Frequency of MC4R Pathway Variants in a Large US Cohort of Pediatric and Adult Patients With Severe Obesity

Eric Bend,[1] Tiago Antao,[2] Ilia Ichetovkin,[2] Rick Norton,[2] Alastair S. Garfield,[2,*] Ida Hatoum Moeller[2]

[1]PreventionGenetics, Marshfield, WI, USA; [2]Rhythm Pharmaceuticals, Inc., Boston, MA, USA

P2-179

Presenting Author:
Ida Hatoum Moeller
imoeller@rhythmtx.com

Summary

- In our large US-based cohort of individuals with severe, early-onset obesity, 25.4% of individuals carry a potentially clinically relevant variant in one or more of 11 MC4R pathway-related genes.
- No differences in variant frequency were observed across age groups, indicating that genetic testing for obesity-related genes is appropriate in both pediatric and adult patients with severe obesity.
- The studied population is highly enriched for patients with severe, early-onset obesity. The estimates of variant frequency found here may not be indicative of the broader obese population.

Introduction

- The melanocortin-4 receptor (MC4R) pathway is critical for the regulation of hunger and energy balance (Figure 1).[1-4]
- Variants in a number of genes within this pathway have well-established associations with severe obesity.[1]
- However, the overall frequency of rare variants in these genes has not been assessed systematically in a clinically relevant population, and it is unknown whether variant frequency differs depending on the age of ascertainment.
- Genetic testing can improve diagnosis of rare genetic diseases of obesity and identify patients who may benefit from targeted therapeutic intervention and may therefore contribute to our understanding of the etiology of these patients' phenotypes.

Figure 1. MC4R pathway diagram.[1-6]

