Abstract No. 278

Real-world Data from Electronic Monitoring of Adherence to Growth Hormone Treatment in Children with Growth Disorders: A Descriptive Analysis

Ekaterina Koledova[1], Vincenzo Tornincasa[2], Paula van Dommelen[3]

[1]Endocrinology Global Medical, Safety and CMO, Merck Healthcare KGaA, an affiliate of Merck KGaA, 64293 Darmstadt, Germany

[2]Merck Medical Devices & Services, Ares Trading S.A., an affiliate of Merck KGaA, 1267 Coinsins, Switzerland

[3]The Netherlands Organization for Applied Scientific Research TNO, Leiden, The Netherlands

INTRODUCTION

- Several factors can detrimentally affect the motivation of patients treated with recombinant human growth hormone (r-hGH).[1,2]
- While traditional approaches to adherence monitoring rely on incidental sources or proxy methods,[3] new eHealth-based ecosystems with automatic adherence recording and data transmissions allow proactive close monitoring of adherence and provide targeted support for individuals and groups of patients.[4,5]
- Although many existing eHealth applications aim to address non-adherence they do not deliver the full potential of an adherence decision support system (ADSS). An ADSS is a form of clinical decision support that provides healthcare professionals (HCPs), patients and care providers with knowledge and person-specific information, presented at the appropriate times, to enhance health and health care.[6]
- The easypod™ electromechanical injection device was developed for patients with growth hormone deficiency to meet the requirements for a reliable, easily used, convenient injection device as part of an ADSS.[7]
- The easypod device sits within the easypod connect ecosystem, through which injection data are transmitted to a secure cloud on the internet via a home station, as part of the ADSS (**Figure 1**).
- Most patients using the easypod connect ecosystem maintained an adherence rate ≥80% for >3 years and poor adherence was linked with suboptimal clinical outcomes.[8,9]
- However, data on adherence and engagement with the device in a real-world setting were not available until very recently.

AIMS

- To perform a real-world observational analysis of the easypod connect platform to gather insights about recorded adherence and engagement to treatment in children with growth disorders from the start of treatment up to 48 months and the effect of age and sex on adherence.

METHODS

- This easypod connect ecosystem was used to collect longitudinal data to study adherence patterns in children transmitting >10 injections between January 2007 and February 2019.
- Adherence (mg injected/mg prescribed) was categorised as high (≥85%), intermediate (>56%–<85%) or low (≤56%), assessed at seven time points and explored by puberty status (nominal cut-offs at 10 years for girls and 12 years for boys) and sex.
- Dosage and frequency as per easypod settings were defined by the care team (HCPs) and data transmissions were instigated by the child, their guardian or by HCPs.
- Adherence was recorded for children transmitting data at each time point; no imputation was made for missing data or withdrawal.
- For each adherence cohort, the mean number of transmissions was calculated as a proxy measure of engagement in disease management.
- Descriptive statistics were used to describe differences over time in adherence (low/intermediate/high), by puberty status (pre-pubertal/pubertal), sex and mean transmissions.

RESULTS

- Longitudinal records were available for 13,553 children between January 2007 and February 2019 and different adherence patterns arose from the data analysis.
- Overall, more children had high adherence (N=9,578) than intermediate (N=2,989) or low adherence (N=986).
- The proportion of children with high adherence decreased from 87% (N=12,964) to 65% (N=957) between Months 1 and 48; however, at each time point up to Month 48, more children had high adherence than low/intermediate adherence (**Figure 2**).
- Compared with children in the low/intermediate adherence categories, children with high adherence also had the highest mean number of transmissions (**Table 1**).
- The proportion of children with high adherence overall was slightly higher in girls than in boys (72% [N=5,766]) vs. 69% [N=7,787]); a similar trend was seen at each time point.
- A higher proportion of pre-pubertal children had high adherence compared with pubertal children; this was seen overall and at each time point (**Figure 3**).
- The proportion of pre-pubertal girls with high adherence was greater than the proportion of pubertal girls (80% [N=1,270] vs. 70% [N=4,496]). Similar results were found in boys (79% [N=2,573] vs. 65% [N=5,214]).

