Year 2 Pharmacokinetic and Pharmacodynamic Modeling of Long-Acting Human Growth Hormone (MOD-4023) in Growth Hormone Deficient Children

Dennis M. Fisher, MD*; Michal Jaron–Mendelson, PhD†; Shelly Vander, MSc†; Ronit Koren, PhD†; Gili Hart, PhD†
*P Less Than, San Francisco; †OPKO Biologics, Nes Ziona, Israel

INTRODUCTION

OPKO Biologics is developing long-acting versions of existing therapeutic proteins utilizing CTP technology. This technology fuses the C-terminus peptide of human chorionic gonadotropin (hCG) to one or both ends of a target protein. CTP technology has been validated clinically and proven as a safe and efficient way to increase the half-life of several therapeutic proteins while maintaining their biological activity.

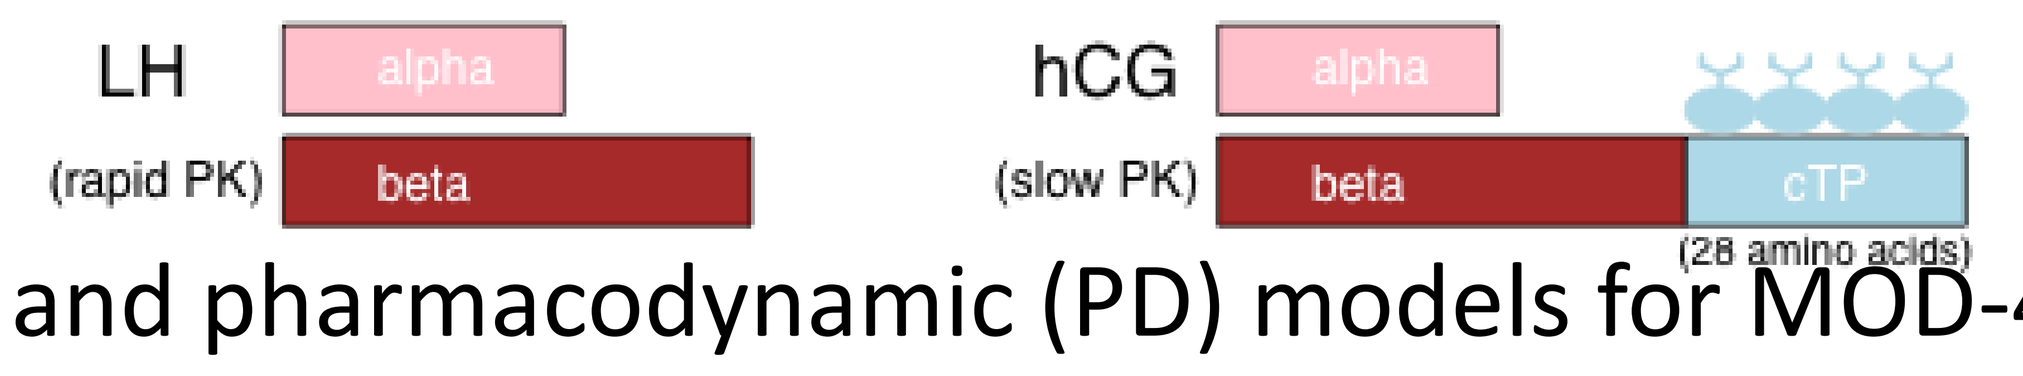
LH
(rapid PK)
alpha
beta
hCG
(slow PK)
alpha
beta
cTP
(28 amino acids)

At ESPE2015 (Barcelona), we reported pharmacokinetic (PK) and pharmacodynamic (PD) models for MOD-4023 and its effect on IGF-1 (and IGF-1 SDS) from clinical trial CP-4-004 (treatment-naive GH-deficient children aged 3-11 years receiving subcutaneous MOD-4023 weekly for up to one year). Thirty-eight (38) of the forty-two (42) subjects in that trial have now continued treatment through at least Year 2. We now report whether the model developed from data obtained during Year 1 predicts the time course of MOD-4023, IGF-1, and IGF-1 SDS during Year 2.

