Evaluating Safety, Efficacy, and Pharmacokinetics of Weekly TransCon CNP in Children with Achondroplasia: Design of the ACcomplisH Trial

Ciara McDonnell[1], Melita Irving[2], Yuri Zarate[3], Hanne B. Hove[4], Wolfgang Högler[5], Daniel Hoernschemeyer[6], YingZhang[7], DortheViuff[8], Marie-Louise Hartoft-Nielsen[8], Michael Beckert[8], Ravi Savarirayan[9]

[1]Children's Health Ireland at Temple Street; Dublin, IRE; [2]Guy's and St. Thomas' NHS Foundation Trust; London, UK; [3]University of Arkansas for Medical Sciences, Little Rock, AK; [4]Center for Rare Diseases, Department of Pediatrics, Copenhagen University Hospital Rigshospitalet; Copenhagen, Denmark; [5]Department of Paediatrics and Adolescent Medicine, Johannes Kepler University Linz; Linz, Austria; [6]University of Missouri-Columbia; Columbia, MO; [7]Ascendis Pharma Inc, [8]Ascendis Pharma A/S, [9]Royal Children's Hospital and University of Melbourne, Victoria AUS

BACKGROUND

- Achondroplasia (ACH) is the most common short limbed skeletal dysplasia[1]. It is caused by a gain-of-function mutation in the fibroblast growth factor receptor 3 (FGFR3) gene and results in impairment of the endochondral ossification process[2] (Figure 1)
- C-type natriuretic peptide (CNP) promotes chondrocyte development through inhibition of the FGFR3 pathway, specifically through activation of Natriuretic Peptide Receptor Type B (NPR-B). CNP is a potential therapeutic pathway for treating growth failure and achondroplasia, as it inhibits the overactive signalling through the mutated FGFR3 receptor causing ACH[3,4,5] (Figure 1)
- ACH is associated with significant morbidity with many patients requiring intervention for obstructive sleep apnea and many have clinical signs and symptoms related to spinal stenosis (Figure 2)
- As reported by caregivers of children with ACH, there is an impact on symptoms and daily functioning (Figure 3)

Figure 1. CNP Can Counteract the Pathology of ACH

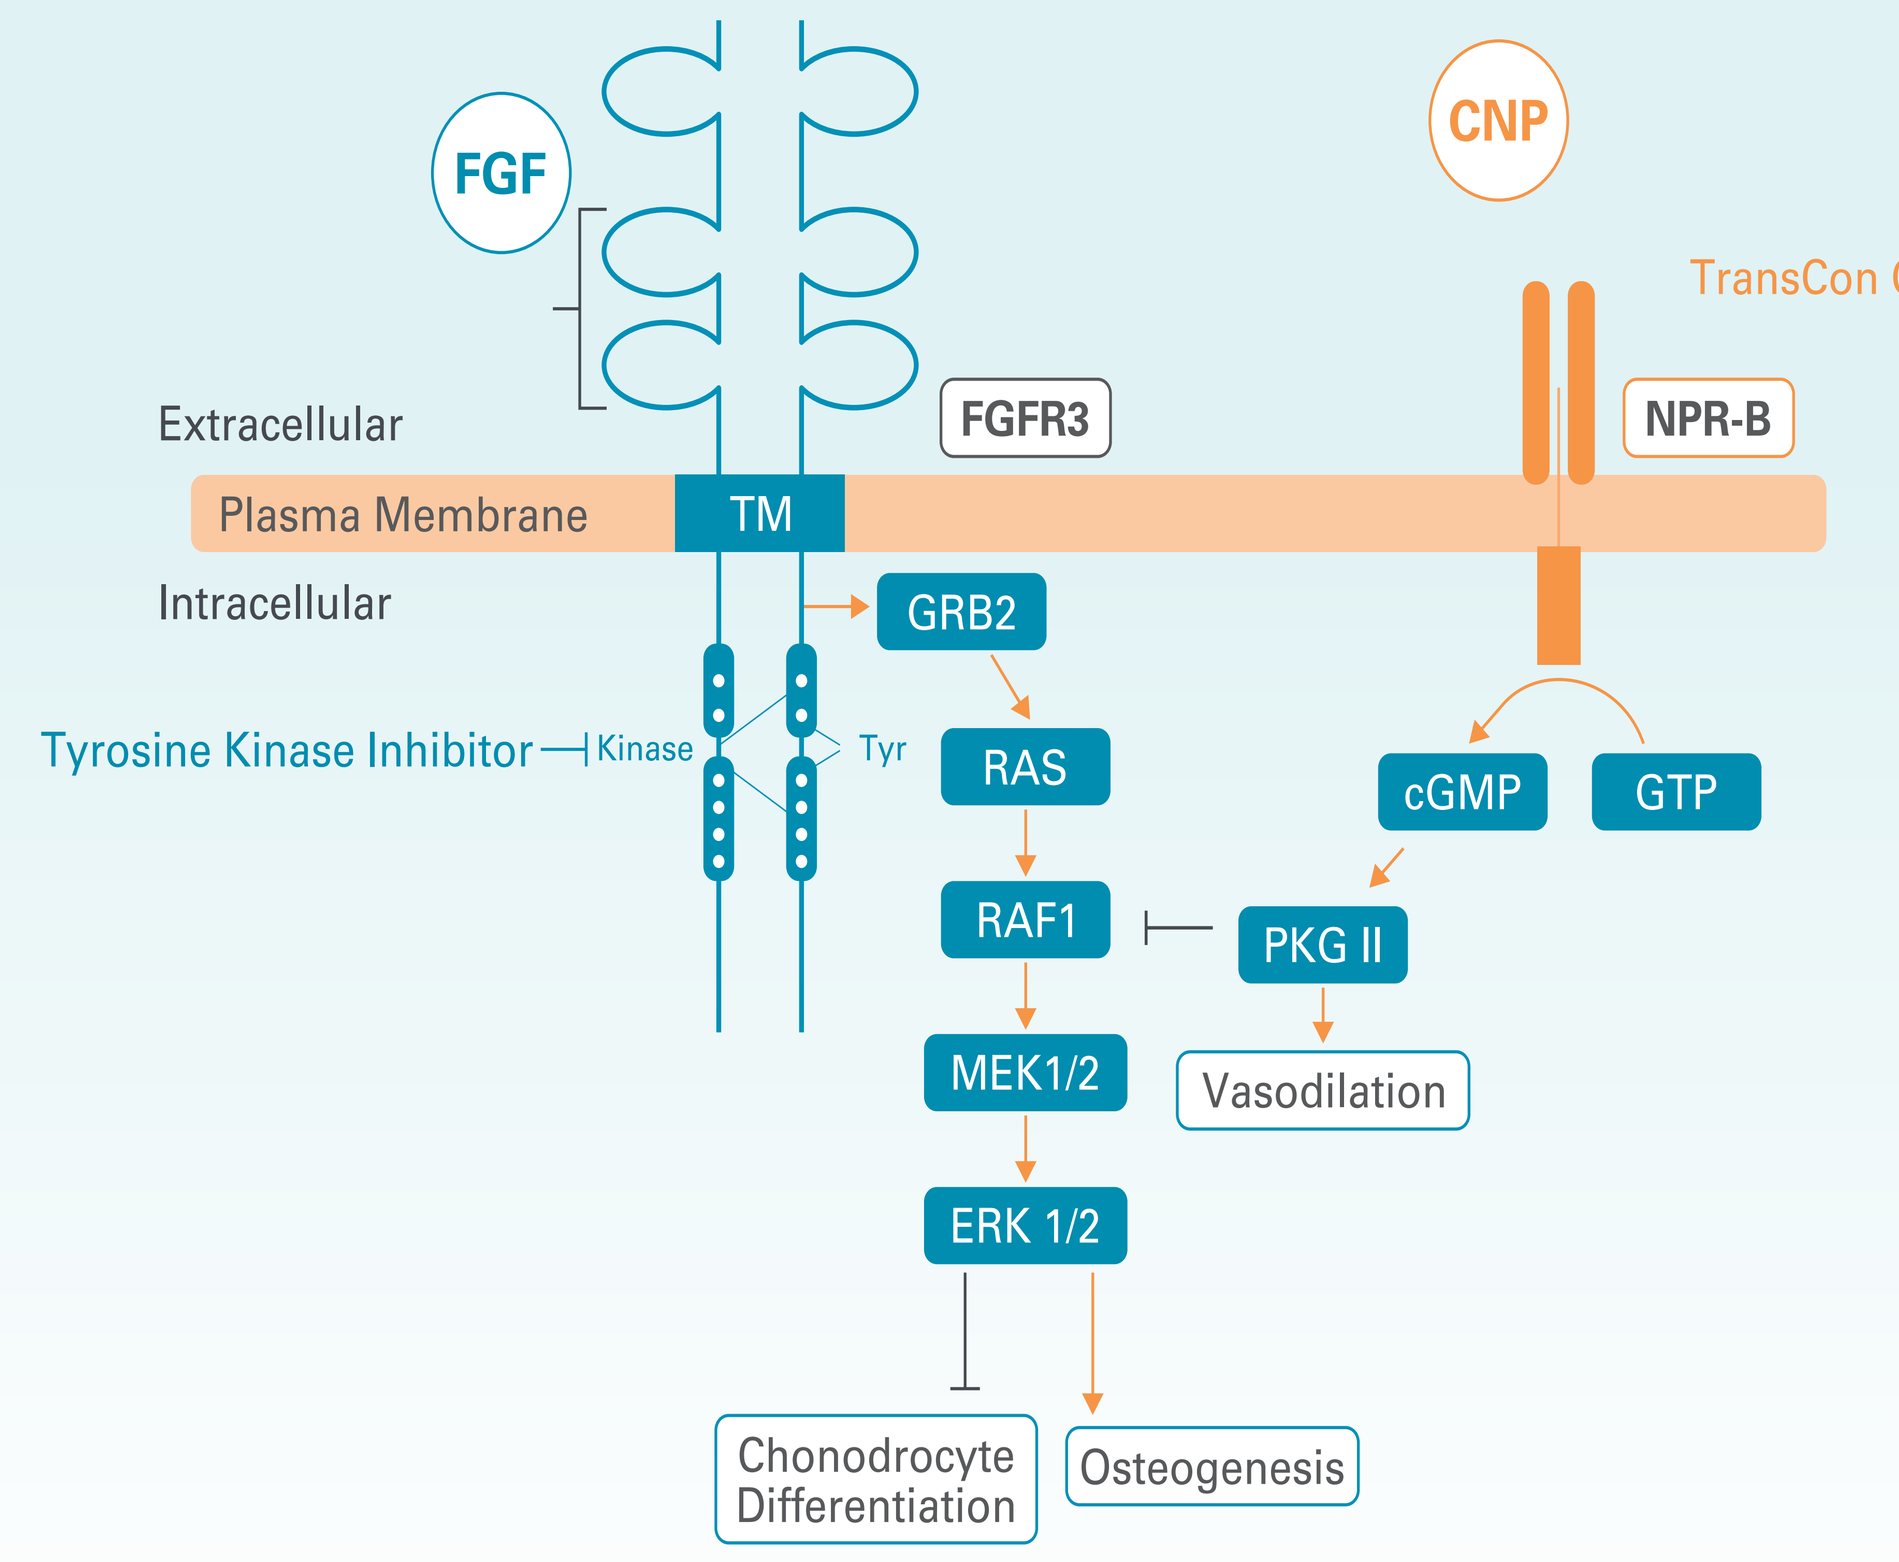
FGF
CNP
TransCon CNP
Extracellular
FGFR3
NPR-B
Plasma Membrane
TM
Intracellular
GRB2
Tyrosine Kinase Inhibitor
Kinase
Tyr
RAS
cGMP
GTP
RAF1
PKG II
MEK1/2
Vasodilation
ERK 1/2
Chonodrocyte Differentiation
Osteogenesis

