A hybrid Fc-fused human growth hormone, GX-H9, shows a potential for weekly and twice-monthly administration in clinical studies.

Eun Jig Lee, MD PhD[1], Jochen Schopohl, MD[2], Aryaev Mykola, MD[3], Tae Kyung Kim, PhD[4], Misoo Kang[5], Jung-Won Woo, PhD[4], Woo Ick Jang, MD PhD[5], Young-Chul Sung, PhD[4] and H. Michael Keyoung, MD, PhD[4]

[1]Yonsei University College of Medicine, Korea, [2]University of Munich Medizinsche Klinik, Germany, [3]Odessa National Medical University, Ukraine [4]Genexine, Inc., Korea, [5]Handok, Inc., Korea

BACKGROUND

- Recombinant human growth hormone (rhGH) is standard treatment for children and adults with growth hormone deficiency. However, current rhGH therapy involves daily subcutaneous (sc) injections with years of treatment. The challenge of daily sc rhGH injections has proven to limit compliance, often reducing the ability to maintain height velocities or optimal clinical outcomes. Thus, long-acting rhGH should improve ease of use, compliance, and consequently efficacy.
- GX-H9 is a chimeric protein composed of rhGH fused to a hybrid Fc. The hybrid Fc (hyFc), is a novel Fc-based platform technology for generating long-acting proteins with a hybrid of IgD Fc and IgG4 Fc, which extends the half-life and the bioactivity of fused proteins.

Long-acting rhGH (GX-H9) Structure

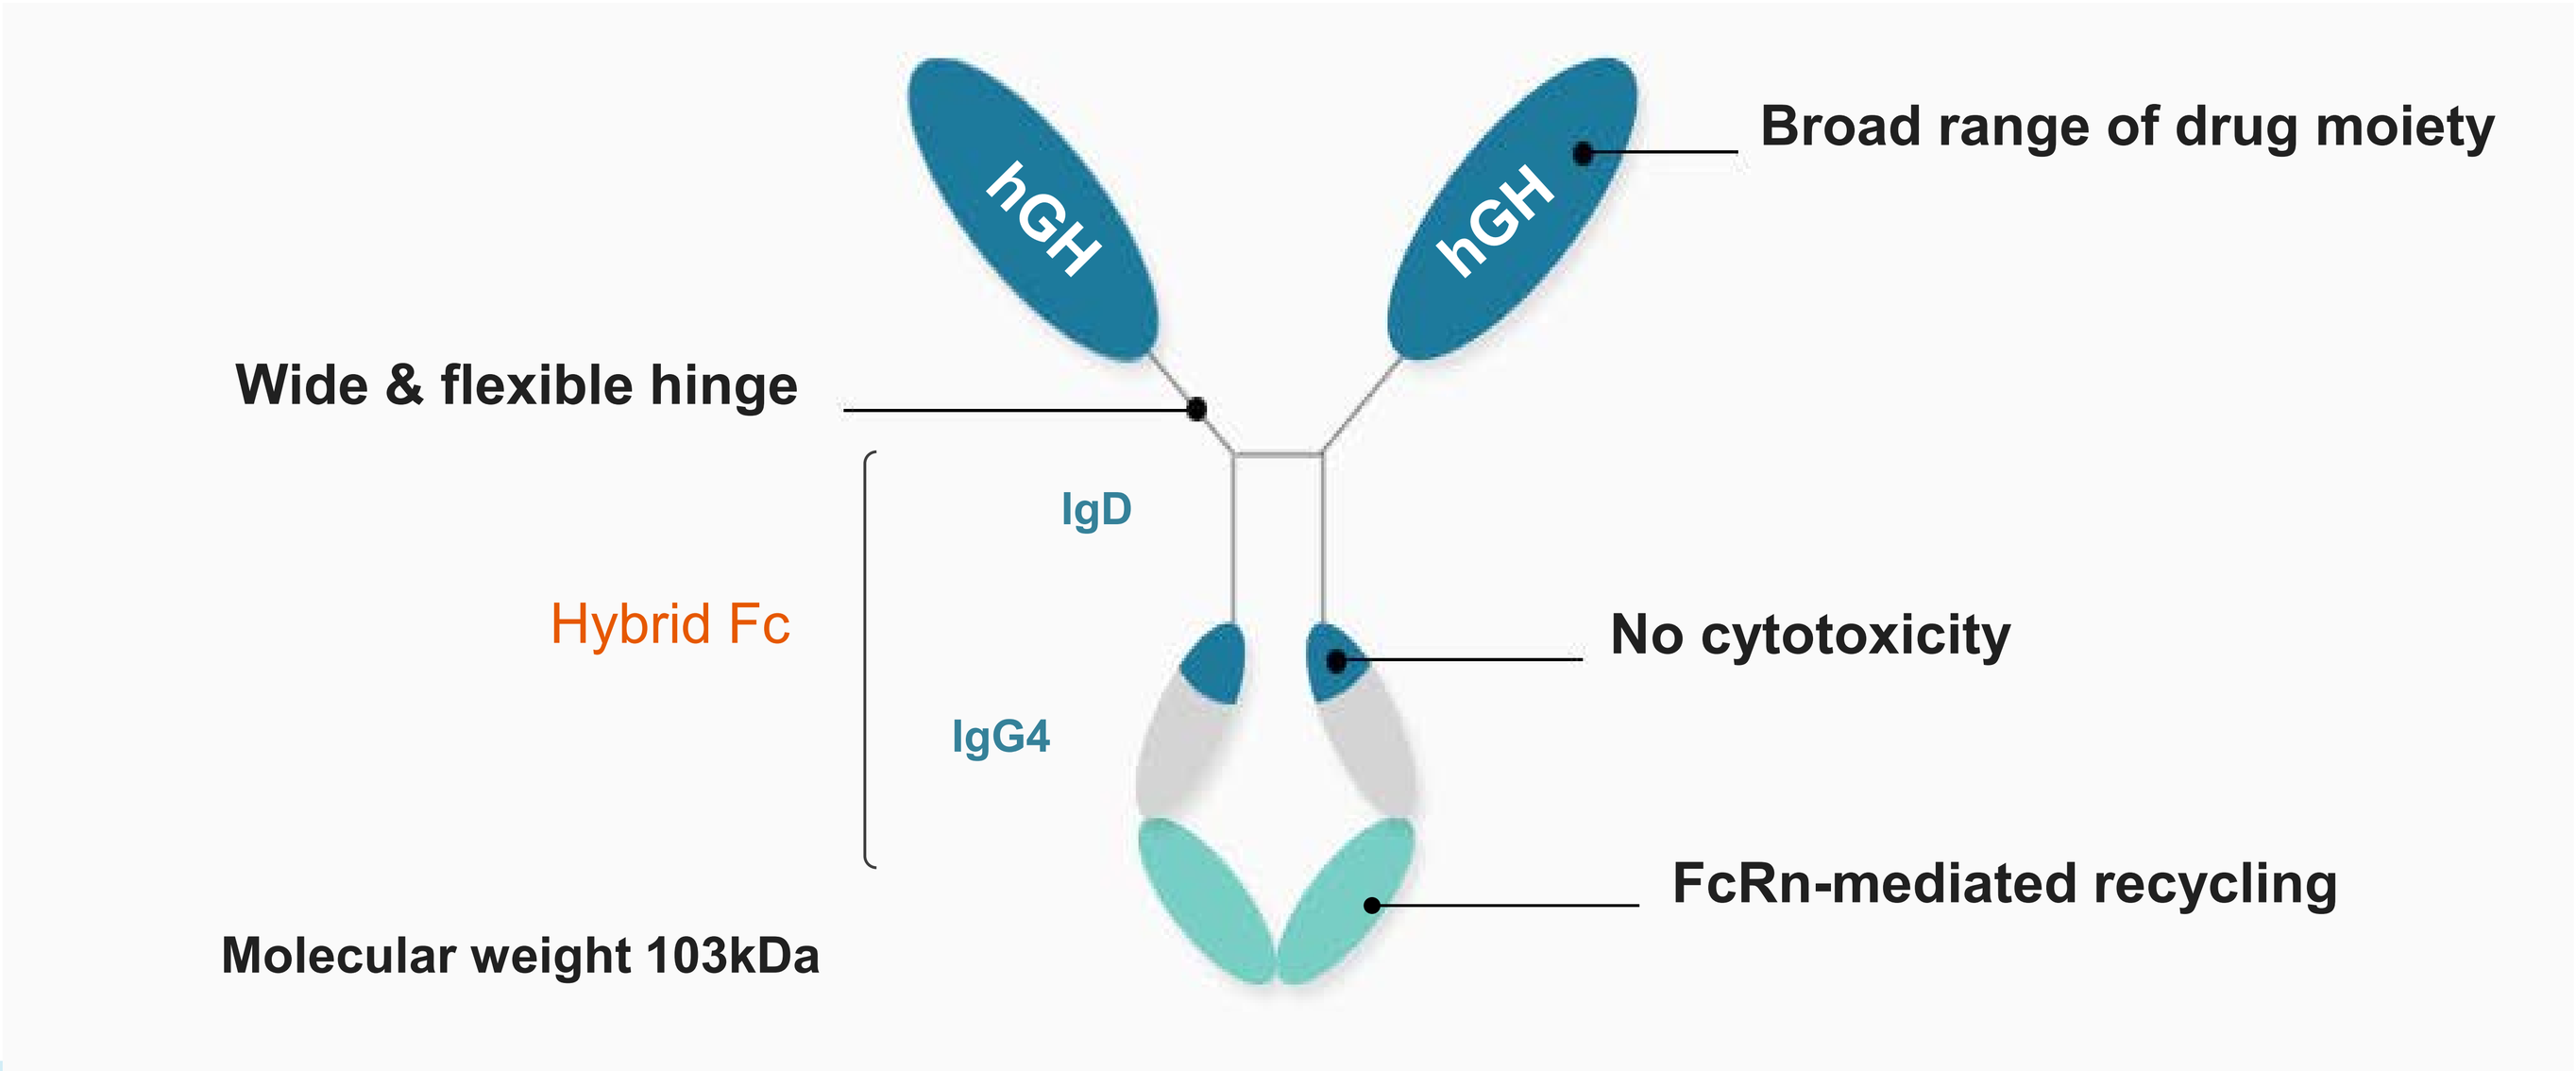
hGH
hGH
Broad range of drug moiety
Wide & flexible hinge
IgD
Hybrid Fc
IgG4
No cytotoxicity
FcRn-mediated recycling
Molecular weight 103kDa

Human Ig Isotypes	IgG1	IgG4	IgD
Hinge flexibility	++	+	++++
Binding of FcyR phagocytes (ADCC)	++++	++	-
Binding of C1q(CDC)	++	-	-
Binding of FcRn	++++	++++	-
In vivo serum half-life (days)	21	21	3

METHODS & RESULTS – Phase 1 Healthy Volunteer Study

- **Study Design (Single Ascending Dose, SAD)**

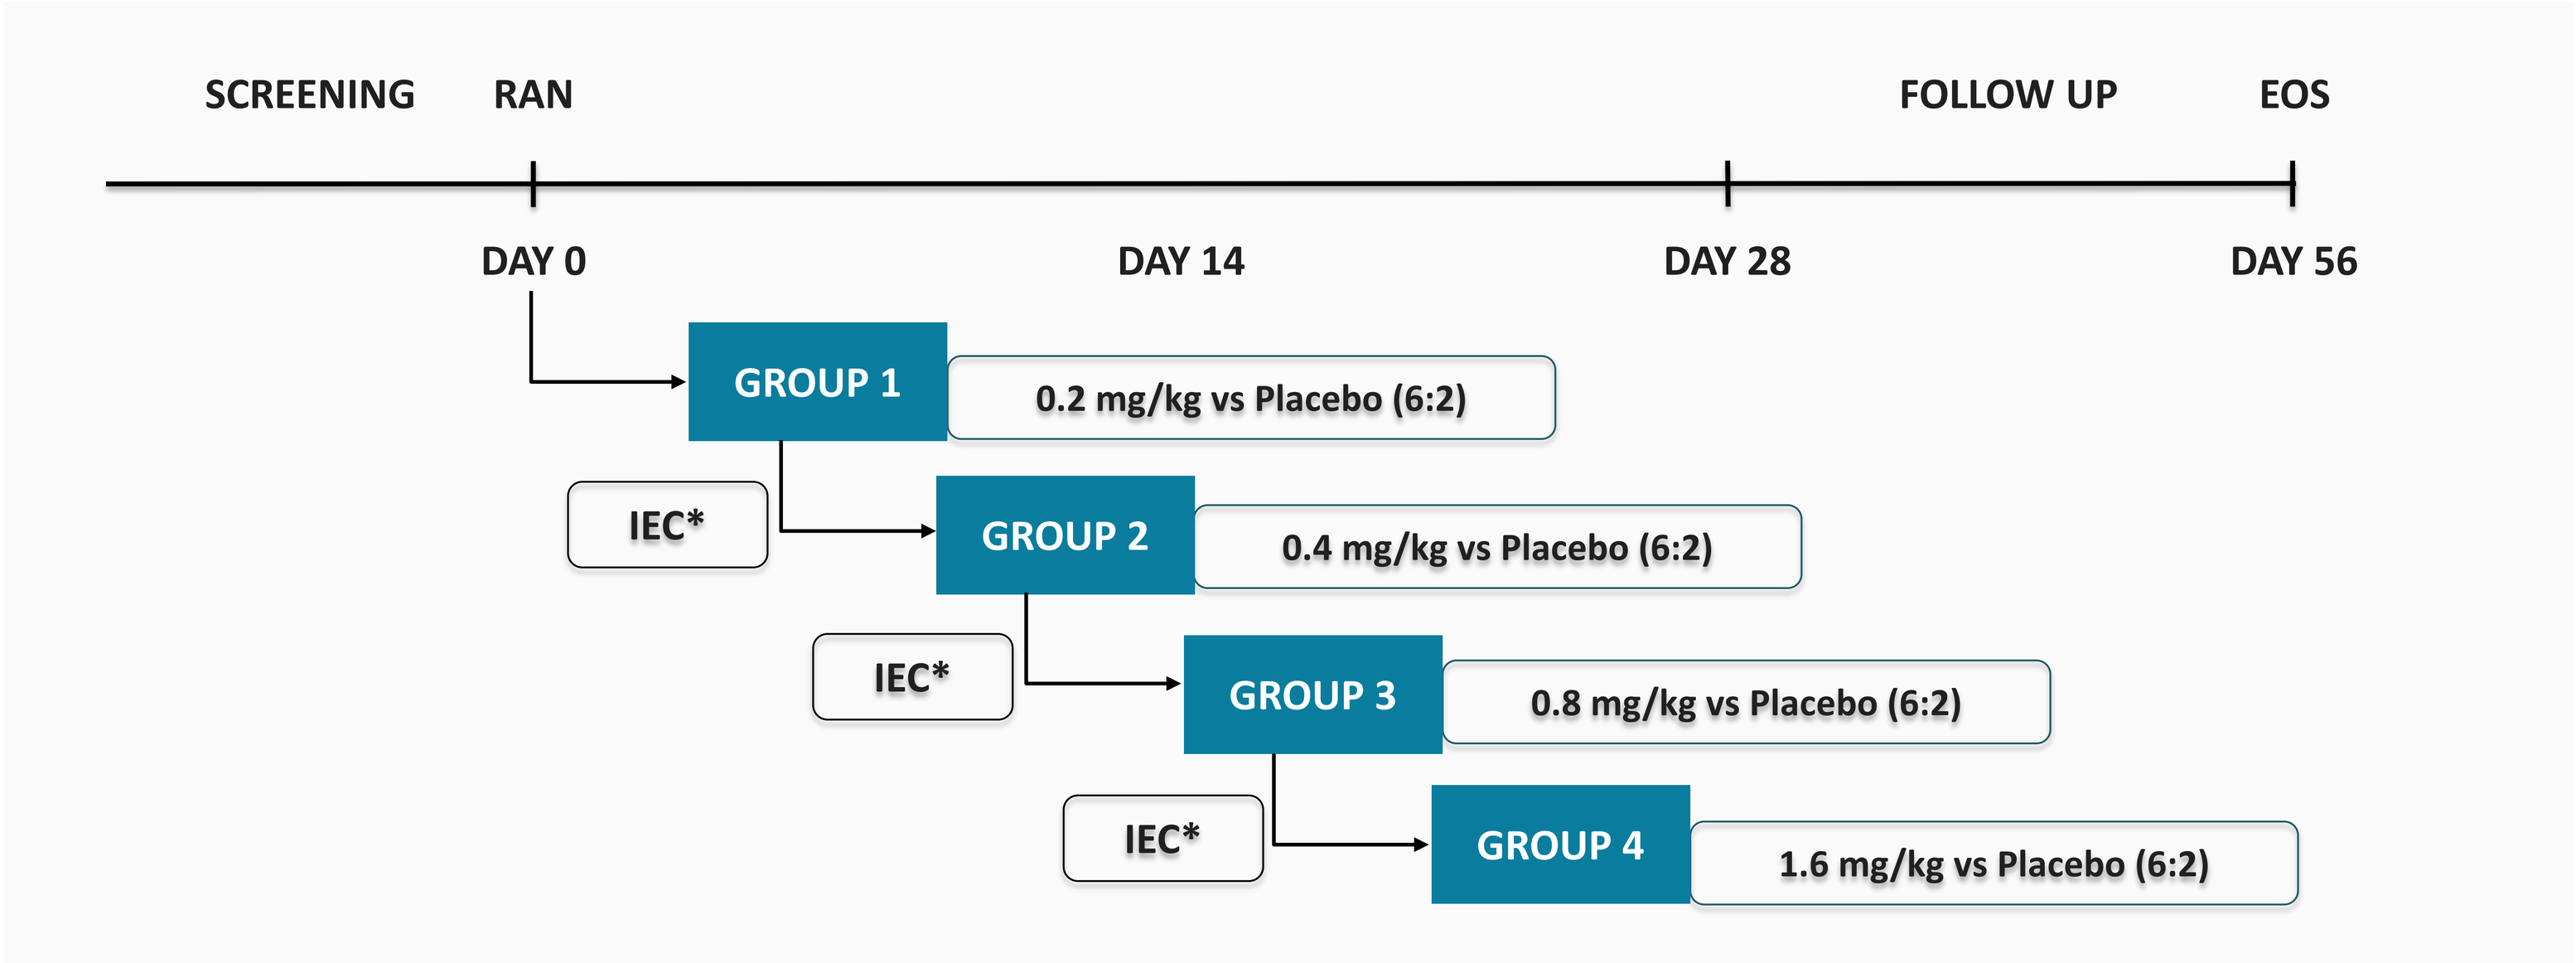
SCREENING
RAN
FOLLOW UP
EOS
DAY 0
DAY 14
DAY 28
DAY 56
GROUP 1
0.2 mg/kg vs Placebo (6:2)
IEC*
GROUP 2
0.4 mg/kg vs Placebo (6:2)
IEC*
GROUP 3
0.8 mg/kg vs Placebo (6:2)
IEC*
GROUP 4
1.6 mg/kg vs Placebo (6:2)

**After day 14 of last patients in each group, available safety and tolerability data will be reviewed by the PI, Sponsor, and the IEC*

- **Results – Pharmacokinetic & Pharmacodynamics**

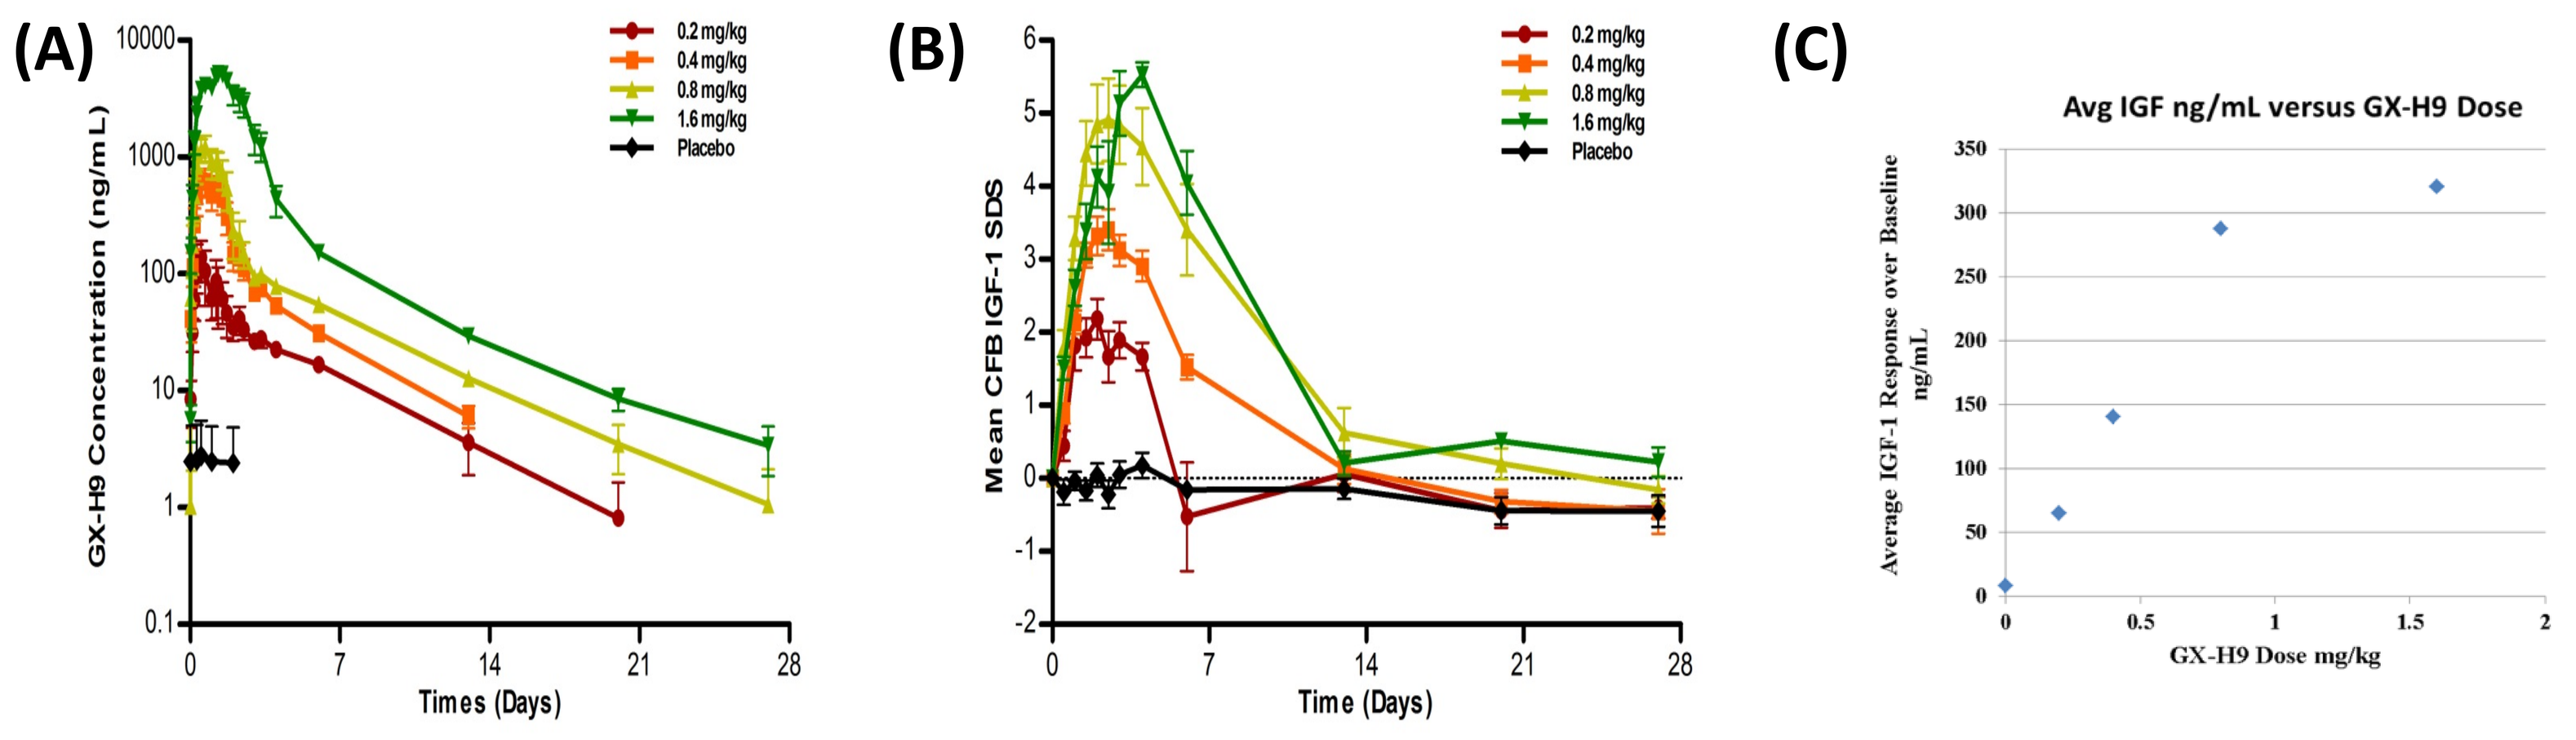
(A)
GX-H9 Concentration (ng/mL)
Times (Days)
0.2 mg/kg
0.4 mg/kg
0.8 mg/kg
1.6 mg/kg
Placebo
(B)
Mean CFB IGF-1 SDS
Time (Days)
(C)
Avg IGF ng/mL versus GX-H9 Dose
Average IGF-1 Response over Baseline ng/mL
GX-H9 Dose mg/kg

PK and PD study of GX-H9. Healthy subjects received a single dose of either 0.2, 0.4, 0.8, 1.6 mg/kg GX-H9 or placebo.

(A) Serum GX-H9 Concentration-Time profiles following single subcutaneous administration.
(B) Mean change from baseline in IGF-1 SDS for GX-H9 and placebo dosing groups. Baseline was defined as the Day 1 pre-dose value (CFB; change from baseline)
(C) Dose-IGF-1 (ng /ml) relationship after single injection of GX-H9

METHODS & RESULTS – Phase 2 Adult GHD Study

- **Study Design (Multiple Ascending Dose, MAD)**

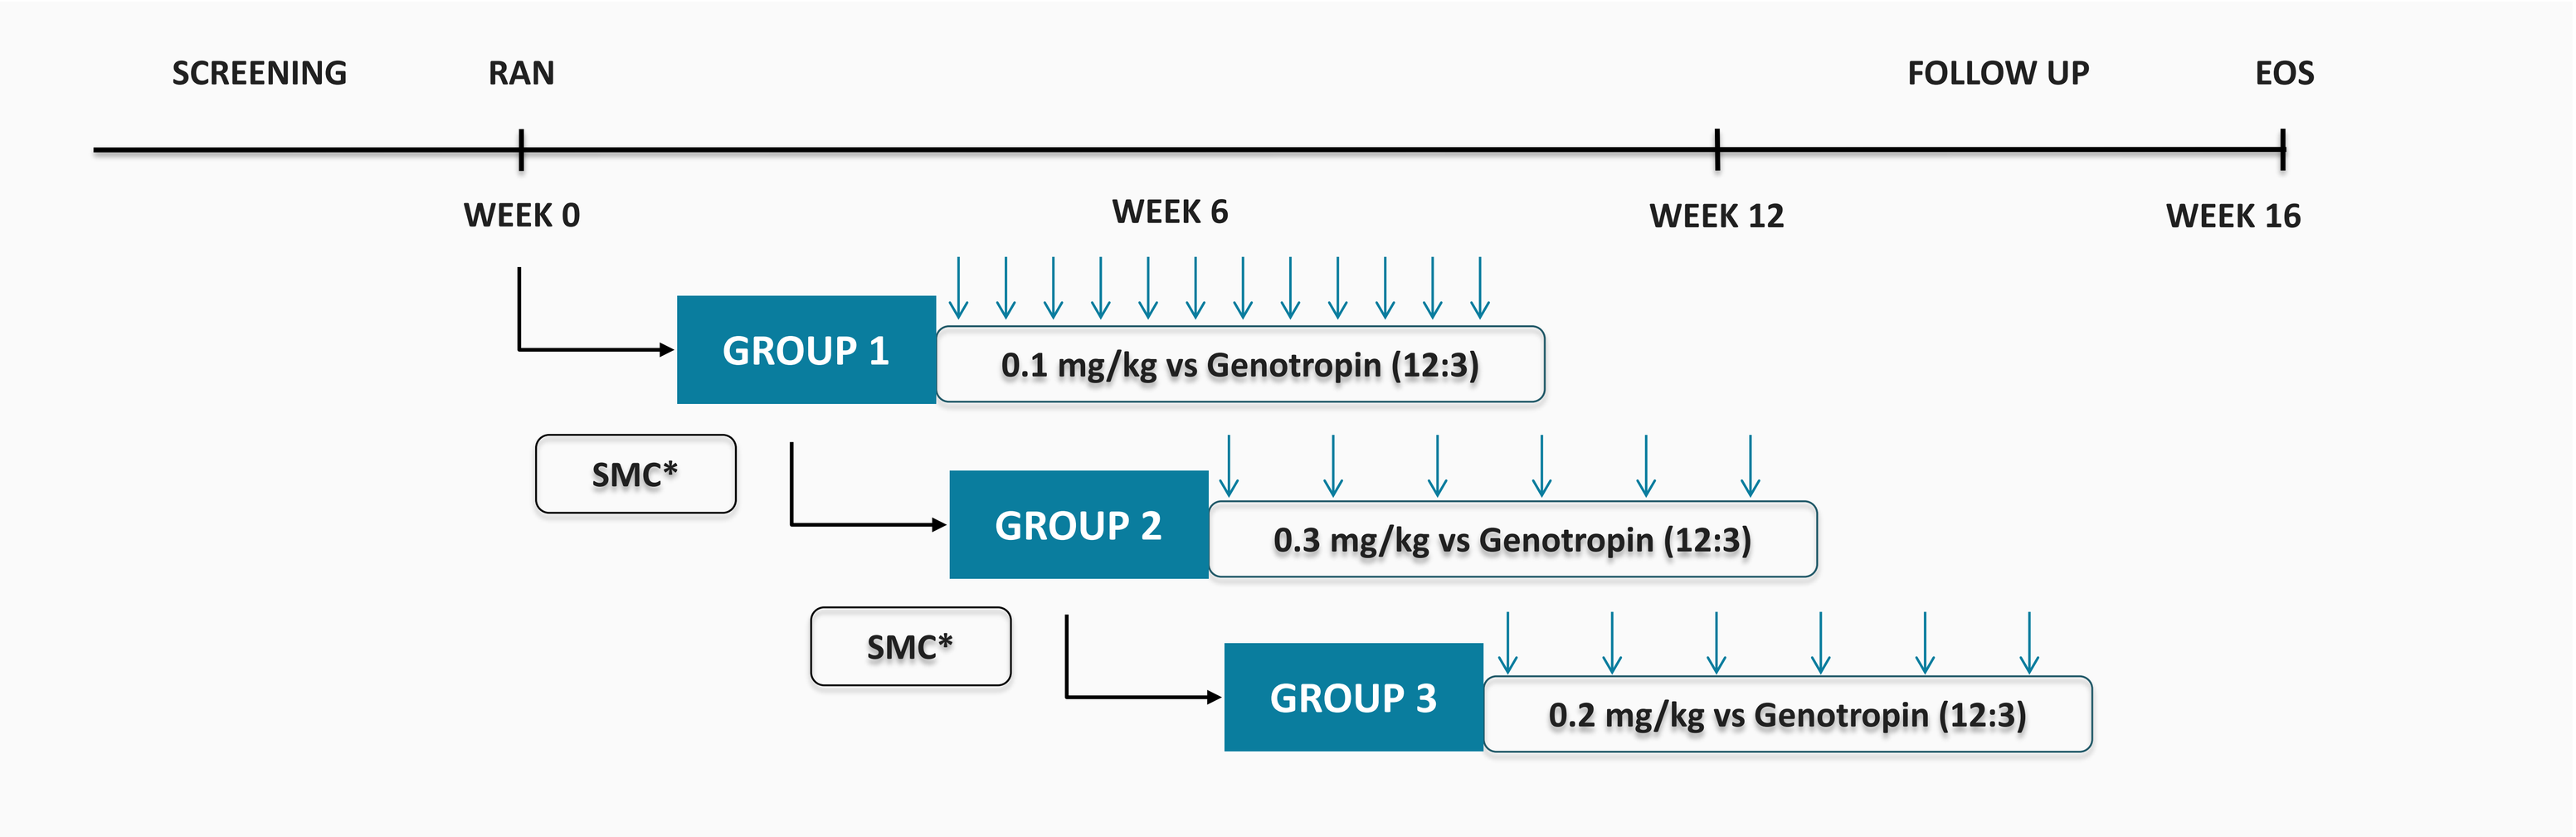
SCREENING
RAN
FOLLOW UP
EOS
WEEK 0
WEEK 6
WEEK 12
WEEK 16
GROUP 1
0.1 mg/kg vs Genotropin (12:3)
SMC*
GROUP 2
0.3 mg/kg vs Genotropin (12:3)
SMC*
GROUP 3
0.2 mg/kg vs Genotropin (12:3)

**After week 6 of last patients in each group, available safety and tolerability data will be reviewed by the PI, Sponsor, and the SMC*

- **Results – Pharmacokinetic & Pharmacodynamics**

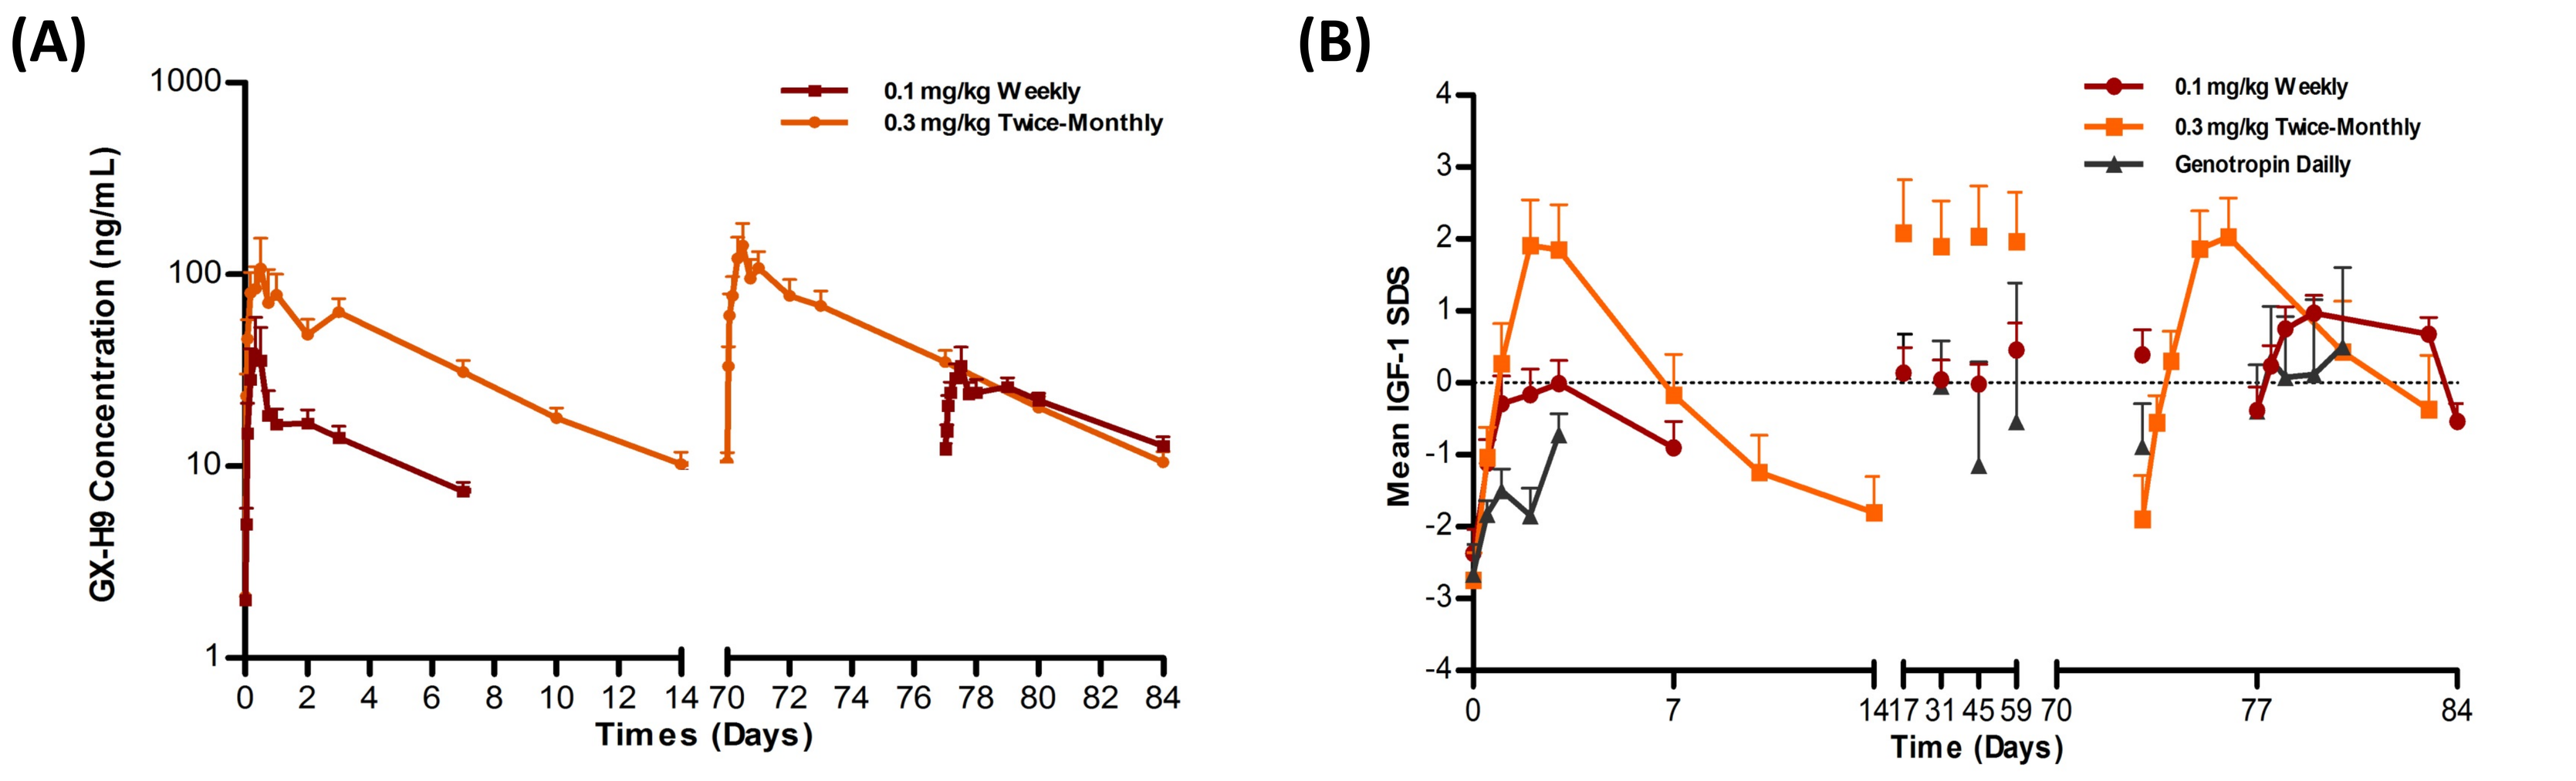
(A)
GX-H9 Concentration (ng/mL)
0.1 mg/kg Weekly
0.3 mg/kg Twice-Monthly
Times (Days)
(B)
Mean IGF-1 SDS
0.1 mg/kg Weekly
0.3 mg/kg Twice-Monthly
Genotropin Dailly
Time (Days)

PK and PD of GX-H9 in AGHD. Subjects received 0.1 mg/kg GX-H9 weekly or 0.3 mg/kg GX-H9 twice-monthly for 12 weeks subcutaneously.

(A) Serum GX-H9 Concentration-Time profiles following GX-H9 administration.
(B) Mean IGF-1 SDS for GX-H9 dosing groups. Baseline was defined as the Day 0 pre-dose value.

METHODS & RESULTS – Phase 2 Pediatric GHD Study

- **Study Design (Single Dose + Multiple Dose)**

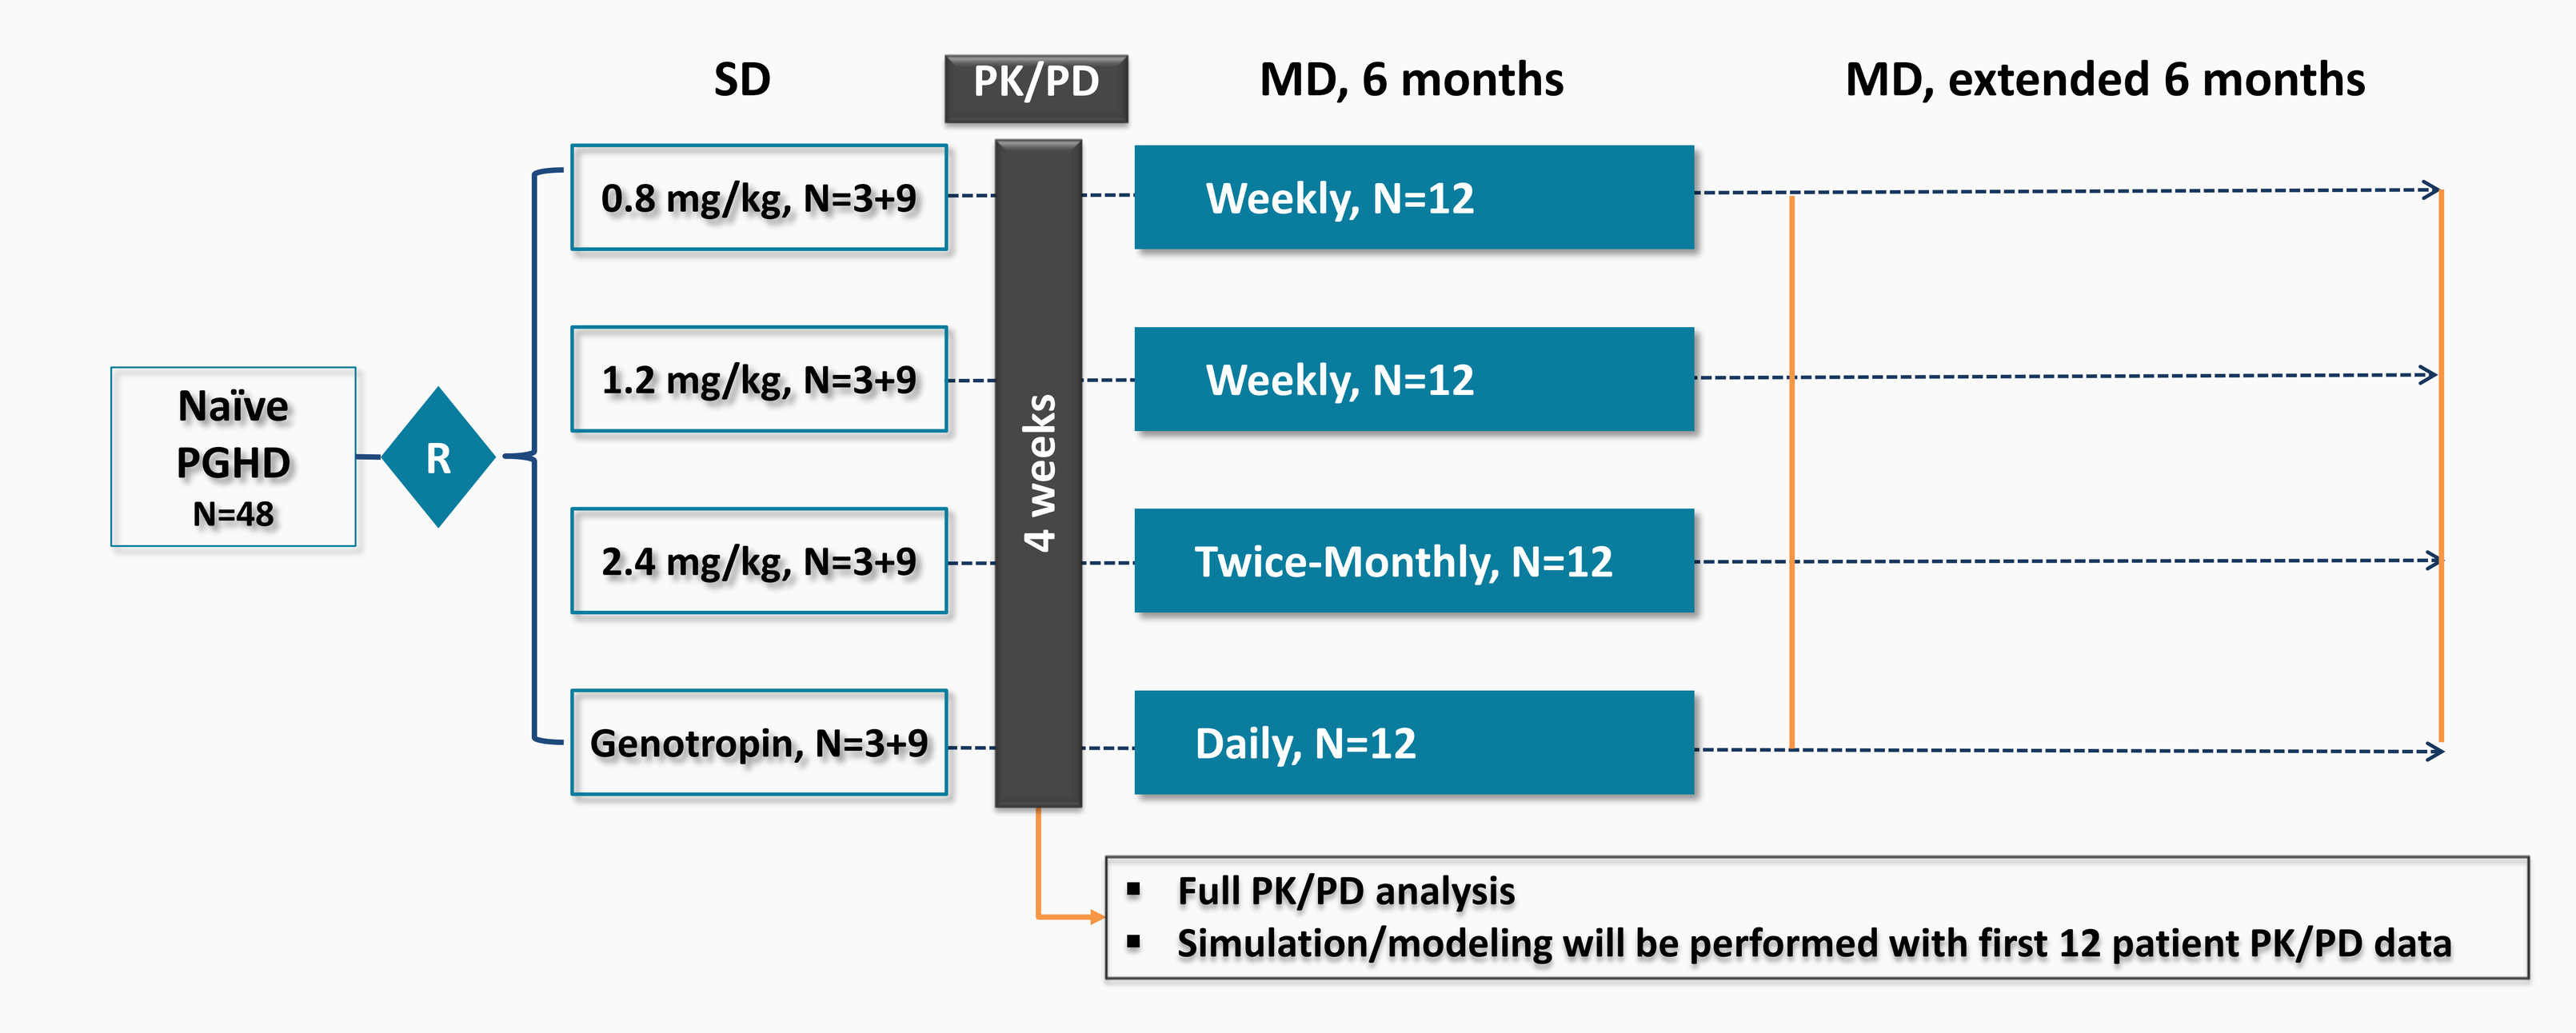
SD
PK/PD
MD, 6 months
MD, extended 6 months
Naïve PGHD N=48
R
0.8 mg/kg, N=3+9
1.2 mg/kg, N=3+9
2.4 mg/kg, N=3+9
Genotropin, N=3+9
4 weeks
Weekly, N=12
Weekly, N=12
Twice-Monthly, N=12
Daily, N=12
Full PK/PD analysis
Simulation/modeling will be performed with first 12 patient PK/PD data

- **Results – Pharmacokinetic & Pharmacodynamics**

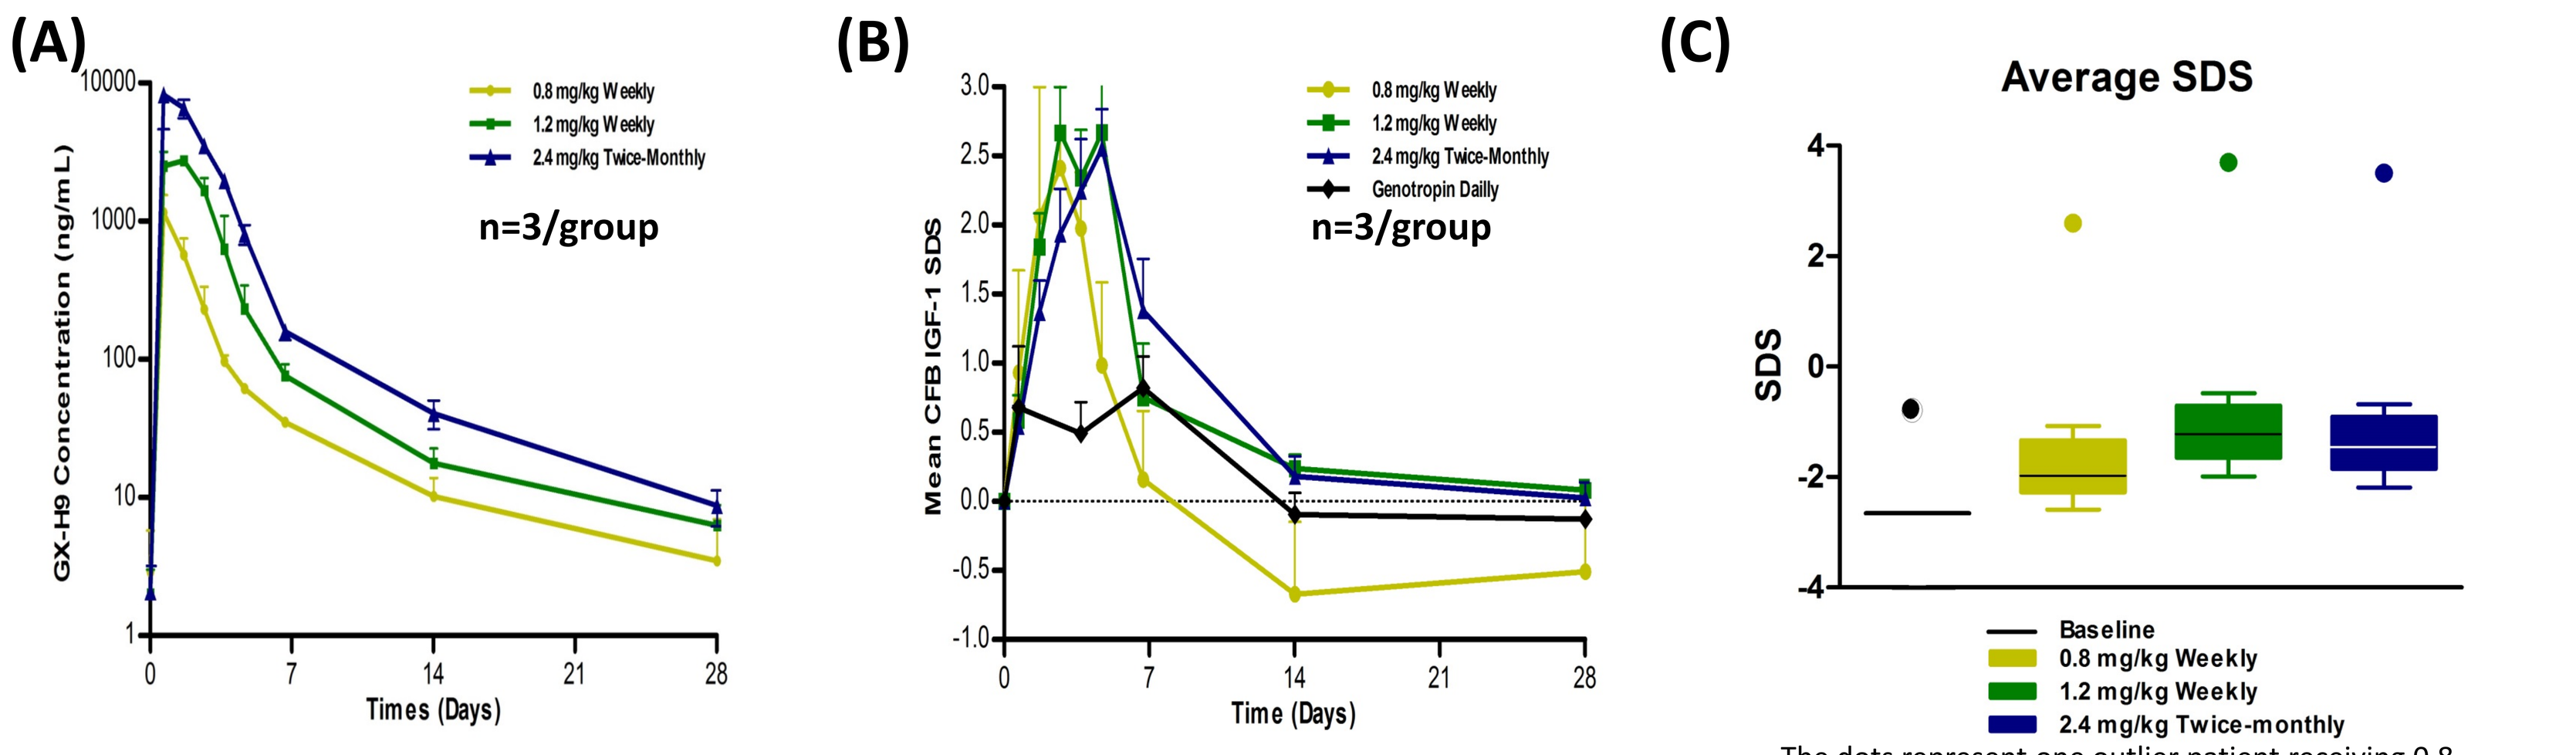
(A)
GX-H9 Concentration (ng/mL)
0.8 mg/kg Weekly
1.2 mg/kg Weekly
2.4 mg/kg Twice-Monthly
n=3/group
Times (Days)
(B)
Mean CFB IGF-1 SDS
Genotropin Dailly
n=3/group
Time (Days)
(C)
Average SDS
SDS
Baseline
0.8 mg/kg Weekly
1.2 mg/kg Weekly
2.4 mg/kg Twice-monthly

The dots represent one outlier patient receiving 0.8 mg/kg/week who had the baseline IGF-1 value close to -1 SDS.

PK and PD of GX-H9 in PGHD. Subjects received single dose of 0.8 , 1.2 or 2.4 mg/kg of GX-H9, followed up to week 4

(A) Serum GX-H9 Concentration-Time profiles following GX-H9 administration.
(B) Mean change from baseline in IGF-1 SDS for GX-H9 dosing groups. Each point indicates potential C_{max} value of IGF-1 SDS after injection. Baseline was defined as the Day 0 pre-dose value (CFB; change from baseline)
(C) Boxplot of individual IGF1-SDS values after single dosing (three simulated regimens)

CONCLUSION

- GX-H9 was safe and well tolerated in healthy volunteers and patients with GHD (adult and limited injections to pediatric)
- Dose dependent PK/PD profiles were observed
- No formation of treatment-emergent ADA was found so far
- Phase 2 adult and pediatric studies showed comparable safety profile with 1st generation hGH
- Weekly and twice-monthly administration of GX-H9 showed potential for both weekly and twice-monthly treatment

ACKNOWLEDGEMENTS

- This study was sponsored by Genexine, Inc. and Handok.
- This research was supported by Korea Drug Development Fund (KDDF) funded by MSIP, MOTIE and MOHW (Grant No. KDDF-201308-07 for Phase 1 Study, KDDF-201502-11 for Phase 2 AGHD Study, & KDDF-201509-12 for Phase 2 PGHD Study)

Disclosure: TK Kim, JW Woo, YC Sung, HM Keyoung are employees and stockholders of Genexine, Inc. MS Kang, WI Jang are employees of Handok.

55ESPE 8--4-RFC
Growth: Clinical
Jungwon Woo
DOI: 10.3252/pso.eu.55ESPE.2016
Poster presented at: