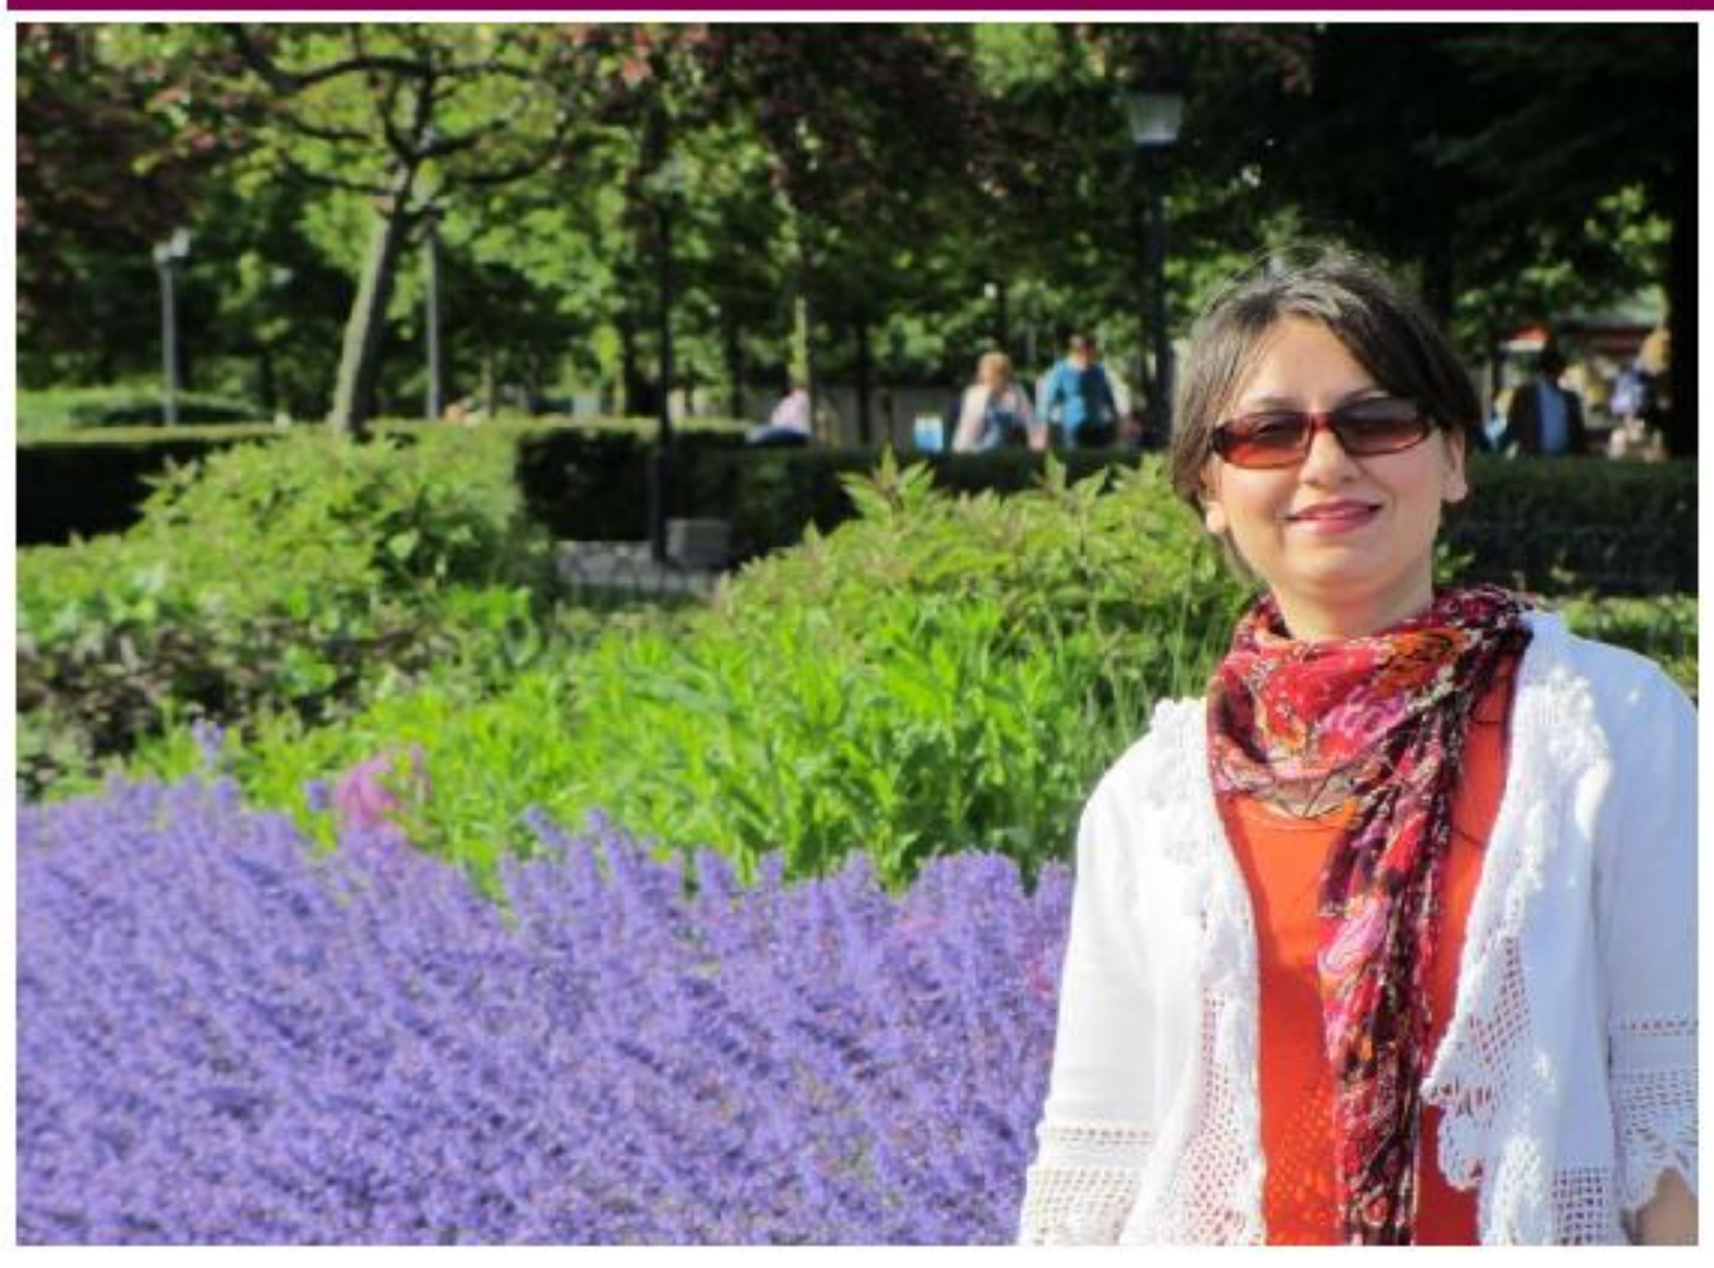

Karolinska Institutet
Maryam Iravani, PhD student
[1]Department of Women's and Children's Health, Karolinska University Hospital, Solna, Stockholm, Sweden
[2]Centre for Bone and Arthritis Research, Institute of Medicine, University of Gothenburg,Gothenburg, Sweden
E-mail: maryam.iravani@.ki.se
Telephone: +46 8 51772382
Fax: + 46 8 51775128
Website: www.ki.se/kbh

Karolinska Institutet

ESPE 2015 232--P2
Bone, Growth Plate and Mineral Metabolism
Maryam Iravani
DOI: 10.3252/pso.eu.54espe.2015
Poster presented at: ESPE 2015 BARCELONA

PosterSessionOnline.com