DISCUSSION AND CONCLUSIONS

- **This analysis shows the feasibility of monitoring hormone treatment adherence in large-scale ADSS observational databases**
- **In a large real-world setting, we showed that most children treated with r-hGH via the easypod device had high adherence, but this can be affected by factors such as age and puberty**
- **There was a trend for children/parents/guardians who were most engaged with working with the device to transmit data more often and to have high adherence**
- **In common with other real-world research, there may be missing information, high levels of variability in treatment regimens, and there were several confounding or child-specific variables that could influence adherence but were not assessed**
- **Combining easypod real-world adherence data with electronic health records can support the creation of an ADSS that can facilitate the assessment of outcomes and adherence plans to support integrated care**

Table 1. Overall Demographics

	Adherence ≥85% (N=9,578)	Adherence >56%–<85% (N=2,989)	Adherence ≤56% (N=986)
Boys, mean (SD) age, years	12.8 (5.3)	15.0 (6.6)	15.3 (7.6)
Number of boys <12 years / ≥12 years	2026 / 3377	417 / 1384	130 / 453
Girls, mean (SD) age, years	12.2 (5.2)	14.3 (7.9)	15.8 (10.6)
Number of girls <10 years / ≥10 years	1018 / 3157	195 / 993	57 / 346
Mean (SD) number of transmissions	12.5 (24.9)	7.2 (15.3)	3.5 (5.7)
Mean (SD) global adherence duration, days	599 (530)	694 (565)	630 (539)

REFERENCES

1. Hartmann K, et al. *Horm Res Paediatr.* 2013;80:1-5.
2. Rosenfeld R, et al. *Endocr Pract.* 2008;14:143-54.
3. Bozzolo M, et al. *BMC Endocr Disord*. 2011;11:4.
4. Stoyanov G, et al. CPEG; February 18-20; Edmonton, Alberta, Canada: CPEG; 2016
5. Kindig M, et al. *Sci Rep.* 2017;7:3866.
6. HealthIT.gov; https://www.healthit.gov/topic/safety/clinical-decision-support (accessed 27 August 2019).
7. Dahlgren J. *Expert Rev Med Devices*. 2008;5:297-304.
8. Koledova E, et al. *Endocr Connect* 2018;7:914-23.
9. Van Dommelen P, et al. *PLoS ONE* 2018;13:e0206009.

