GH and IGFs
P1-80

Clinical characteristics, puberty pattern and adult or near-adult-height data in a group of patients with growth failure due to severe primary IGF-1 deficiency (GROWPATI study)

Athanasia Stoupa[1,2], Christine Loraud[1], Isabelle Flechtner[1], Magali Viaud[1,3], Graziella Pinto[1], Dinane Samara-Boustani[1,3], Caroline Thalassinos[1], Frédéric Brioude[4], Irène Netchine[4], Serge Amselem[5], Marie Legendre[5], Michel Polak[1,2,3,6]

1 Pediatric Endocrinology, Diabetology and Gynecology Department, Necker Children's University Hospital, Paris, France, 2 Institut Imagine Affiliate, INSERM U1163 and U1016, Institut Cochin, Paris, France, 3 Centre de référence des maladies endocriniennes rares de la croissance et de développement (CMERCD), 4 Sorbonne Université, INSERM UMR_S938 Centre de Recherche Saint-Antoine, Trousseau Hospital Paris, France, 5 Sorbonne Université, INSERM UMR_S933, Trousseau Hospital, Genetics Department, Paris, France, 6 Paris Descartes University, Paris, France

Background

Severe primary insulin-growth factor-1 (IGF1) deficiency (SPIGF1D) is a rare cause of growth retardation. Diagnostic criteria include age- and sex-dependent low basal IGF1 levels (<2.5th percentile), height ≤-3SDS, absence of growth hormone (GH) deficiency and of any secondary causes of growth failure.

Aims

Description of pubertal onset and growth spurt, data on adult or near-adult-height in a subgroup of patients diagnosed with growth retardation due to SPIGF1D.

Patients and Methods

- **Thirty patients (M/F:17/13) with SPIGF1D (*historical study cohort*) out of 2546 patients referred for growth failure to Paediatric Endocrinology Department of Necker Children's University Hospital, in Paris between 2004-2009 (*Teissier et al, EJE, 2014*).**
- **Nineteen patients with SPIFGD (*new cohort*) among patients referred with growth retardation between 2016-2019.**
- **Puberty and adult height data, if available, were studied for a subgroup of patients from both cohorts (n=19, 11M/8F).**

Results

Characteristics of SPIGF1D patients

49 patients with SPIGF1D

- SGA, n=29
- Constitutional bone disease, n=5
- Noonan Sd, n=1
- Silver-Russell Sd, n=2
- Laron Sd, n=1
- Heterozygous *GHR* mutations, n=2

Puberty pattern

- Pubertal onset: mean age for Tanner 2: 12.5 years (M, n=11) 12 years (F, n=8). Mean age of menarche was 13.6 years with regular menses.
- Two boys had advanced evolutive central puberty, treated by GnRH agonist.

Adult Height (AH)/Near-adult height data

- Data were available for 5 boys: mean (SD)/ -2.2 SDS(0.3) and 4 girls: -2 SDS(1), except a female patient with Laron syndrome (AH:128.5cm).
- AH are shown by dots in the growth charts for boys and girls.
- Predicted adult heights for boys and girls with available AH were -1.5 SDS(0.4) and -1.8 SDS(0.6), respectively. These patients have been treated by growth hormone or, for Laron syndrome, by Increlex® (recombinant human IGF1).