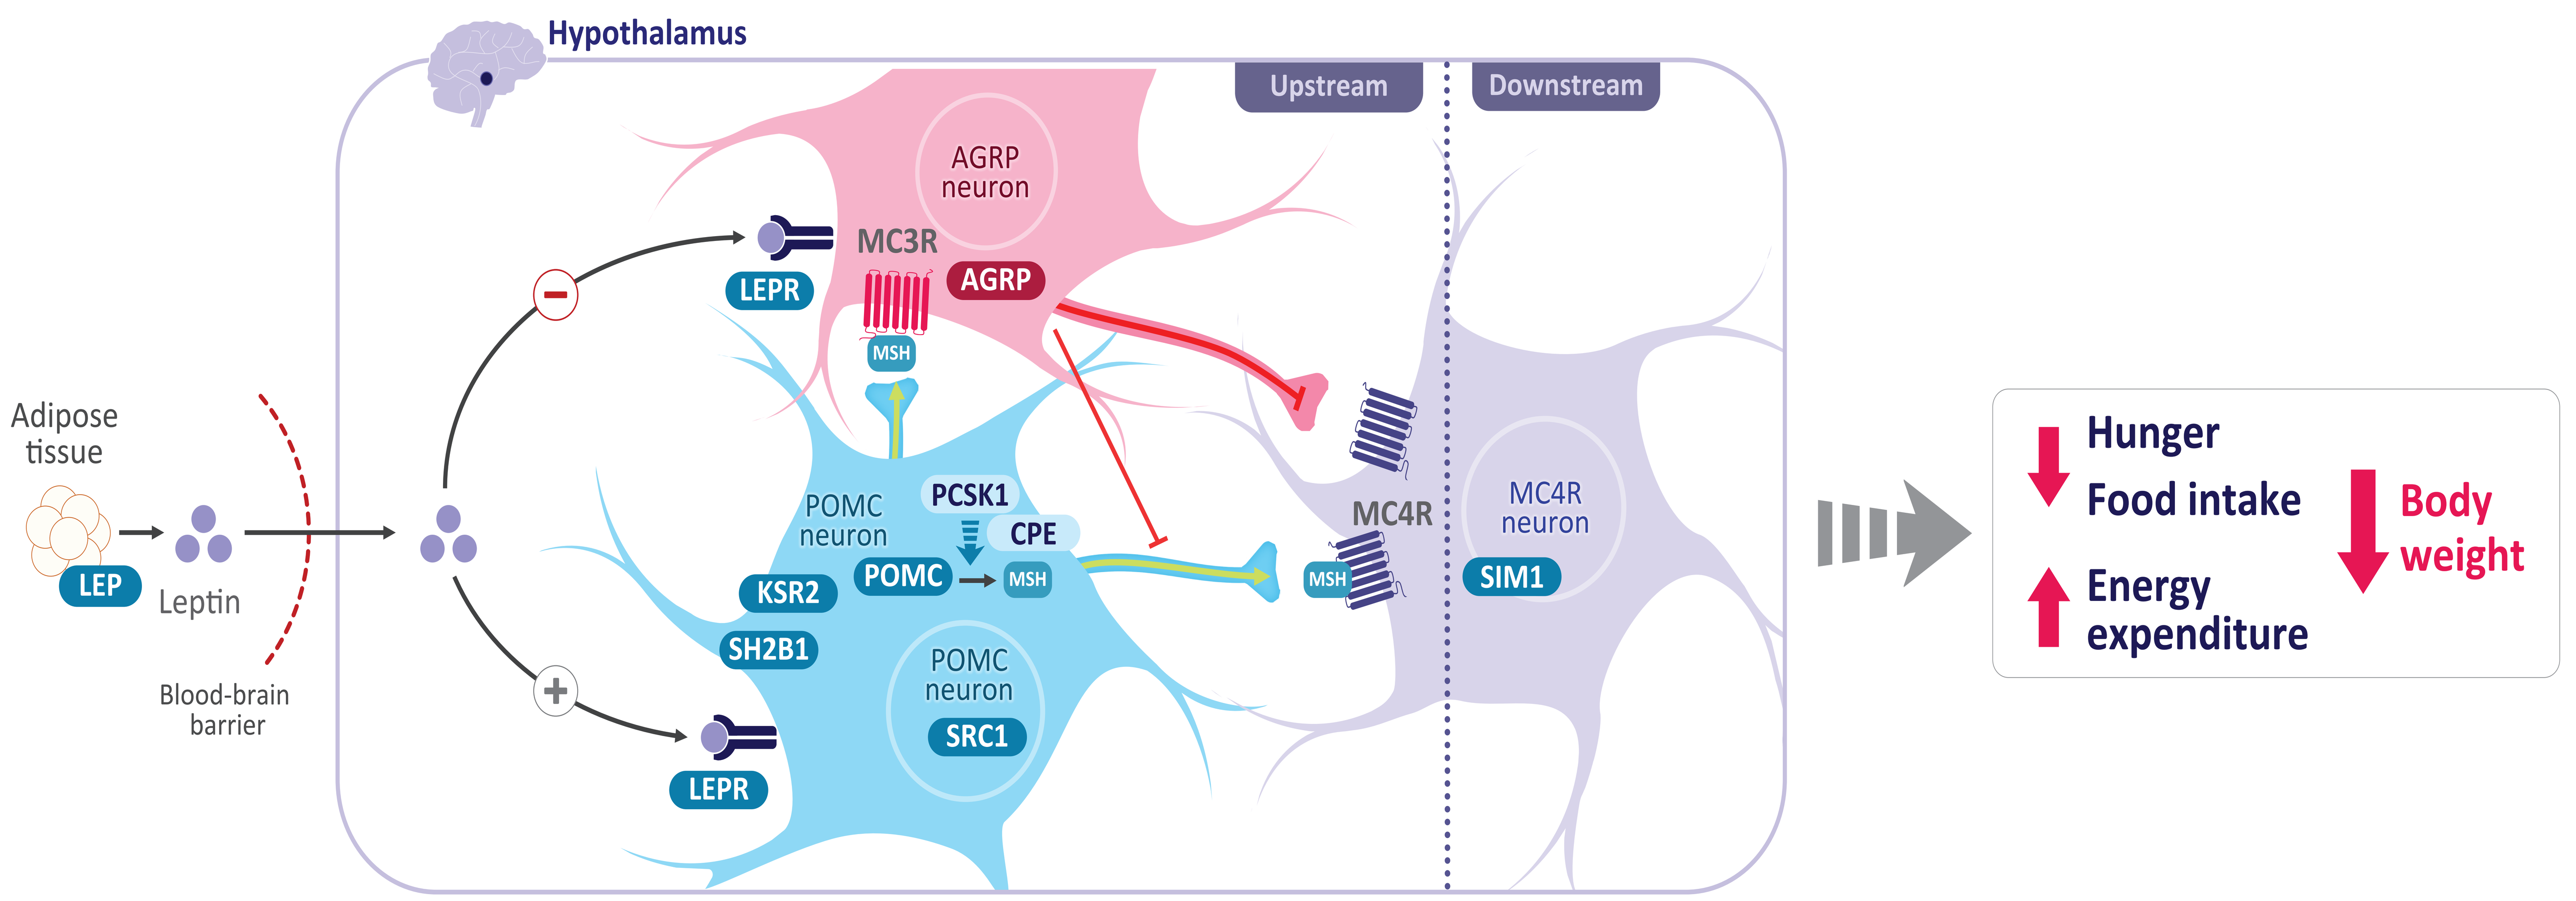
Hypothalamus
Upstream
Downstream
AGRP neuron
MC3R
AGRP
LEPR
MSH
Adipose tissue
LEP
Leptin
Blood-brain barrier
POMC neuron
PCSK1
CPE
POMC
MSH
KSR2
SH2B1
POMC neuron
SRC1
LEPR
MC4R
MSH
MC4R neuron
SIM1
Hunger
Food intake
Energy expenditure
Body weight