CNP does not alter the function of FGF receptors or change endogenous levels of FGF ligands, hereby reducing the risk of interfering with normal FGF biology

FGF, fibroblast growth factor; FGFR3, fibroblast growth factor receptor 3; CNP, C-type natriuretic peptide; NPR-B, natriuretic peptide receptor B; GRB2, Growth factor receptor-bound protein 2; cGMP, cyclic guanosine monophosphate; GTP, guanosine 5'-triphosphate; PKG II, protein kinase G II; RAF, rapidly accelerated fibrosarcoma kinases; MEK, Mitogen-activated protein kinase; ERK, extracellular signal-regulated kinase

Figure 2. Achondroplasia Morbidity[6]

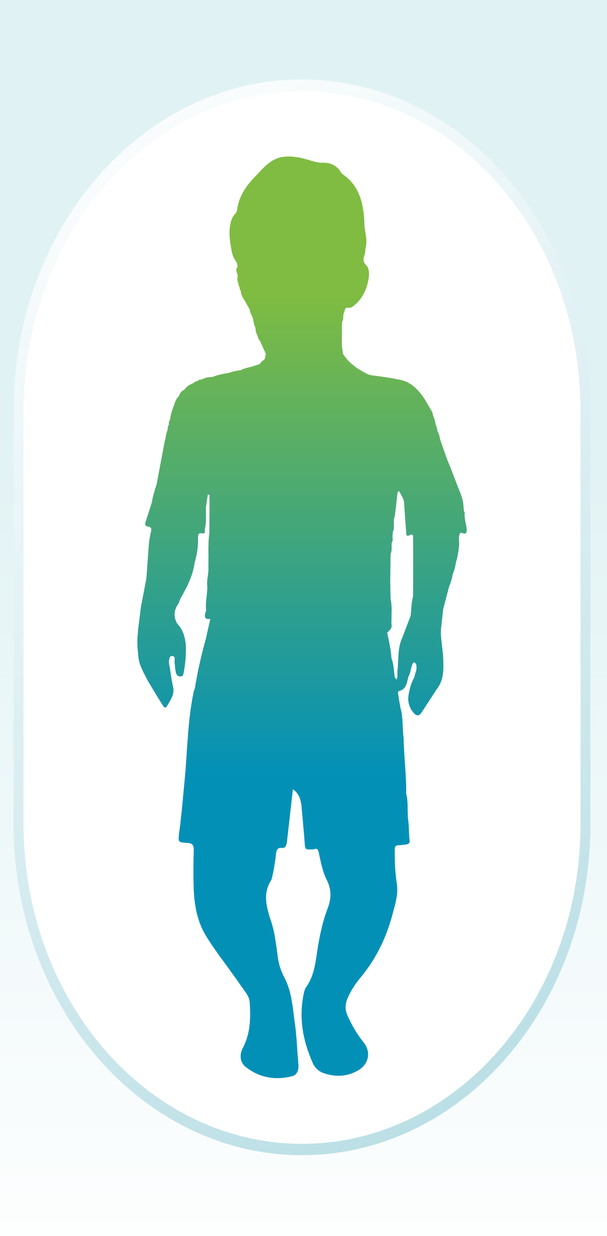

- Up to **85%** of patients require intervention for obstructive sleep apnea and respiratory insufficiency
- **25%** of children have hearing loss increasing to >50% in adulthood
- **22%** have osteotomy
- **15-30%** have fixed kyphotic deformity
- Up to **28%** require cervicomedullary decompression by age 4
- **10%** of children have neurological signs of spinal stenosis
- **80%** of adults have clinical signs and symptoms related to spinal stenosis