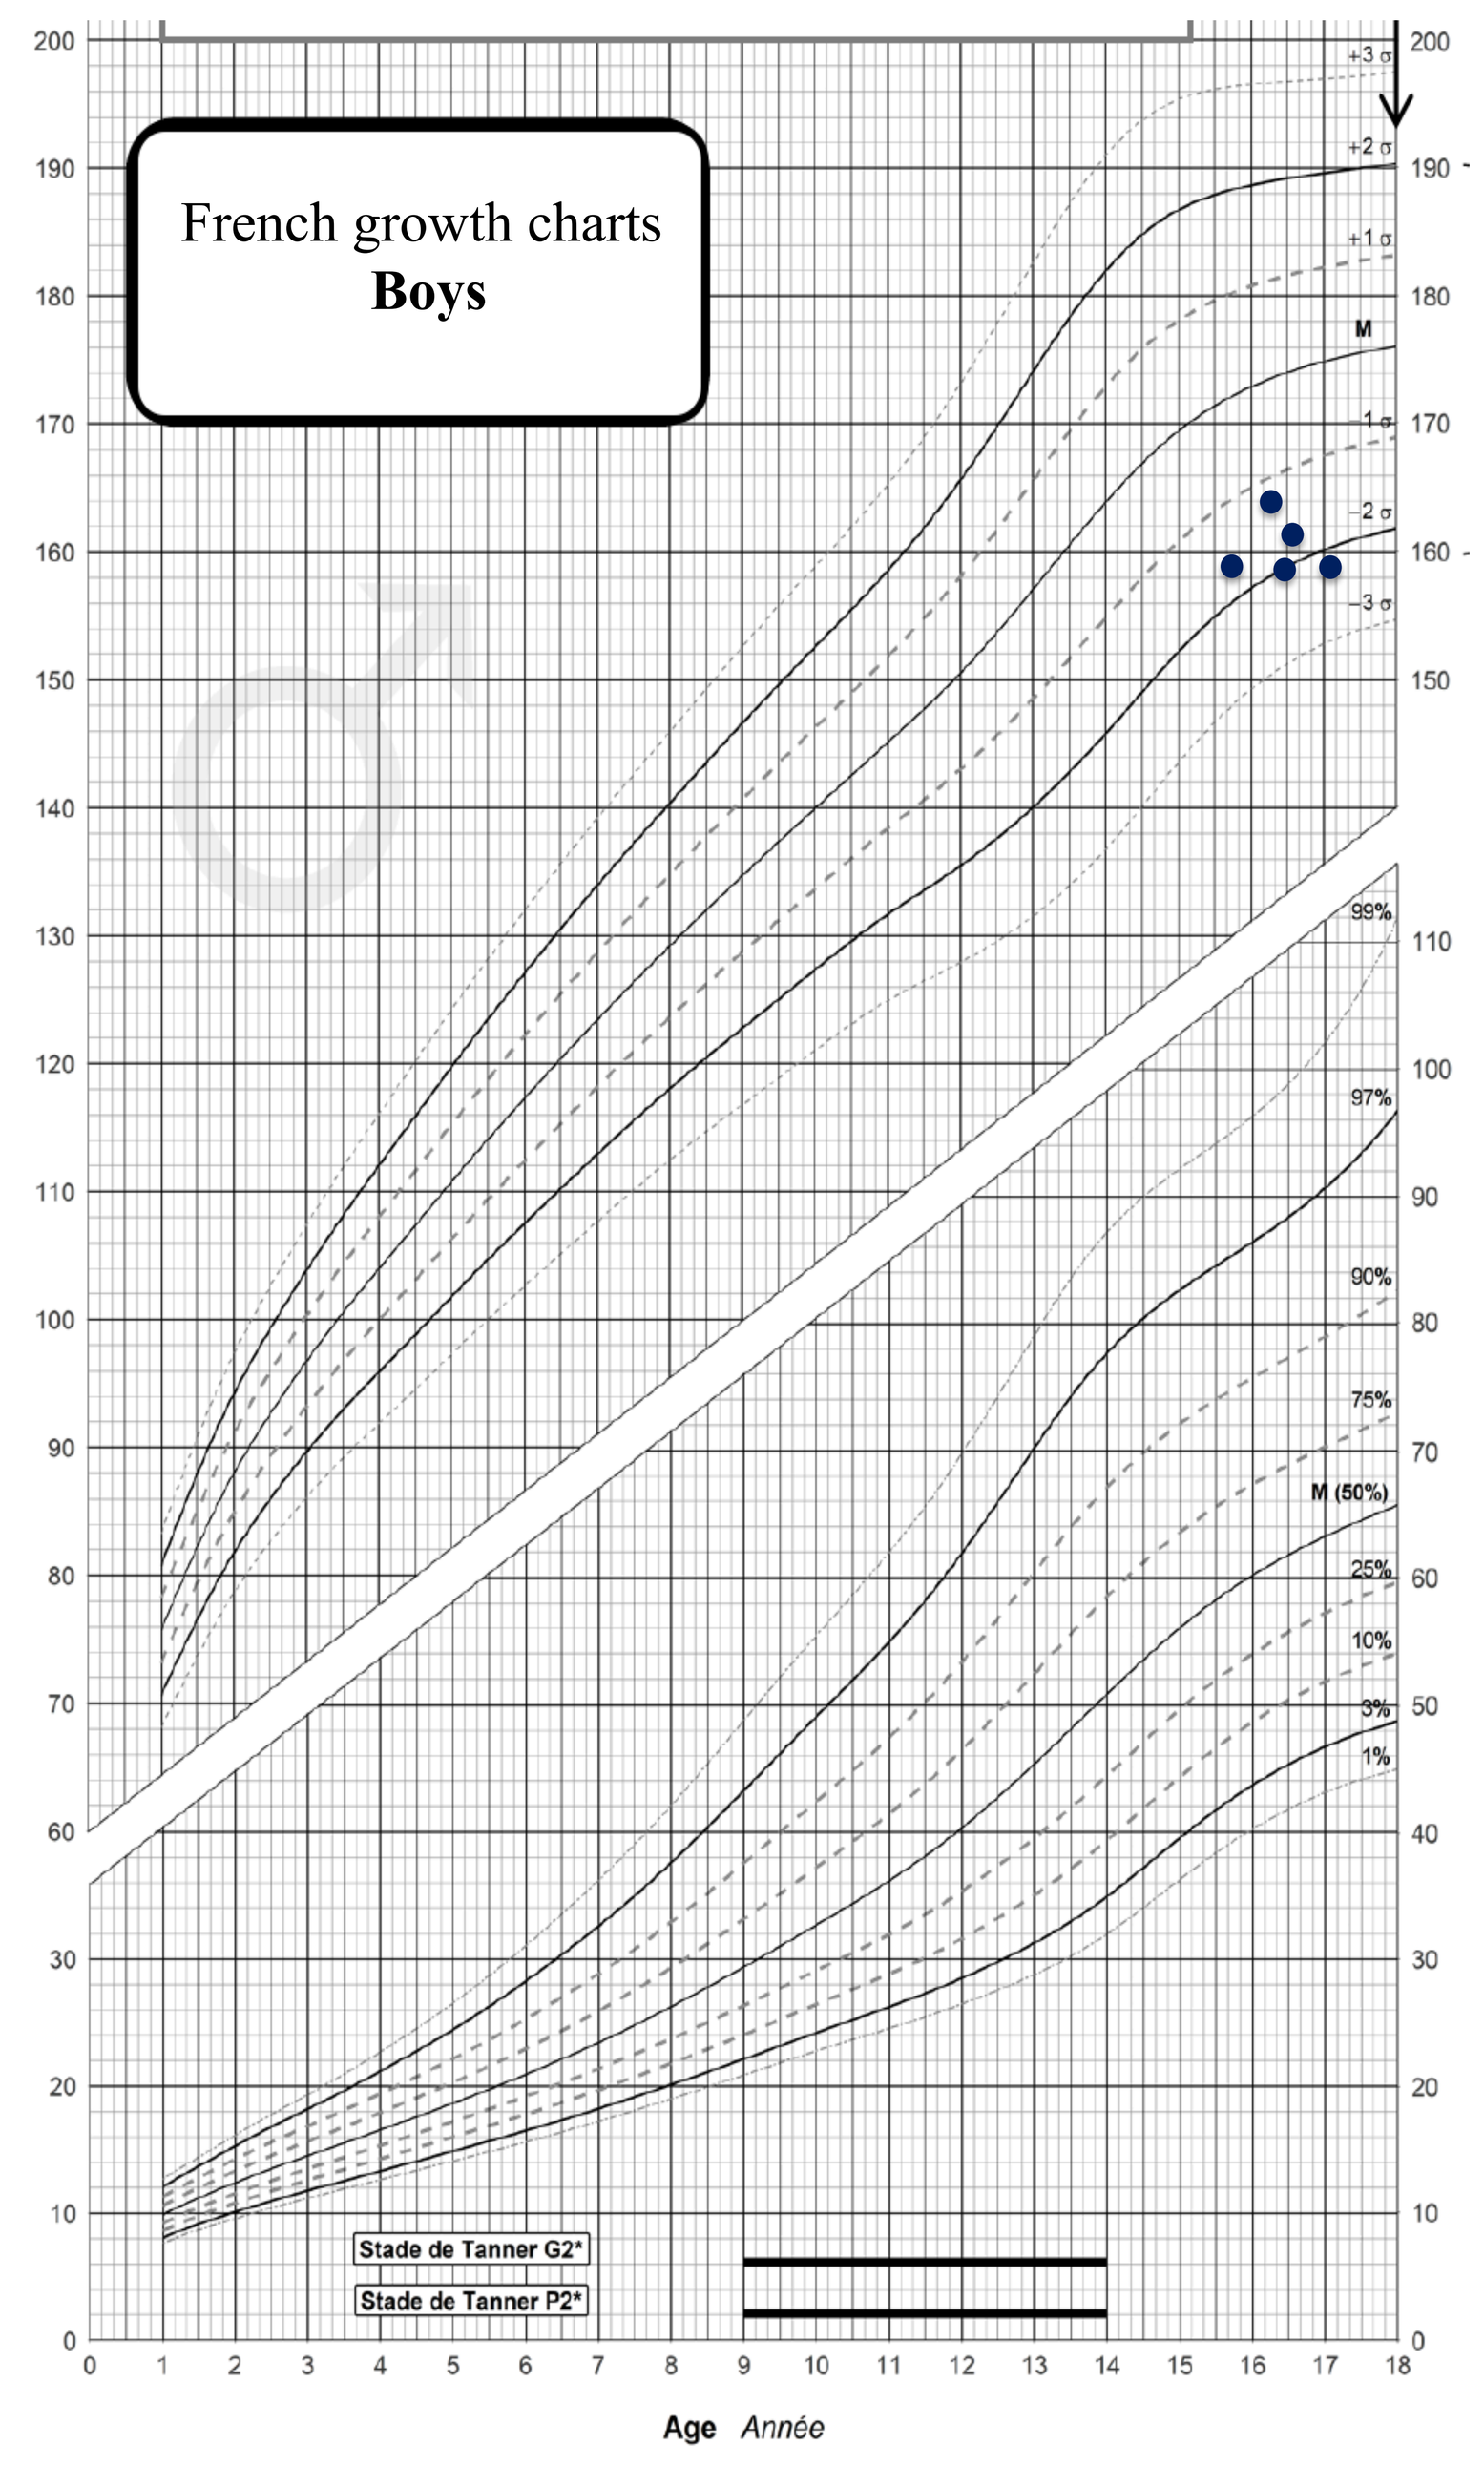
French growth charts
Boys
Stade de Tanner G2*
Stade de Tanner P2*
Age Année

