P1-811 Syndromes: Mechanisms and Management

SAHLGRENSKA ACADEMY
INSTITUTE OF CLINICAL SCIENCES

UNIVERSITY OF GOTHENBURG

Correlations between prepubertal and pubertal estrogen levels and final height out-come in growth hormone treated boys with Silver Russell syndrome

Kjersti Kvernebo-Sunnergren[1], Carina Ankarberg-Lindgren[2], Karin Åkesson[1], Jovanna Dahlgren[2]
[1] Department of Pediatrics, Ryhov County Hospital, S-551 85 Jönköping, Sweden
[2] Göteborg Pediatric Growth Research Center, Department of Pediatrics, Institute of Clinical Sciences, Sahlgrenska Academy, University of Gothenburg, S-41685 Göteborg, Sweden
Disclosure statement: This study was supported with unrestricted grants from Pfizer and the Swedish Research Council.

Contact information: kjersti.kvernebo.sunnergren@rjl.se

Introduction: Children with Silver Russell syndrome (SRS) suffer from short stature and delayed bone maturation during infancy. The majority of these children initially respond well to growth hormone (GH) treatment. A catch-up period of skeletal maturation is often seen in early puberty. Later in puberty unexpected poor height acceleration has previously been reported [1,2].

Objective: To evaluate the association between prepubertal and pubertal estrogen levels and final height (FH) out-come in GH treated boys with SRS.

Conclusion: There is an association between high levels of E2 at the age of 10-12 and low E1/E2 ratio at the age of 12 leading to impaired FH out-come in GH treated boys with SRS.

Take home message: FH out-come in GH treated boys with SRS depends on prepubertal and pubertal estrogen levels.

Results:

