Abstract No. 137

Positive Impact on Adherence Through Educational Activities of the Argentina Patient Support Program in Children with Low Adherence to Treatment with Recombinant Growth Hormone (easypod connect Electronic Autoinjector Device)

A. Assefi[1], C. Chareca[1], F. Roca[2], L. Celis Ayala[2] , A. Rubstein[1], C. Von Schulz Hausmann[1]
[1]Medical Department, Merck S.A, Argentina; [2]Patient Support Program, Medical Department, Merck S.A, Argentina

INTRODUCTION

- The long-term benefits of recombinant human growth hormone (r-hGH) therapy depend on good adherence to treatment.[1]
- In daily clinical practice, the rate of non-adherence to r-hGH treatment might be as low as 36% to 49%.[1]
- Clinical evidence has found that non-adherent patients show lower physical and psychological benefits to r-hGH treatment, together with an increased risk of cardio–metabolic complications.[1]
- Implementation of educational tools focused on strengthening and empowering the patient on the treatment is a relevant topic in the patient–physician relationship, and the availability of electronic devices that improve the comfort of r-hGH application and objectively record the adherence, represent two effective strategies to optimize the therapeutic benefits.[1]
- The Merck Patient Support Program (PSP) carried out educational actions focused on patients with low adherence and their parents, to raise their awareness of the importance of good adherence (≥80% of the number of doses prescribed) in achieving desired efficacy.
- We report the results of an observational study to evaluate (with the easypod™ connect electronic autoinjector device) the impact of educational advice from nursing staff on automatically recorded adherence to treatment with r-hGH in patients with low adherence.

OBJECTIVE

- The objective of the study was to assess whether educational training focused on compliance to r-hGH administration and medical indications improves adherence to treatment in patients with low adherence (<80%) registered with the easypod connect electronic autoinjector device.

METHODS

- This was a 12-month observational, retrospective cohort study.
- Adherence rate data were collected from the PSP database from April to September 2015. Patients with low adherence (<80%) who were visited by PSP nurse to receive an educational training visit were selected. Their adherence was measured during 6 months, before and after the educational visit.
- The intervention comprised a visit from the PSP staff for educational training and an educational chat of 15–30 mins to raise awareness about the importance of following the medical instructions and complying with the regime of applications to achieve height objective and minimize possible risks.
- Patient demographic data were tabulated and graphed (N=80).
- Statistical analyses were carried out with the STATA 15.0 software.
- Continuous variables were presented as mean and median with their respective 95% CI; categorical variables were presented as proportion with 95% CI.
- Comparison of adherence (continuous non-parametric pared data) before and after the educational intervention were evaluated with the Wilcoxon signed-rank test.

RESULTS

- Data from 80 patients with low adherence (<80%) were analyzed (**Figure 1**).
- Gender distribution was 65% male, 35% female, age ranged between 2–18 years (mean: 11.77), diagnoses were growth hormone deficiency (71.25%), small for gestational age (20%), Turner syndrome (7.50%) and chronic renal disease (1.25%) (**Figure 2**).
- Duration of treatment was 0.4–11.13 years (mean 4.34).
- Currently there are studies that demonstrate a negative correlation between the years of treatment, the age of patients (especially teenagers), type of device, etc. and adherence;[2] however, there are few data on low-cost strategies that could improve patient adherence.