Key elements from the Year 1 analysis were:

- Apparent clearance was proportional to weight (supporting OPKO's decision to administer initial doses based on weight).
- No other covariates affected systemic pharmacokinetics in children.
- An indirect pharmacodynamic model proposed by Sun *et al.* (MOD-4023 concentration [Cp] affects IGF-1 input; sigmoid Emax relationship between Cp and effect) fit the IGF-1 data well. Calculations were performed using IGF-1 values rather than IGF-1 SDS because IGF-1 SDS is a non-linear transform of IGF-1.
- Baseline IGF-1 (the trough of the IGF-1 profile during each dosing interval) increased ~ 21% per hour, consistent with known increases in IGF-1 with maturation.

METHODS

During Year 2, samples were obtained at months 3, 6, 9, and 12 typically 4 days post-dose. MOD-4023 and IGF-1 concentrations were determined using electrochemiluminescence and IDS-iSYS assays, respectively. The individual (*post hoc*) parameters from the model for Year 1 were applied to predict MOD-4023 and IGF-1 concentrations at each sampling time in Year 2. IGF-1 SDS was calculated from IGF-1 concentrations using Bidlingmaier's table (2) that account for age and gender. Comparisons between observed and predicted values were explored graphically.

RESULTS

Data from a representative subject are shown in **Figure 1**. Observed values are displayed *vs.* predicted values in **Figure 2**. Individual and median of the ratio between observed and predicted values (MOD-4023, IGF-1) or difference (IGF-1 SDS) for each set of samples is displayed in **Figure 3**.

The model from Year 1 generally predicted the Year 2 data in an unbiased manner. Differences between predictions and observations were moderate in magnitude. Deviations between observed and predicted MOD-4023 Cp were largest at low Cp, possibly a result of a larger coefficient of variation of the assay at low Cp values. For IGF-1 and IGF-1 SDS, deviations were also largest at low values. For IGF-1 SDS, the deviation between predictions and observations was largest at Sample 4 (12 months), approximately 0.5 SDS units.

CONCLUSIONS

PK and PD models developed from Year 1 data for MOD-4023 treatment in children generally predicted Year 2 MOD-4023, IGF-1, and IGF-1 SDS in an unbiased manner. However, variability was moderate.

REFERENCES

1. Sun *et al*. JPET 1999; 289:1523
2. Bidlingmaier *et al.* J Clin Endocrinol Metab 2014; 99:1712

MOD-4023 | IGF-1 | IGF-1 SDS

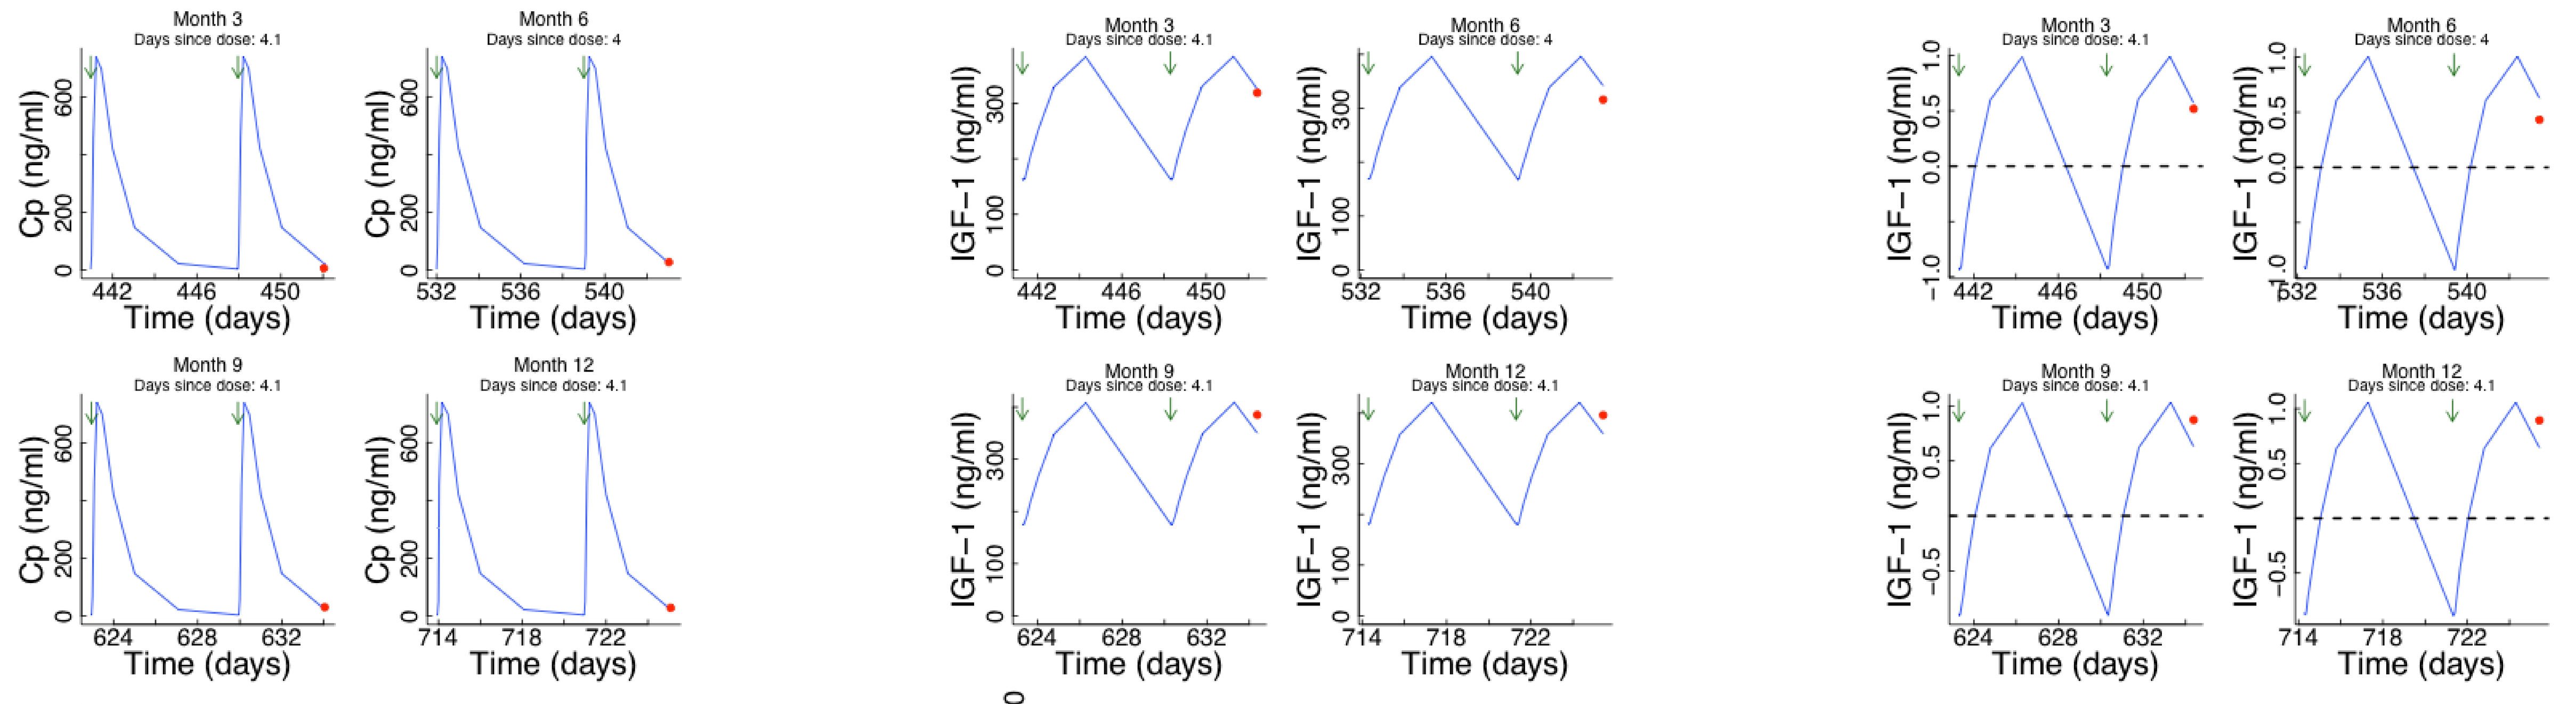
Month 3
Days since dose: 4.1
Month 6
Days since dose: 4
Month 9
Days since dose: 4.1
Month 12
Days since dose: 4.1
Cp (ng/ml)
IGF–1 (ng/ml)
Time (days)

Figure 1. Values at each of 4 sampling periods for a representative subject. The blue line is the profile predicted from Year 1 data. The red circle is the measured value. Arrows indicate doses.

