Oral Administration of CRN04777, a Nonpeptide Selective SST5 Agonist, Suppresses Insulin Secretion and Rescues Hypoglycemia in Neonatal Rat Models of Congenital Hyperinsulinism

Melissa A. Fowler, Jian Zhao, Emmanuel Sturchler, Jon Athanacio, Taylor A. Kredel, Agnes Antwan, Claudia Yan, Christine Staley, Jenny Chen, Ana K. Kusnetzow, R. Scott Struthers, Yun Fei Zhu, Stephen F. Betz, Stacy Markison

Crinetics Pharmaceuticals, San Diego, CA.

CRN04777 is an orally administered nonpeptide that is a potent and selective agonist of somatostatin 5 (SST5) receptors and is currently under development for the treatment of congenital hyperinsulinism (HI), the most common cause of persistent hypoglycemia in newborns and infants. Congenital HI arises from mutations within the insulin secretion pathway and is characterized by excessive and/or inappropriate insulin secretion by pancreatic islet beta cells. This excess insulin can result in profound hypoglycemia which can lead to neurological impairment, coma, and even death if not treated promptly.

The neuropeptide somatostatin is an important modulator of hormonal signaling in the pancreas, and SST5 activation has been shown to suppress insulin and raise blood glucose, thus providing a potential treatment mechanism for patients with congenital HI. We characterized the ability of CRN04777 to raise blood glucose and lower plasma insulin in preclinical models using neonatal (PND7-11) rats.

Oral administration of CRN04777 in neonatal rats suppressed insulin secretion and dose-dependently increased blood glucose levels for up to 6 hours after a single dose. Furthermore, the CRN04777 mediated increase in blood glucose was sustained over 5 days of repeated oral administration.

The most common mutations underlying congenital HI are found in the genes encoding the ATP-sensitive potassium (K_{ATP}) channel, which controls the membrane potential of pancreatic beta cell, and children with K_{ATP}-dependent HI are typically unresponsive to diazoxide, the only FDA-approved drug for HI. Sulfonylurea drugs, such as glyburide, close K_{ATP} channels and can be used to induce a hyperinsulinemic hypoglycemic state in rats that mimics some of the characteristics of K_{ATP}-HI. As expected, a single administration of glyburide in neonatal rats induced a pronounced increase in plasma insulin levels, with a concomitant decrease in blood glucose that persisted for several hours. Oral administration of CRN04777 rescued the glyburide-induced hyperinsulinemic hypoglycemia, lowering insulin levels and restoring blood glucose levels to >60 mg/dL.

These findings provide evidence that CRN04777 is an efficacious nonpeptide SST5 agonist that suppresses insulin and increases blood glucose in neonatal rats. In addition, oral administration of CRN04777 rescued hypoglycemia in a neonatal rat model of congenital HI, demonstrating its potential as an oral therapeutic for children with congenital HI.