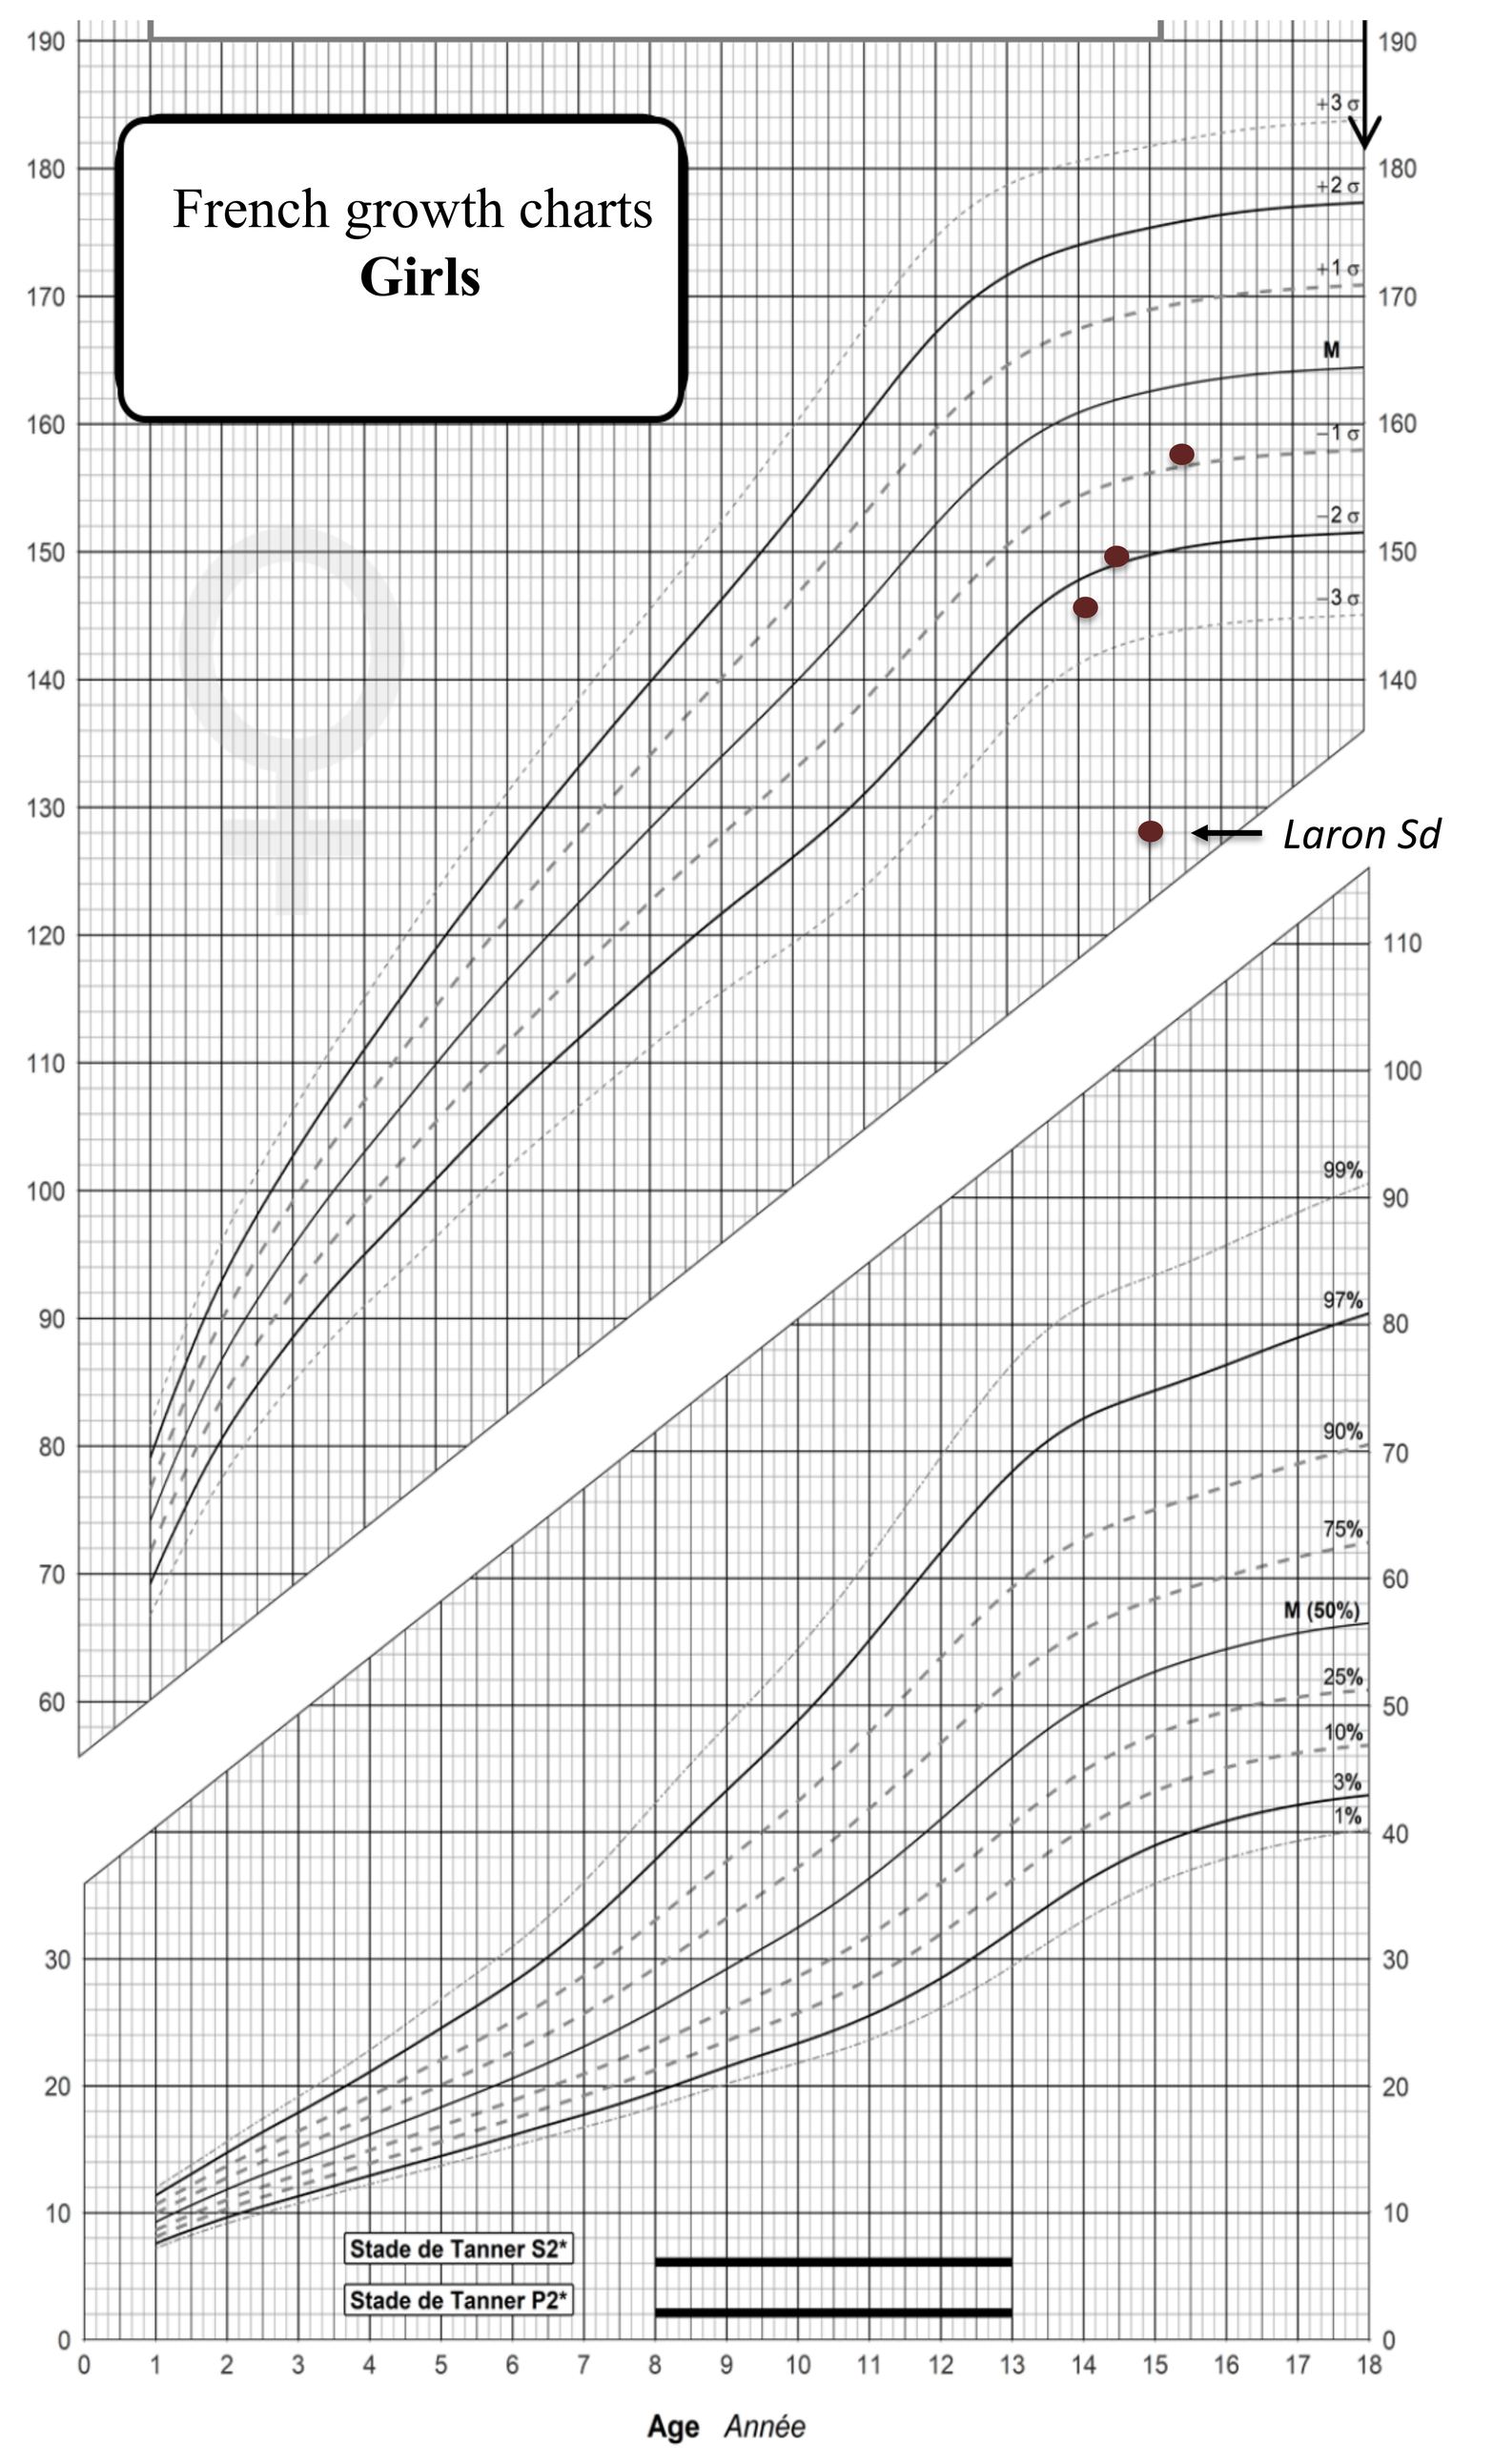
French growth charts
Girls
Laron Sd
Stade de Tanner S2*
Stade de Tanner P2*
Age Année

Conclusions

- **Final heights in our patients were below predicted adult heights, and height velocity during puberty varied.**
- **Long-term follow-up and genetic investigations are necessary for providing more insights in the SPIF1D.**

This work was in part supported by IPSEN laboratory

58ESPE P1-080
GH and IGFs
Athanasia Stoupa
Poster presented at: ESPE 2019 Vienna
PosterSessionOnline.com