Figure 1. Patient Flowchart

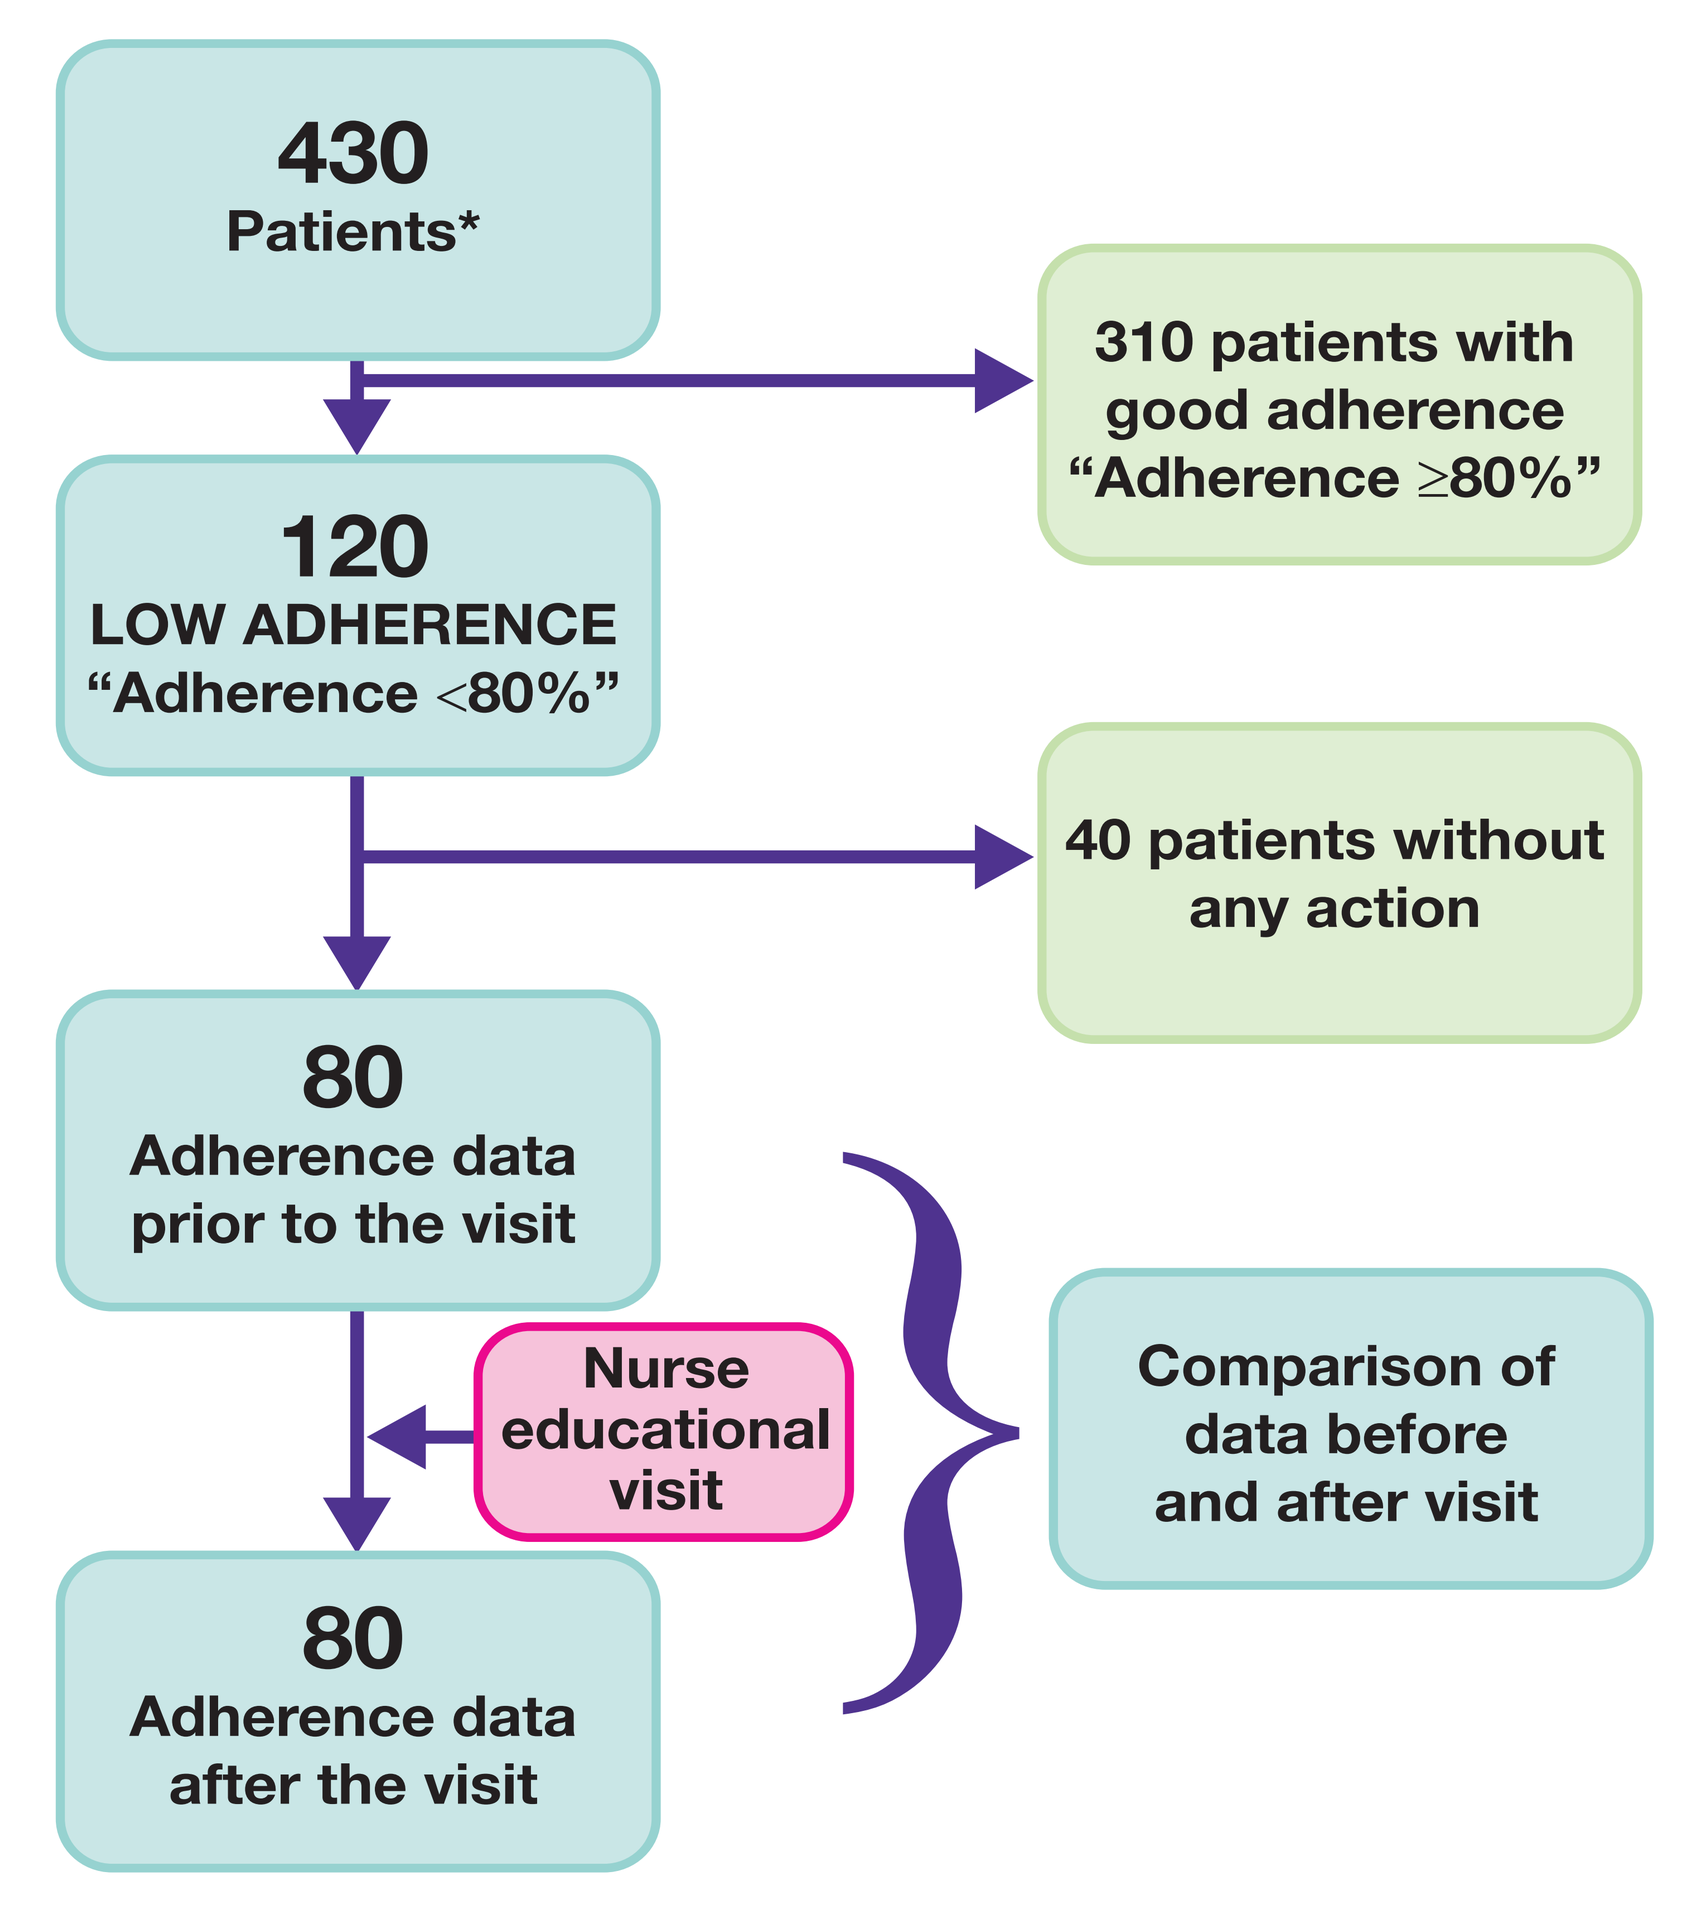
430
Patients*
310 patients with good adherence "Adherence ≥80%"
120
LOW ADHERENCE
"Adherence <80%"
40 patients without any action
80
Adherence data prior to the visit
Nurse educational visit
80
Adherence data after the visit
Comparison of data before and after visit