Figure 2. Proportion of Children in Each Adherence Category at Each Time Point

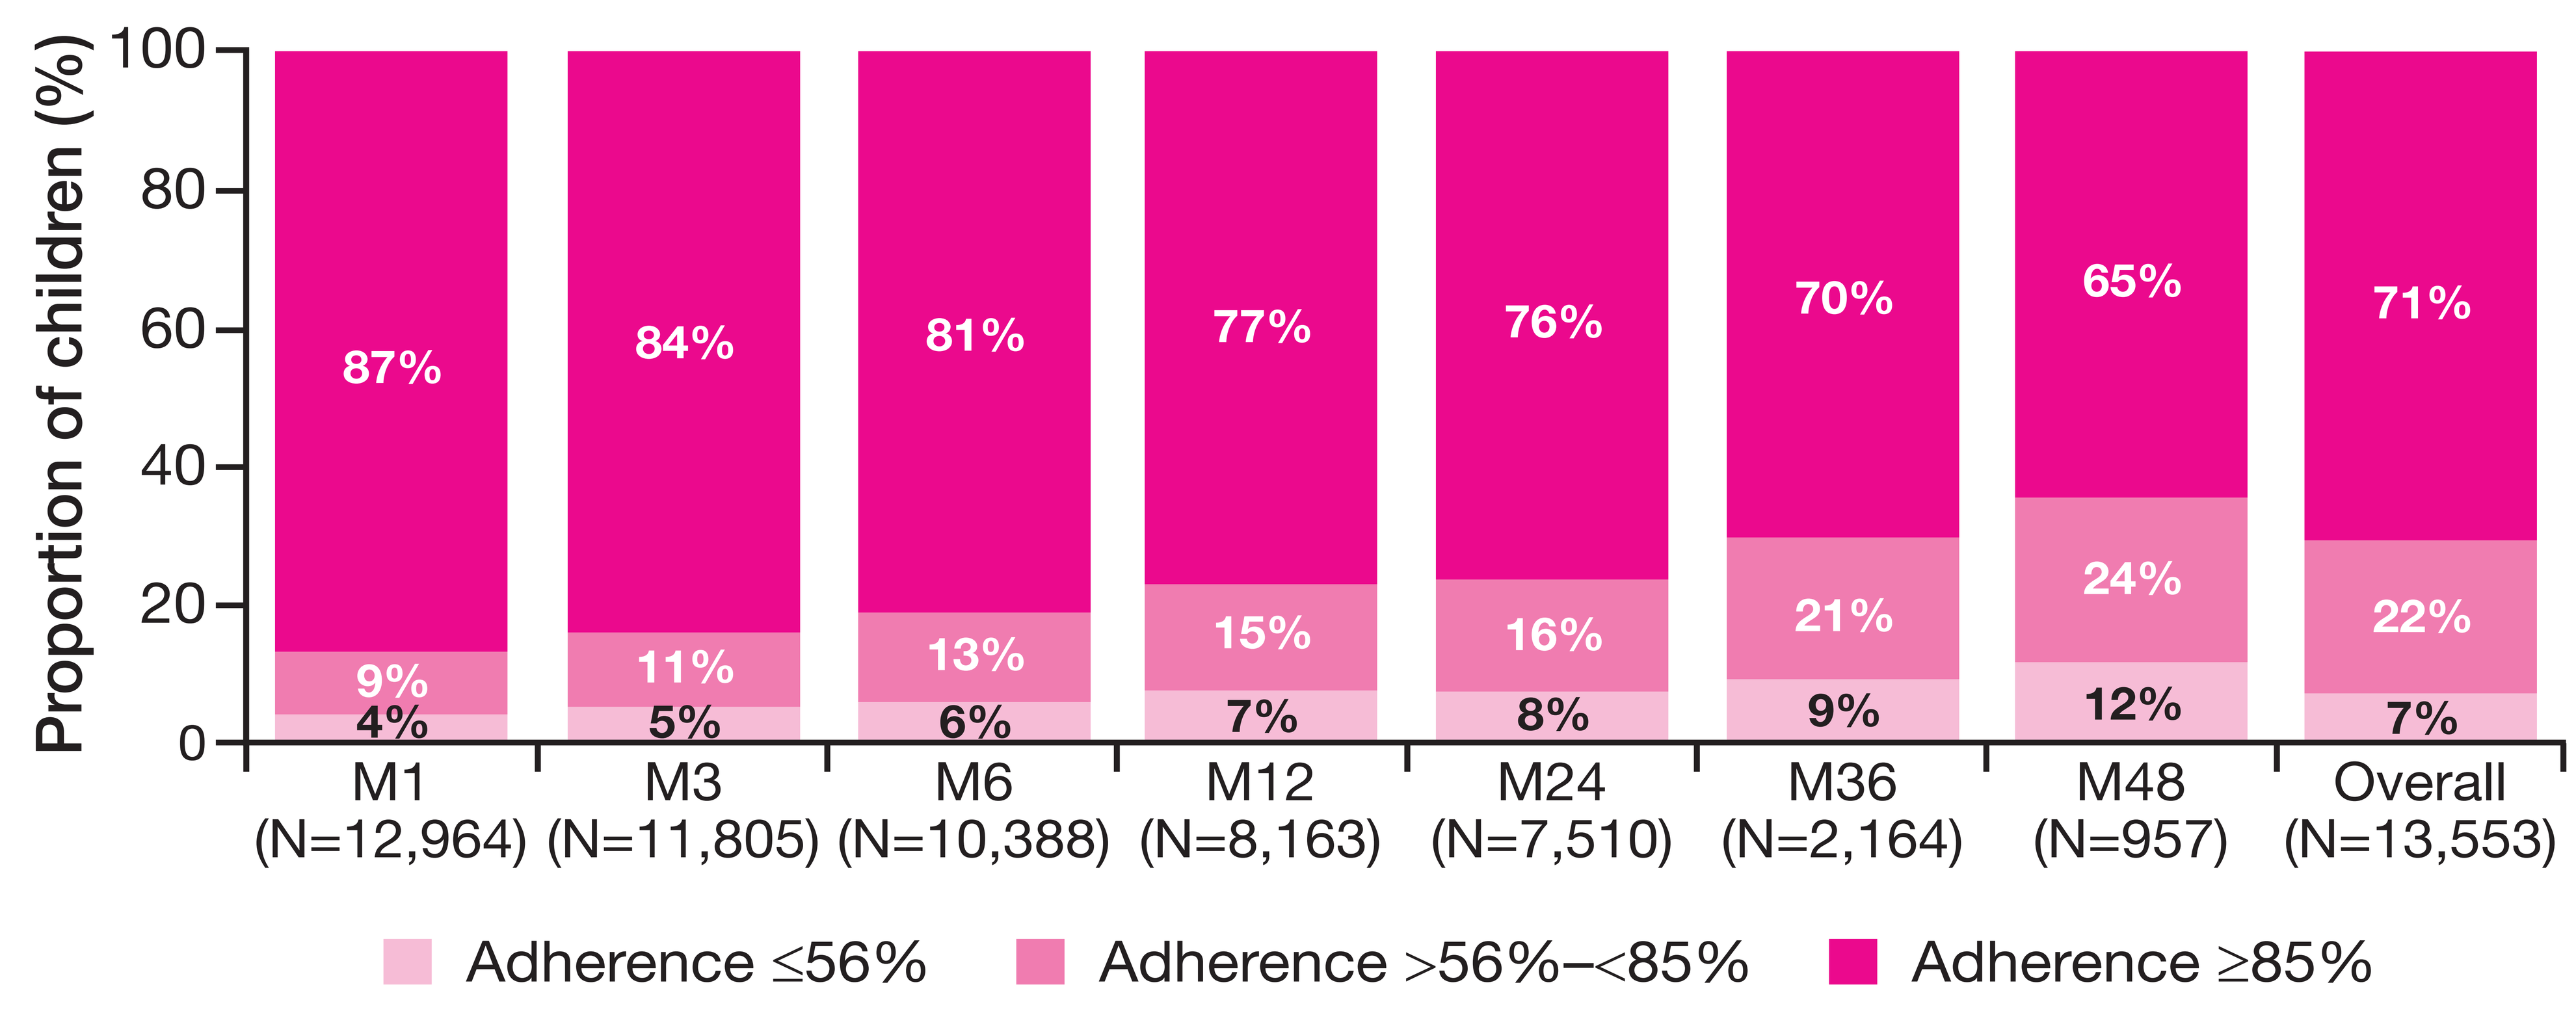

Proportion of children (%)
100
80
60
40
20
0
87%
9%
4%
84%
11%
5%
81%
13%
6%
77%
15%
7%
76%
16%
8%
70%
21%
9%
65%
24%
12%
71%
22%
7%
M1 (N=12,964)
M3 (N=11,805)
M6 (N=10,388)
M12 (N=8,163)
M24 (N=7,510)
M36 (N=2,164)
M48 (N=957)
Overall (N=13,553)
Adherence ≤56%
Adherence >56%–<85%
Adherence ≥85%

Figure 3. Proportion of Children in Each Adherence Category at Each Time Point According to Age

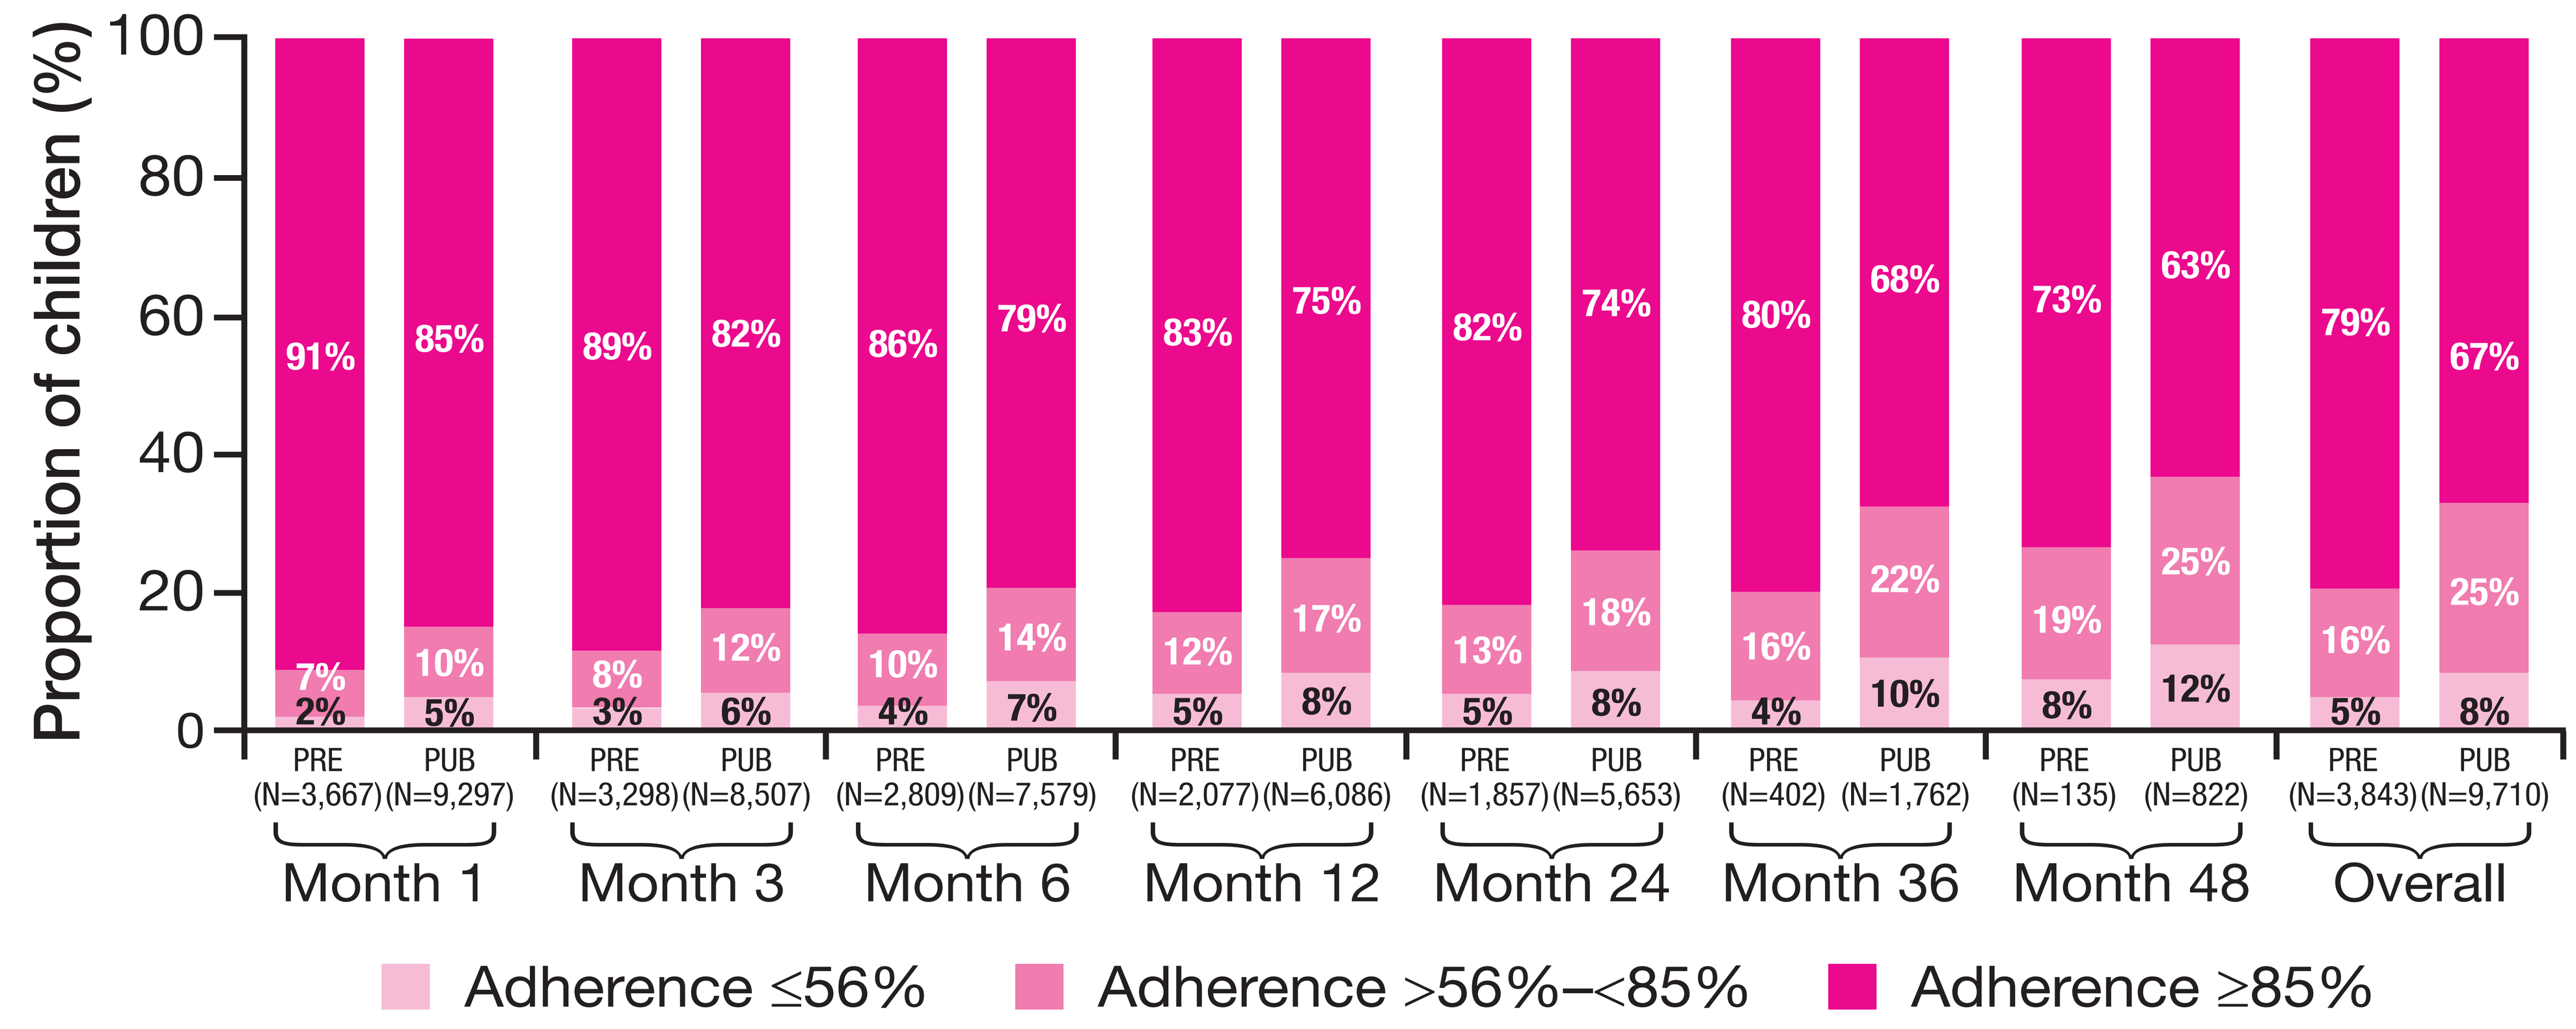

Proportion of children (%)
91% 7% 2%
85% 10% 5%
89% 8% 3%
82% 12% 6%
86% 10% 4%
79% 14% 7%
83% 12% 5%
75% 17% 8%
82% 13% 5%
74% 18% 8%
80% 16% 4%
68% 22% 10%
73% 19% 8%
63% 25% 12%
79% 16% 5%
67% 25% 8%
PRE (N=3,667) PUB (N=9,297)
PRE (N=3,298) PUB (N=8,507)
PRE (N=2,809) PUB (N=7,579)
PRE (N=2,077) PUB (N=6,086)
PRE (N=1,857) PUB (N=5,653)
PRE (N=402) PUB (N=1,762)
PRE (N=135) PUB (N=822)
PRE (N=3,843) PUB (N=9,710)
Month 1
Month 3
Month 6
Month 12
Month 24
Month 36
Month 48
Overall
Adherence ≤56%
Adherence >56%–<85%
Adherence ≥85%

Children categorized according to nominal puberty cut-off ages (10 years for girls and 12 years for boys). PRE, prepubertal. PUB, pubertal.

Figure 1. The easypod connect Ecosystem

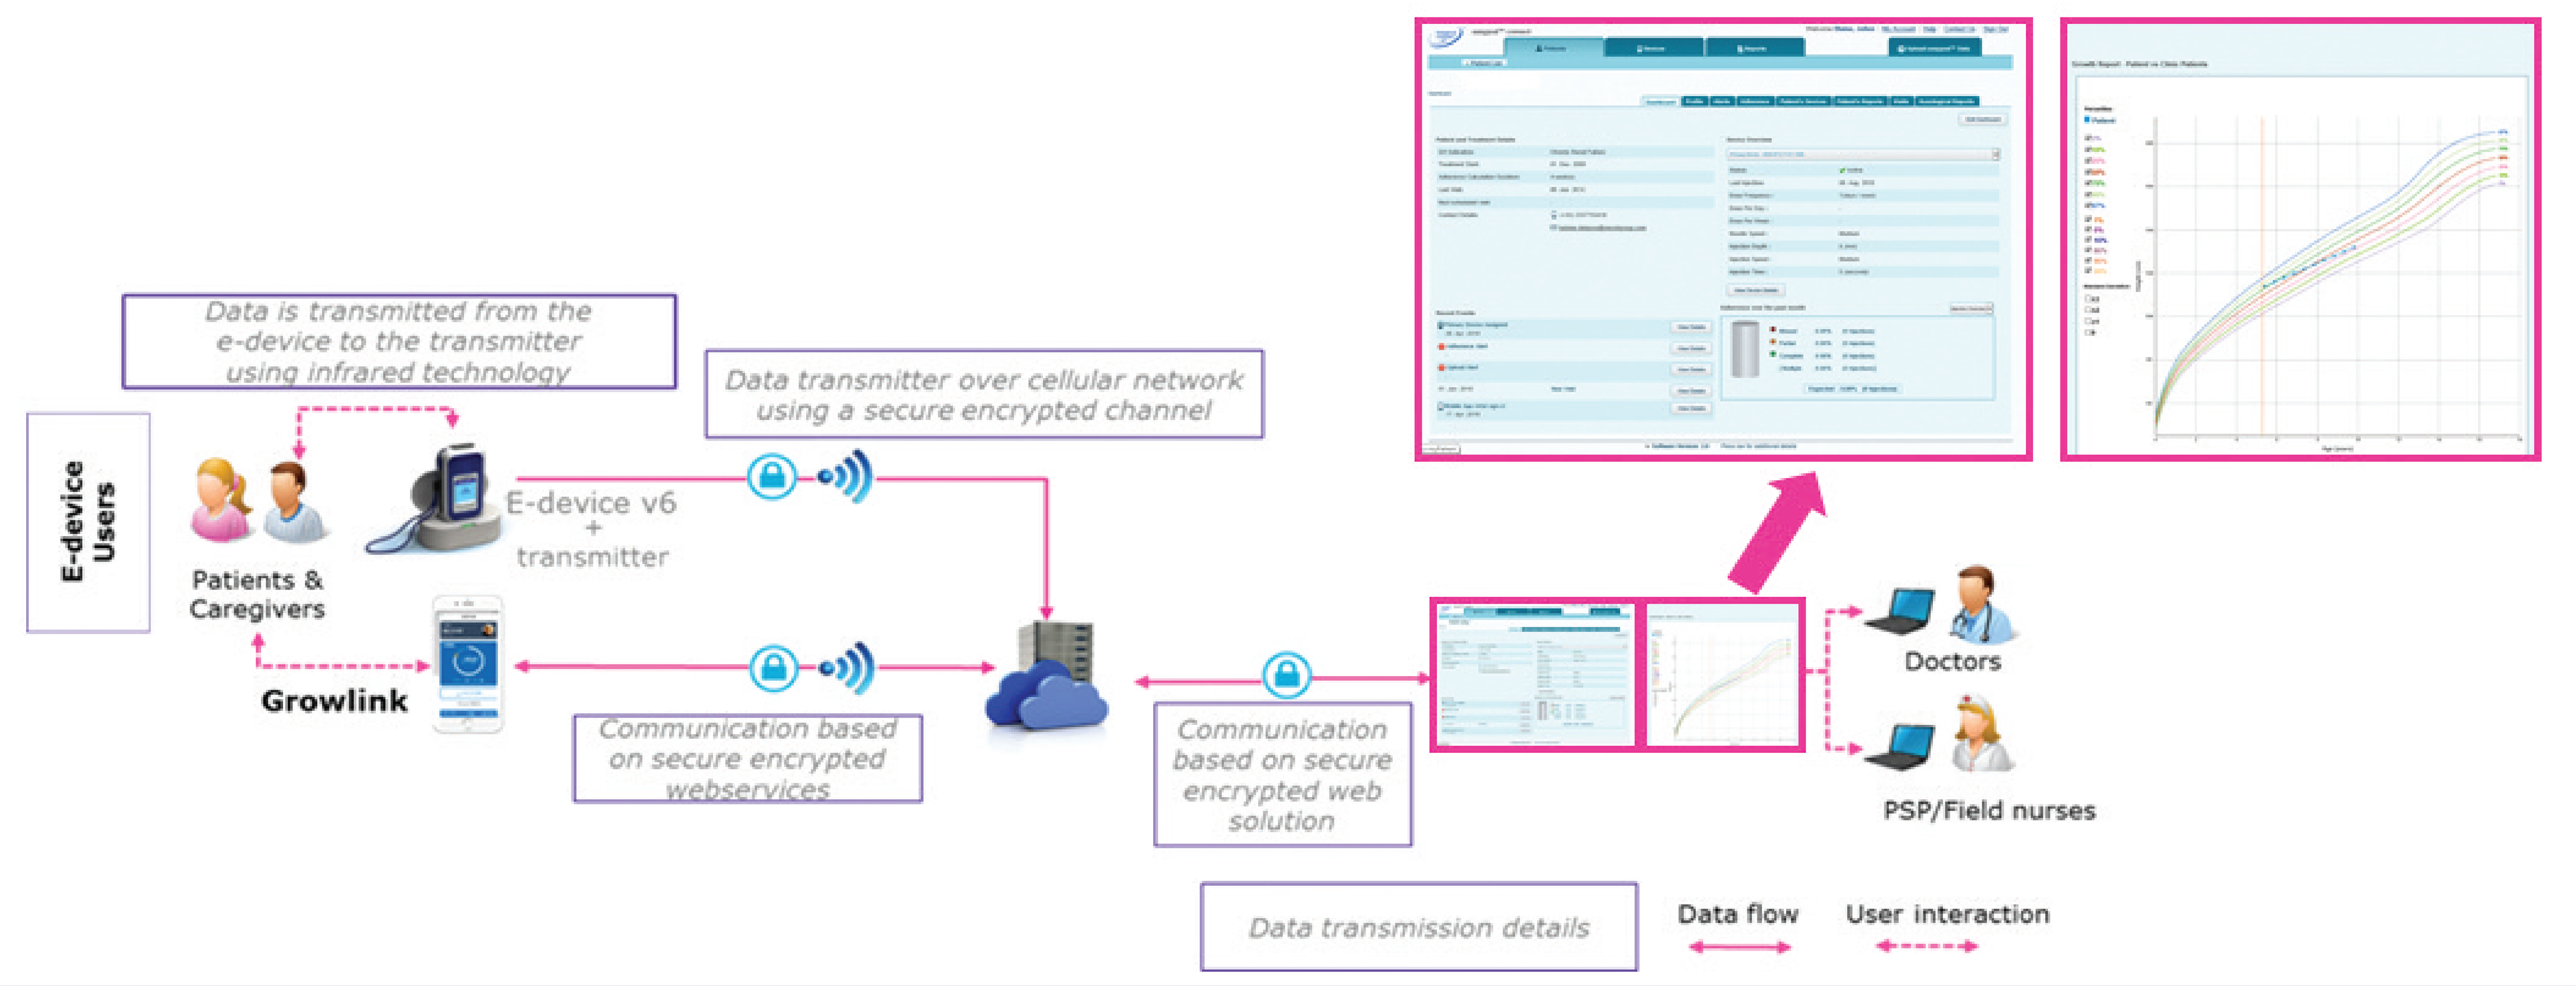

Data is transmitted from the e-device to the transmitter using infrared technology
Data transmitter over cellular network using a secure encrypted channel
E-device Users
Patients & Caregivers
E-device v6 + transmitter
Growlink
Communication based on secure encrypted webservices
Communication based on secure encrypted web solution
Doctors
PSP/Field nurses
Data transmission details
Data flow
User interaction

The easypod connect ecosystem comprises the connected pen device and online portal, as part of an adherence decision support system (ADSS)

ACKNOWLEDGMENTS

The authors would like to thank Jorge Cancela (previously of Ares Trading, an affiliate of Merck KGaA, Darmstadt, Germany) for designing the analysis, interpreting the data and for providing critical comments, and Luis Fernandez (Salumedia, Seville, Spain) for his valuable advice and input on digital therapeutics for monitoring adherence in the real world, Merat Bagha (Geneva, Switzerland) for providing critical comments and Ruben Michael Dezeure (Ares Trading, an affiliate of Merck KGaA, Darmstadt, Germany) for performing the statistical analysis. Medical writing assistance was provided by Steven Goodrick, inScience Communications, UK, and sponsored by Merck Healthcare KGaA, Darmstadt, Germany. The authors would like to acknowledge the children, their families and the healthcare professionals who participated in the analysis.

DISCLOSURES

EK is an employee of Merck Healthcare KGaA, Darmstadt, Germany. VT is an employee of Ares Trading, an affiliate of Merck KGaA, Darmstadt Germany. PvD has a consultancy agreement with Merck Healthcare KGaA, Darmstadt, Germany.

GET POSTER PDF
Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

A Plain English summary of this poster is available, using this QR code

Presented at ESPE 2019, 19-21 September, 2019; Vienna, Austria

58ESPE P1-220 GH and IGFs Ekaterina Koledova

Poster presented at: ESPE 2019 VIENNA

PosterSessionOnline.com