P1-P212

Real-world safety data in a cohort of children with Noonan syndrome treated with growth hormone: final results from NordiNet® International Outcome Study (IOS) and the ANSWER Program

Pétur Benedikt Júlíusson[1]; Jovanna Dahlgren[2]; M. Jennifer Abuzzahab[3]; Birgitte Tønnes Pedersen[4]; Sebastian Röhrich[5]; Alicia Romano[6]

[1]University of Bergen, Bergen, Norway; [2]University of Gothenburg, Gothenburg, Sweden; [3]Children's Minnesota, Saint Paul, MN, USA; [4]Novo Nordisk A/S, Søborg, Denmark; [5]Novo Nordisk Health Care AG, Zurich, Switzerland; [6]New York Medical College, Valhalla, NY, USA

Objective

To describe real-world safety data on growth hormone (GH) therapy in paediatric patients with Noonan syndrome (NS) who were enrolled in NordiNet® IOS and the ANSWER Program.

Introduction

- Patients with NS have a high prevalence of cardiac defects[1] and a higher risk than the general population for leukaemia and certain solid tumours.[2]
- Current safety data do not indicate an association of GH therapy with worsening of congenital cardiac defects or an increased risk for malignancies in individuals with NS; however, data are limited.[2]

Methods

- The long-term effectiveness and safety of Norditropin® (somatropin; Novo Nordisk A/S, Denmark), as prescribed in clinical practice, were evaluated in two complementary, non-interventional, multicentre studies (**Figure 1**).[3]
- We report here safety results for 412 paediatric patients with NS. Patients for whom a valid Norditropin® exposure was not recorded were excluded from the safety evaluation (**Figure 1**).

Figure 1 • Countries included, total number of patients and numbers of patients with Noonan syndrome enrolled in the two studies

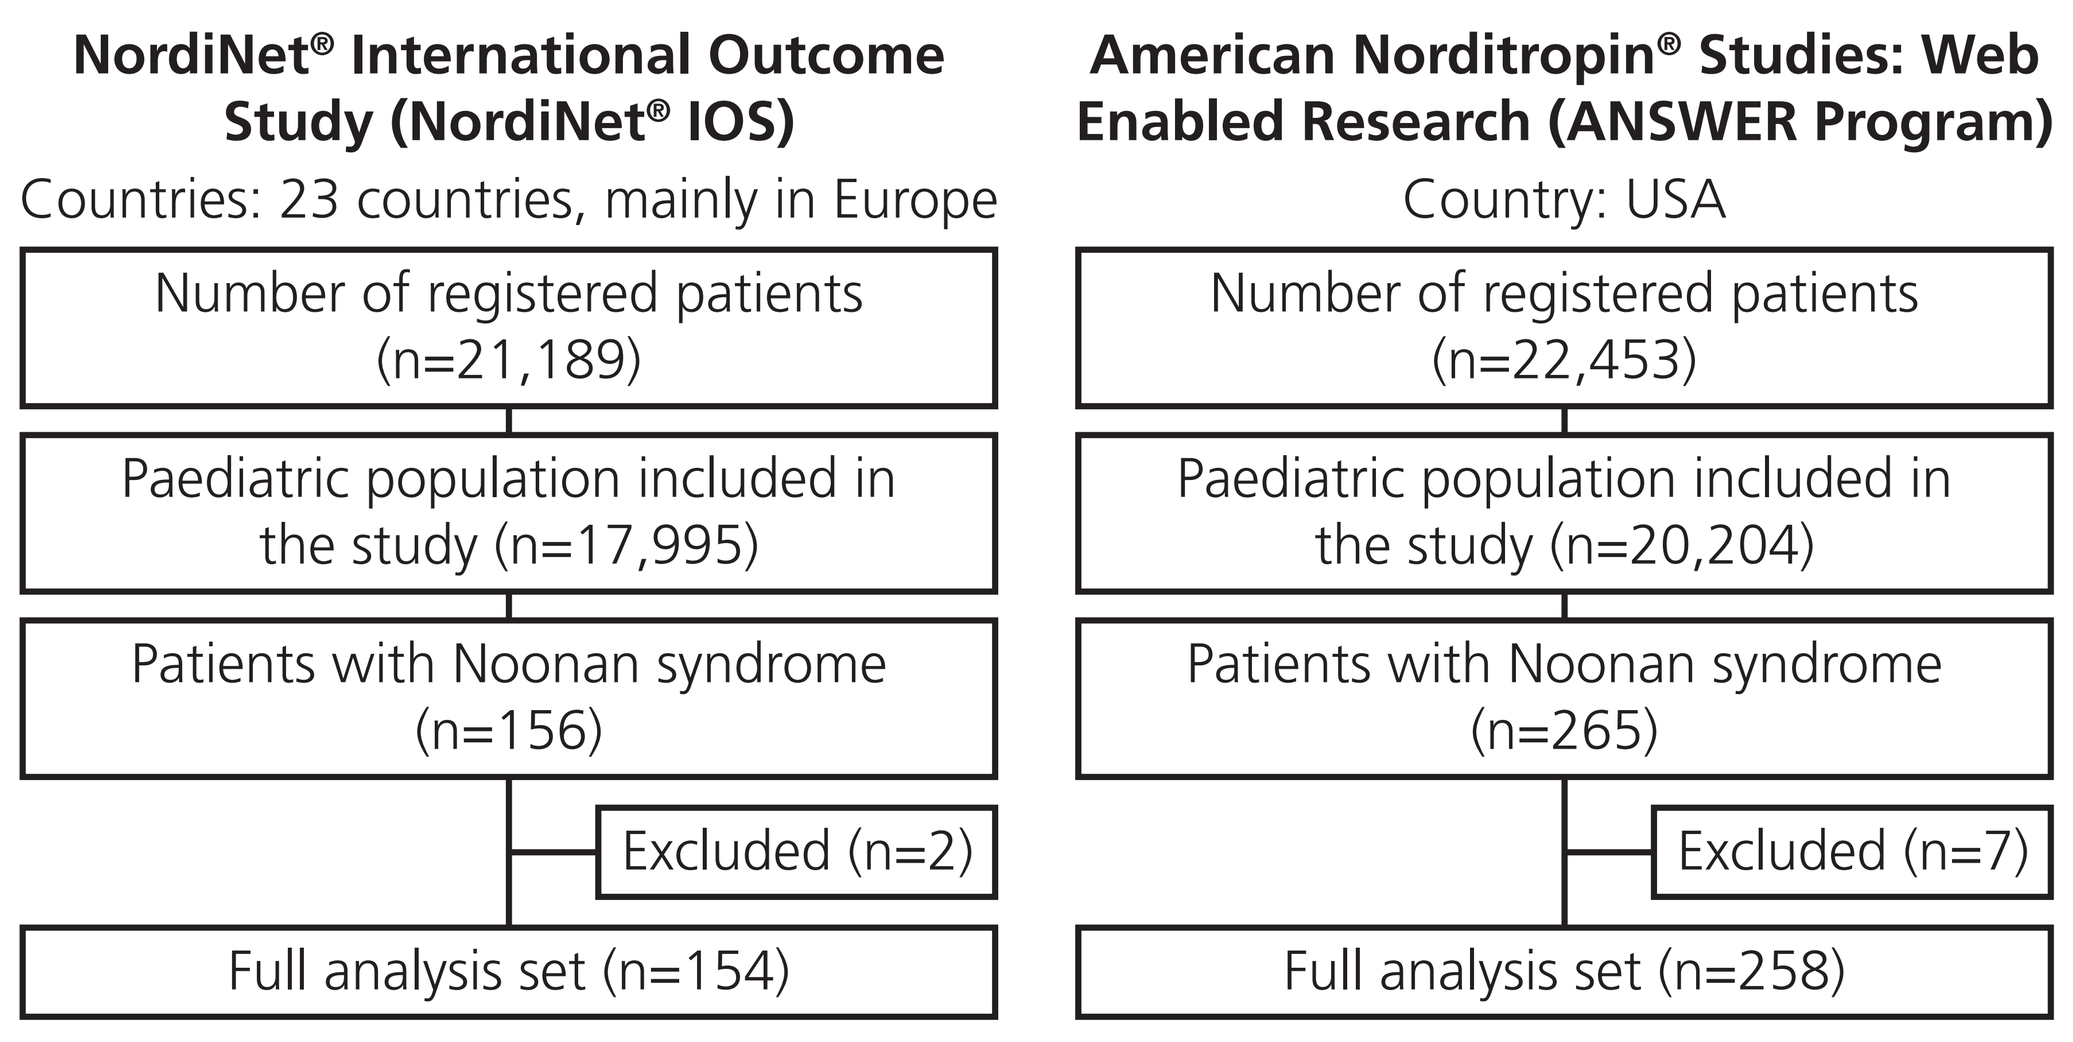
NordiNet® International Outcome Study (NordiNet® IOS)
Countries: 23 countries, mainly in Europe
Number of registered patients (n=21,189)
Paediatric population included in the study (n=17,995)
Patients with Noonan syndrome (n=156)
Excluded (n=2)
Full analysis set (n=154)
American Norditropin® Studies: Web Enabled Research (ANSWER Program)
Country: USA
Number of registered patients (n=22,453)
Paediatric population included in the study (n=20,204)
Patients with Noonan syndrome (n=265)
Excluded (n=7)
Full analysis set (n=258)

The full analysis set includes all patients with available birthdate information who were <18 years of age and with at least one Norditropin® prescription recorded.

- Safety data are presented descriptively for all patients with NS included in the full analysis set (FAS).
- Data were collected and reported in line with local or national practice. Safety events could be observed by the physician or reported by the patient.
- Safety events were reported as serious adverse reactions (SARs) and non-serious adverse reactions (NSARs); events judged as possibly or probably related to use of Norditropin®, by the reporter or Novo Nordisk or both. Serious adverse events (SAEs) not related to GH therapy were also analysed for NordiNet® IOS.
- Safety events were coded using the Medical Dictionary for Regulatory Activities (MedDRA).

Results

Patient characteristics

- Baseline characteristics and exposure information for all patients and for patients with safety events are presented in **Table 1**.
- Genotypes were only available for patients included in the ANSWER Program. Of 258 patients, data on mutations were available for 61 patients as follows: *PTPN11*, n=56; *KRAS*, n=2; *SOS1*, n=2; *RAF1*, n=5; *SHOC2*, n=1 (one patient could have more than one mutation).
- Cardiovascular (CV) comorbidities were reported in 35 (8.5%) patients prior to GH start; comorbidities reported in three or more patients are shown in **Table 2**.
- After start of GH treatment, (potentially pre-existing) CV comorbidities were reported in five patients: unspecified CV disease (n=3), pulmonary valve stenosis (n=1) and ruptured abdominal aortic aneurysm (n=1).
 - These events were all reported as comorbidities rather than AEs. The ruptured abdominal aortic aneurysm occurred after the start of GH treatment and should have been considered as an SAE, based on medical judgement (see 'Specific patients' below).

Safety outcomes

- Overall, 31 safety events were reported in 21 patients (**Table 3**); of these, 68% (21/31) were NSARs.
- Most patients with safety events reported a single event (16/21). One patient reported two SARs (see 'Specific patients' below).
- Aside from the comorbidities listed above, no cardiac SARs, NSARs or SAEs not related to GH therapy were reported.
- Under the MedDRA term 'Neoplasms, benign, malignant and unspecified' four events were reported in three patients (**Table 3**).

Specific patients

- The patient with two SARs (**Table 3**) reported a brain neoplasm (metastatic fourth ventricular pilocytic astrocytoma) and metastases to spine, which were considered as possibly related to GH treatment by the reporter. This patient (*PTPN11* mutation) had a history of headaches, which may suggest an underlying condition. A temporal association between reported brain neoplasm and GH treatment cannot be excluded, but further medical assessment was not undertaken.
- One patient (*PTPN11* mutation) had a ruptured abdominal aortic aneurysm diagnosed 26 months after start of follow-up. This patient was also diagnosed with Crohn's disease (22 months after start of follow-up) and with a glioneuronal tumour 1 week after the aneurysm diagnosis.

Table 1 • Baseline characteristics of patients (all patients and patients with safety events) and GH exposure information

	All patients		*Patients with safety events*	
	Number of patients	*Mean (SD)**	*Number of patients*	*Mean (SD)**
Female/male, n and %	120/292	29.1%/70.9%	8/13	38.1%/61.9%
Age (years)	412	9.48 (3.92)	21	9.90 (4.13)
Height SDS†	371	–2.65 (0.95)	16	–3.14 (0.82)
Weight SDS†	308	–2.03 (1.31)	13	–2.57 (1.54)
Bone age/chronological age	163	0.83 (0.19)	7	0.86 (0.10)
IGF-I SDS‡	162	–1.13 (1.62)	6	–1.10 (2.14)
GH dose at baseline (µg/kg/day)	403	43.9 (13.7)	19	42.4 (17.3)
GH-naïve at baseline, yes (%)	282	68.4%	12	57.1%
Duration of treatment follow-up (years)	412	3.1 (2.6)	21	3.06 (2.17)
GH dose during childhood (µg/kg/day)	412	46.6 (13.6)	21	47.6 (16.8)

*Unless otherwise specified. †Height and weight SDS were calculated using age- and gender-specific national references. ‡Brabant G *et al. Horm Res* 2003;60:53–60. GH, growth hormone; IGF, insulin-like growth factor; SD, standard deviation; SDS, standard deviation score.

Table 2 • Cardiac comorbidities reported in three or more patients at baseline

Comorbidity	*Number of patients*
Pulmonary valve stenosis	13
Congenital pulmonary valve stenosis	6
Atrial septal defect/atrioventricular septal defect	4/1
Unspecified cardiovascular disease	4
Cardiac murmur	3
Congenital malformation of the heart	3
Other hypertrophic cardiomyopathy	3

Patients could have more than one diagnosis.

Table 3 • Summary of safety events

	Number of safety events (number of patients)			*Total number of safety events (total number of patients)*
Preferred term	*NSAR*	*SAR*	*SAE not related to GH*	
Scoliosis	2 (2)	–	1 (1)	3 (3)
Headache	6 (6)	–	–	6 (6)
Myalgia	2 (2)	–	–	2 (2)
Arthralgia	5 (3)	–	–	5 (3)
Brain neoplasm	–	1 (1)	–	1 (1)
Metastases to spine	–	1 (1)	–	1 (1)
Giant cell epulis	–	–	1 (1)	1 (1)
Epilepsy	–	–	1 (1)	1 (1)
Glioneuronal tumour	–	–	2 (2)	2 (2)
Condition aggravated	–	–	1 (1)	1 (1)
Spinal fusion surgery	–	–	1 (1)	1 (1)
Moyamoya disease	–	–	1 (1)	1 (1)
Other*	6 (5)	–	–	6 (5)
Total	21 (15)	2 (1)	8 (5)	31 (21)

*Other NSARs were oedema, injection site erythema, growing pains, muscle spasms, condition-aggravated (patient with arthralgia) and injection site extravasation. GH, growth hormone; NSAR, non-serious adverse reaction; SAE, serious adverse event; SAR, serious adverse reaction.

Discussion

- In the current analysis, one cardiac safety event (ruptured abdominal aortic aneurysm) was reported. A recent randomised, double-blind, clinical trial of Norditropin®, in which cardiac function was monitored (n=51), showed no evidence of a negative effect of GH on cardiac function or structure.[4] Furthermore, previous reports indicate that long-term GH treatment does not appear to have negative effects on the heart, in particular, ventricular wall thickness.[1,2]
- As there was no requirement in the protocol to report cardiac comorbidities at baseline, these baseline data may have been under-reported.
- The underlying pathophysiology of NS includes dysregulation of the Ras/mitogen-activated protein kinase signalling pathway, which may increase the intrinsic risk of cancer development.[5] NS is associated with a higher risk of benign and malignant proliferative disorders,[6] including solid tumours,[7] and glioneural tumour and astrocytoma have both been reported in patients with NS.[6] The available data on GH and cancer risk give no cause for concern, but underlying susceptibility to tumour growth should be considered when GH therapy is started.[2]

Conclusions

- Real-world data from NordiNet® IOS and the ANSWER Program support a favourable safety profile of GH therapy in patients with NS, specifically with regard to cardiac safety events.
- As with other real-world studies, assessment of safety events may be difficult owing to the unknown natural history of and/or under-reporting of comorbidities.

References

1. Noordam C *et al. Horm Res* 2009;72(Suppl 2):49–51.
2. Noonan JA *et al. Horm Res Paediatr* 2015;83:157–66.
3. Höybye C *et al. Clin Epidemiol* 2013;26:119–27.
4. Ozono K *et al. Endocr J* 2018;65:159–74.
5. Smpoku P *et al. Clin Genet* 2015;88:516–22.
6. Jongmans M *et al. Eur J Hum Genet* 2011;19:870–4.
7. Kratz CP *et al. Br J Cancer* 2015;112:1392–7.

Conflict of interest disclosures

PBJ received a lecture fee from Novo Nordisk for a presentation at a Novo Nordisk meeting. JD received lecture fees (2007) and unrestricted grants from Novo Nordisk for a genetic study in NS. MJA is on the speakers' bureau for Novo Nordisk and participates as PI on study NN8640-4231. BTP is an employee of, and owns stocks/shares in, Novo Nordisk A/S. SR is an employee of Novo Nordisk Health Care AG. AR is on the speakers' bureau for Genentech, Novo Nordisk and Genzyme, and is a consultant for Genentech and Novo Nordisk.

This study was supported by Novo Nordisk. NordiNet® IOS is registered at ClinicalTrials.gov (NCT00960128). The ANSWER Program is registered at ClinicalTrials.gov (NCT01009905). The authors thank the investigators and patients participating in this study. The authors take full responsibility for the content of the poster but are grateful to Watermeadow Medical (supported by Novo Nordisk) for writing assistance.
Presented at the 57th Annual Meeting of the European Society for Paediatric Endocrinology, Athens, Greece, 27–29 September 2018.

57ESPE 212--P1
Pituitary, neuroendocrinology and puberty
Pétur Benedikt Júlíusson
Poster presented at: 57th ESPE 2018 Meeting ATHENS GREECE 27-29 September 2018
PosterSessionOnline.com