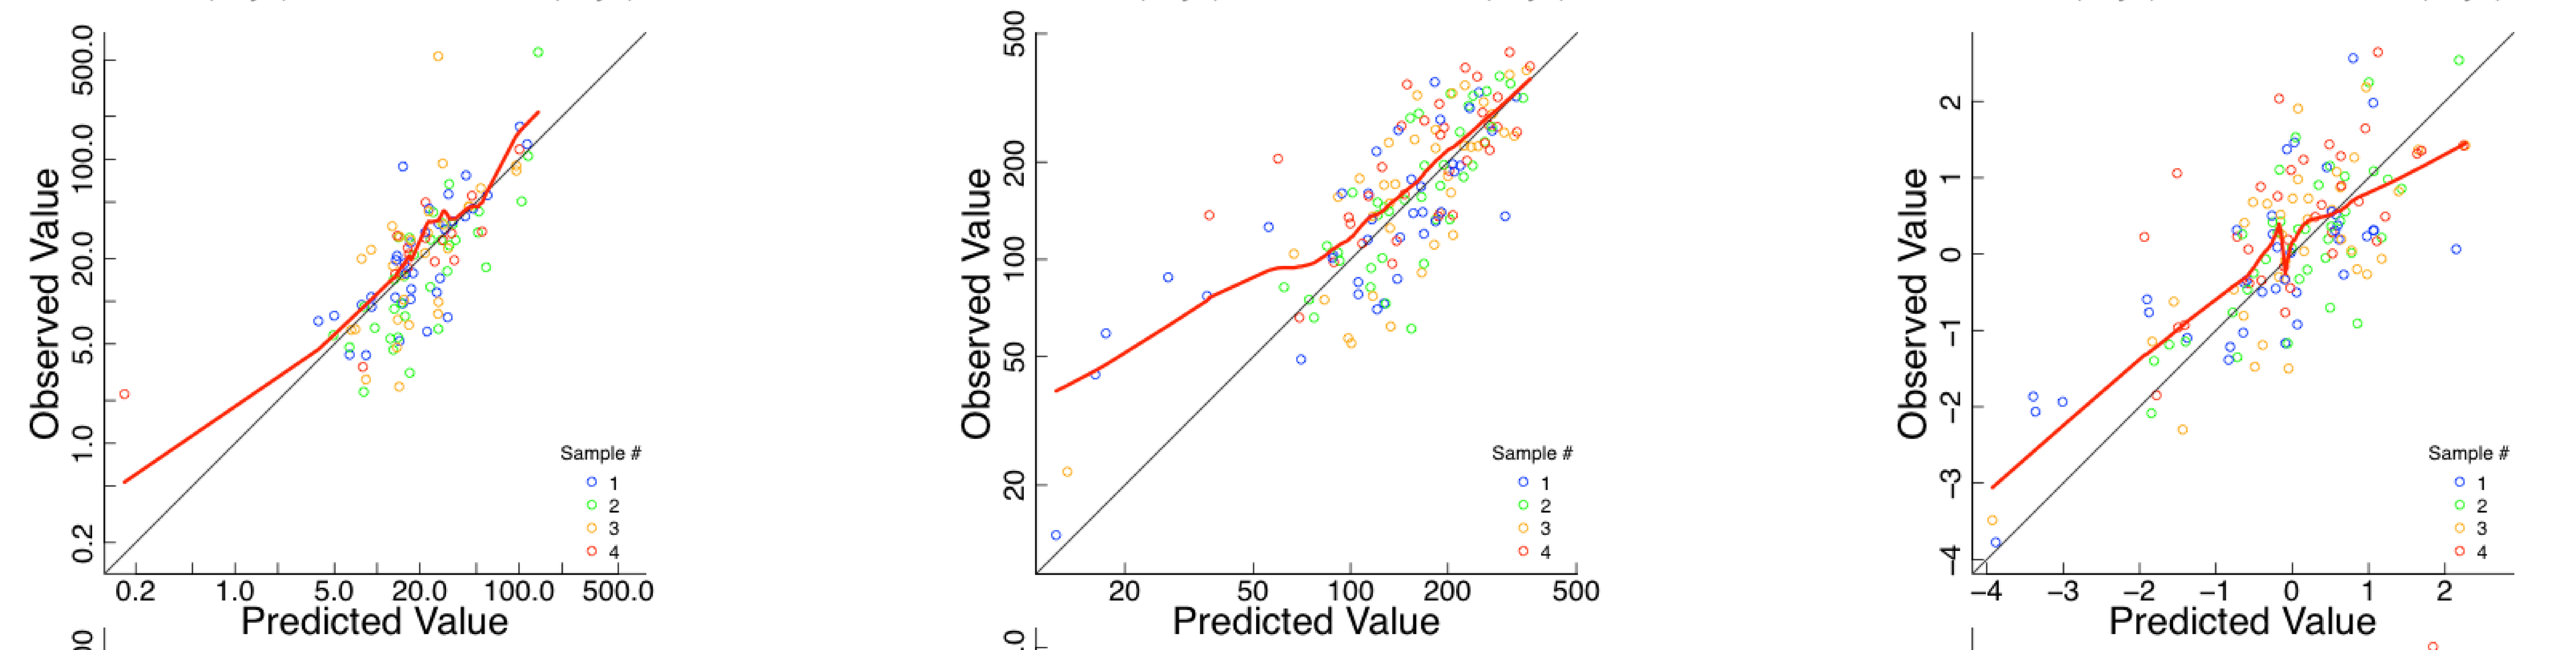
Observed Value
Predicted Value
Sample #
1
2
3
4