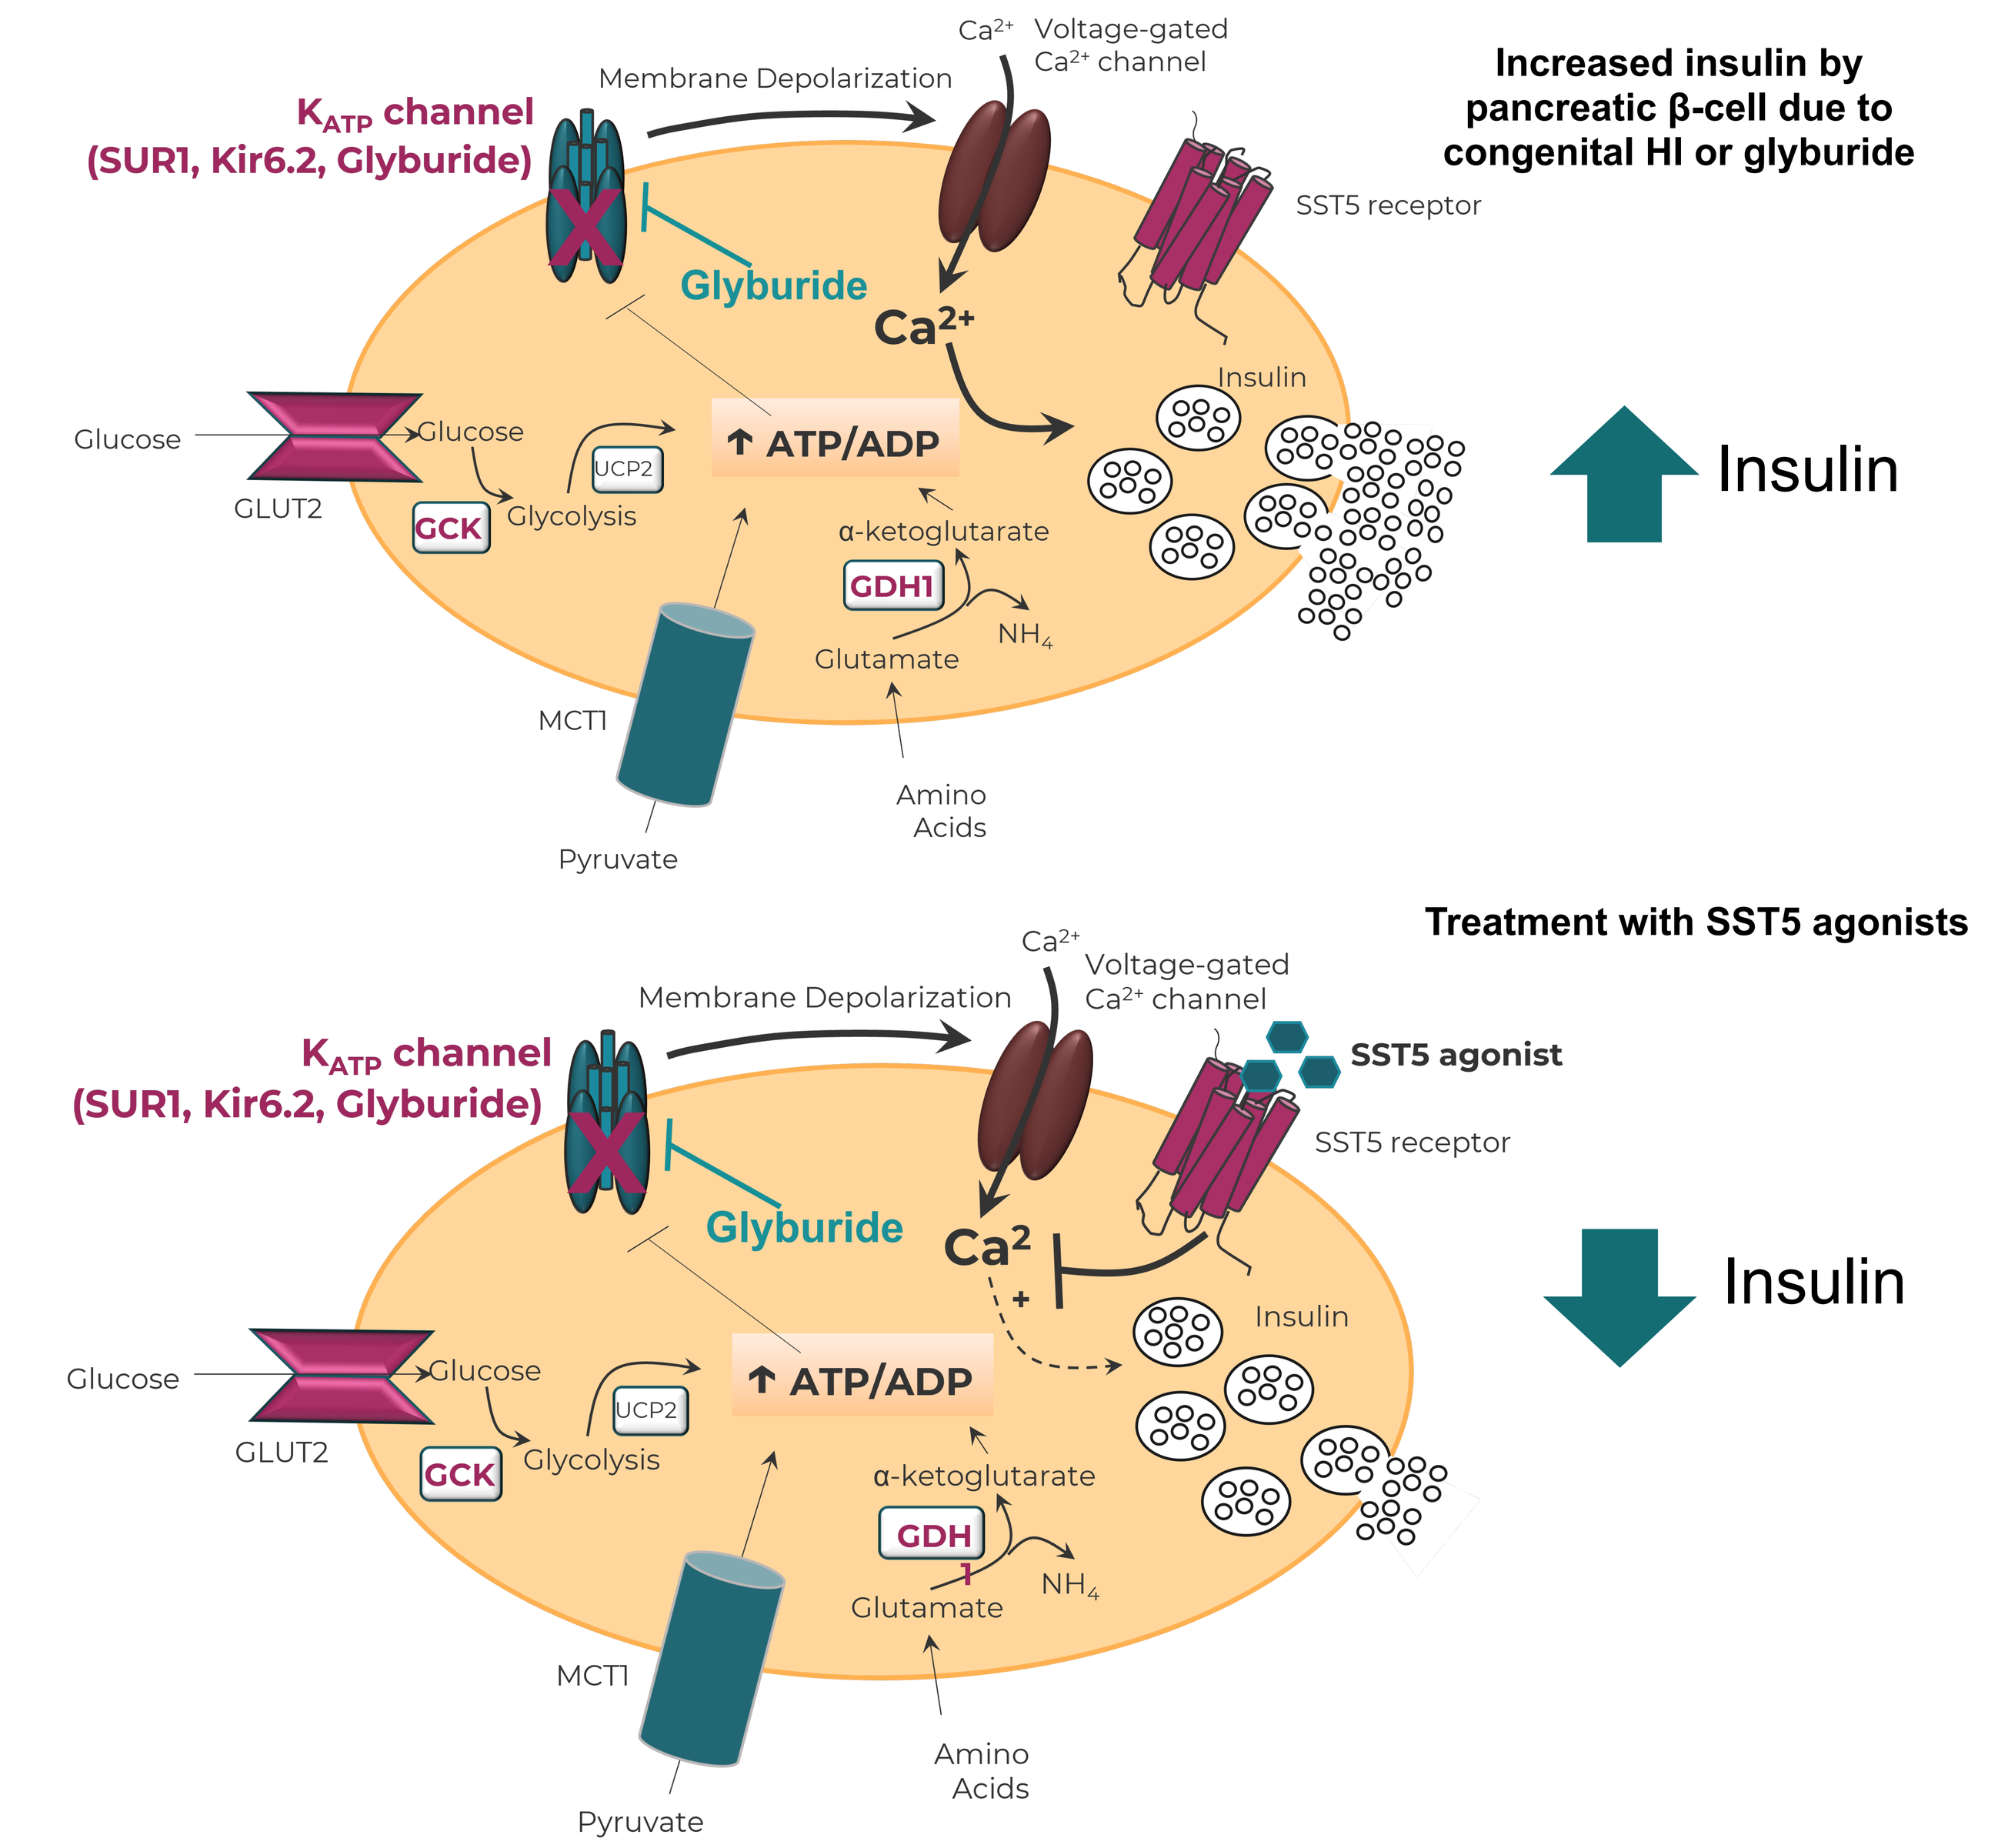
Increased insulin by pancreatic β-cell due to congenital HI or glyburide
K_{ATP} channel (SUR1, Kir6.2, Glyburide)
Membrane Depolarization
Voltage-gated Ca^{2+} channel
SST5 receptor
Glyburide
Ca^{2+}
Insulin
Glucose
GLUT2
GCK
Glycolysis
UCP2
↑ ATP/ADP
α-ketoglutarate
GDH1
NH_4
Glutamate
MCT1
Pyruvate
Amino Acids
Insulin
Treatment with SST5 agonists
SST5 agonist
Insulin

Figure 1. Treatment with SST5 agonists inhibits abnormal insulin secretion from pancreatic β-cells in congenital HI. The most common mutations in congenital HI are highlighted (GCK, GDH1, K_{ATP} channel- SUR1 & Kir6.2). Sulfonylurea drugs, such as glyburide, close K_{ATP} channels inducing insulin secretion and mimicking some of the characteristics of K_{ATP}-HI.

CRN04777 is a Potent and Selective SST5 Agonist

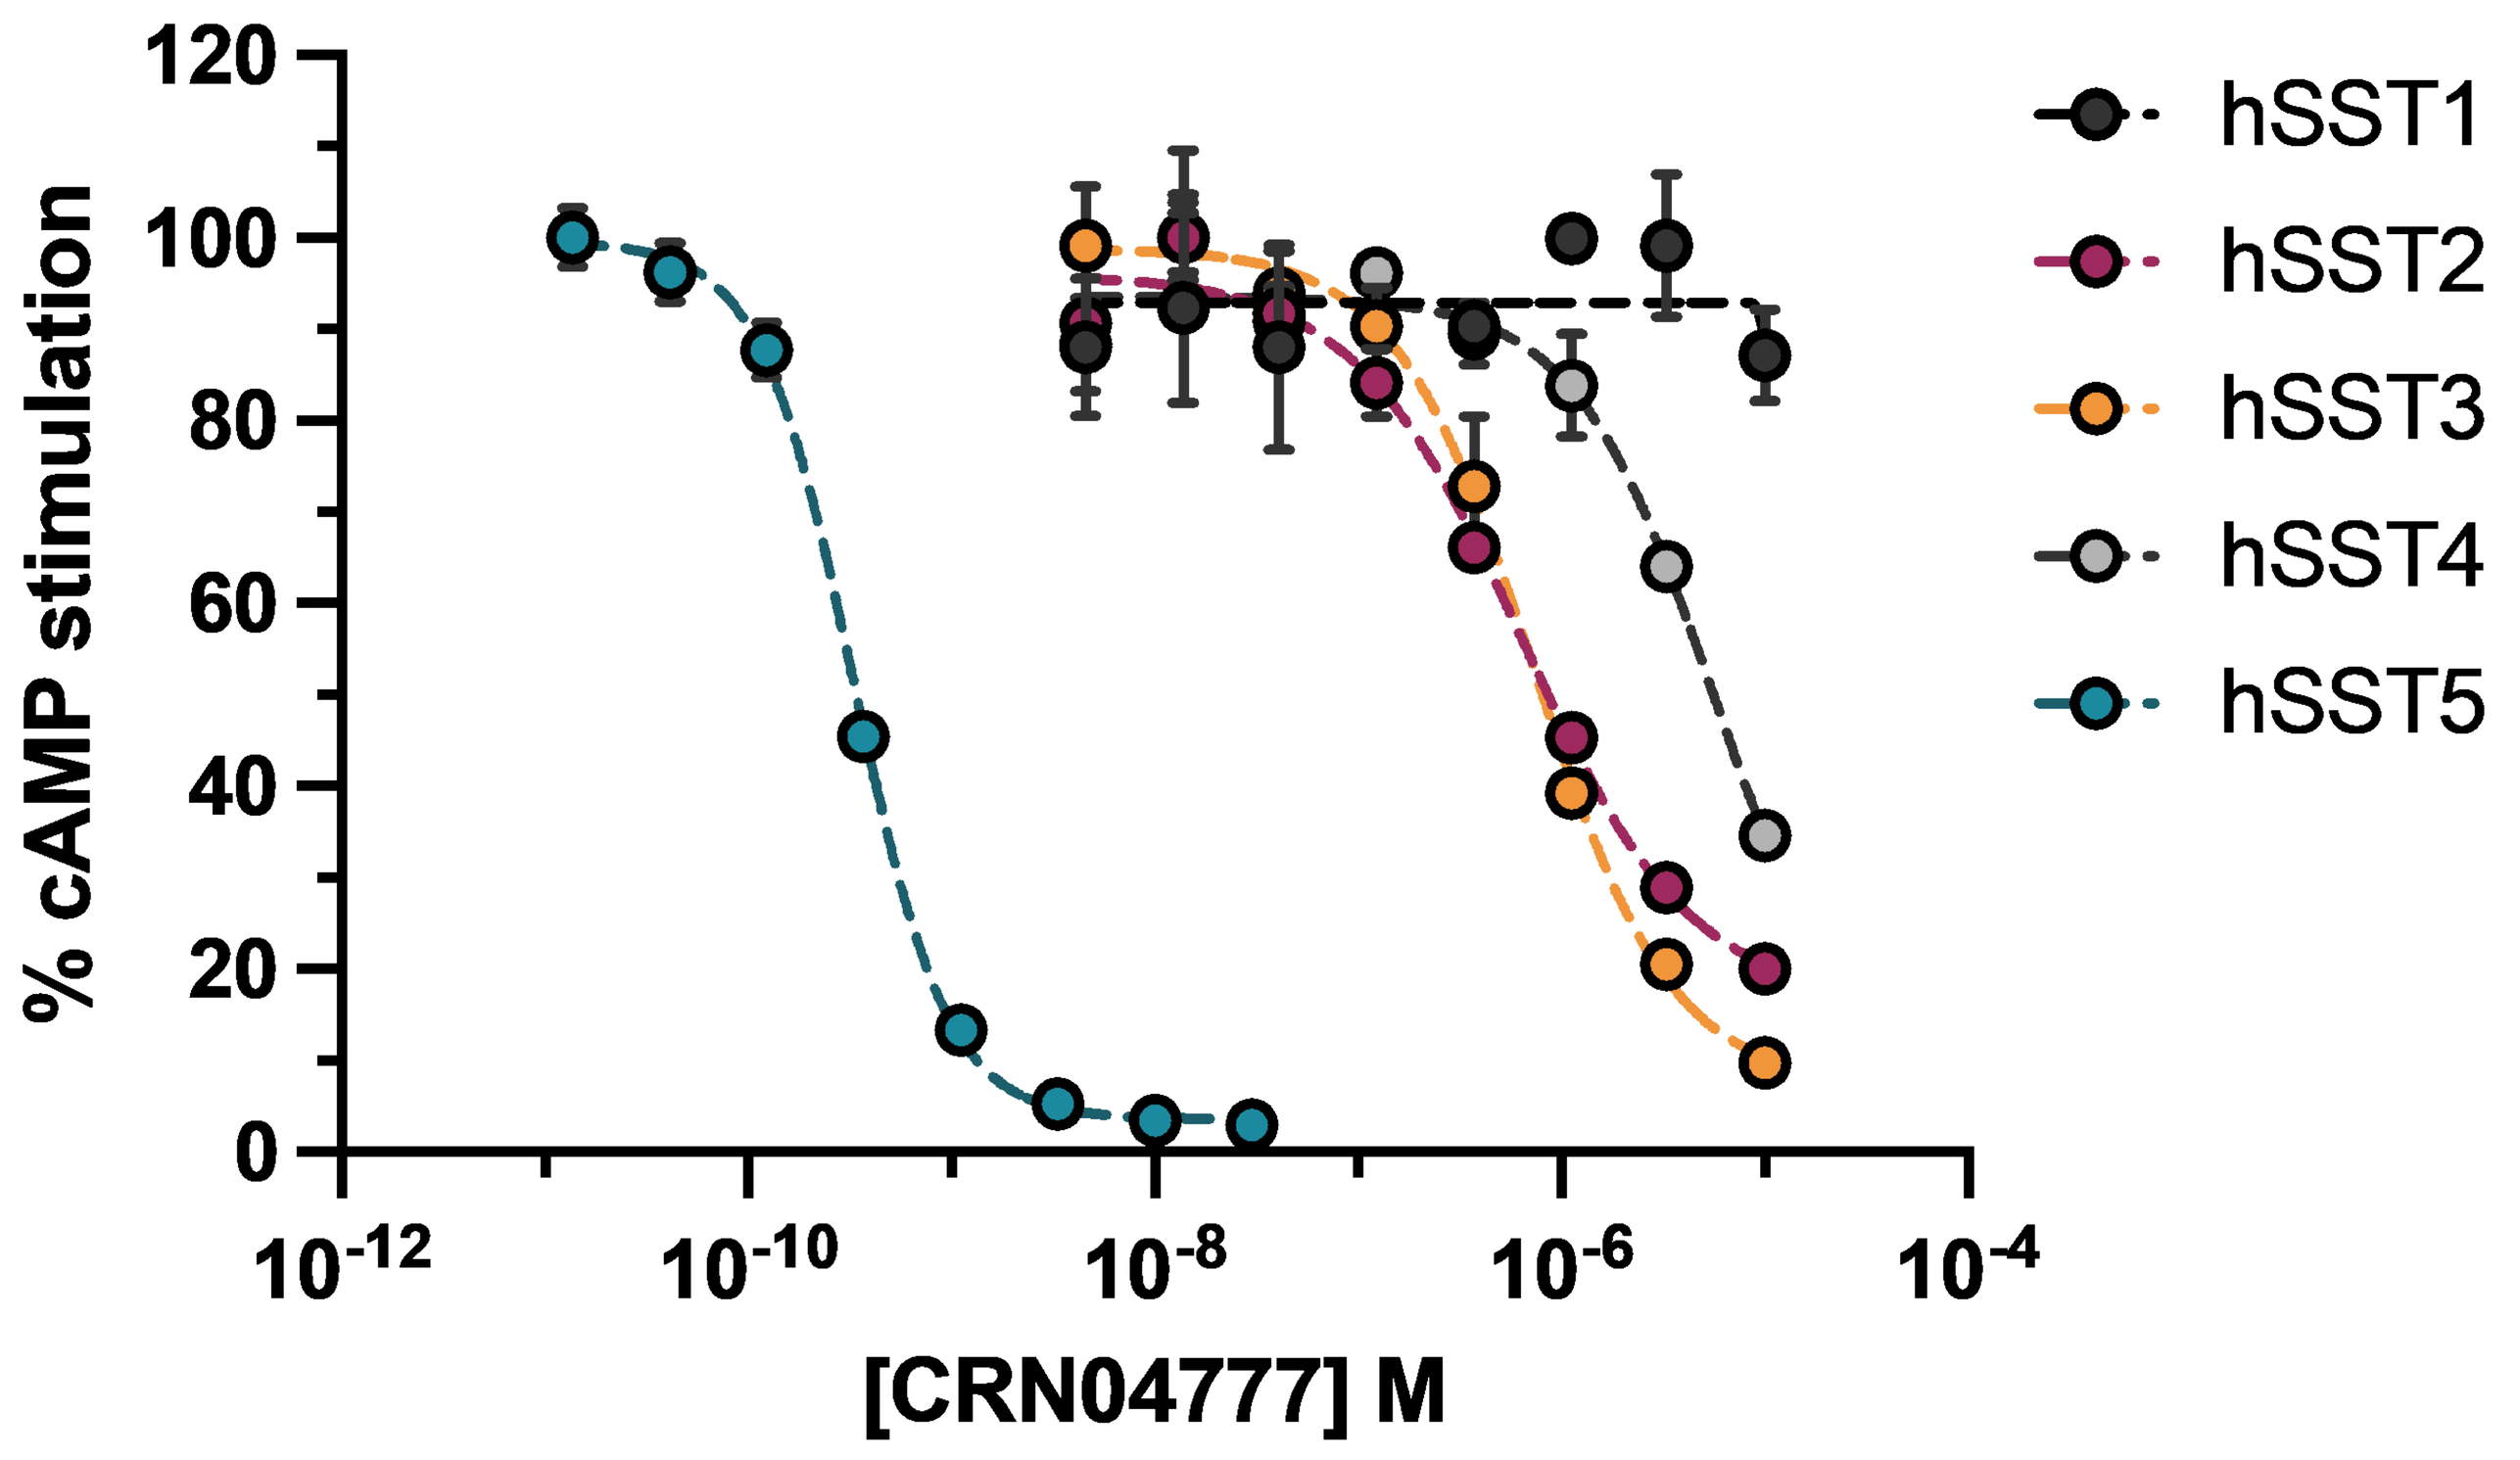
% cAMP stimulation
[CRN04777] M
hSST1
hSST2
hSST3
hSST4
hSST5