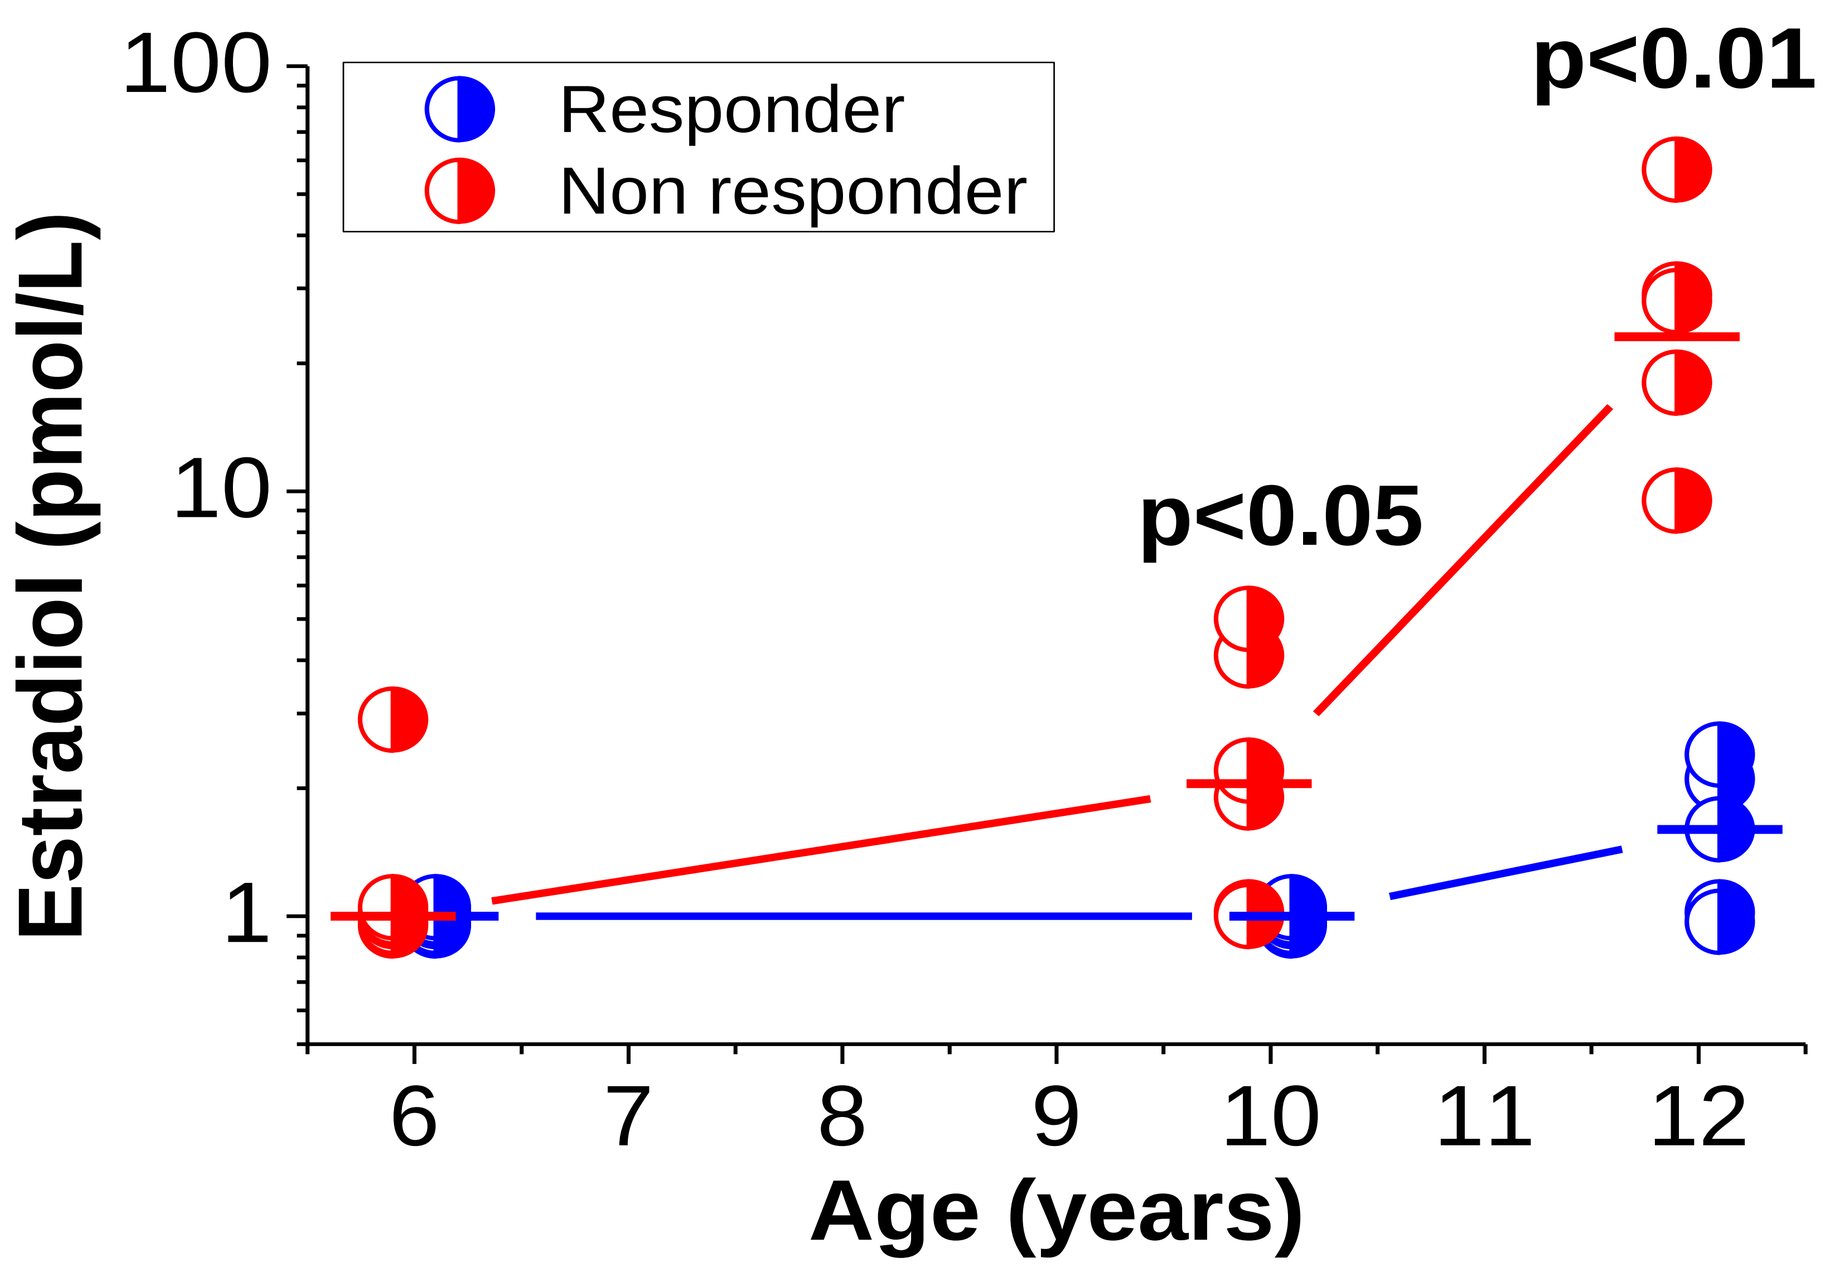
Responder
Non responder
p<0.01
p<0.05
Estradiol (pmol/L)
Age (years)