Figure 2. Observed *vs*. predicted values at each of the 4 (color-coded) sampling times. The red line (Supersmoother) generally tracks the line of identity.

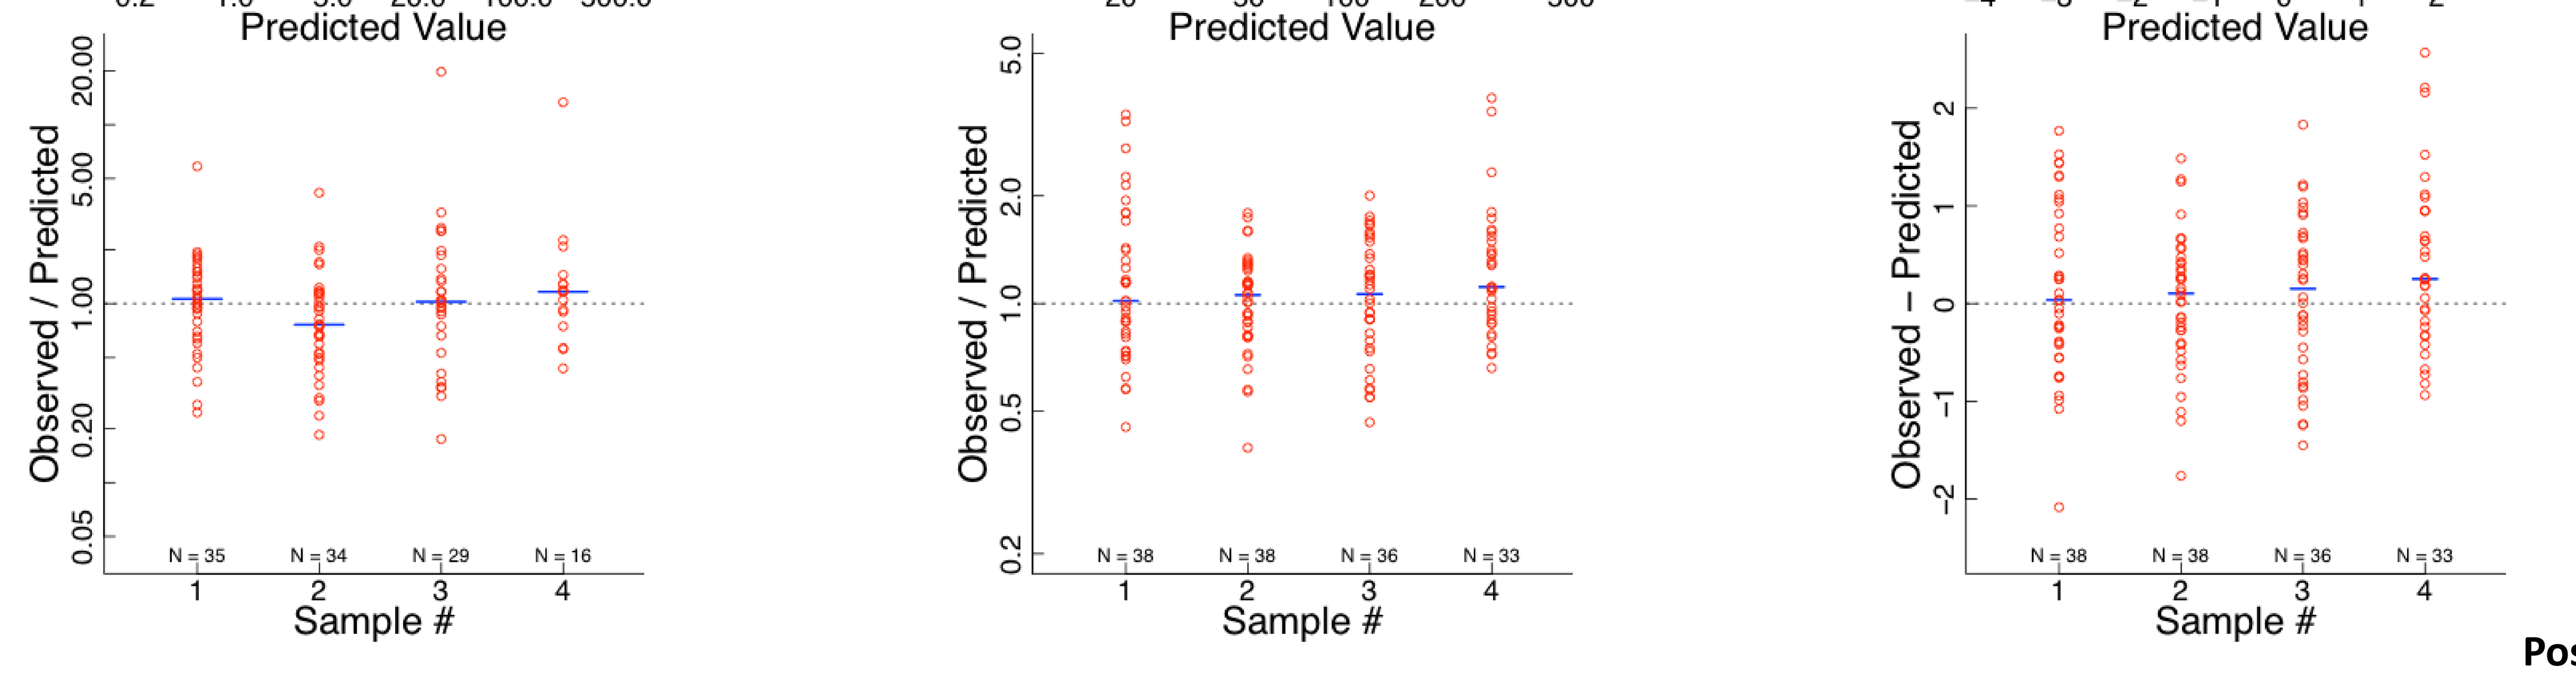
Observed / Predicted
Observed – Predicted
Sample #
N = 35
N = 34
N = 29
N = 16
N = 38
N = 38
N = 36
N = 33

Figure 3. Individual values and mean of ratio (MOD-4023, IGF-1) or difference (IGF-1 SDS) for each set of samples.

Disclosure Statement
DMF: Consultant, OPKO Biologics. Nothing to Disclose: MJM, SV, RK, GH.

Poster P1-625

55ESPE 625--P1
Growth
Gili Hart
DOI: 10.3252/pso.eu.55ESPE.2016
Poster presented at: