Optimal Sampling of IGF-1 during Weekly Administration of a Long-Acting Human Growth Hormone (MOD-4023)

Dennis M. Fisher, MD*; Michal Jaron–Mendelson, PhD†; Shelly Vander, MSc†; Ronit Koren, PhD†; Gili Hart, PhD†

*P Less Than, San Francisco; †OPKO Biologics, Nes Ziona, Israel

INTRODUCTION

OPKO Biologics is developing long-acting versions of existing therapeutic proteins utilizing CTP technology. This technology fuses the C-terminus peptide of human chorionic gonadotropin (hCG) to one or both ends of a target protein. CTP technology has been validated clinically and proven as a safe and efficient way to increase the half-life of several therapeutic proteins while maintaining their biological activity.

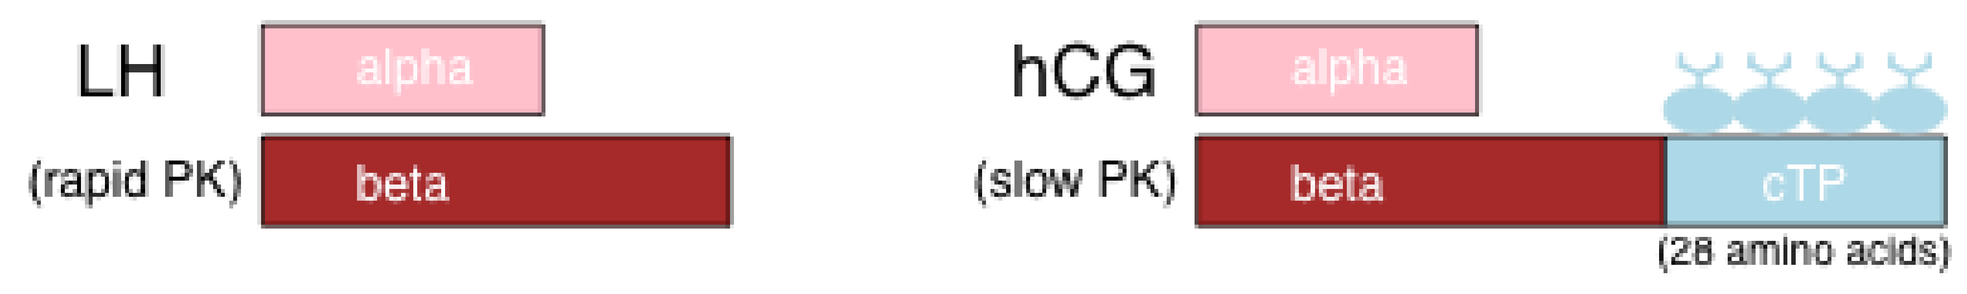
LH
(rapid PK)
alpha
beta
hCG
(slow PK)
alpha
beta
cTP
(28 amino acids)

MOD-4023, application of the CTP technology to human growth hormone (hGH), is being developed for the treatment of short stature in children. The goal is to develop a product that allows weekly dosing while maintaining a small clinically-tolerable injection volume.

During Phase 3 clinical trials and following future approval of MOD-4023 for clinical use, IGF-1 SDS values will be used to assess safety of weekly therapy and to determine any potential need for dose adjustment. During the one-week dosing interval, IGF1 (and resulting IGF-1 SDS) values fluctuate. Therefore, it is important to understand the relationship between IGF1 values obtained at different timepoints in the dosing interval to the peak and mean IGF−1 SDS during the dosing interval.

We used pharmacokinetic (PK) and pharmacodynamic (PD) modeling and simulation to determine the optimal time to sample IGF-1.

METHODS: TRIAL DESIGN

In clinical trial CP-4-004, treatment-naive GH-deficient children aged 3-11 years received subcutaneous MOD-4023 weekly for up to one year. Sparse samples (MOD-4023, IGF-1) were obtained after the second steady state dose and at later timepoints.

METHODS: PHARMACOKINETIC / PHARMACODYNAMIC ANALYSIS

- Mixed-effects (population) methods with NONMEM (Icon Development Solutions) were needed because of sparse sampling
- PK model: linear 2-compartment model with first-order absorption and absorption lag
- PD model:
 — Indirect model (1) relates drug concentration to IGF-1 input; sigmoid Emax relationship between drug concentration and effect
 — Based on IGF-1 rather than IGF-1 SDS (IGF-1 SDS is non-linear function of IGF-1)
- Body size, age, gender, organ function evaluated as covariates for all PK and PD parameters

METHODS: PHARMACOKINETIC / PHARMACODYNAMIC SIMULATION

- For each subject, the time course of MOD-4023 and IGF-1 was simulated following 11 weekly doses. The dose escalation scheme employed in the trial was implemented, followed by equal-sized steady state doses.
- IGF-1 SDS values estimated based on IGF-1 values, age, and gender, using Bidlingmaier's reference tables (2)
- For each subject, the simulated values for each of MOD-4023 and IGF-1 SDS were examined to determine:
 — mean values over the dosing interval (determined using linear trapezoids)
 — peak values
 — values at each of Day 1 (24 hours post-dose) through Day 7 (end of dosing interval)
- Graphics were prepared to examine the relationship between the values at each day *vs.* peak / mean values

RESULTS

Peak IGF-1 SDS: Samples obtained on **Day 2** (48 hours post-dose) predicted **peak** IGF-1 SDS well (**Figure 1**): correlation was high (r > 0.99) and deviation from the line of unity was minimal. Values obtained on all other days (Day 4 displayed) underestimated peak IGF-1 SDS. However, there was a strong linear relationship between the value on any day and peak IGF-1 SDS; the r value and intercept/slope for those relationships are displayed in **Table 1**.

Mean IGF-1 SDS: Samples obtained on **Day 4** (96 hours post-dose) predicted **mean** IGF-1 SDS well (**Figure 2**): correlation was high (r > 0.99) and deviation from the line of unity was minimal. Values obtained on all Day 2 (displayed) and Day 3 overestimated mean IGF-1 SDS whereas those obtained on Days 6 or 7 underestimated mean IGF-1 SDS. However, there was a strong linear relationship between the value on any day and mean IGF-1 SDS; the r value and intercept/slope for those relationships are displayed in **Table 2.**

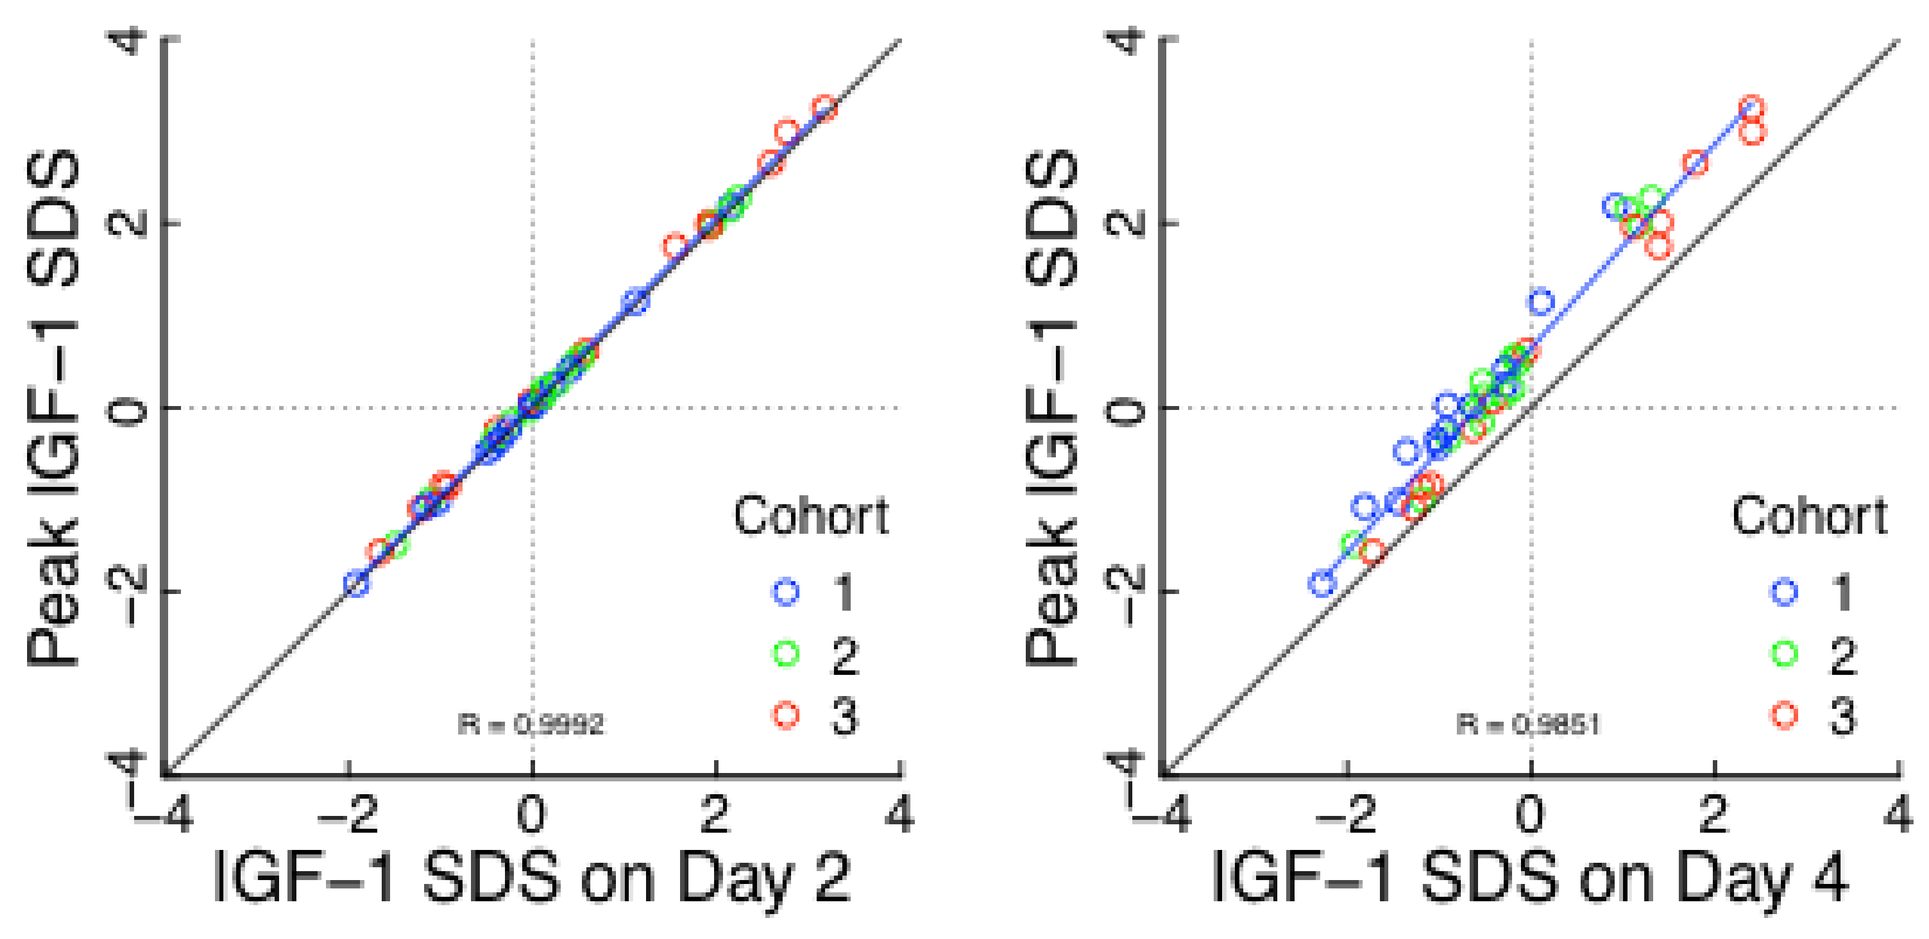
Peak IGF−1 SDS
IGF−1 SDS on Day 2
R = 0.9992
Cohort
1
2
3
Peak IGF−1 SDS
IGF−1 SDS on Day 4
R = 0.9851
Cohort
1
2
3

Figure 1. Peak IGF-1 for each subject is plotted again the value at Day 2 (left) or Day 4 (right)

Table 1. The relationship (r, intercept, and slope of a linear regression) between peak IGF1- SDS and the value obtained at Day 1 – Day 7 is shown.

DAY	r	Intercept	Slope
1	0.9948	0.5765	1.1471
2	0.9992	0.0485	1.0059
3	0.9937	0.1886	1.0141
4	0.9851	0.6417	1.1099
5	0.9811	1.2377	1.2515
6	0.9778	1.9332	1.4213
7	0.9730	2.7216	1.6147

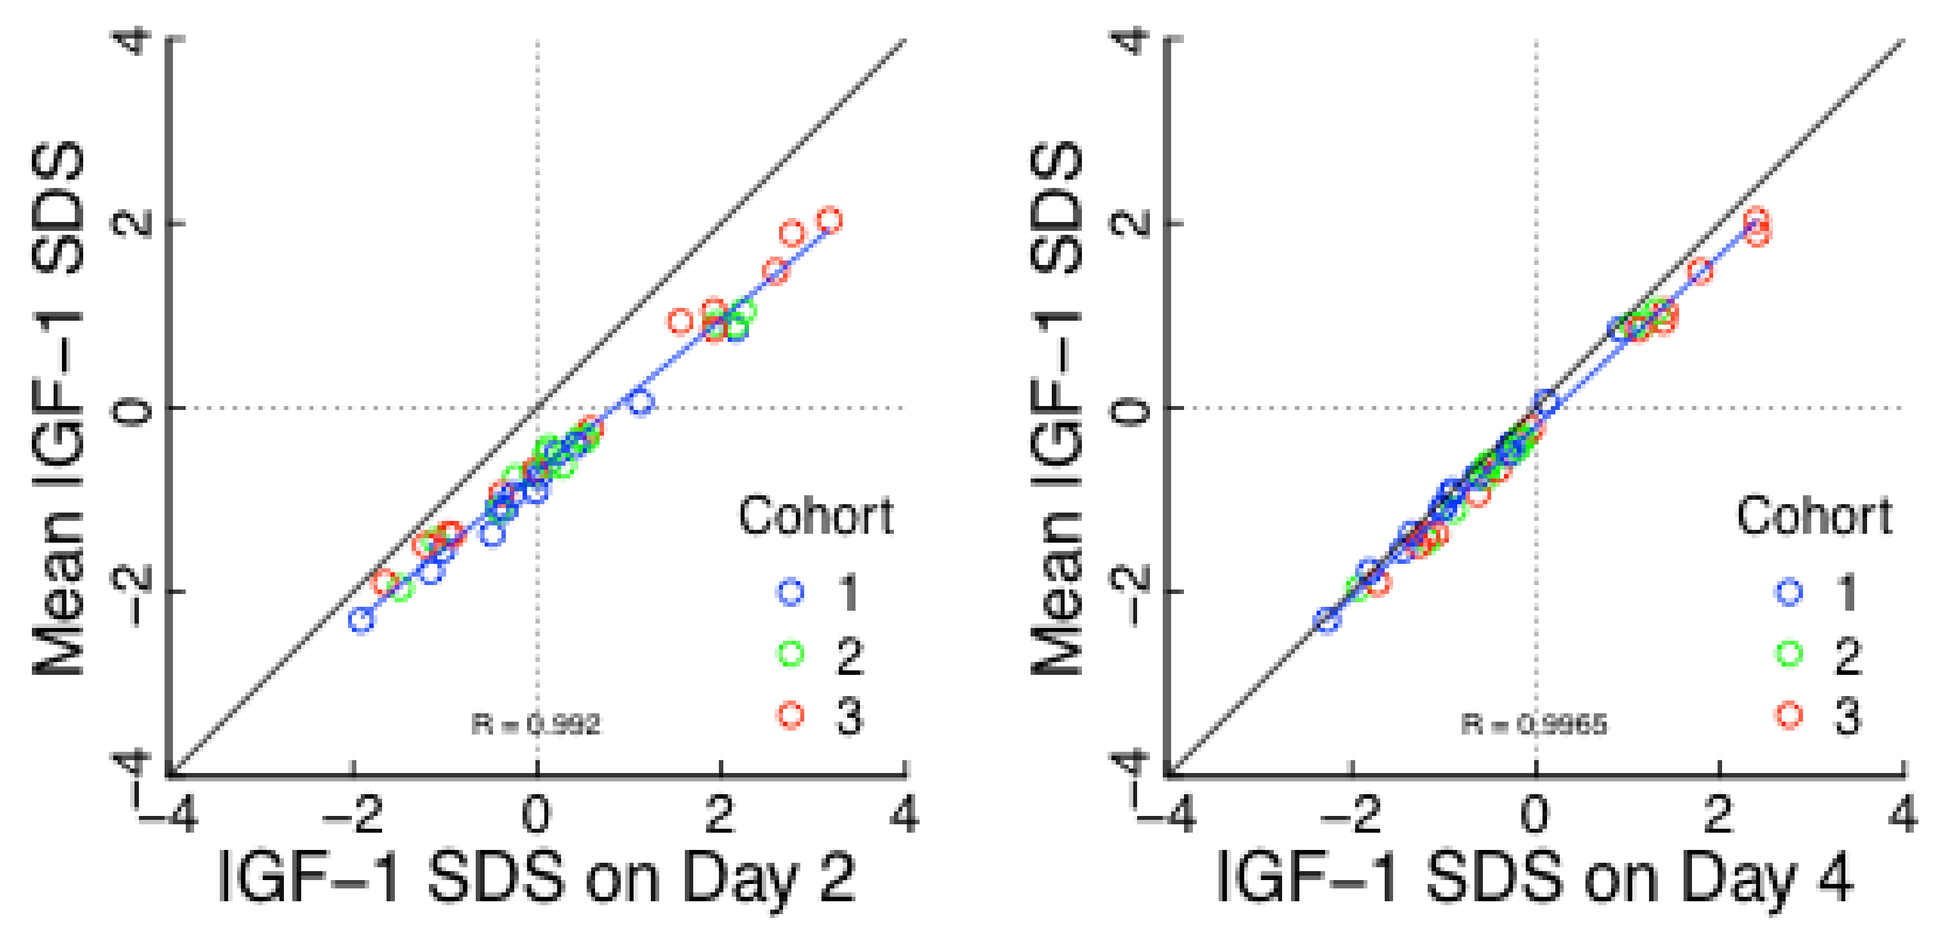
Mean IGF−1 SDS
IGF−1 SDS on Day 2
R = 0.992
Cohort
1
2
3
Mean IGF−1 SDS
IGF−1 SDS on Day 4
R = 0.9965
Cohort
1
2
3

Figure 2. Mean IGF-1 for each subject is plotted again the value at Day 2 (left) or Day 4 (right)

Table 2. The relationship (r, intercept, and slope of a linear regression) between mean IGF1- SDS and the value obtained at Day 1 – Day 7 is shown.

DAY	r	Intercept	Slope
1	0.9839	-0.2611	0.9396
2	0.9920	-0.6946	0.8271
3	0.9988	-0.5812	0.8442
4	0.9965	-0.2025	0.9298
5	0.9952	0.2988	1.0513
6	0.9937	0.8855	1.1962
7	0.9906	1.5528	1.3616

For both peak and mean IGF-1 SDS, the relationship to values on any sampling day did not differ between cohorts.

CONCLUSIONS

- IGF-1 SDS, determined from an IGF-1 sample obtained at **Day 2**, provides a direct estimate of **peak** IGF-1 SDS. In contrast, a sample at **Day 4** provides a direct estimate of **mean** IGF-1 SDS.
- Samples obtained at other days can be used to estimate peak or mean IGF-1 SDS using the linear relationships displayed in **Table 1** and **Table 2**.
- These values of peak and/or mean IGF-1 SDS can be used to adjust dosing of MOD-4023.
- Similar results (not reported) were obtained from a study in GH-deficient adults (CP-4-003).

REFERENCES

1. Sun *et al*. JPET 1999; 289:1523
2. Bidlingmaier *et al.* J Clin Endocrinol Metab 2014; 99:1712

Disclosure Statement

DMF: Consultant, OPKO Biologics. Nothing to Disclose: MJM, SV, RK, GH

Poster RFC8.5

55ESPE 8--5-RFC
Growth: Clinical
Gili Hart
DOI: 10.3252/pso.eu.55ESPE.2016
Poster presented at: ESPE 2016 PARIS
PosterSessionOnline.com