Non-responders had significantly higher levels of E2 (median 2 versus 1 pmol/L, p<0,05 and 23,1 versus 1,6 pmol/L, p<0,01) at the age 10 and 12 compared to responders.

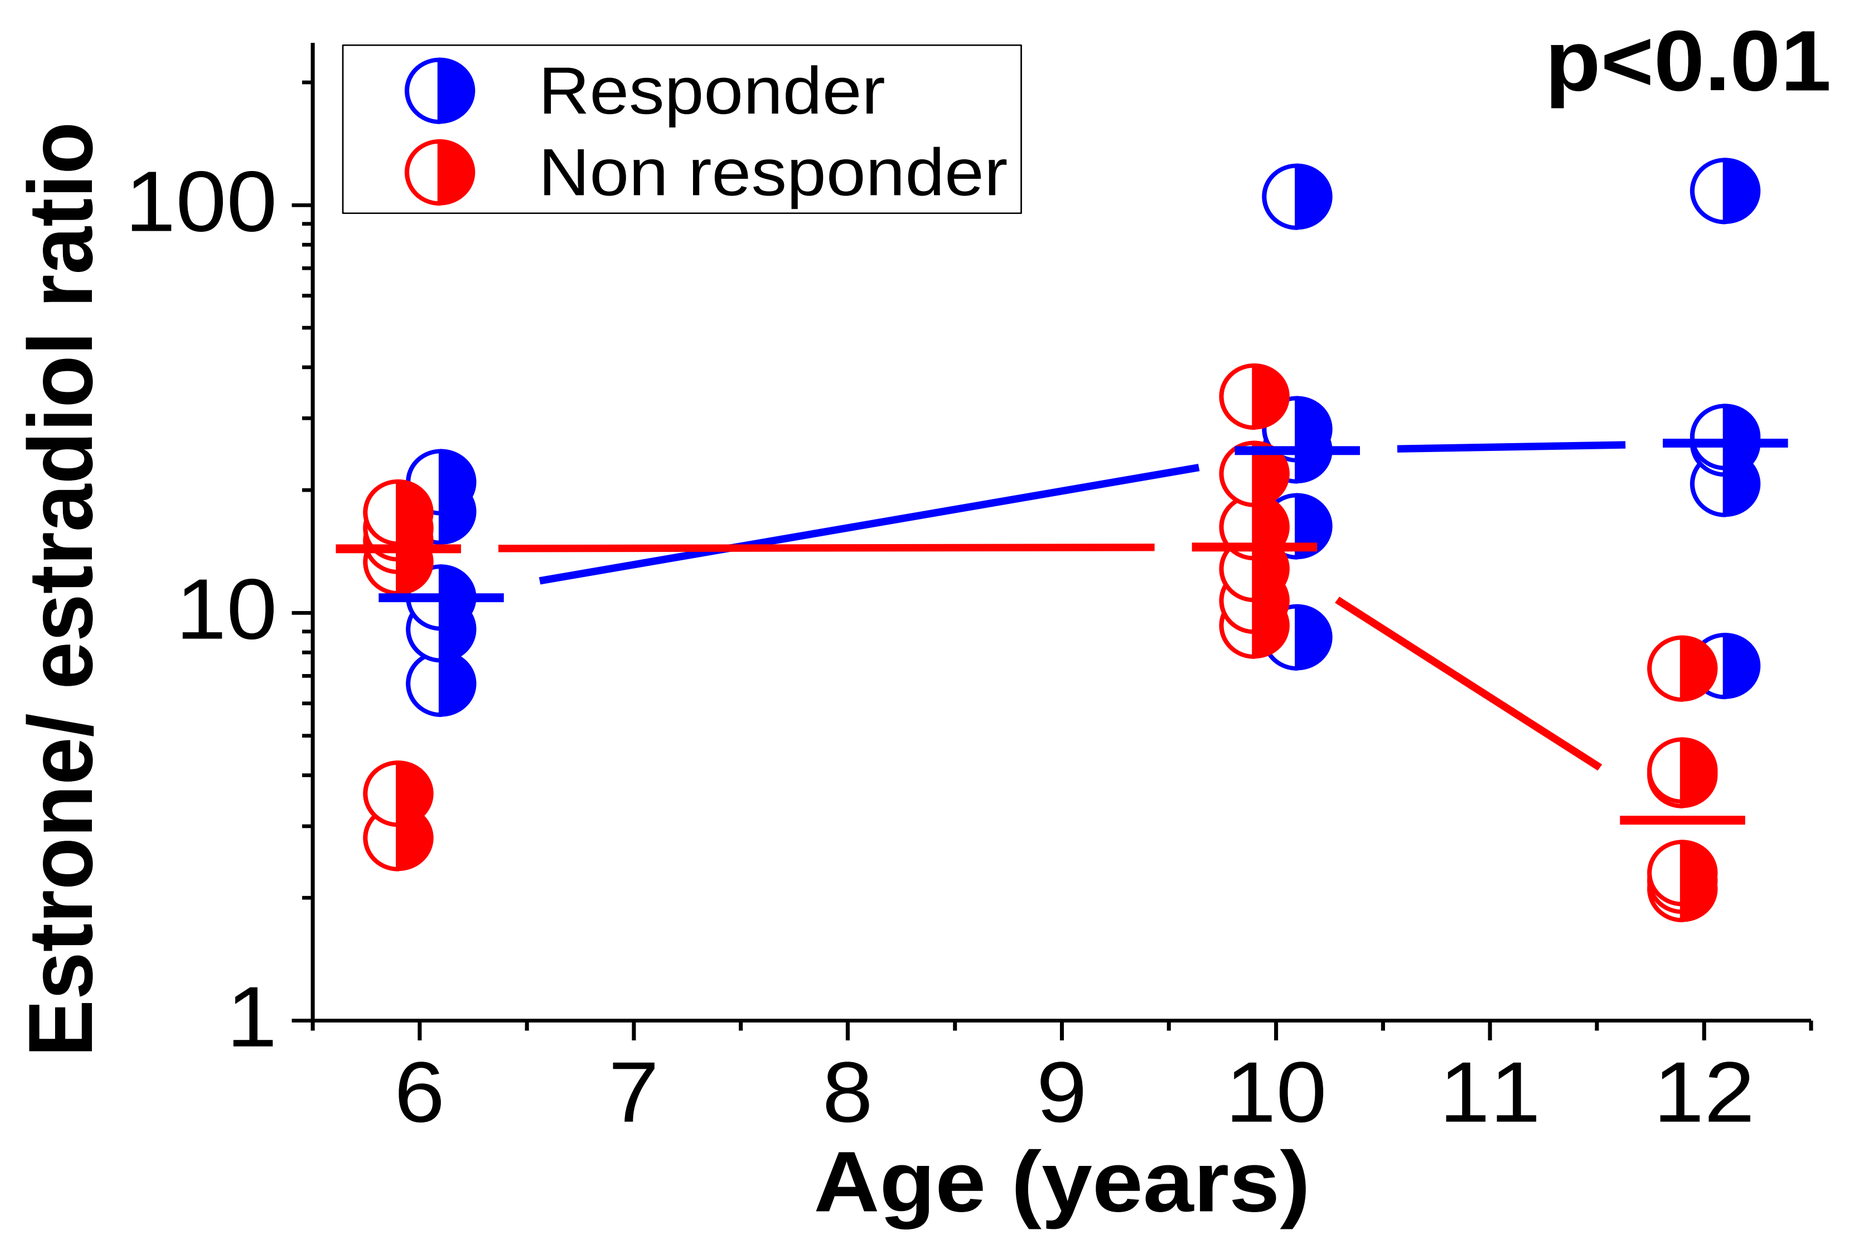
Responder
Non responder
p<0.01
Estrone/ estradiol ratio
Age (years)

Non-responders had significantly lower E1/E2 ratio (3,2 versus 26,1, p<0,01) at the age of 12 compared to responders.

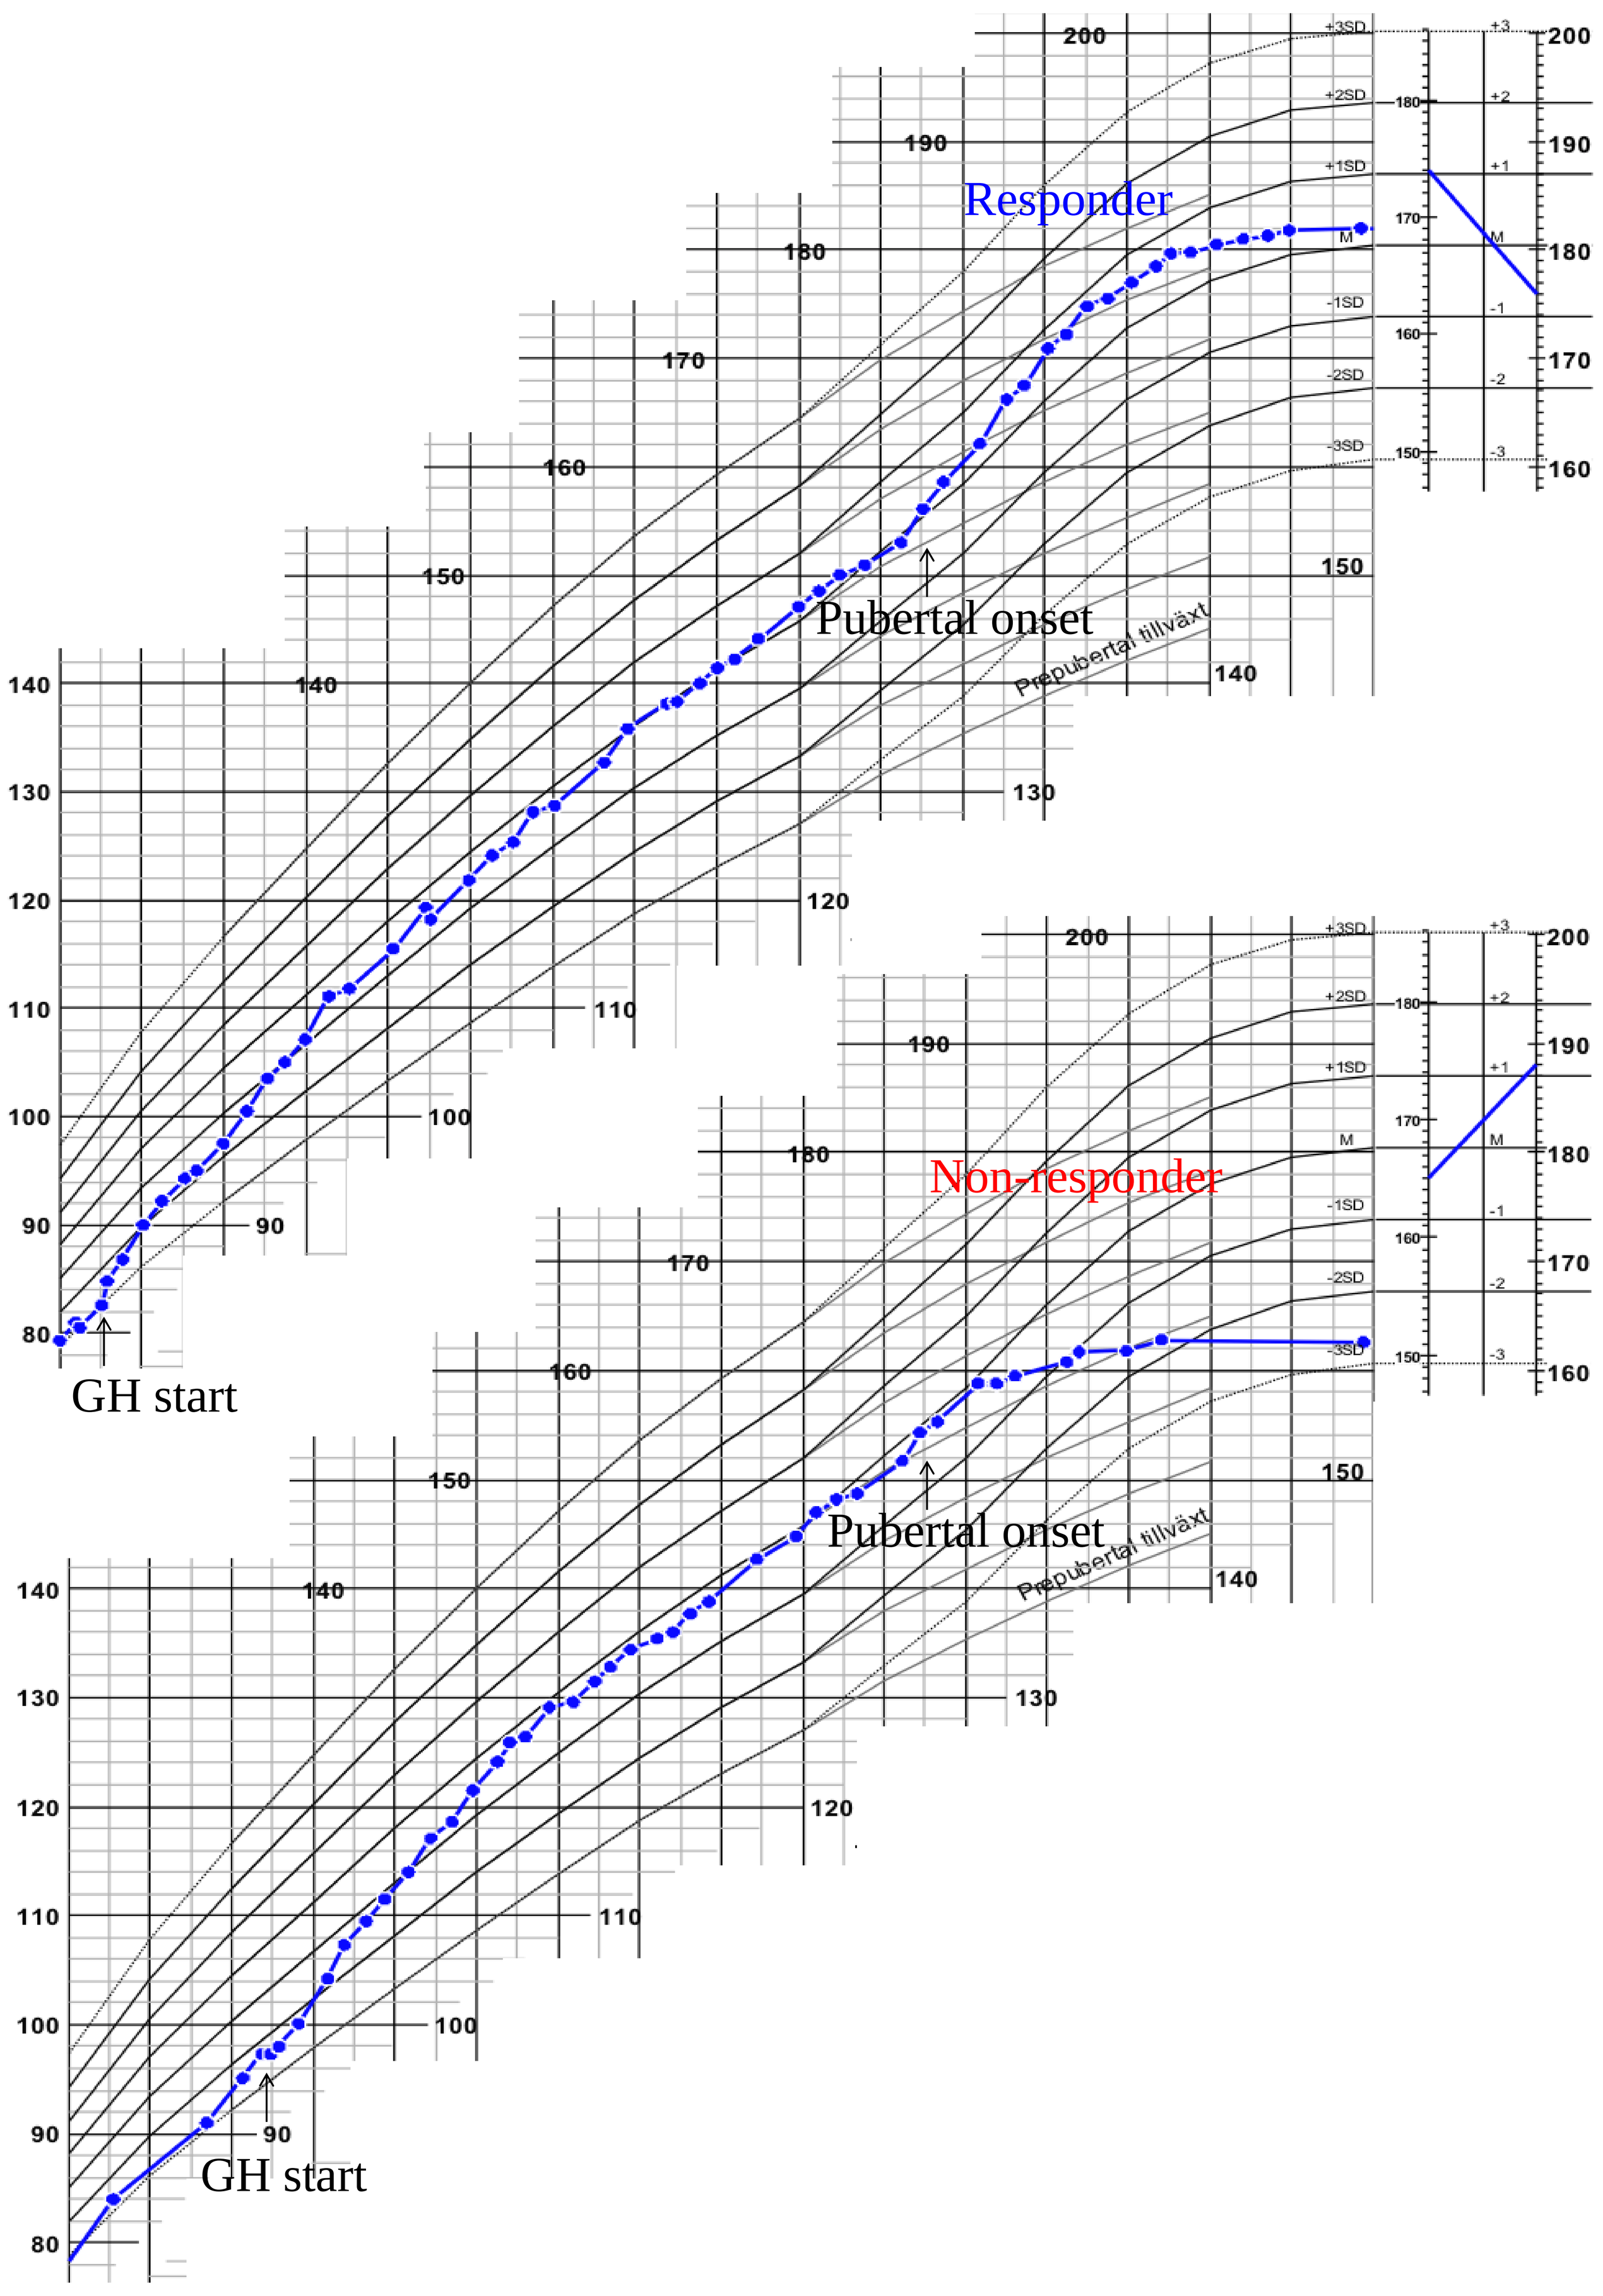
Responder
Pubertal onset
GH start
Non-responder
Pubertal onset
GH start
Prepubertal tillväxt

Results:

Presented as median (range)	Responders (n=5)	Non-responders (n=6)
Age (yrs) at GH start	2,9 (2,0-6,0)	3,5 (2,7-4,5)
Height SDS from TH at GH start	-2,3 (-3,3- -1,2)	-3,0 (-5,3--1,5)
Age (yrs) at pubertal onset (testes ≥3 mL)	12,9 (10,5-14,9)	11,5 (11,0-11,8)
Height SDS from TH at pubertal onset	0,5 (-0,7-+0,2)	-0,2 (-1,2-+0,8)
Final height SDS from TH	-0,32 (+0,25--0,81)	-1,8 (-1,3--3,3)

Subjects: 11 GH treated boys with SRS were divided into two groups depending on FH. Subjects with FH ≤ -1 SDS from target height (TH) were considered responders and subjects with FH > -1 SDS from TH were considered non-responders. At the age of 10 all subjects were prepubertal.

Methods: Serum concentrations of estrone (E1) and estradiol (E2) were determined yearly until FH by gas chromatography-tandem mass spectrometry with lower limit of detection (LoD) 9 pmol/L and 2 pmol/L, respectively. E2 concentrations below LoD were set to 1 pmol/L. Wilcoxon signed-rank test was used for statistical analysis.

Discussion: An increase in E2 secretion before puberty is most likely derived from steroid secretion from the adrenal gland. The E2 levels may be sufficient to accelerate skeletal maturation but far lower than what is normally seen during puberty which may explain why there is no enhanced prepubertal growth in the non-responder group.

[1]Tanner, J. M., Lejarraga, H., & Cameron, N. (1975). The natural history of the Silver-Russell syndrome: a longitudinal study of thirty-nine cases. *Pediatr Res*, *9*(8), 611-623.
[2]Wollmann, H. A., Kirchner, T., Enders, H., Preece, M. A., & Ranke, M. B. (1995). Growth and symptoms in Silver-Russell syndrome: review on the basis of 386 patients. *Eur J Pediatr, 154*(12), 958-968.

55ESPE 811--P1
Syndromes : Mechanisms and Management
Kjersti Kvernebo
DOI: 10.3252/pso.eu.55ESPE.2016
Poster presented at: ESPE 2016 PARIS
PosterSessionOnline.com