Figure 2: Distribution of Non-adherent Patients by Diagnosis

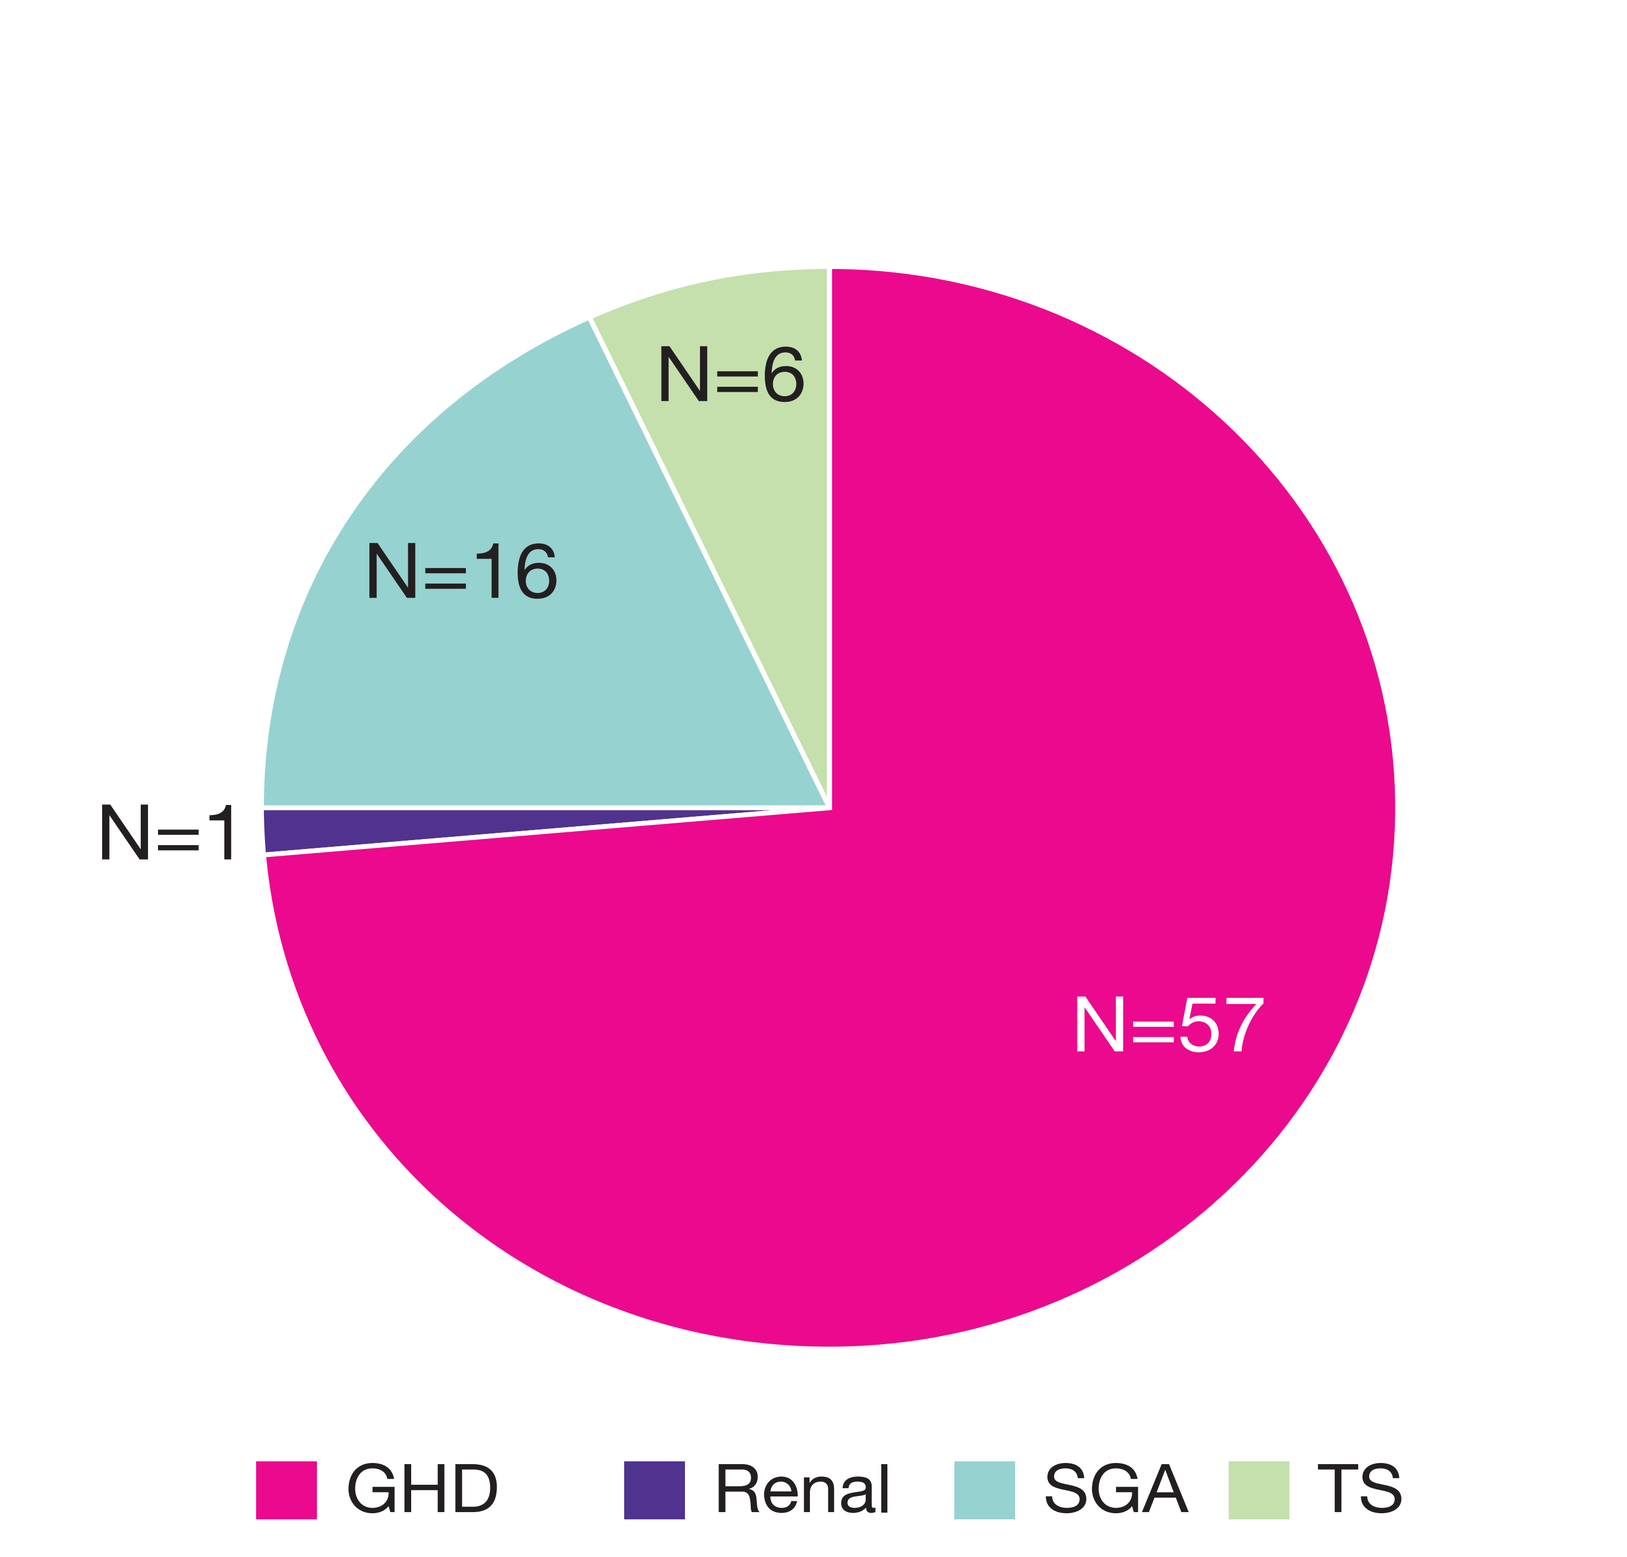
N=6
N=16
N=1
N=57
GHD
Renal
SGA
TS

- In our study we have designed a simple and low-cost strategy that gives educational training to poorly adherent patients on the importance of complying with the recommendations and indications of their attending doctor to achieve the goals of treatment.
- This educational activity was done through the PSP nurses who regularly visit the patients in the course of treatment with r-hGH using the easypod electronic autoinjector device.
- The purpose of our study was to assess the effect of the intervention on adherence by comparing the adherence data before and after the intervention (**Figure 3**).
- Because the variable adherence rates were continuous, paired and with non-parametric distribution, we performed Wilcoxon signed-rank test that evaluates the sign and the ranking of the differences and analyzes their magnitude. The test showed significant differences in the median adherence after the intervention (z=4.432, *P*=0.0000).
- At baseline, median adherence was 67%; after the intervention it increased to 76%, a significant median improvement of 9% (*P*=0.0000 Wilcoxon signed-rank test). Therefore 34% (29/80) of the patients with low adherence increased their adherence to values considered as good adherence (≥80%). Both changes were clinically relevant. (**Figure 4**).
- After the intervention, an increase in adherence to applications was observed (**Figure 5**).
- The variables analyzed were age, medical coverage, city, diagnosis, gender and treatment time. No significant differences or correlations were observed in the regression model for this population

Figure 3: Comparison of Adherence Before and After the Educational Intervention

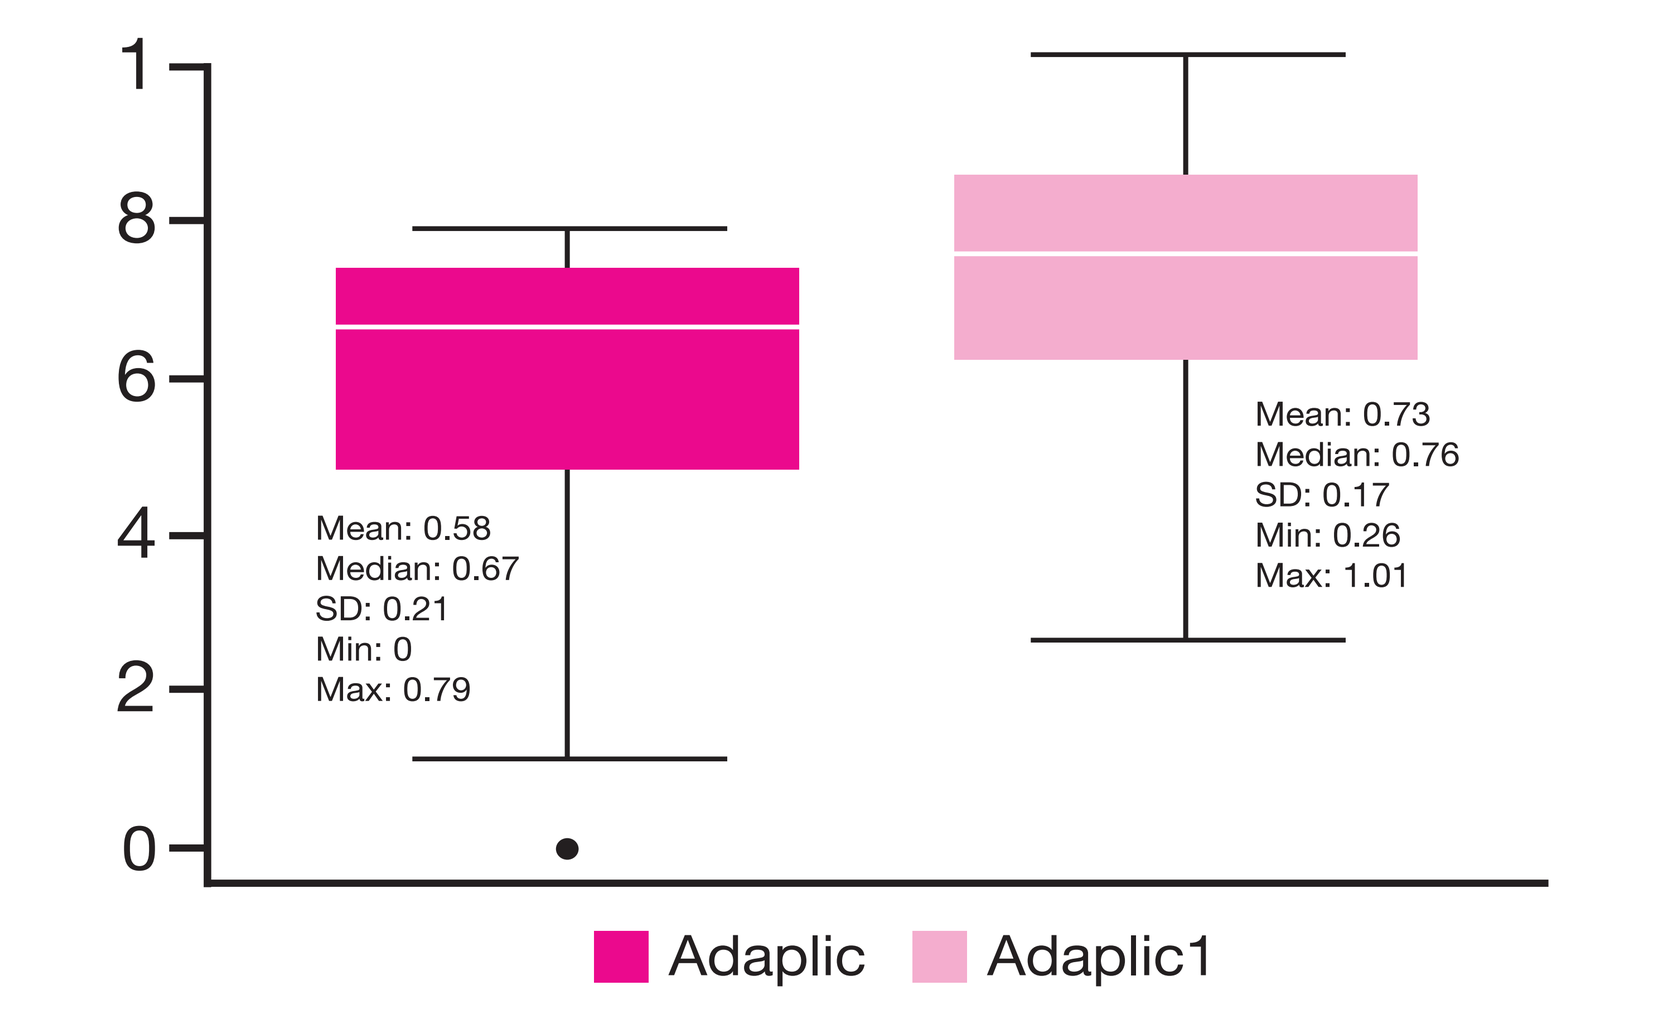
1
8
6
4
2
0
Mean: 0.58
Median: 0.67
SD: 0.21
Min: 0
Max: 0.79
Mean: 0.73
Median: 0.76
SD: 0.17
Min: 0.26
Max: 1.01
Adaplic
Adaplic1

Figure 4: Percentage of Patients who Achieved Good Adherence

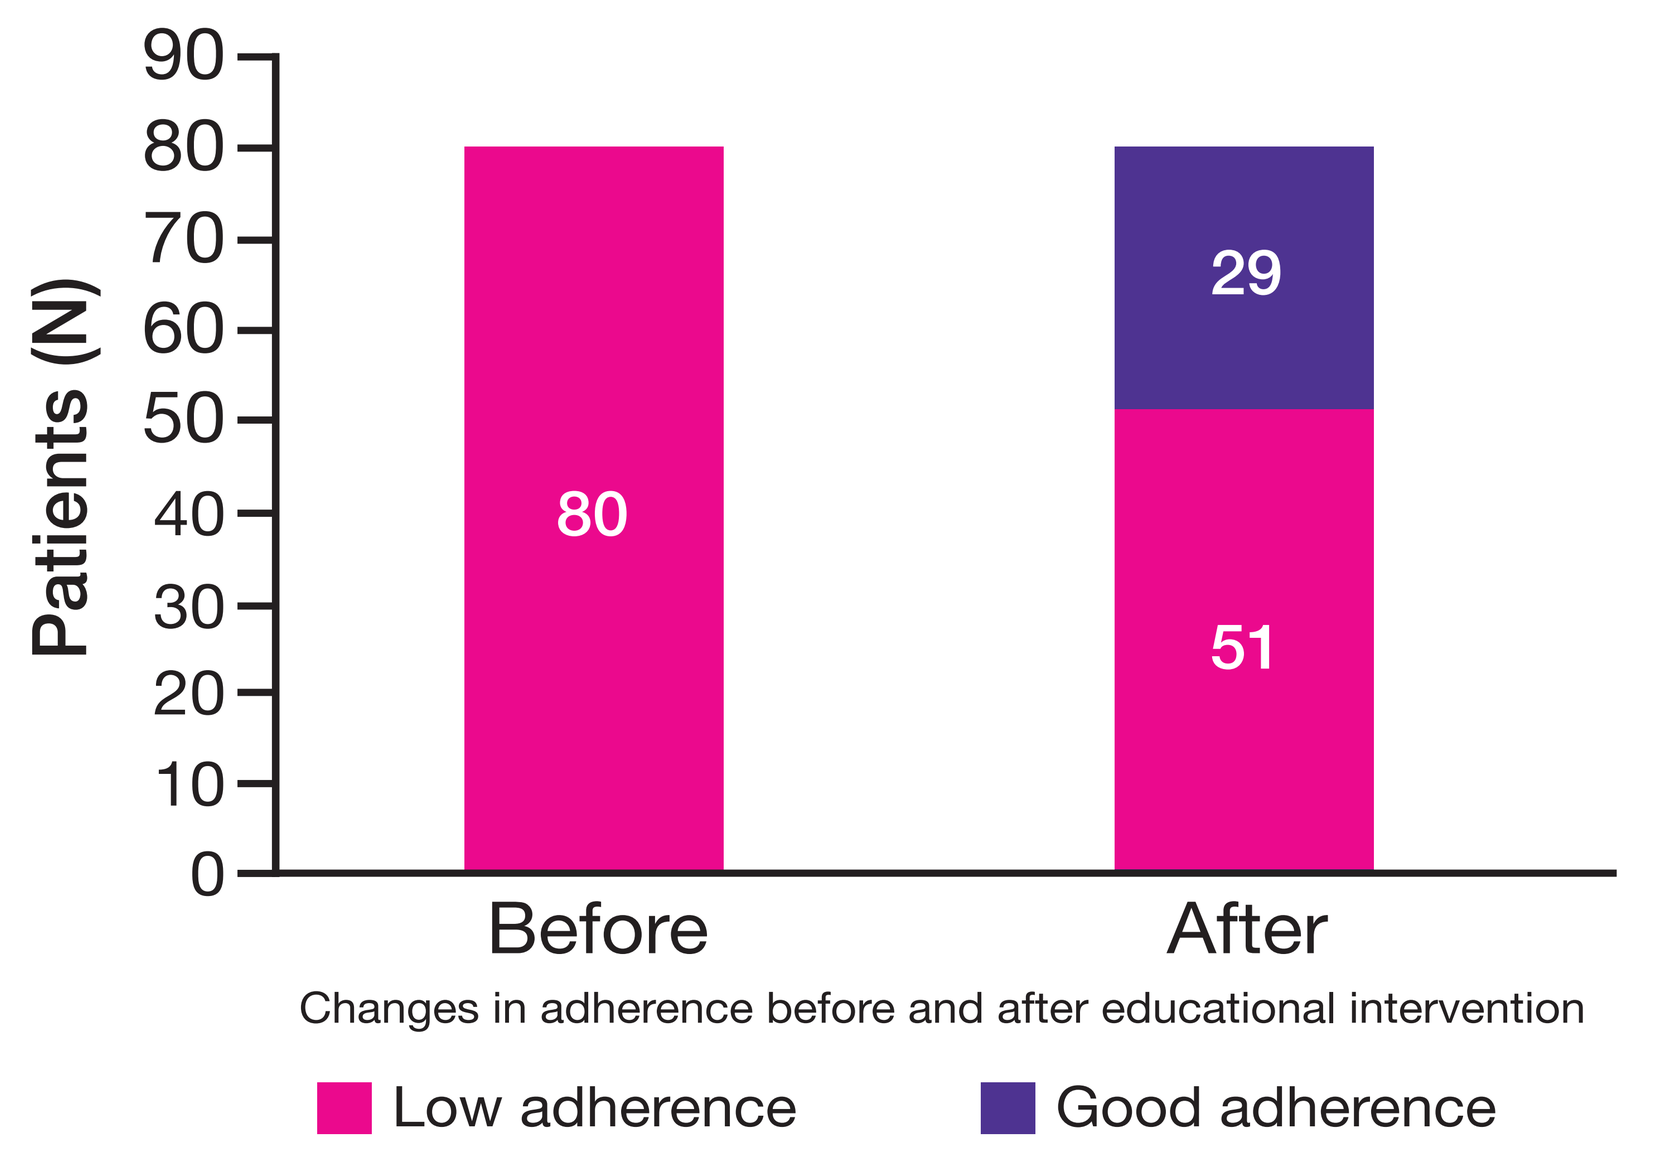
Patients (N)
90
80
70
60
50
40
30
20
10
0
80
29
51
Before
After
Changes in adherence before and after educational intervention
Low adherence
Good adherence

Figure 5: Histogram of Adherence Before and After the Educational Intervention by PSP Nurse

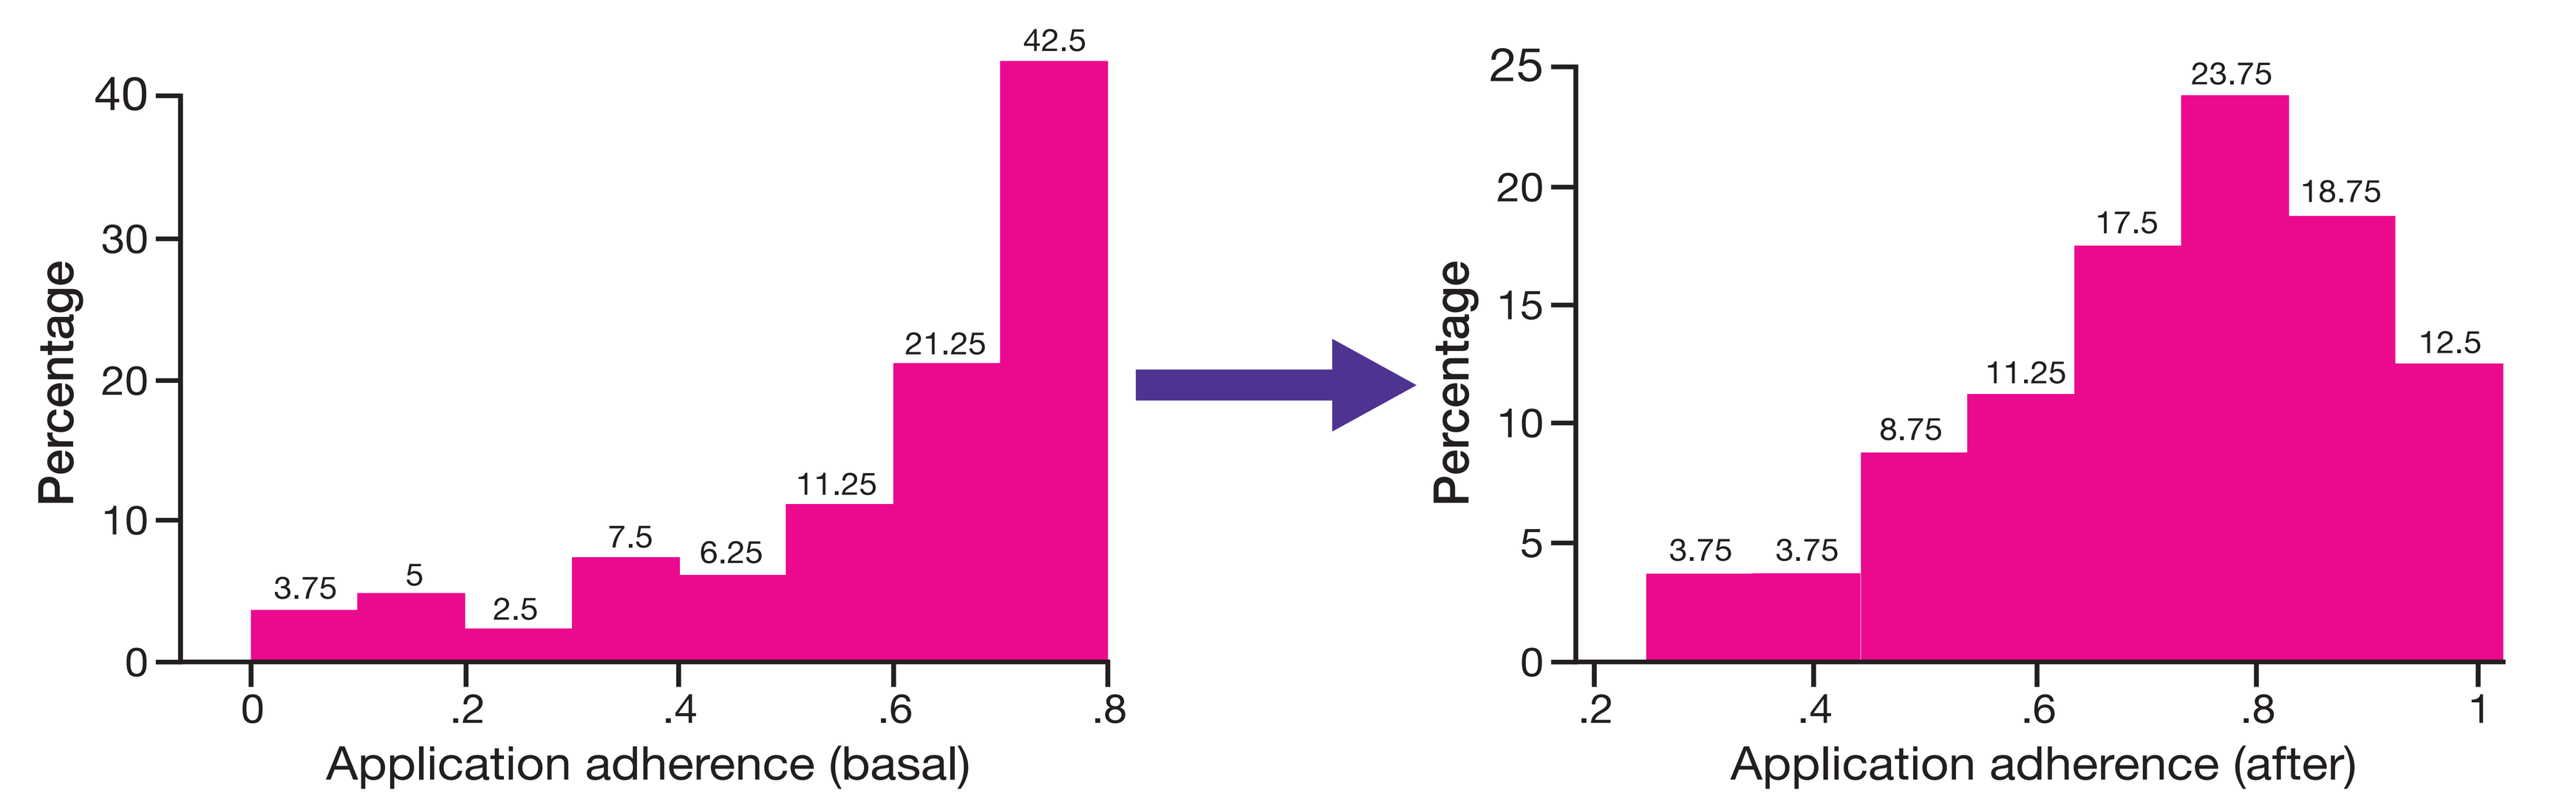
Percentage
40
30
20
10
0
3.75
5
2.5
7.5
6.25
11.25
21.25
42.5
0
.2
.4
.6
.8
Application adherence (basal)
Percentage
25
20
15
10
5
0
3.75
3.75
8.75
11.25
17.5
23.75
18.75
12.5
.2
.4
.6
.8
1
Application adherence (after)

CONCLUSIONS

- **Low adherence to r-hGH therapy is multifactorial and there are few effective methods to improve it**
- **We conclude that educational intervention is a simple and low-cost method to improve adherence rate in patients with low adherence**
- **We recommend this type of action in this sub-population to reduce the gap between the indication/recommendation of the specialist and the patients' behavior**

REFERENCES

1. Haverkamp F et al. *Clin Ther*. 2008;30:307-16.
2. Koledova E et al. *Endocr Connect*. 2018 Aug;7(8):914–923.

ACKNOWLEDGMENTS

We would like to thank the entire PSP team that carried out all the educational activities for the patients, and Jesica Benitez who collaborated in the statistical analysis of the data

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

Presented at ESPE 2019, 19-21 September, 2019; Vienna, Austria

58ESPE P1-082
GH and IGFs
Cinthia Chareca
Poster presented at:
PosterSessionOnline.com