Changes in body composition in male adolescents with severe GH deficiency are diagnostic during transition

Gerhard Binder, Bettina Becker, Jana Leonie Bauer, Roland Schweizer
University Children's Hospital, Pediatric Endocrinology,
Hoppe-Seyler-Strasse 1, 72076 Tübingen, Germany

Objective

Restarting rhGH in adolescents with childhood-onset GHD is usually based on retest GH, IGF-1, additional pituitary hormone deficiencies and pituitary morphology. Short-term changes in body composition in adolescents with childhood-onset GHD when off rhGH may contribute to the identification of those in need of treatment continuation.

Patients and Measurements

This was a prospective longitudinal single-center study. The body composition of 58 adolescents (12 females) with low-likelihood severe GHD of adolescence was measured by DXA 6 months before and at rhGH discontinuation, as well as 6 and 12 months thereafter. The adolescents were re-examined after 3 months off rhGH using IGF-1 and GHRH-arginine test. Severe GHD of adolescence was defined both by stimulated GH < 16 ng/ml and IGF-1 < -1.90 SDS.

Patients' characteristics (n=58)

Familial target height	SDS	-0.45 ± 0.71
Age at diagnosis	y	5.4 ± 2.8
Bone age at diagnosis	y	4.0 ± 2.5
Height at diagnosis	SDS	-2.85 ± 0.72
GH maximum at diagnosis	ng/ml	5.0 ± 2.2
IGF-1 at diagnosis	SDS	-4.71 ± 1.86
Age at stop of rhGH	y	16.4 ± 1.2
Height at stop of rhGH	SDS	-0.48 ± 0.86
Malformation of the pituitary	n	9
MPHD	n	7

Results

Severe GHD of adolescence had 7 out of 58 patients. Because of the low number of recruited females, we included 5 additional females from the pilot study.

After six months off rhGH males with severe GHD of adolescence gained significantly more fat mass (FM) and lost more lean body mass (LBM) than healthy individuals.

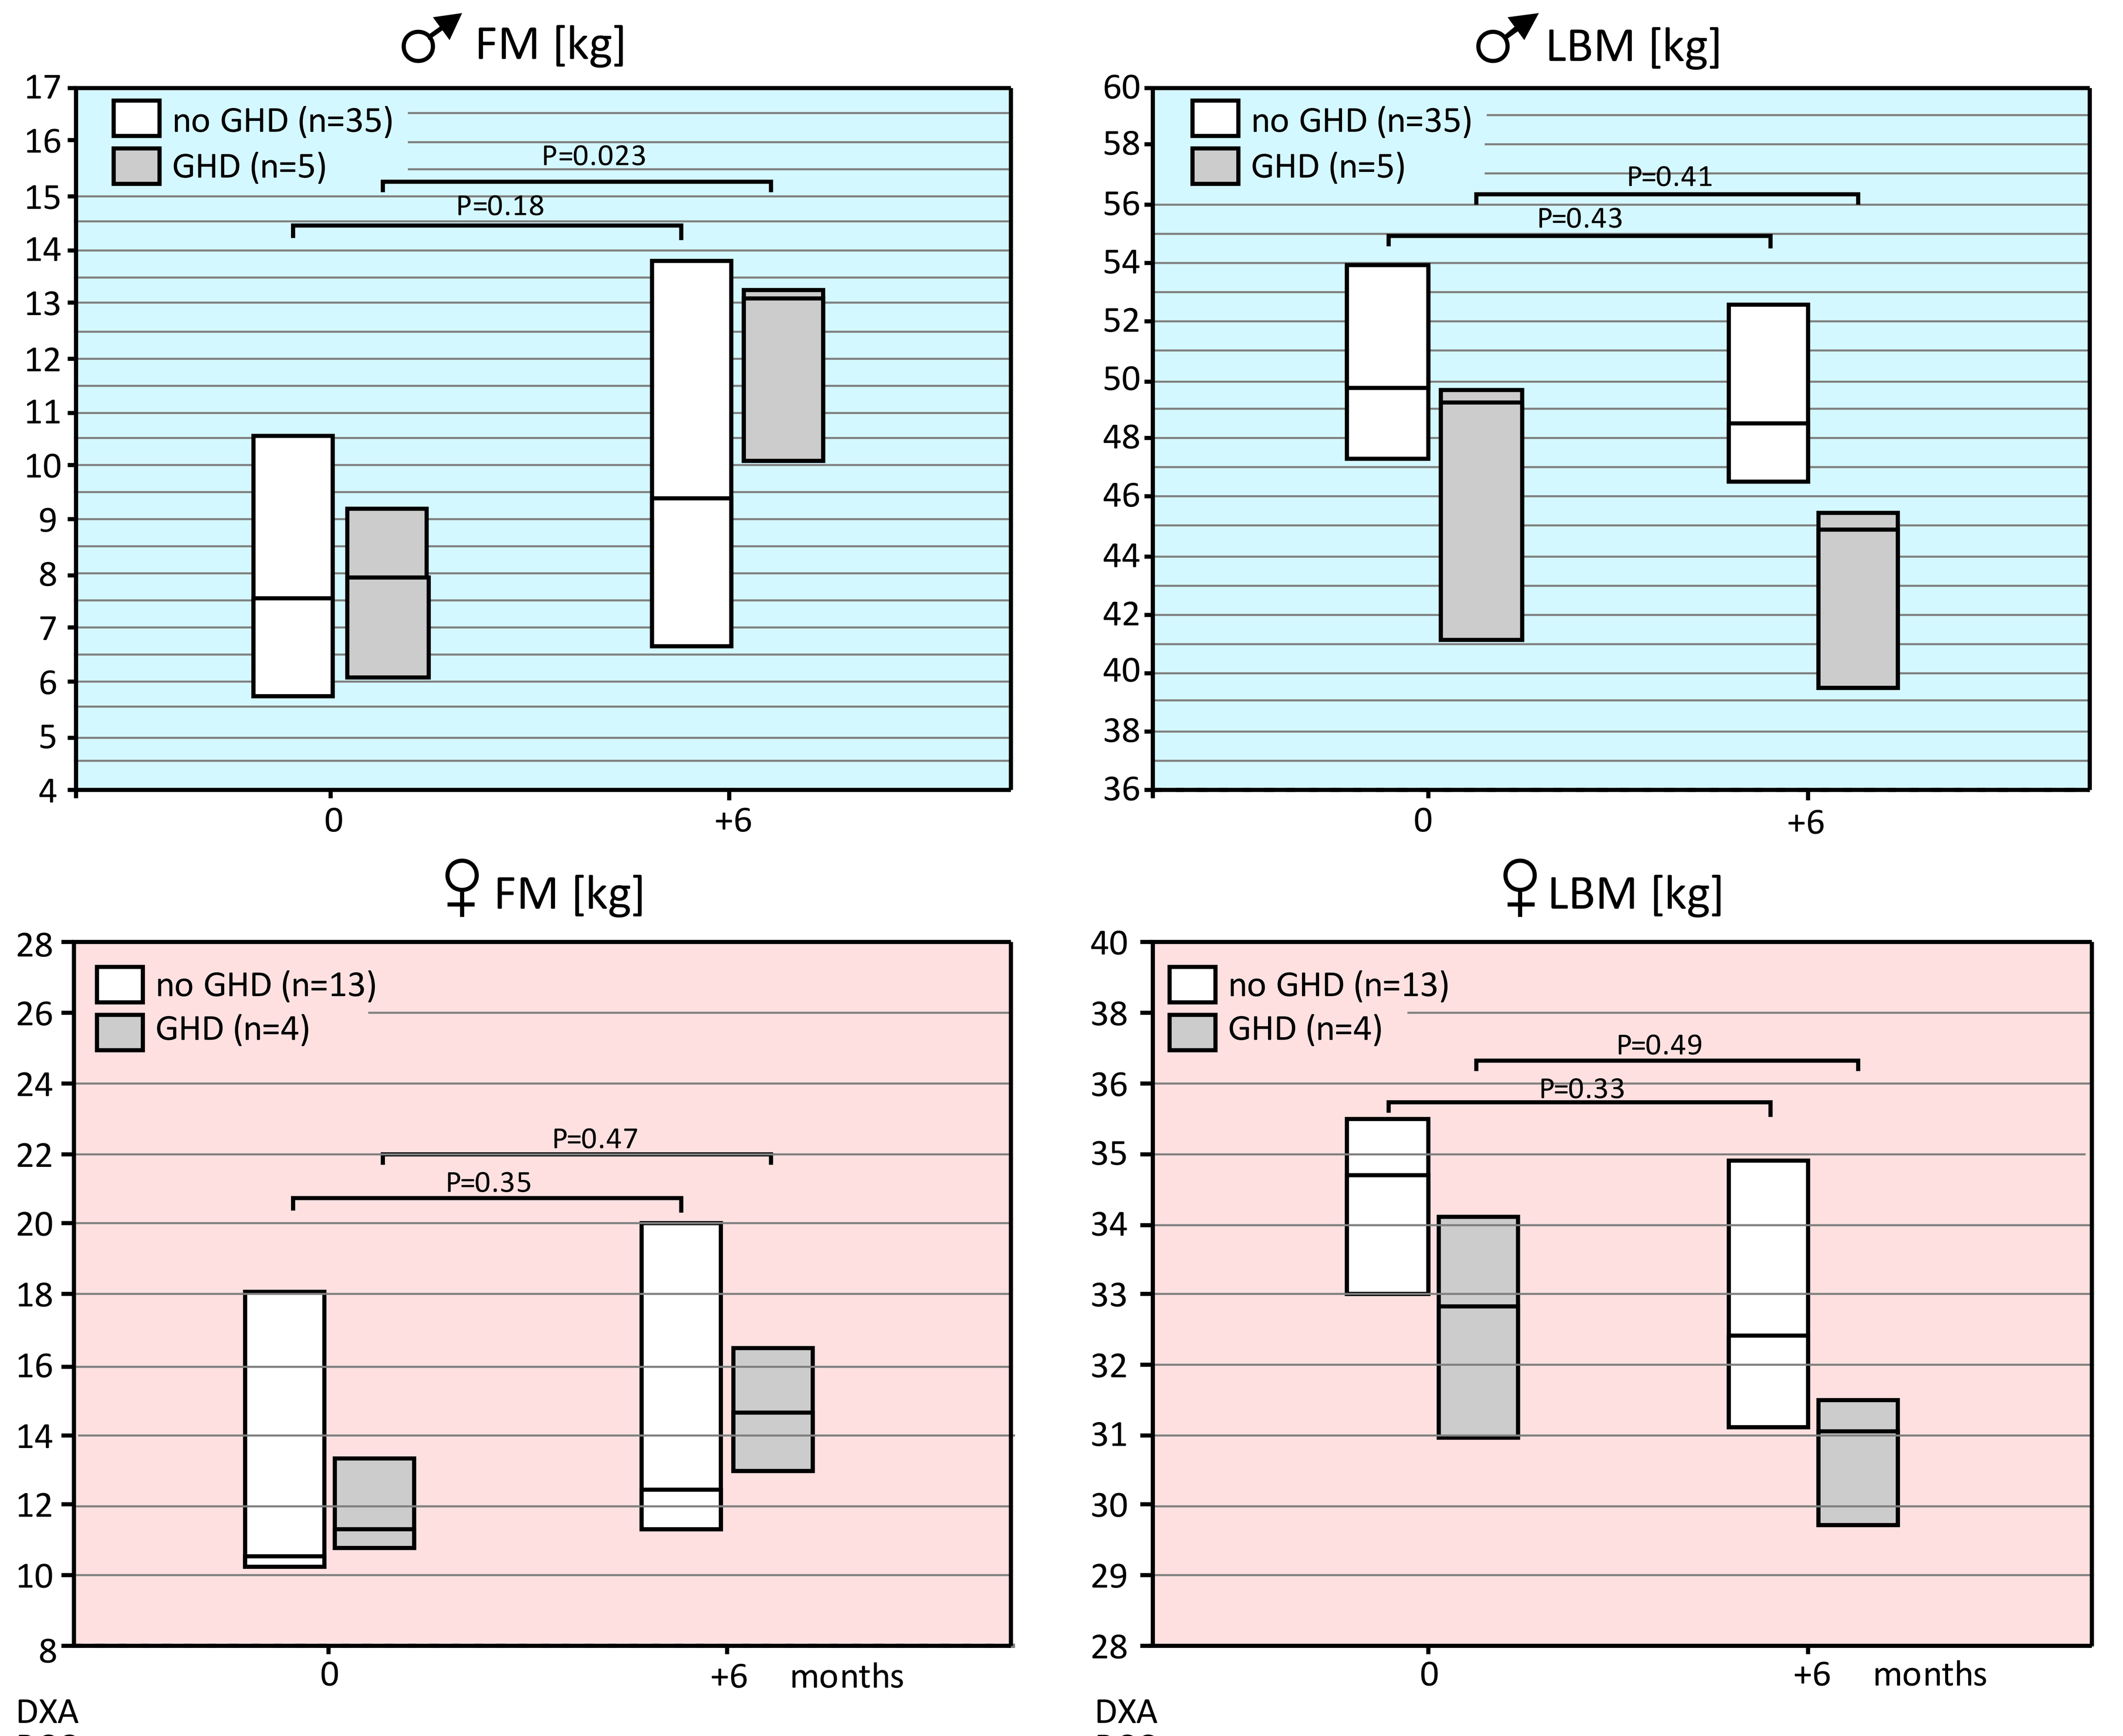
♂ FM [kg]
no GHD (n=35)
GHD (n=5)
P=0.023
P=0.18
♂ LBM [kg]
no GHD (n=35)
GHD (n=5)
P=0.41
P=0.43
♀ FM [kg]
no GHD (n=13)
GHD (n=4)
P=0.47
P=0.35
♀ LBM [kg]
no GHD (n=13)
GHD (n=4)
P=0.49
P=0.33
0
+6
months

These changes were nonsignificant in females with severe GHD of adolescence.

Definition: BCC score = gain in FM + loss in LBM

The BCC score after 6 months correlated highly with the GH peak in males (R=0.54; P<0.001) but not in females (R=0.004; P=0.88).

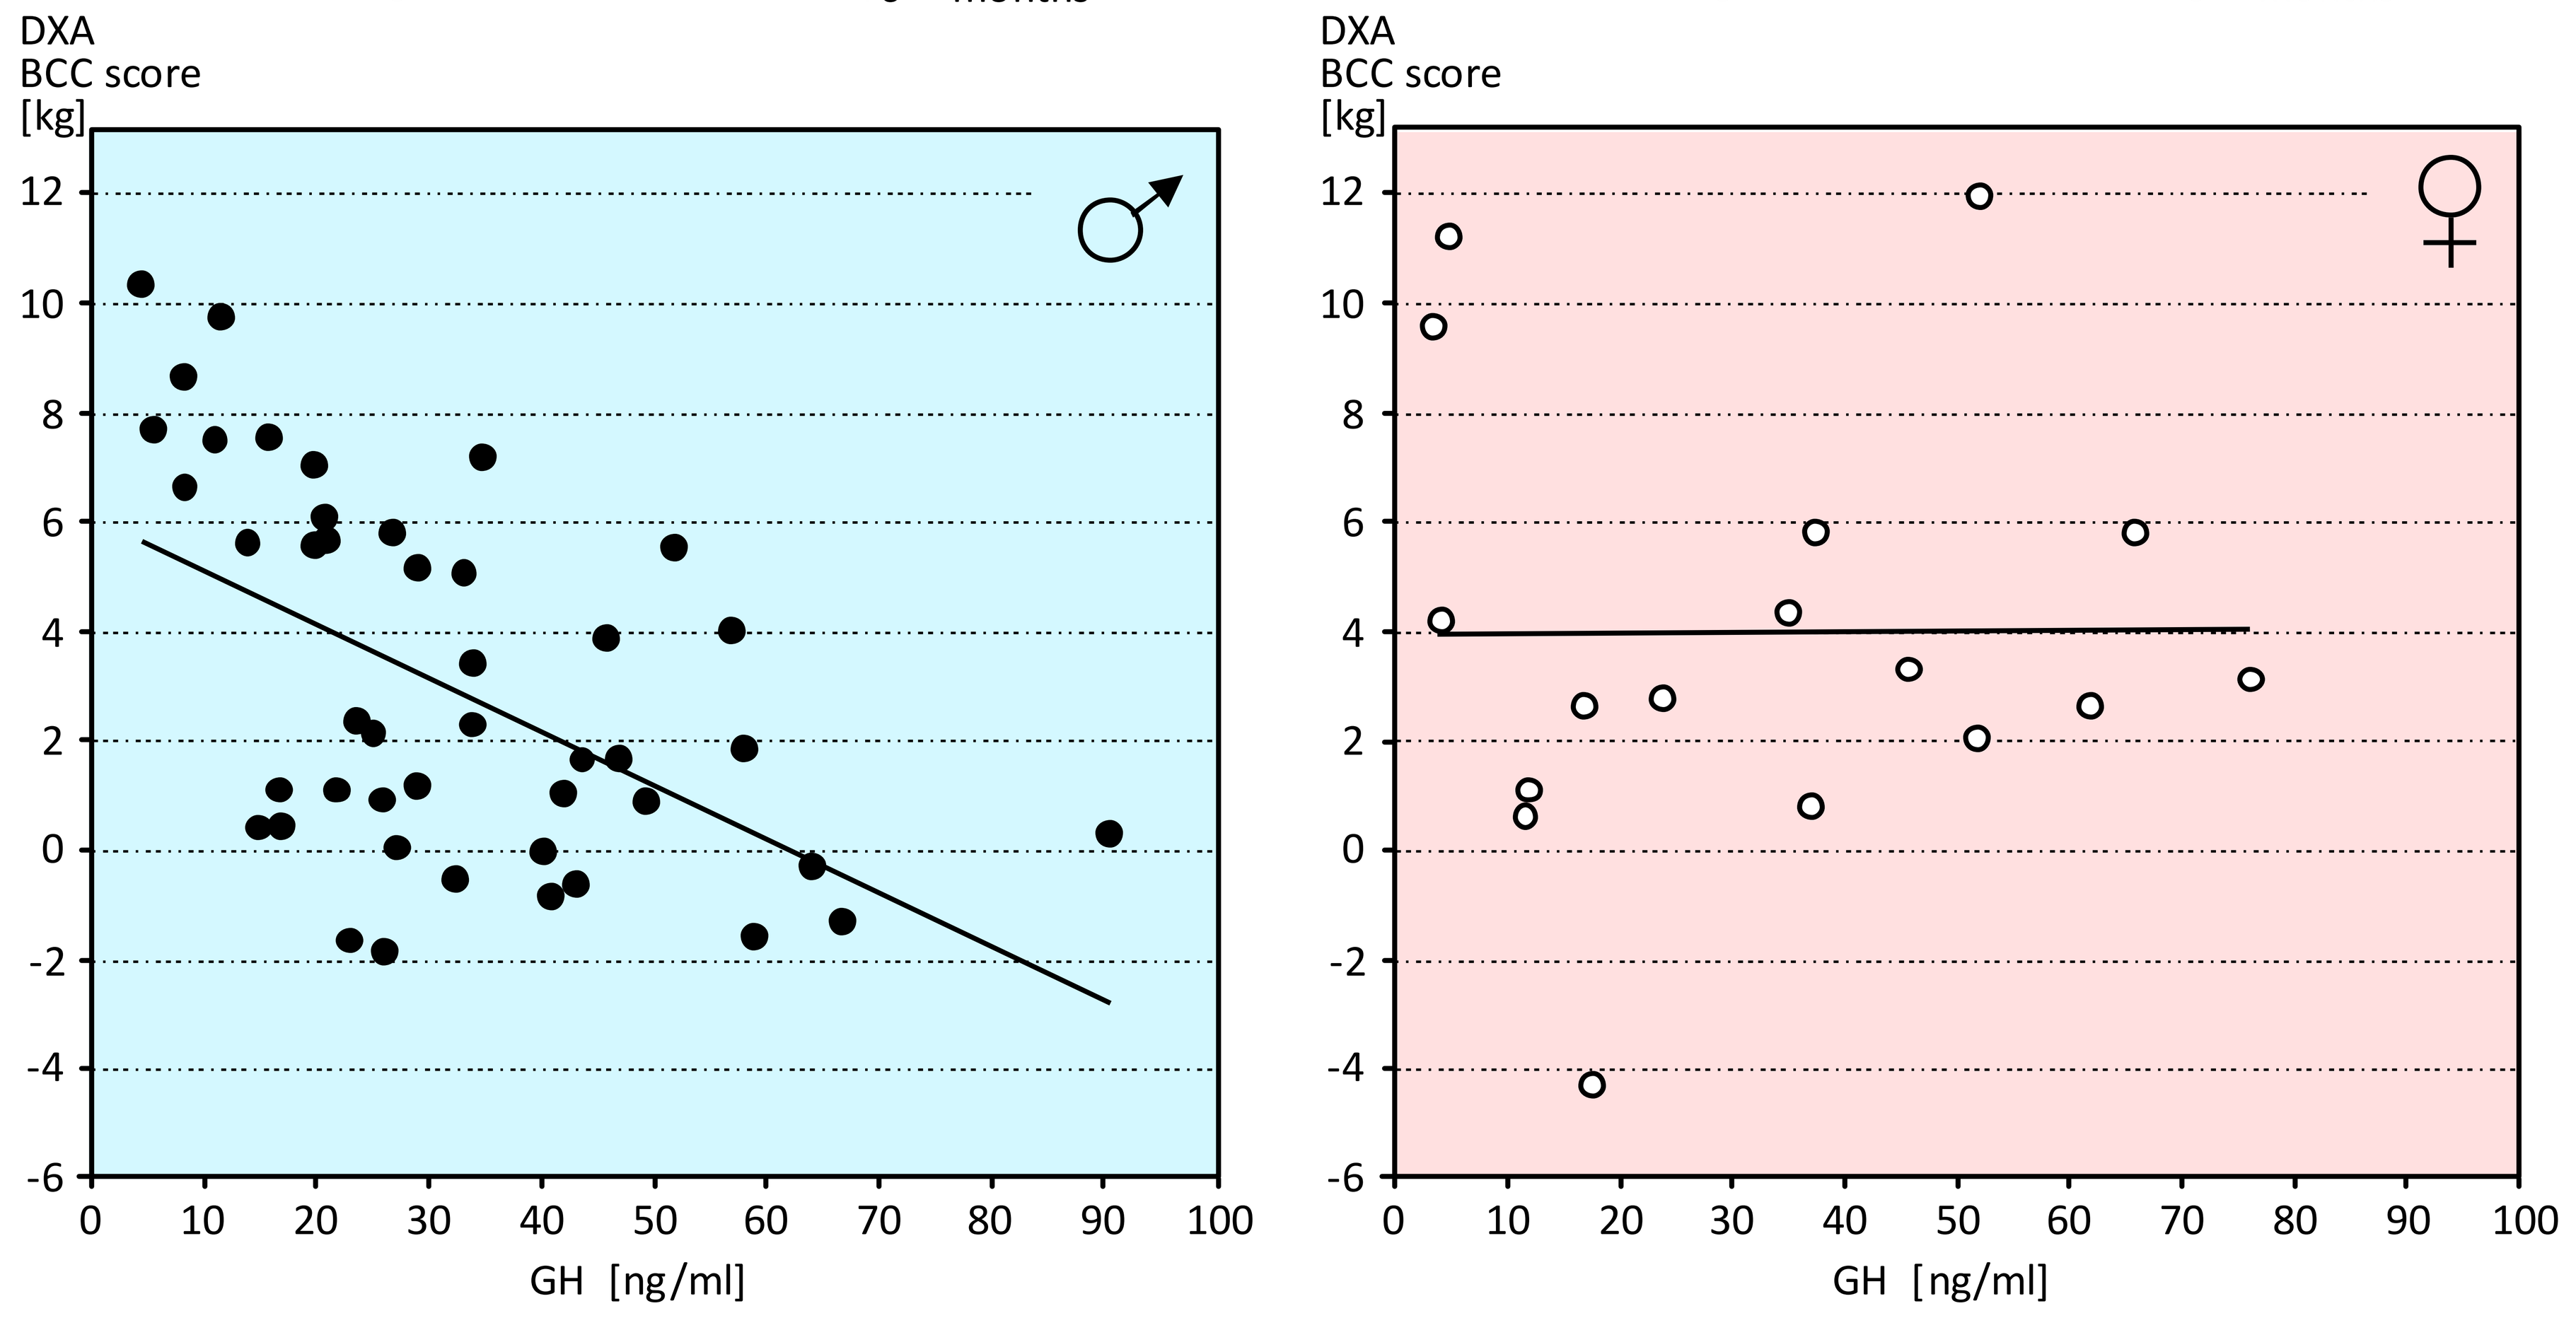
DXA
BCC score
[kg]
♂
♀
GH [ng/ml]

Conclusions

Short-term changes in body composition (BCC score) when off rhGH are good clinical markers of severe GHD in male but not female adolescents.

Acknowledgments

The pilot of this study was supported by an Investigator Initiated Research Program grant from Pfizer Company to G. B. (WI171929).

Contact: gerhard.binder@med.uni-tuebingen.de

57ESPE 327--P2
Pituitary, neuroendocrinology and puberty
Gerhard Binder
Poster presented at:
PosterSessionOnline.com