Methods

- Through May 4, 2021, we sequenced exons and intron-exon boundaries for 40 obesity-related genes in 7,826 individuals with severe obesity (<18 years old, ≥97th percentile BMI for age; ≥18 years old, BMI ≥40 kg/m^2) who participated in the US-based Uncovering Rare Obesity® (URO) diagnostic genetic testing program.
- This Next Generation Sequencing panel included 11 genes associated with non-syndromic obesity that encode proteins that function in the MC4R pathway (*POMC*, *PCSK1*, *LEPR*, *SRC1*, *SH2B1*, *MC4R*, *MC3R*, *CPE*, *LEP*, *KSR2*, and *SIM1*).
- Variants were classified as pathogenic/likely pathogenic (P/LP) or as a variant of uncertain significance (VUS) according to American College of Medical Genetics (ACMG) criteria.
- We additionally included one variant, *PCSK1* p.N221D, for which published functional and population studies suggest a potential contribution to obesity. This variant is classified as "Risk" according to ACMG criteria.
- Patients with variants in 5 of these genes (*POMC*, *PCSK1*, *LEPR*, *SRC1*, and *SH2B1*) have previously been assessed in a trial using the MC4R agonist setmelanotide and demonstrated meaningful weight loss.
- Variants in genes were additionally classified according to their classical mode of inheritance (MOI).
 - Autosomal dominant (AD) genes included *SRC1, SH2B1, MC4R, MC3R, KSR2*, and *SIM1*.
 - Autosomal recessive (AR) genes included *POMC, PCSK1, CPE*, and *LEP*.
- AD variants were considered in line with MOI if an individual had ≥1 variant allele in that gene, while AR variants were considered in line with MOI if ≥2 variants were present in that gene (ie, homozygous or compound heterozygous).

Results

- Among the 7,826 individuals sequenced, 16% were <6, 33% were 6 to 12, 30% were 12 to 18, and 21% were greater than 18 years old (Figure 2).

Figure 2. Number and percent of URO participants by age group.

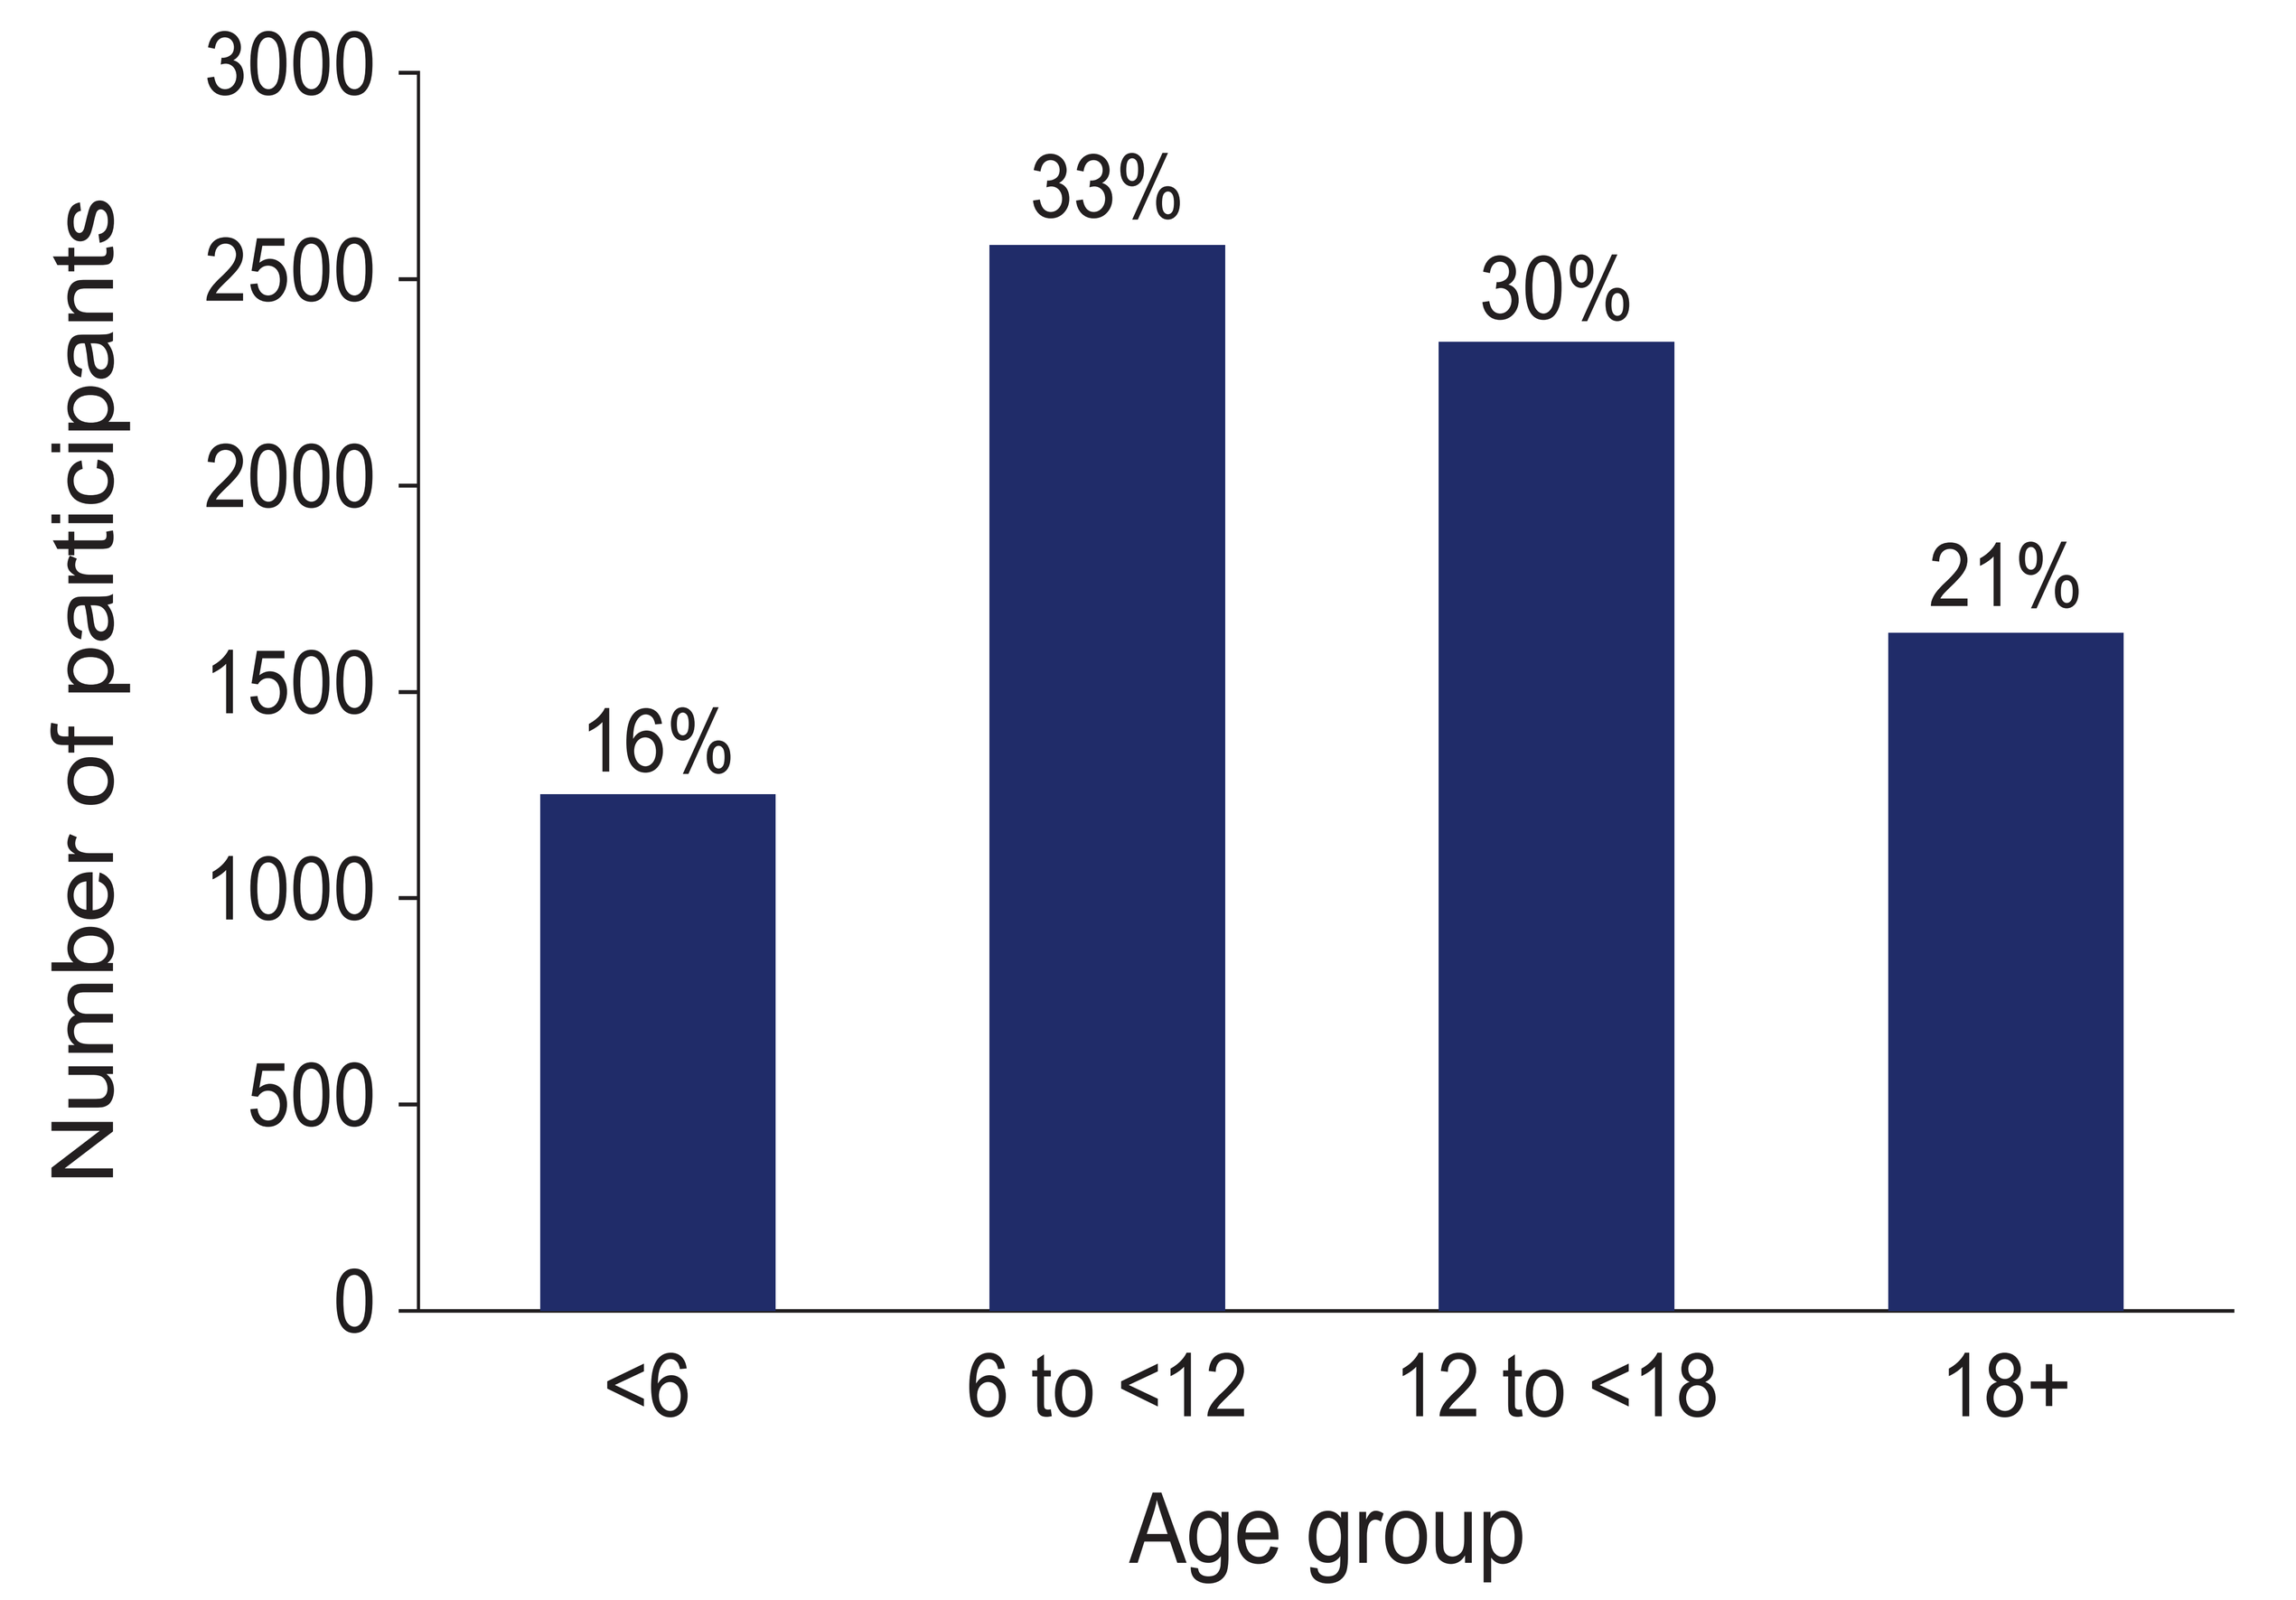
Number of participants
3000
2500
2000
1500
1000
500
0
16%
33%
30%
21%
<6
6 to <12
12 to <18
18+
Age group