Figure 3. Symptoms and Functioning Reported by Caregivers of Children With ACH

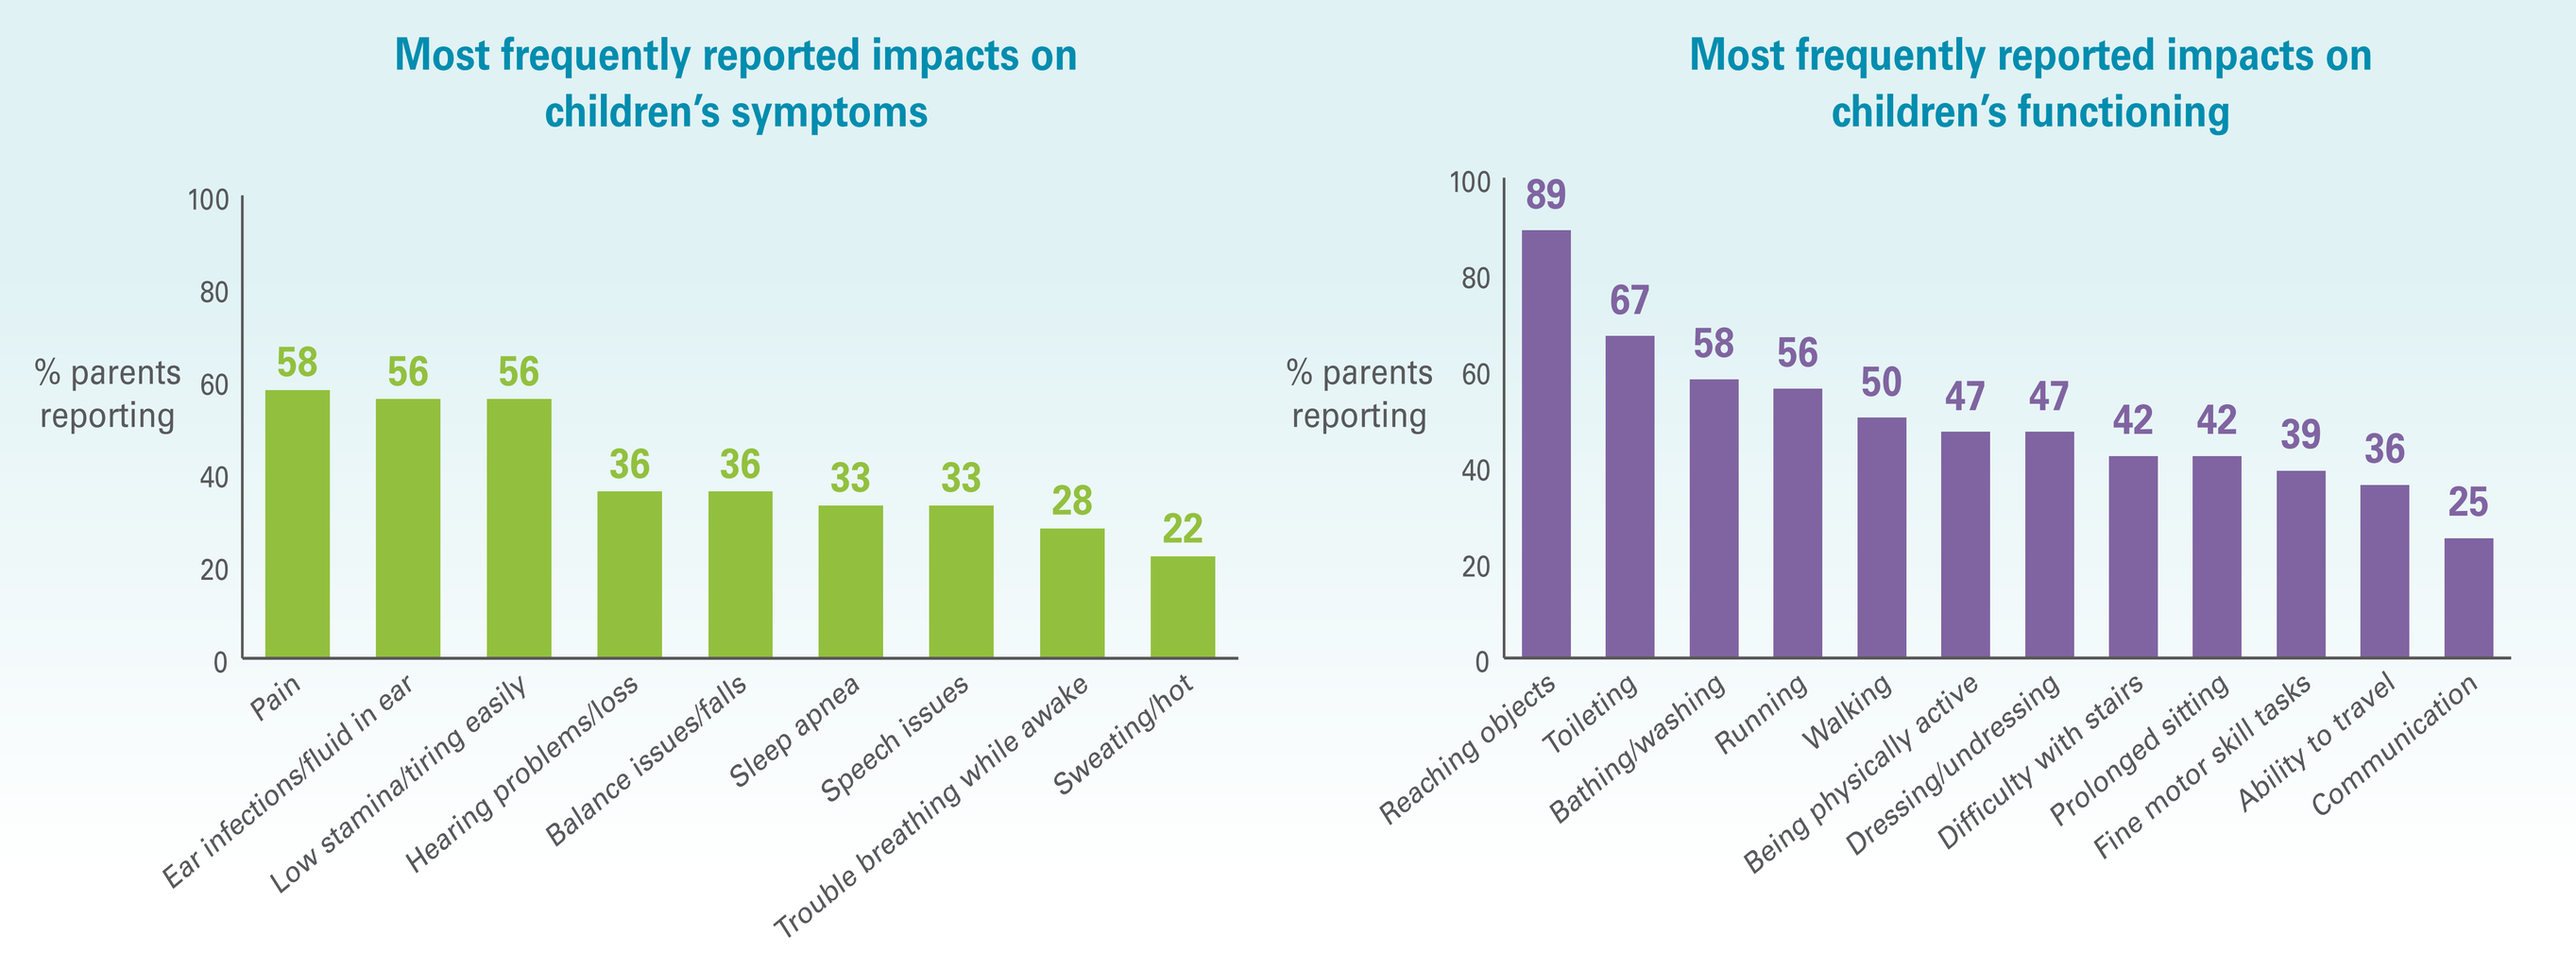
Most frequently reported impacts on children's symptoms
% parents reporting
Pain 58
Ear infections/fluid in ear 56
Low stamina/tiring easily 56
Hearing problems/loss 36
Balance issues/falls 36
Sleep apnea 33
Speech issues 33
Trouble breathing while awake 28
Sweating/hot 22
Most frequently reported impacts on children's functioning
% parents reporting
Reaching objects 89
Toileting 67
Bathing/washing 58
Running 56
Walking 50
Being physically active 47
Dressing/undressing 47
Difficulty with stairs 42
Prolonged sitting 42
Fine motor skill tasks 39
Ability to travel 36
Communication 25

Qualitative analysis of impacts of ACH on children aged 2-12 years was undertaken to inform the development of the Achondroplasia Child Experience Measures (ACEMs): ACEM – Symptom and ACEM – Impact (N = 36)[7]

Figure 4. TransCon CNP Design

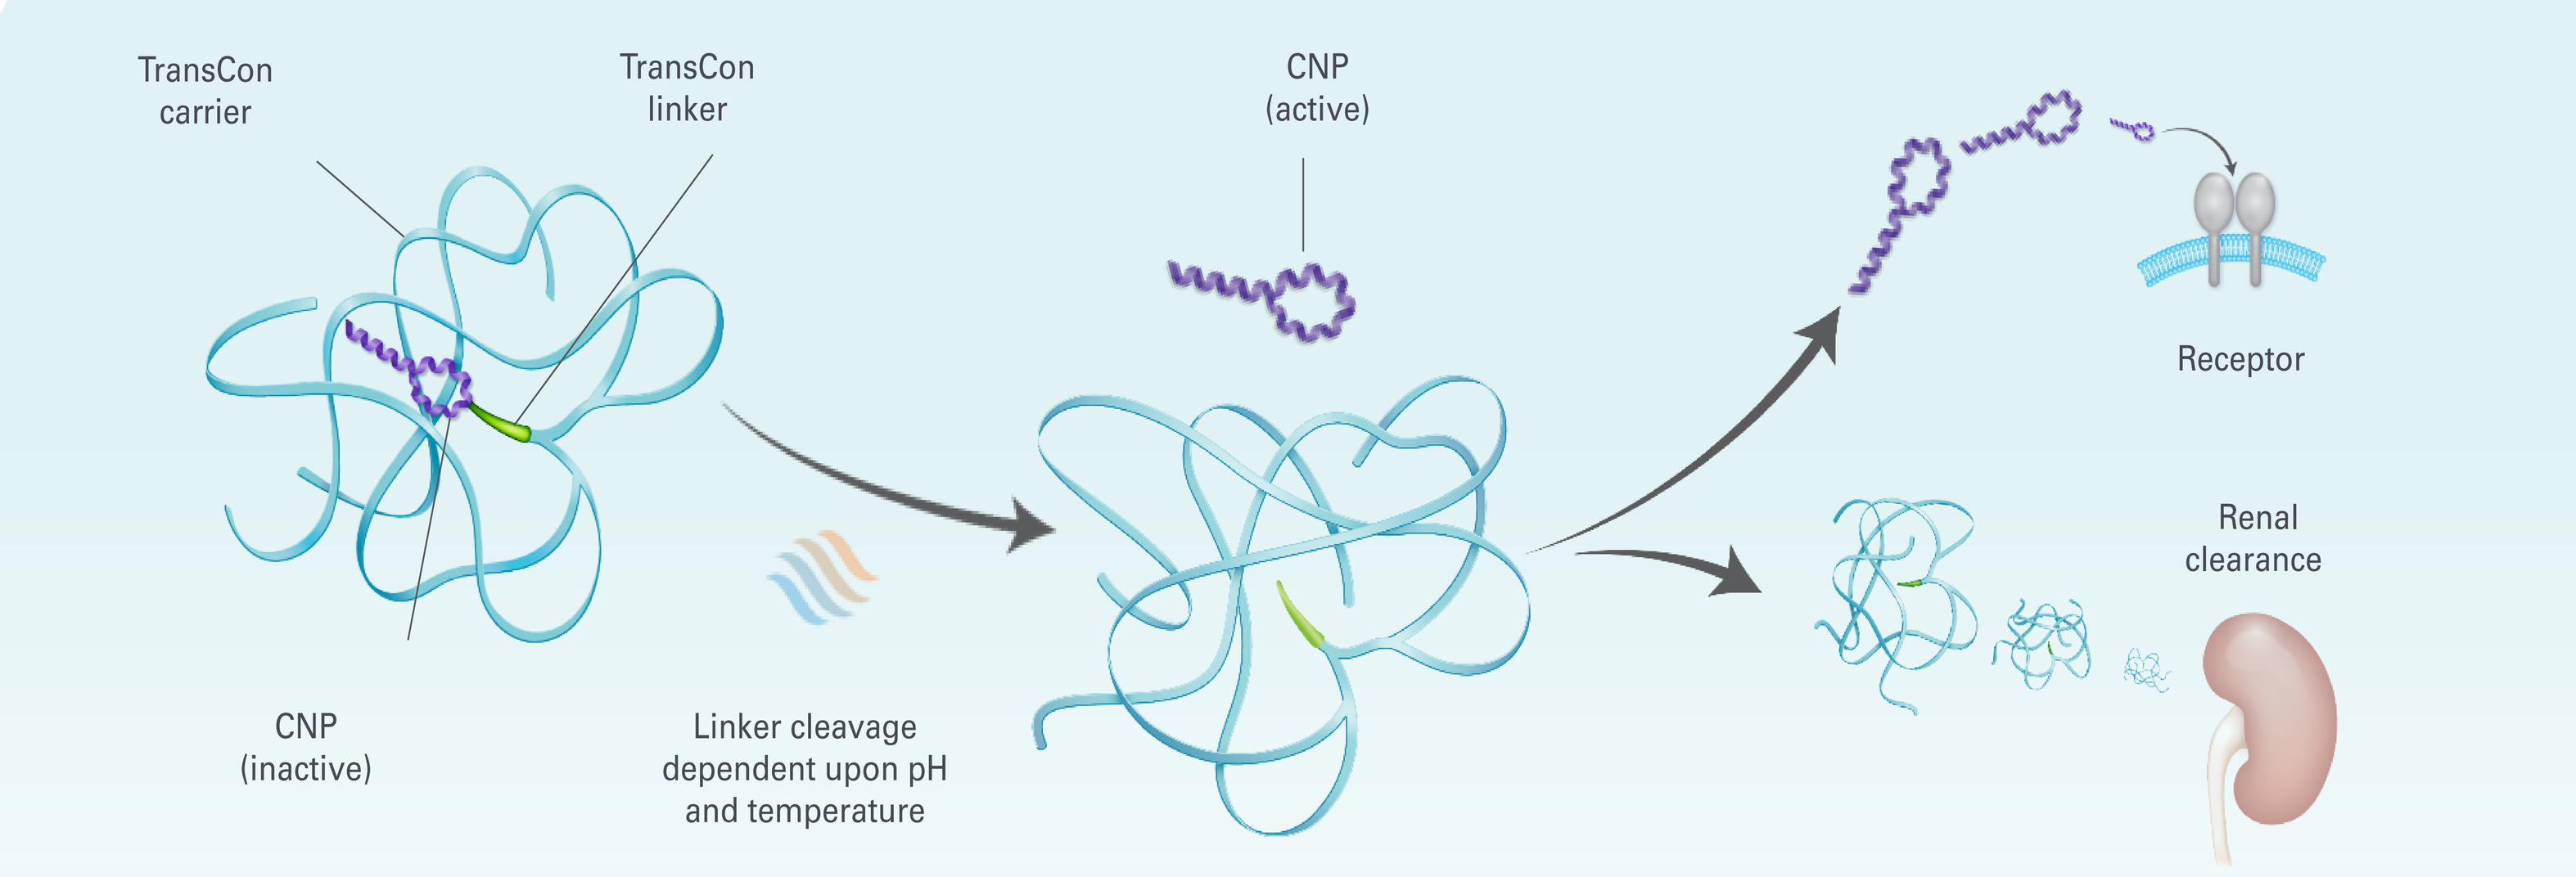
TransCon carrier
TransCon linker
CNP (active)
Receptor
Renal clearance
CNP (inactive)
Linker cleavage dependent upon pH and temperature

- TransCon technology is designed to provide effective shielding of CNP[8]:
 - From neutral endopeptidase degradation in subcutaneous tissue and blood compartment
 - Minimize binding of TransCon CNP to the NPR-C receptor
 - Reduce binding of TransCon CNP to the NPR-B receptor in vasculature to avoid hypotension
- CNP liberated from TransCon CNP maintains small enough size to allow penetration into growth plates

CNP = C-type natriuretic peptide; NPR-B = natriuretic peptide receptor B; NPR-C =natriuretic peptide receptor C

- TransCon CNP is a sustained-release prodrug of CNP. CNP is transiently bound via the TransCon Linker to a chemically inert carrier that prolongs the peptide's overall circulatory half-life. This is achieved by minimizing renal clearance of the TransCon CNP prodrug and shielding of the CNP molecule from proteolytic degradation and from binding to its primary activating and clearance receptors, NPR-B and NPR-C
- Following cleavage of the TransCon Linker under physiological pH and temperature, active CNP-38 peptide is slowly and continuously released
- The Phase 1 clinical trial with TransCon CNP demonstrated that single doses up to 150 µg CNP/kg were well-tolerated in healthy adult male volunteers, with no clinically significant trends observed in clinical laboratory assessments, vital sign measurements, ECG parameters, or physical examination findings[9,10]

Figure 5. Dose Proportional CNP Exposure For 1 Week

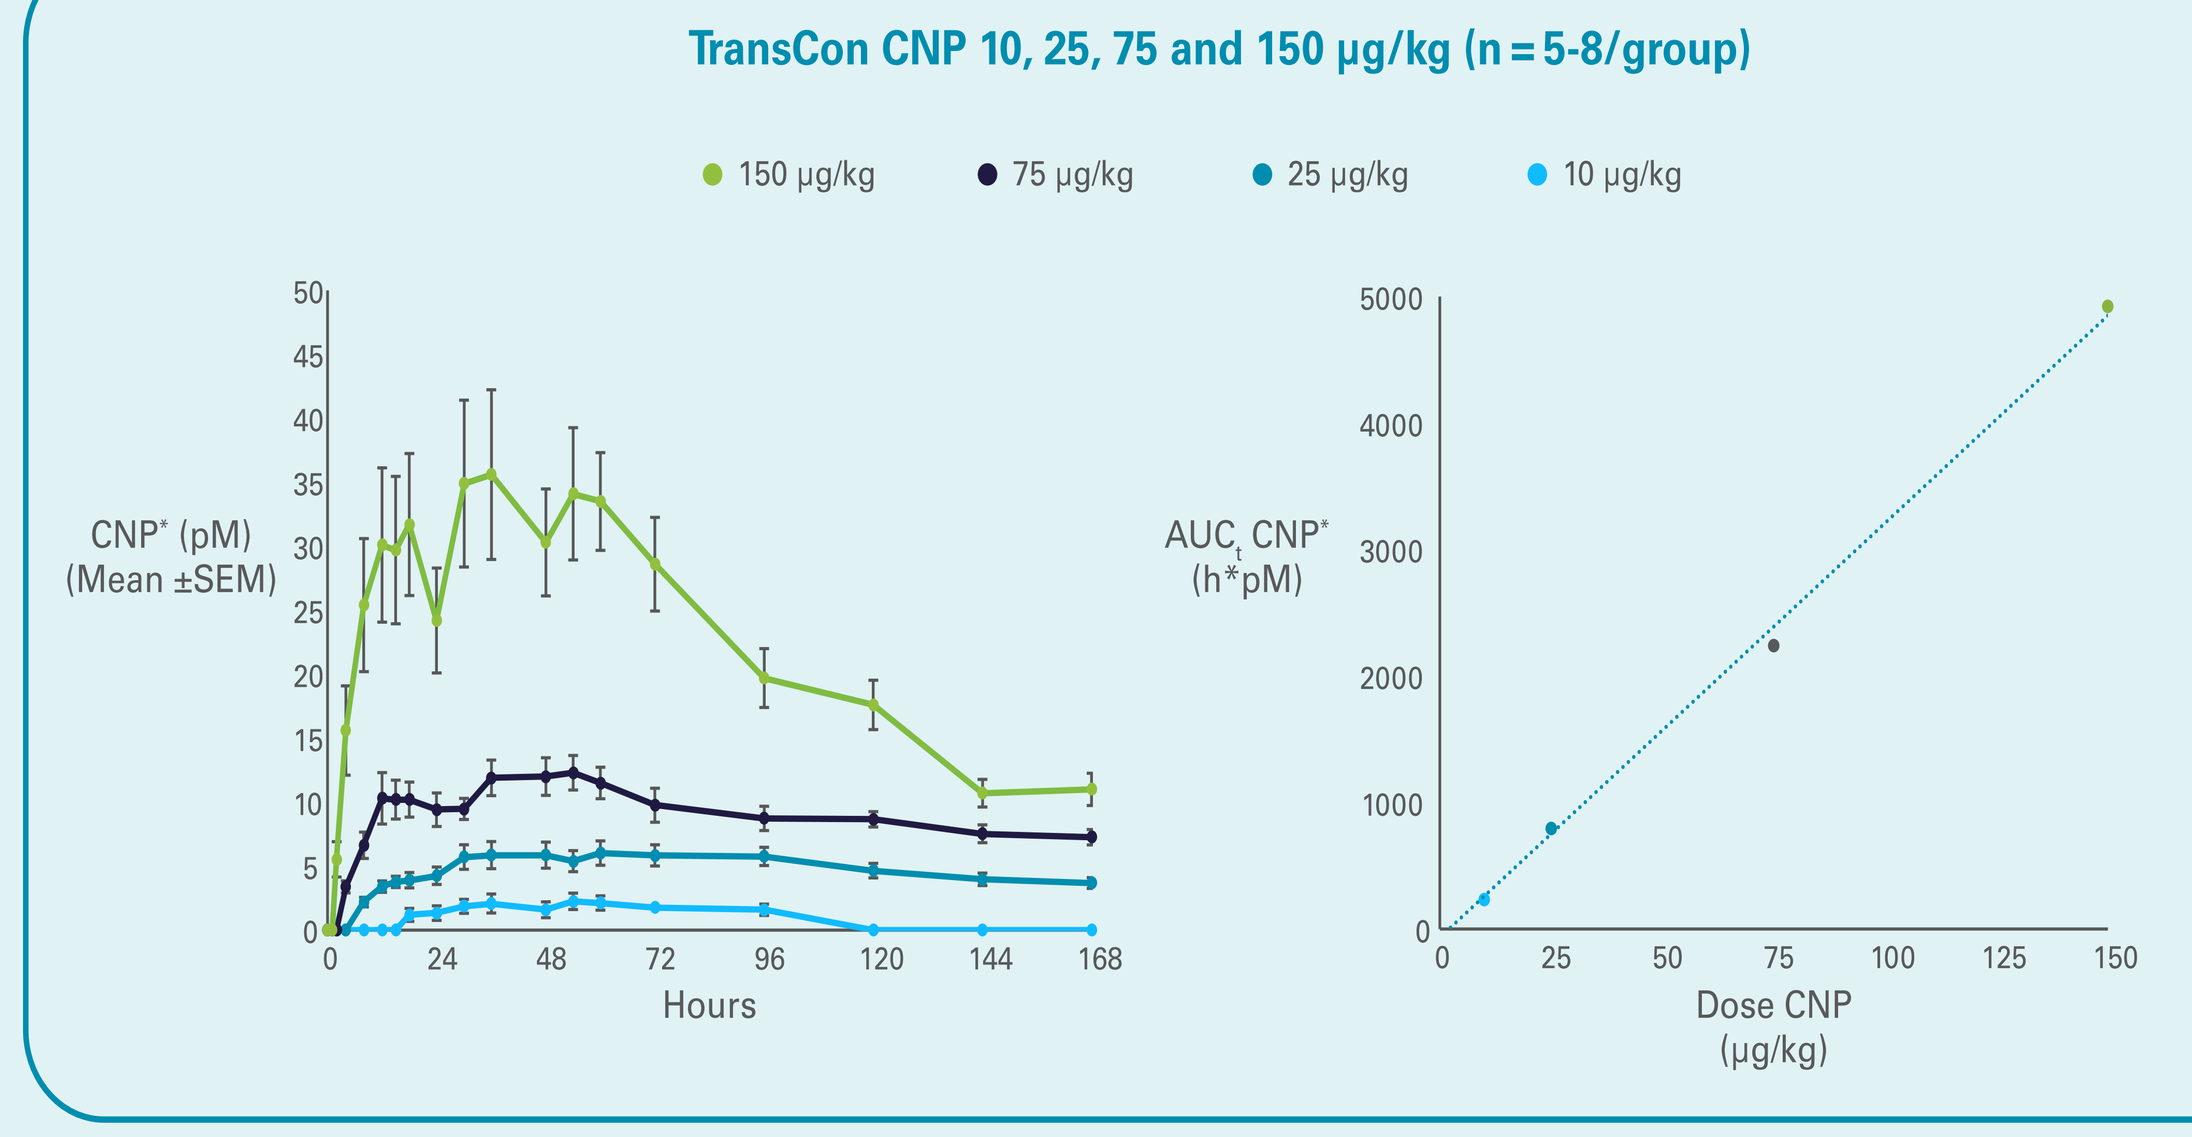
TransCon CNP 10, 25, 75 and 150 µg/kg (n = 5-8/group)
150 µg/kg
75 µg/kg
25 µg/kg
10 µg/kg
CNP* (pM) (Mean ±SEM)
Hours
AUC CNP* (h*pM)
Dose CNP (µg/kg)

*CNP measured as CNP-38

- Phase 1 showed effective CNP $t_{½}$ of approximately 120 hours (native CNP $t_{½}$ of 2–3 minutes)[8,11]
- Dose proportional increase in CNP exposure suggests ability to titrate dosing
- Mean pharmacokinetic (PK) profiles of Free CNP following subcutaneous administration of TransCon CNP. The Free CNP PK-profile following release from the prodrug was characterized by a slow rise to peak plasma concentrations with median T_{max} ranging from 45 to 66 h postdose. After reaching C_{max}, Free CNP concentrations slowly declined. The apparent $t_{½}$ was estimated to be approximately 120 h
- Statistical assessment concluded dose proportionality in exposure to Free CNP over the 10 to 150 µg CNP/kg dose range for C_{max} and $AUC_{0\text{-}t}$ based on regression slope estimates of 0.973 to 1.09 with the 95% CIs encompassing 1

Figure 6. Change from Baseline in Plasma (A) and Urine (B) cGMP

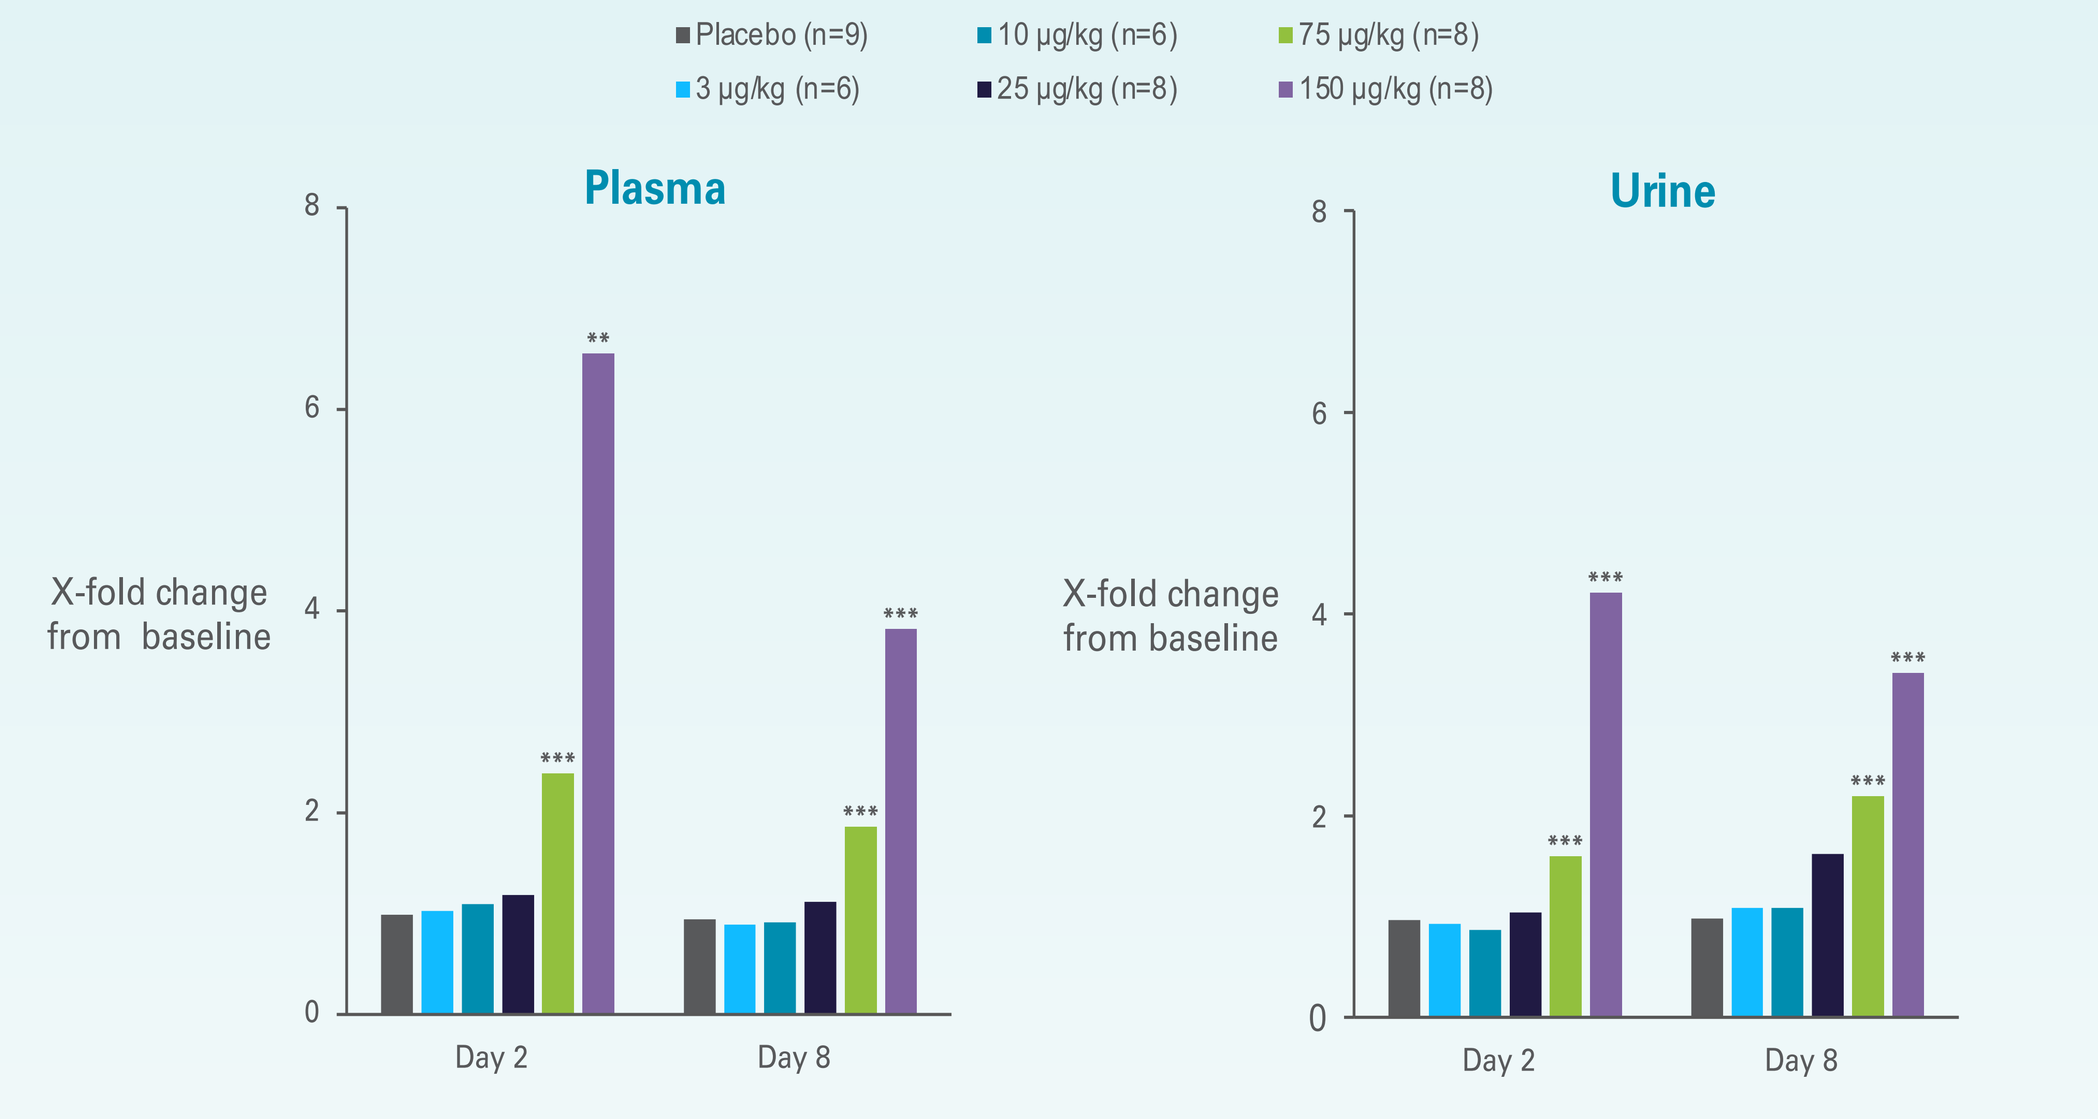
Placebo (n=9)
3 µg/kg (n=6)
10 µg/kg (n=6)
25 µg/kg (n=8)
75 µg/kg (n=8)
150 µg/kg (n=8)
Plasma
Urine
X-fold change from baseline
Day 2
Day 8

P < 0.01, *P < 0.001

cGMP=cyclic guanosine monophosphate.

- cGMP is a secondary messenger of NPR-B activation by CNP
- cGMP levels correlated with dose
- cGMP was measured in plasma and urine samples. Normalized levels of cGMP (fold change from baseline) measured in both plasma (left) and urine (right) samples collected post-dose at Day 2 & 8. ***P* < 0.01. ****P* < 0.001

Figure 7. Mean Resting Blood Pressure and Heart Rate Unchanged from Predose*

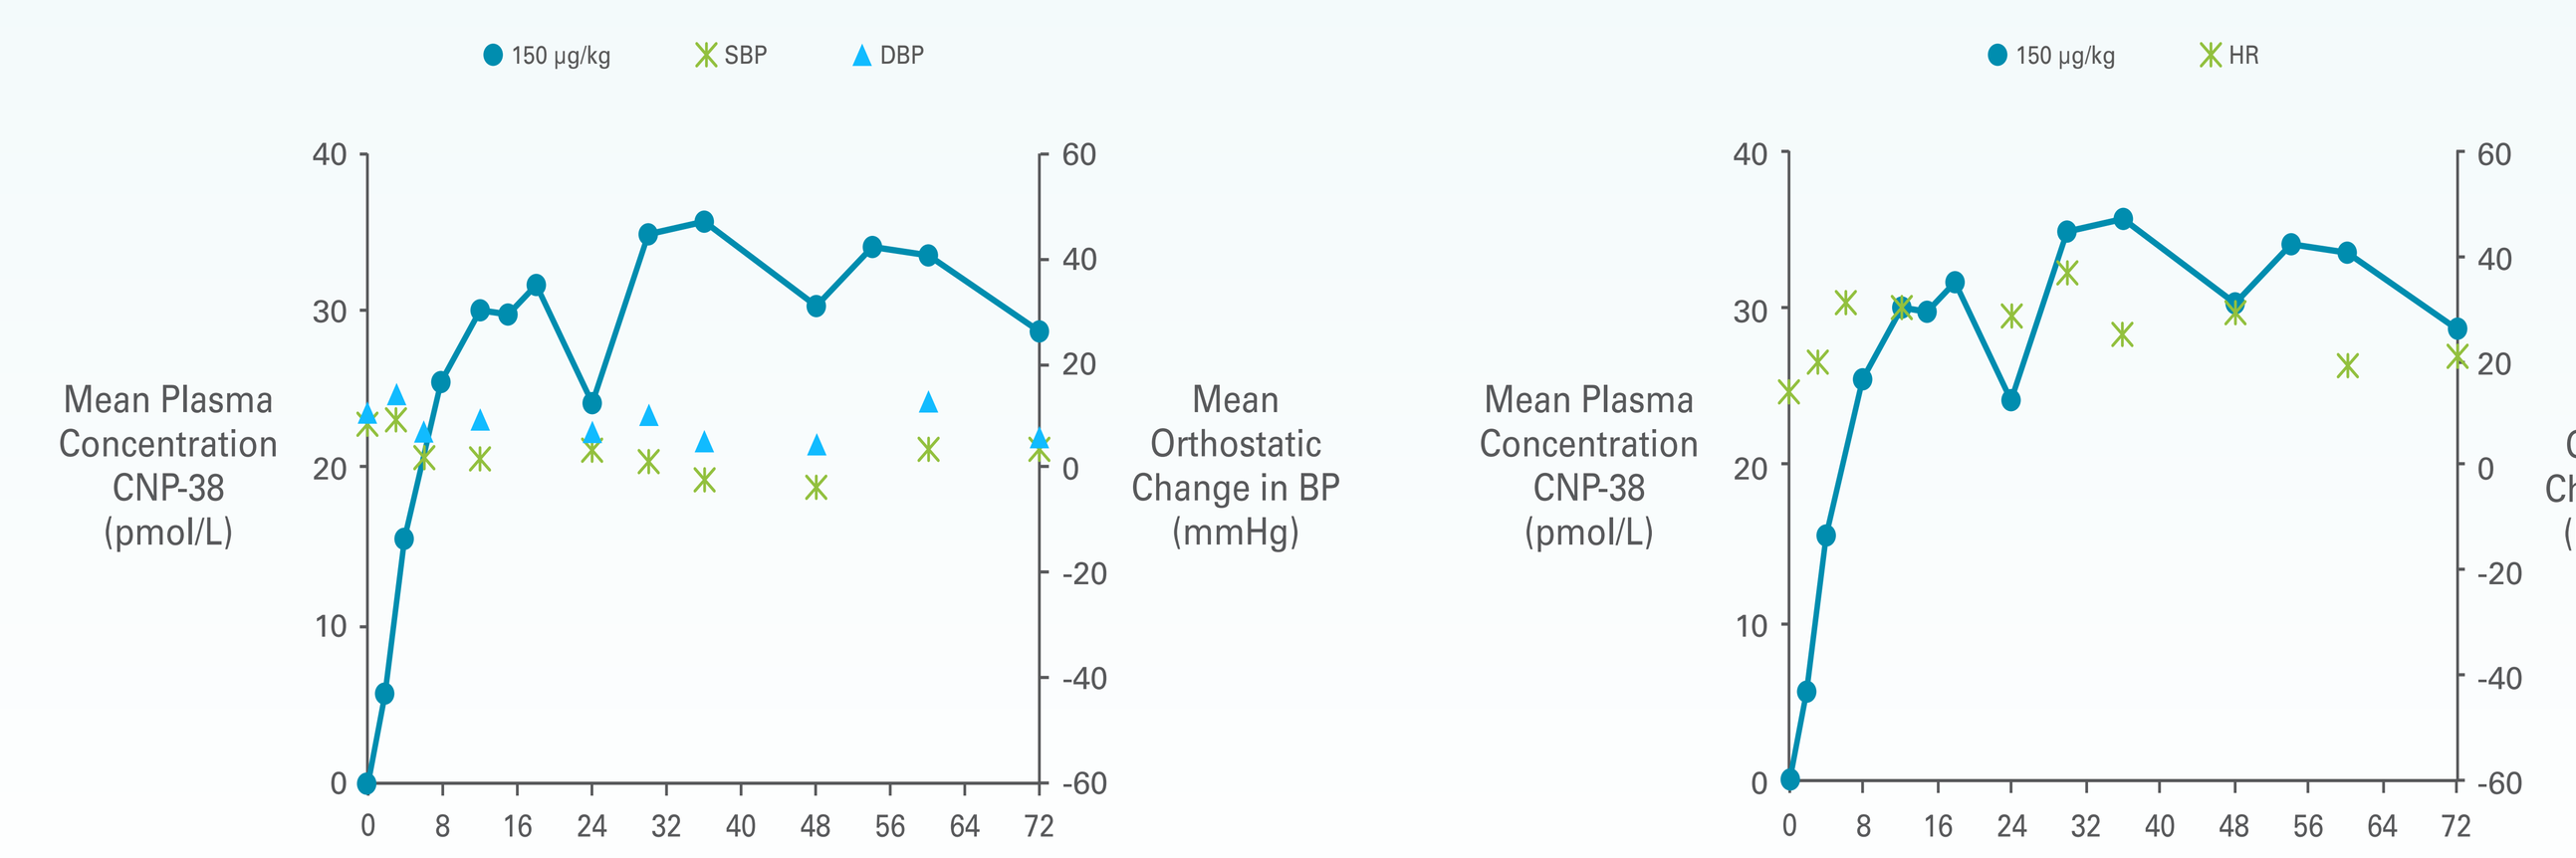
150 µg/kg
SBP
DBP
HR
Mean Plasma Concentration CNP-38 (pmol/L)
Mean Orthostatic Change in BP (mmHg)
Mean Orthostatic Change in HR (beats/min)

*3.0 and 10 µg/kg dose levels are not represented. Data from these cohorts are consistent with placebo. SBP, systolic blood pressure; DBP, diastolic blood pressure; HR, heart rate; pmol/L, picomole/liter

- Assessment of cardiac safety did not reveal any effects on electrocardiogram parameters, including heart rate (HR), PR interval, QRS complex, and QTcF intervals (not shown)
- TransCon CNP was well-tolerated and no serious treatment emergent adverse events or discontinuations were reported
- No clinically relevant dose-dependent effects in mean orthostatic BP or HR changes were observed across treatment groups

METHODS

Figure 8. TransCon CNP: Phase 2 Trial Design

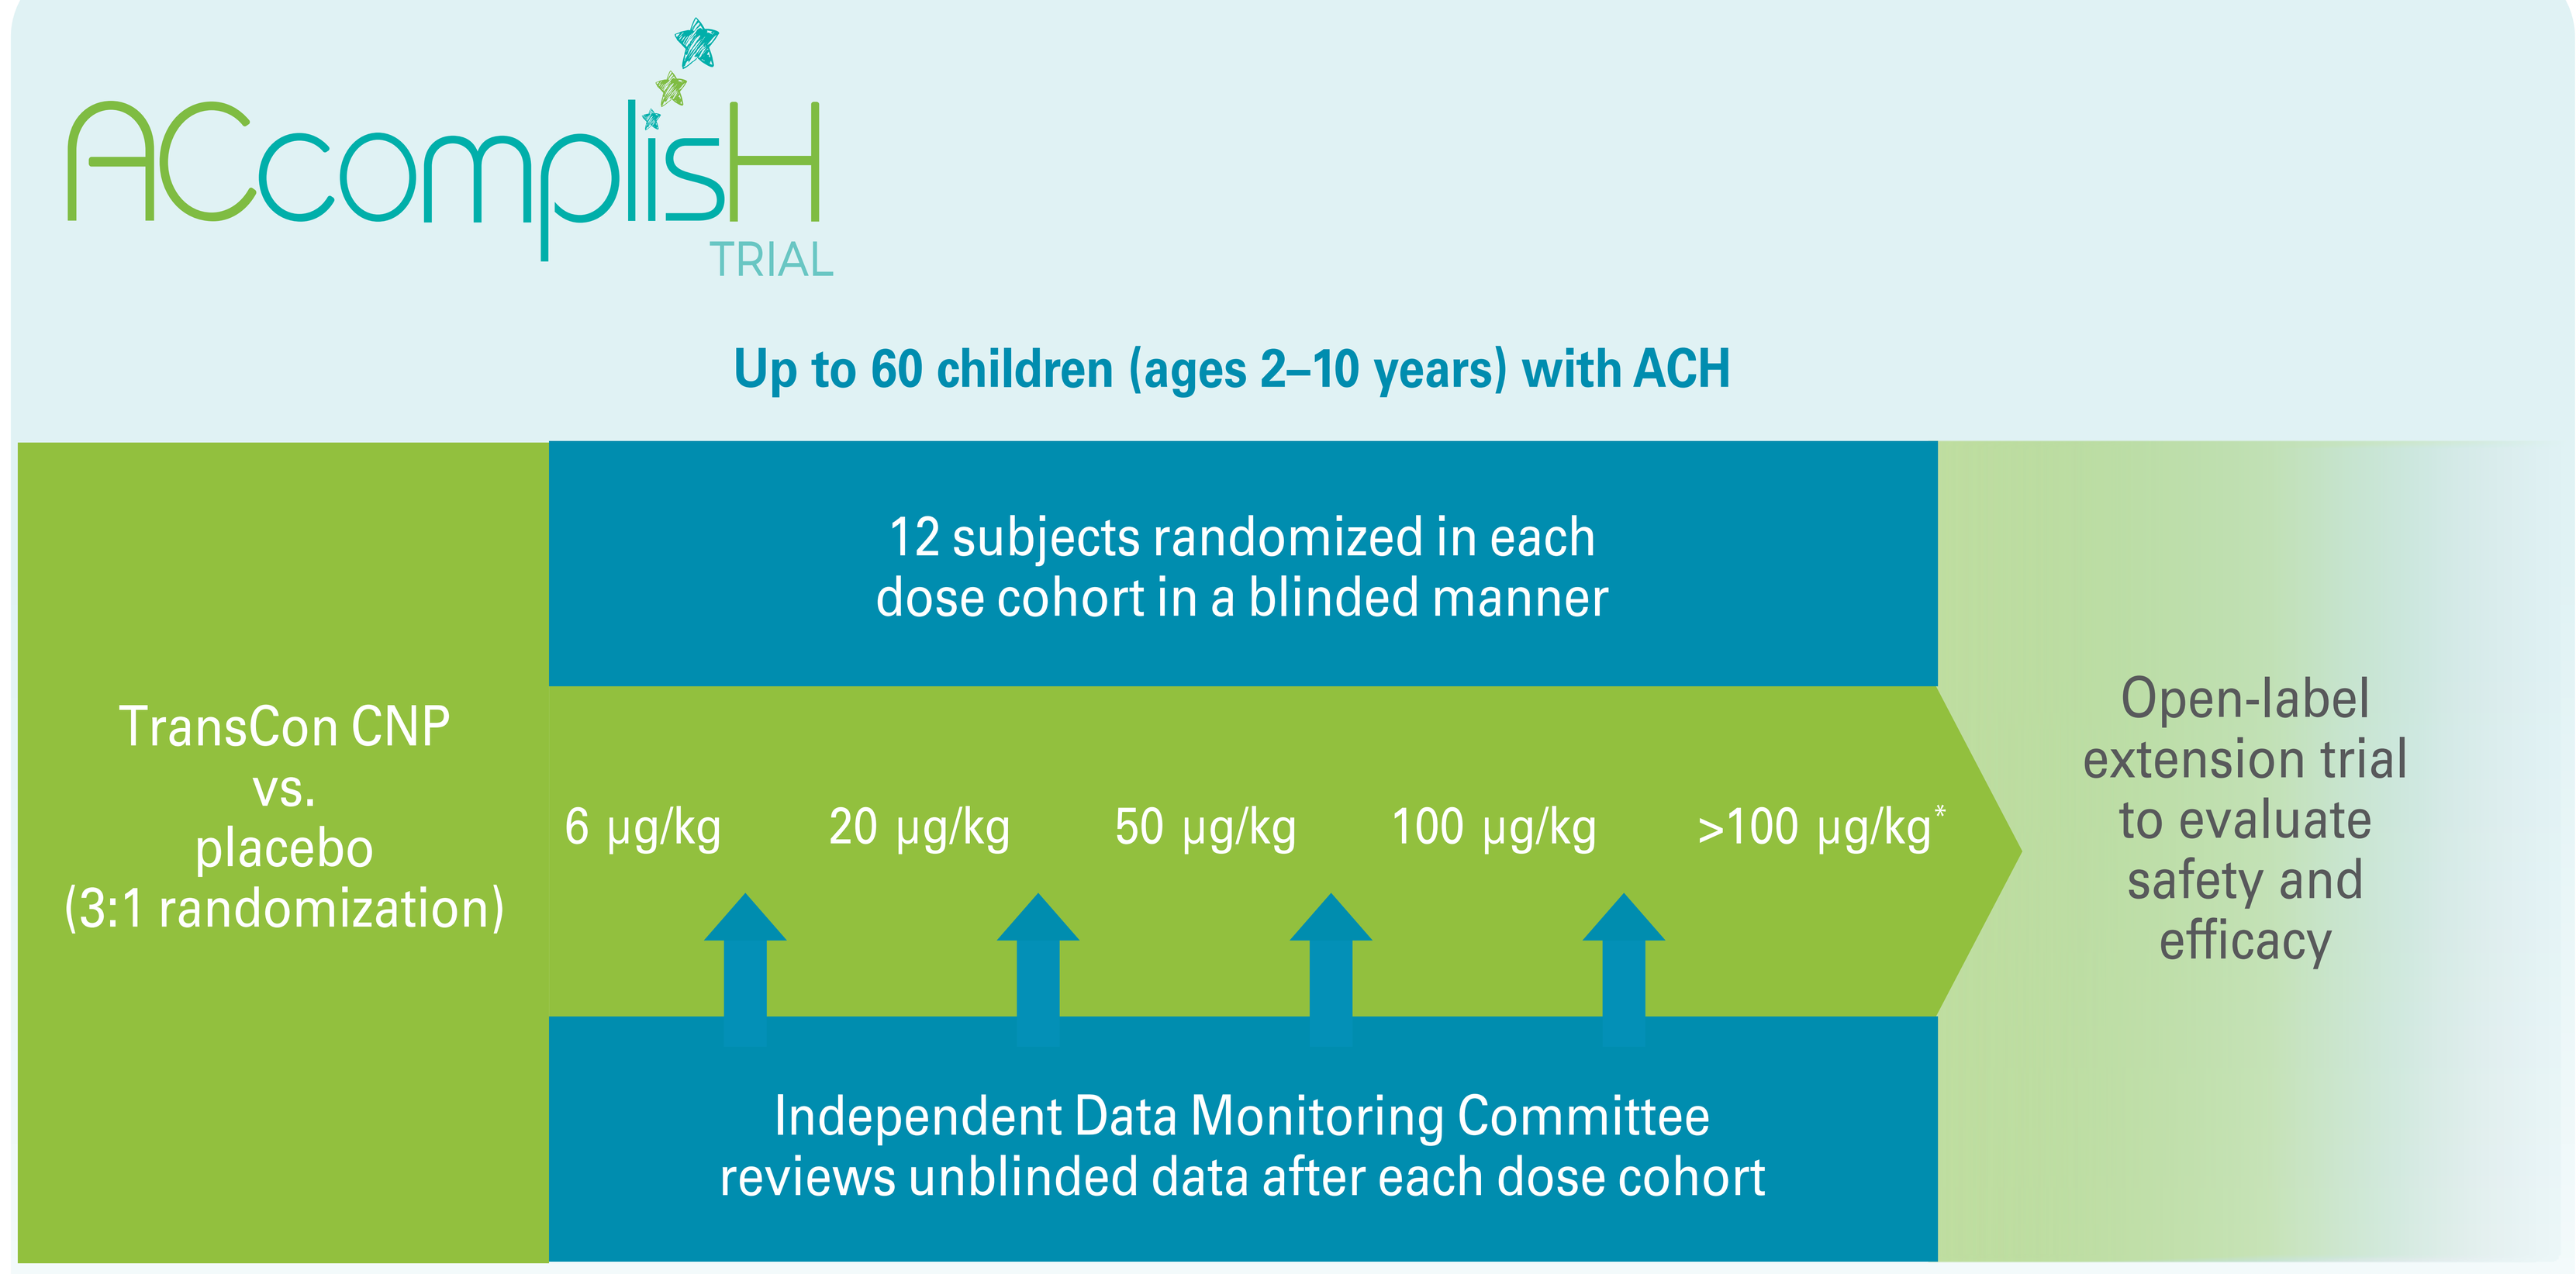
ACcomplisH TRIAL
Up to 60 children (ages 2–10 years) with ACH
TransCon CNP vs. placebo (3:1 randomization)
12 subjects randomized in each dose cohort in a blinded manner
6 µg/kg
20 µg/kg
50 µg/kg
100 µg/kg
>100 µg/kg*
Independent Data Monitoring Committee reviews unblinded data after each dose cohort
Open-label extension trial to evaluate safety and efficacy

*Dose to be determined. If needed, based on emerging data.

- The phase 2 ACcomplisH trial of TransCon CNP in children with ACH will be conducted at approximately 20 sites worldwide
- The primary objective is to evaluate the safety and efficacy of once-weekly TransCon CNP compared to placebo at 12 months in prepubertal ACH children, aged 2-10 years old. Approximately 60 subjects will be randomized 3:1 to receive either weekly TransCon CNP or placebo
- Following screening and verification of clinical ACH with genetic confirmation, subjects will remain within the dose cohort for 52 weeks of treatment. Four staggered parallel cohorts will receive increasing doses of 6 to 100 µg TransCon CNP/kg/week, with an optional 5th cohort at up to 200 µg CNP/kg/week. An independent Data Monitoring Committee will review safety data prior to initiation of a higher dose cohort
- All subjects will continue in an open label-extension trial after Week 52

Primary Efficacy Endpoint

- Annualized height velocity (AHV), as measured after 52 weeks of weekly TransCon CNP treatment or placebo

Secondary Efficacy Endpoint

- Change in upper to lower body segment ratio as measured at 52 weeks of weekly TransCon CNP treatment or placebo

Safety Endpoints

- Incidence of adverse events (AE), lab assessments, vital signs, electrocardiogram, radiographic imaging, and CNP-38 antibodies

Other Exploratory Endpoints

- AHV over time throughout the Open-Label Extension Period, change in upper to lower body segment ratio over time throughout the Open-Label Extension Period, potential biomarkers of pharmacodynamic response to treatment with TransCon CNP, changes in ratio of upper arm to forearm and upper leg to lower leg, anthropometric measurements
- Pharmacokinetics, patient-reported outcome (PRO) and observer-reported outcome (ObsRO) measures

KEY INCLUSION CRITERIA

- Clinical diagnosis of ACH with genetic confirmation
- Age between 2 to10 years old (inclusive) at Screening Visit
- Prepubertal (Tanner Stage 1 breasts for girls or testicular volume < 4mL for boys) at Screening Visit
- Able to stand without assistance
- Caregiver willing and able to administer subcutaneous injections of study drug

KEY EXCLUSION CRITERIA

- Clinically significant findings at Screening that:
 - are expected to require surgical intervention during participation in the trial or
 - are musculoskeletal in nature, such as Salter-Harris fractures and severe hip pain or
 - otherwise are considered by investigator or Medical Monitor/ Medical Expert to make a participant unfit to receive study drug or undergo trial related procedures
- Have received treatment (> 3 months) with human growth hormone or other medications known to affect stature or body proportionality at any time
- Have received any dose of medications intended to affect stature or body proportionality within the previous 6 months of Screening Visit
- Have received any study drug or device intended to affect stature or body proportionality at any time
- History or presence of injury or disease of the growth plate(s), other than ACH, that affects growth potential of long bones

CONCLUSIONS

CONCLUSIONS AND STUDY STATUS

- CNP is targeted specifically to the underlying pathology of ACH through inhibition of FGFR3
- TransCon CNP is designed to provide sustained exposure of CNP
- The Phase 2 ACcomplisH trial is designed to assess the efficacy, safety, and PK of TransCon CNP by weekly administration compared to placebo. This trial will provide valuable long-term data beyond 52 weeks and include open-label extension. The trial is currently ongoing

References

1. Horton WA, et al. *Lancet.* 2007;370:162-72; **2.** Rousseau F, et al. *Nature.*1994;371(6494):252-4; **3.** Bocciardi R, et al. *Hum Mutat.* 2007l;28(7):724-31; **4.** ClinicalTrials.gov identifier NCT04085523. https://clinicaltrials.gov/ct2/show/NCT04085523. Accessed February 22, 2021; **5.** Savarirayan R, et al. *Lancet.* 2020;396(10252):684-692; **6.** Viuff D, et al. Presented at: 9th Annual Meeting of the International Conference on Children's Bone Health; June 22-25, 2019; Salzburg, Austria; **7.** Ota S, et al. Presented at: 14th Annual Meeting of the International Skeletal Dysplasia Society; Sept 11-14, 2019; Oslo, Norway; **8.** Ireland PJ, et al. *Appl Clin Genet.* 2014;7:117-125.; **9.** Pfeiffer KM, et al. *Am J Med Genet.* 2020;1-13; **10.** Breinholt VM et al. J Pharmacol Exp Ther. 2019;370(3):459-471; **11.** Laederich MB, et al. *Curr Opin Pediatr.* 2010; 22:516-523.

PosterSessionOnline.com
Presented at: ESPE 2021
Growth and syndromes (to include Turner syndrome)
Ciara Mcdonnell
P2-332
59ESPE