- 19% of individuals carried ≥1 rare variant in ≥1 of the 11 studied genes, including 3.6% carrying a P/LP or high impact variant and 15.4% carrying a VUS variant.
- Restricting to the 5 genes with prior demonstrated responsiveness to setmelanotide, the yield was 11% for P/LP or VUS variants.
- 11.9% of individuals had variants in line with MOI, including 2.1% with all P/LP variants.
- An additional 6.4% carried the *PCSK1* p.N221D variant.
- Therefore, overall, 25.4% of individuals carried one or more variants in one of the 11 MC4R pathway genes studied, including P/LP, VUS, or Risk variants.
- No differences in variant frequency were observed by age group (Figure 3).

Figure 3. Percent variant frequency by age group.

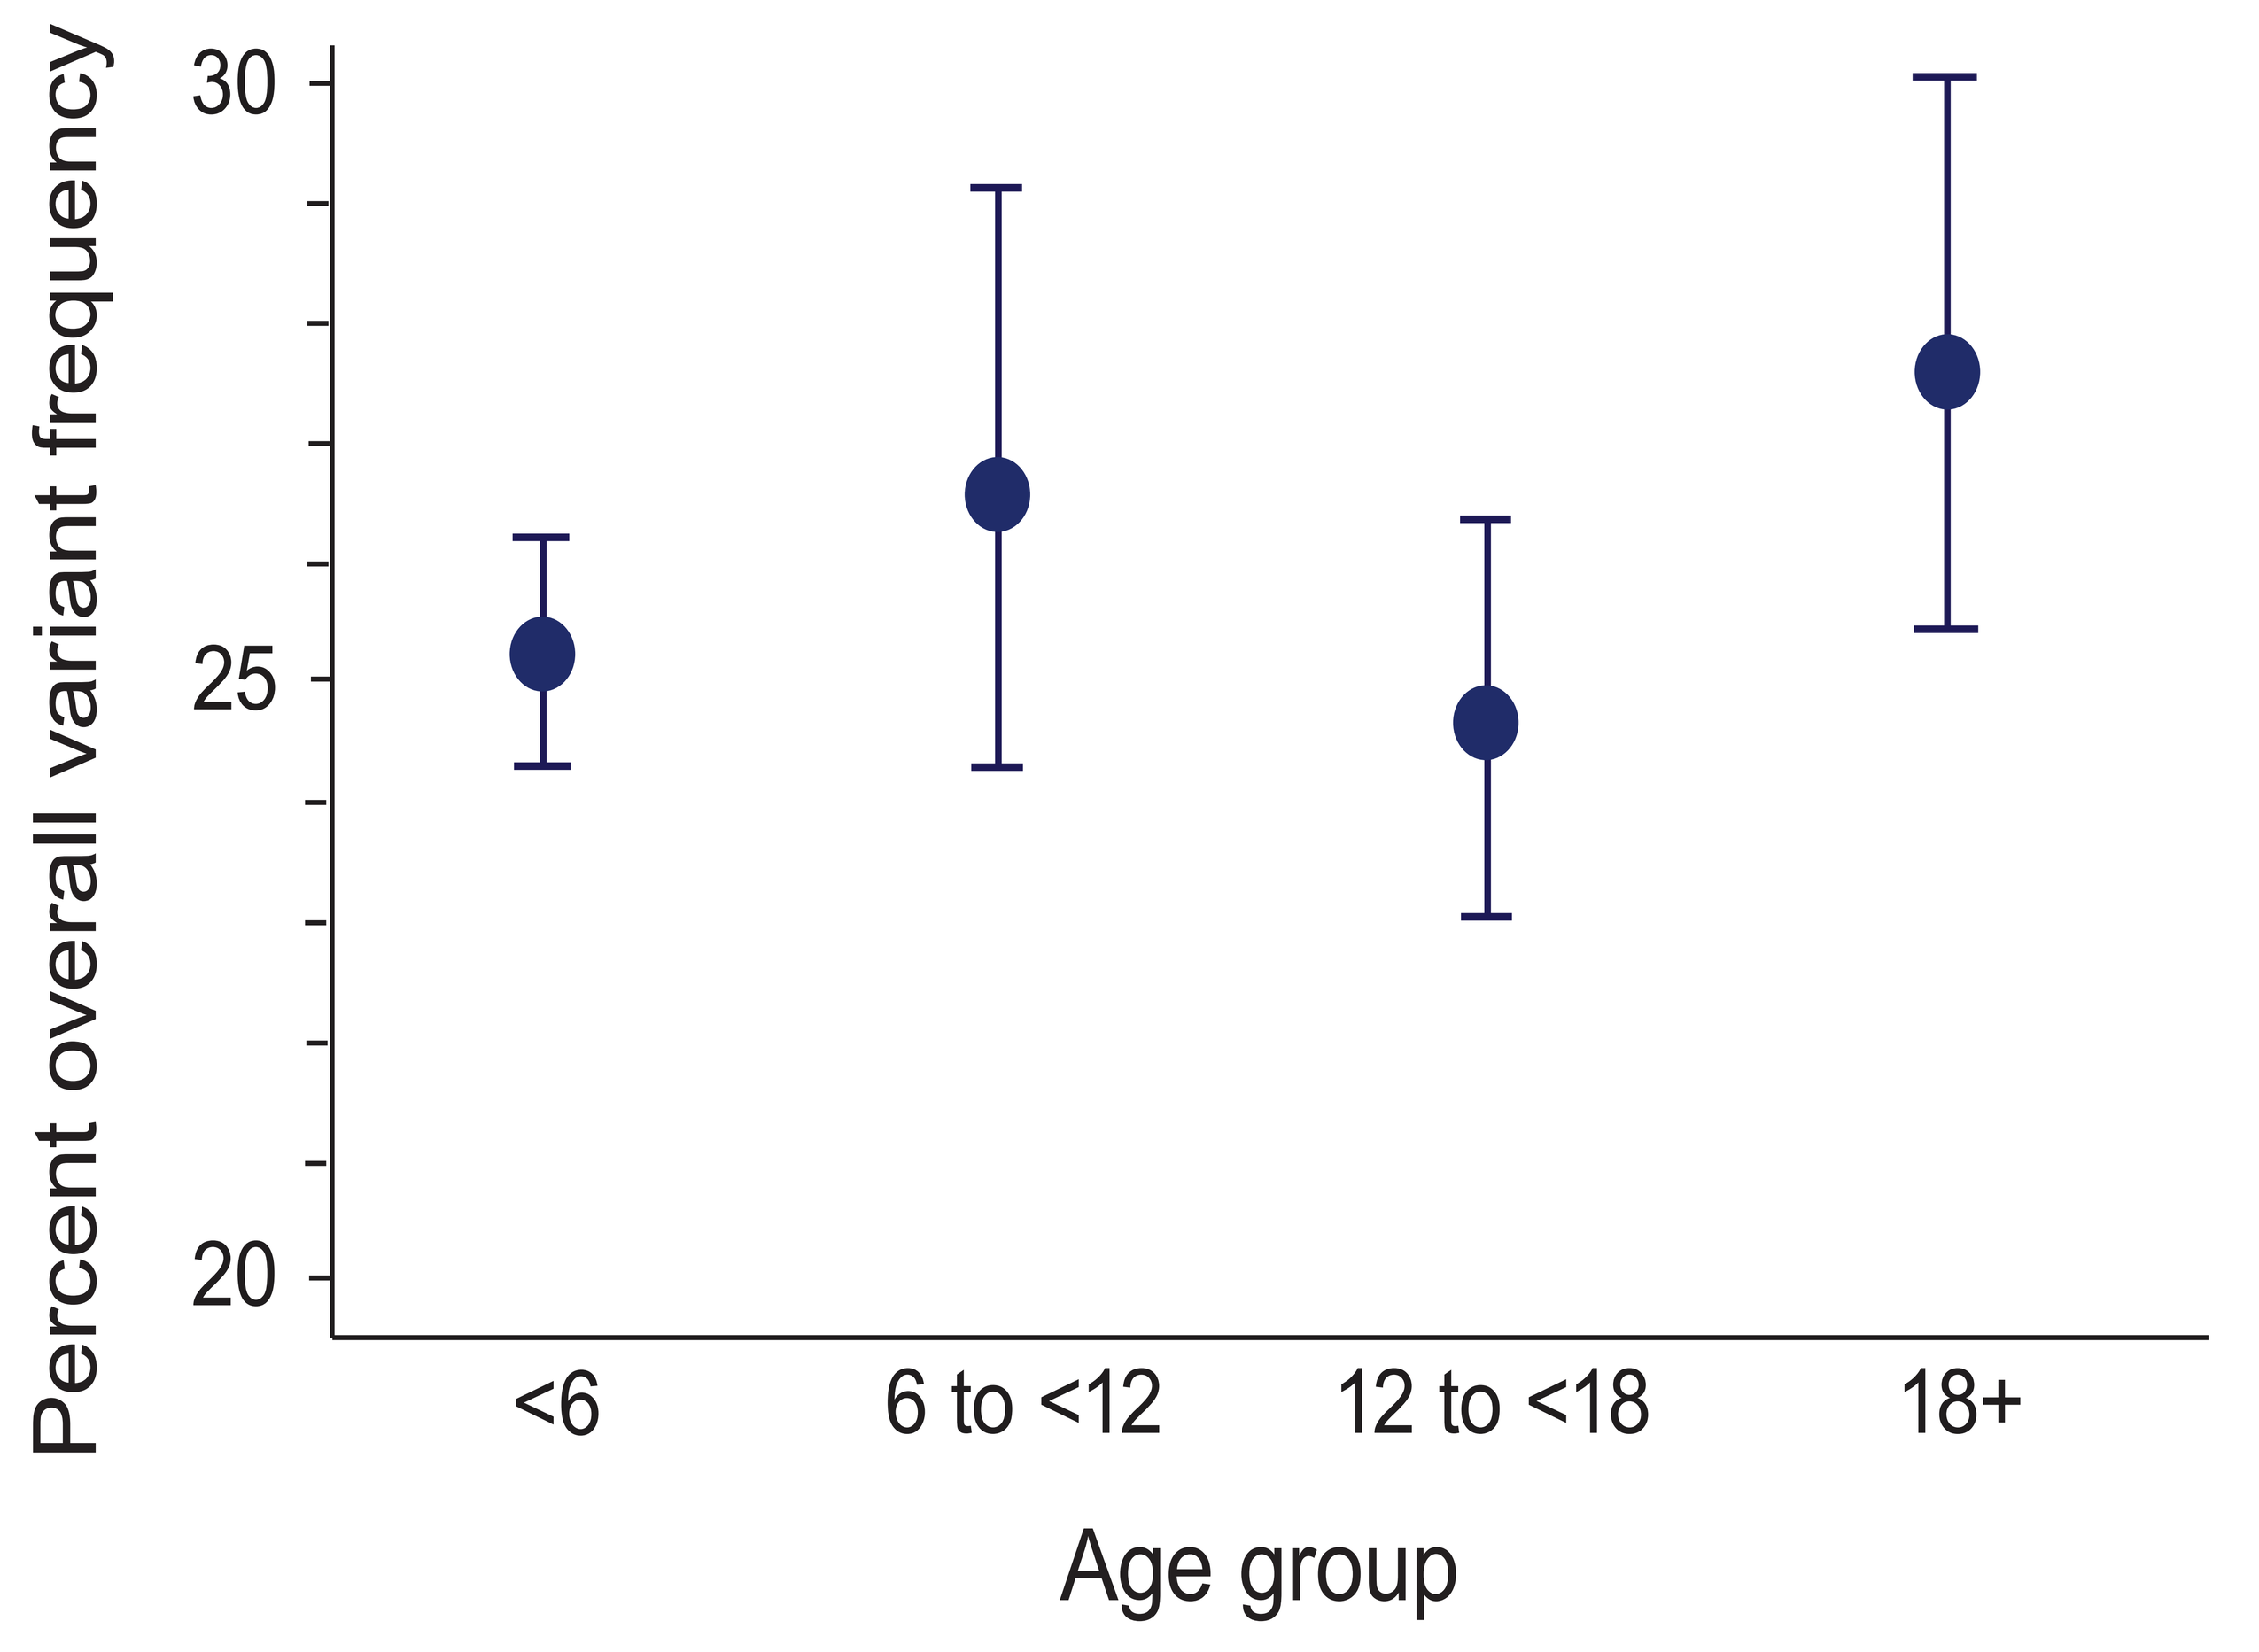
Percent overall variant frequency
30
25
20
<6
6 to <12
12 to <18
18+
Age group

*Alastair Garfield was an employee of Rhythm Pharmaceuticals, Inc., at the time of abstract submission.

Uncovering Rare Obesity is a registered trademark of Rhythm Pharmaceuticals, Inc.

Acknowledgments: This study was sponsored by Rhythm Pharmaceuticals, Inc. The authors would like to thank Megan Vogel for advising on the development of Figure 1.

References: **1.** Huvenne et al. *Obes Facts*. 2016;9:158-173. **2.** Yazdi et al. *PeerJ*. 2015;24:ee856. **3.** Yang et al. 2019:10;1718. **4.** Seo et al. *Hum Mol Genet*. 2009;18:1323-1331. **5.** Revelli et al. *Obesity (Silver Spring)*. 2011;19:1010-1018. **6.** Doche et al. *J Clin Invest*. 2012;122:4732-4736.

European Society for Paediatric Endocrinology • September 22-26, 2021 • Virtual

Presented at: ESPE 2021

Fat, metabolism and obesity
Ida Moeller

P2-179