Avg. EC_{50} (nM) SST5				
Human	Mouse	Rat	Dog	Monkey
0.41	0.50	6.2	3.6	0.87

Figure 2. Graph shows a comparison of activity of CRN04777 at all five human SST receptor subtypes in cell-based assays that measure the activation of G_i and subsequent decrease in cAMP levels. Dose response curves are shown from individual representative experiments. The table shows the activity of CRN04777 at SST5 derived from human, mouse, rat, dog, and monkey.

CRN04777 Dose Dependently Increases Blood Glucose in Male Neonatal Rats For Up to 6 Hours After a Single Dose

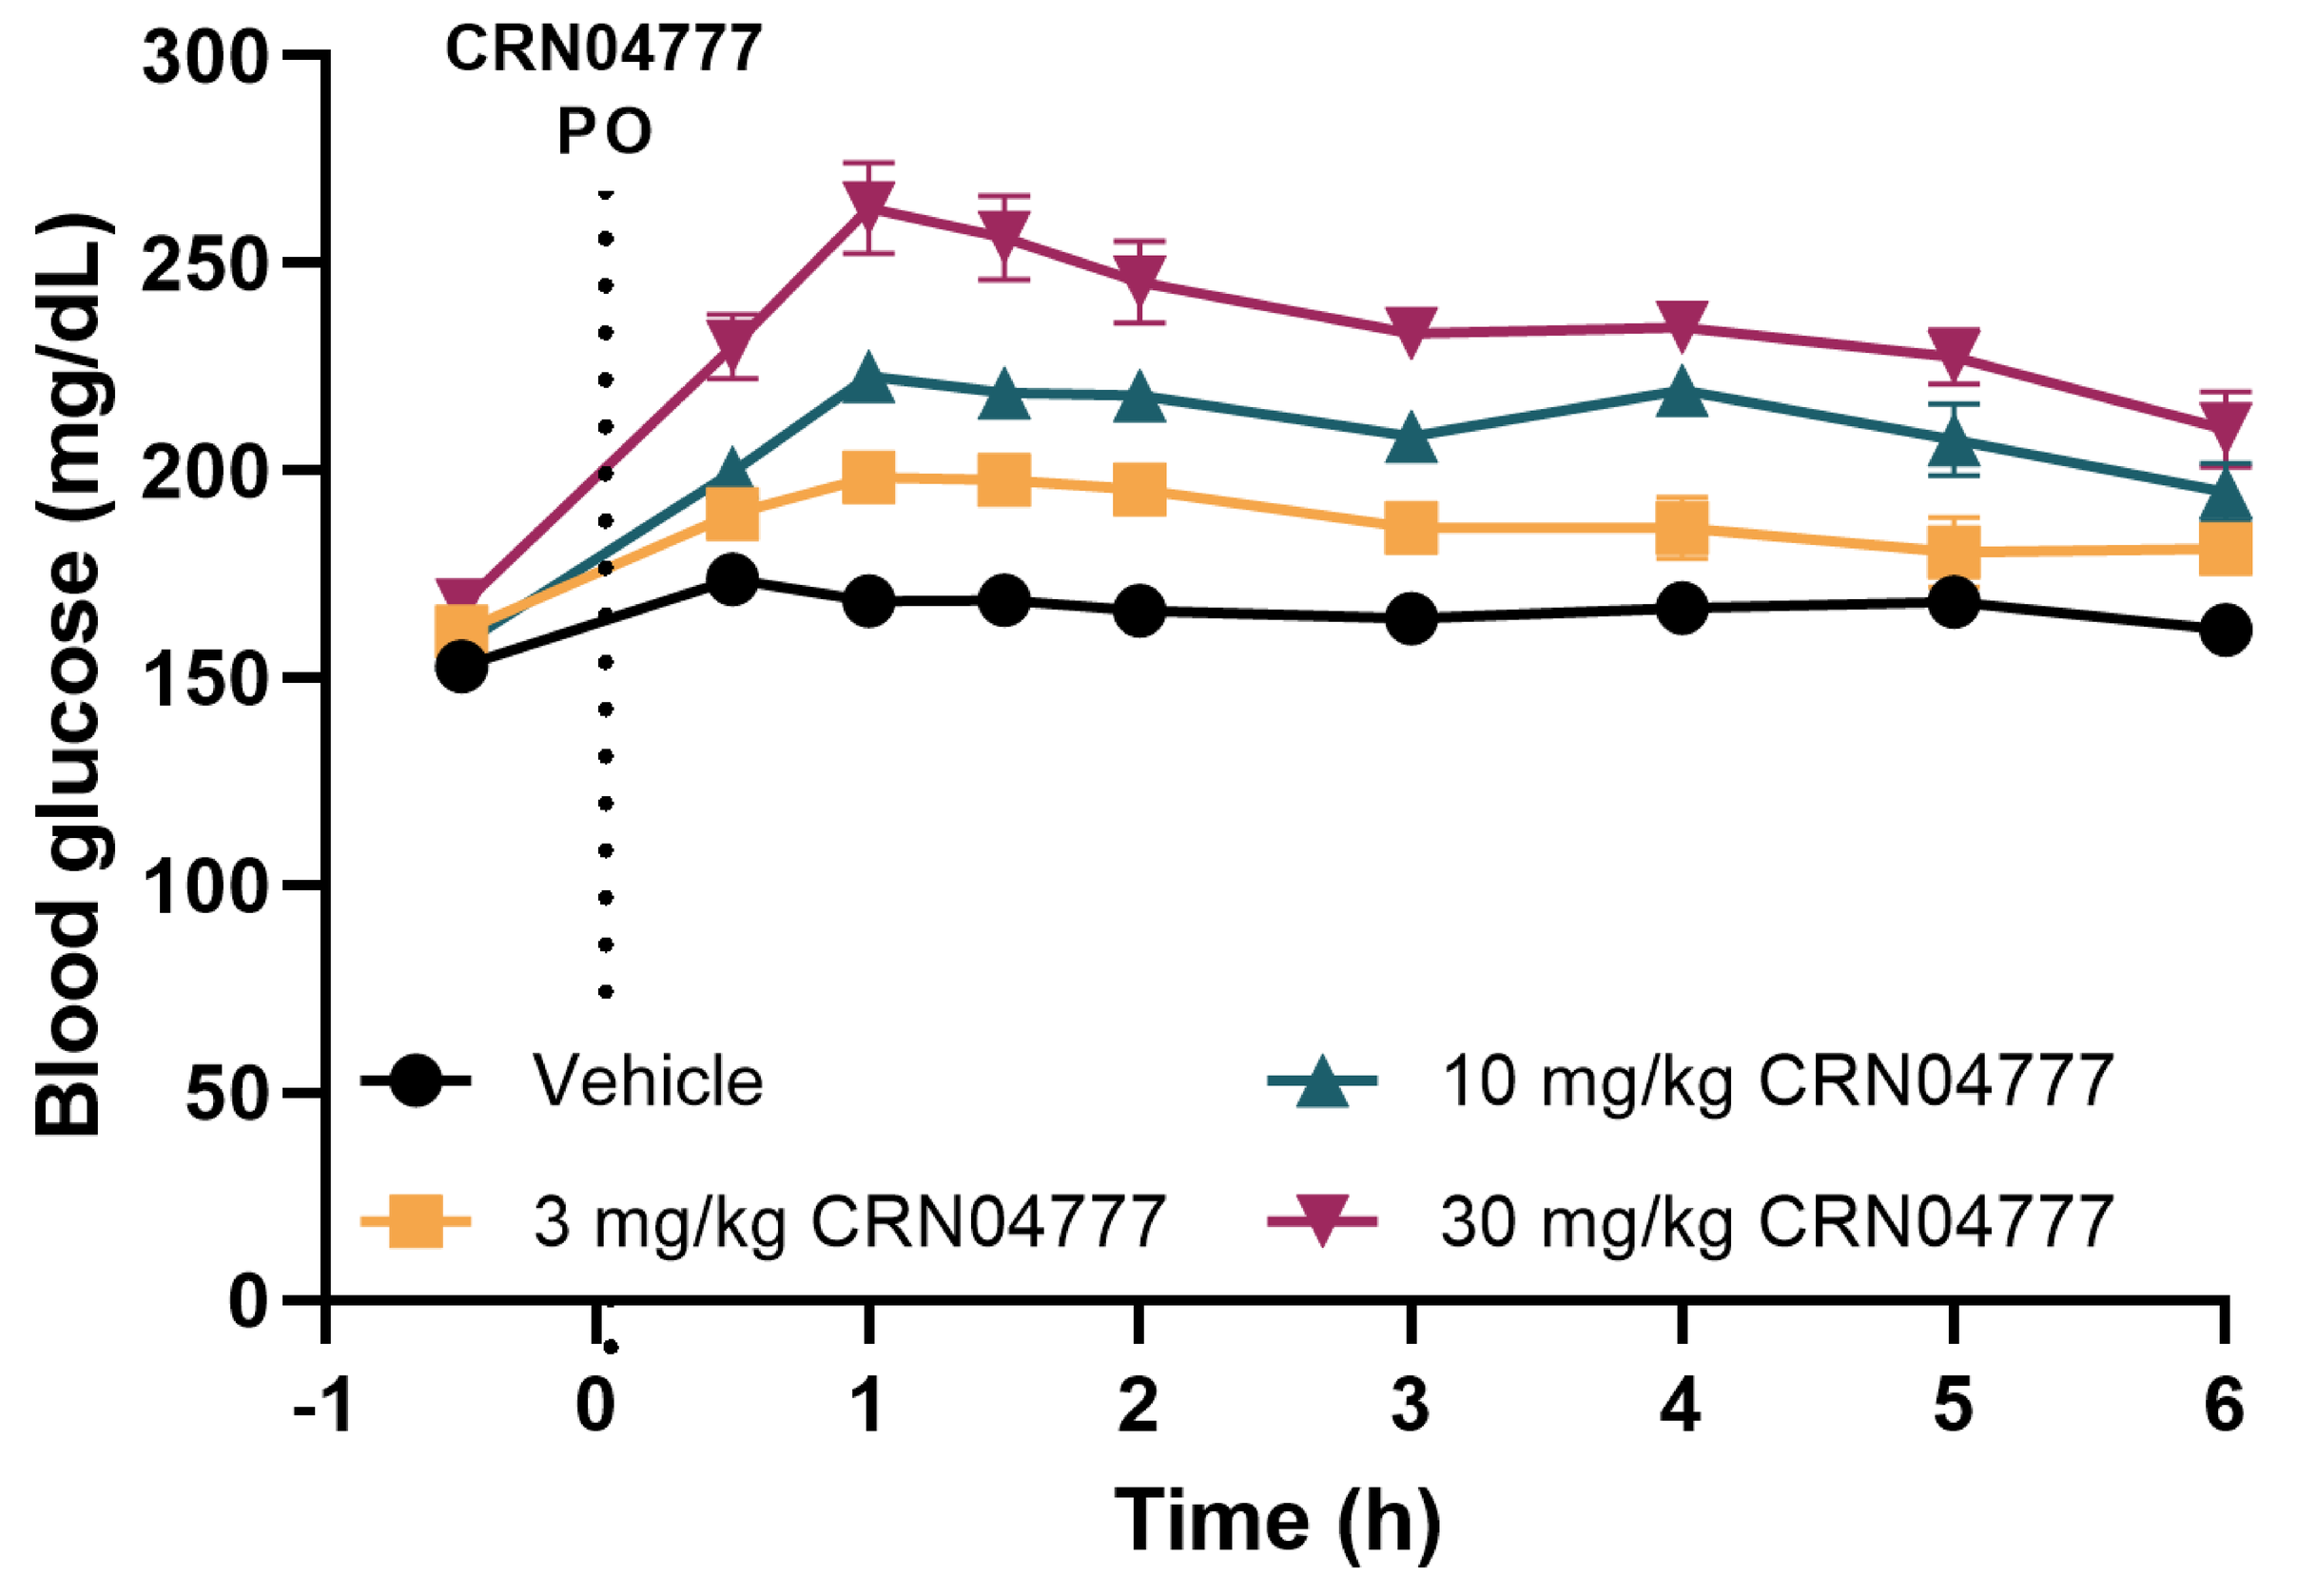
CRN04777 PO
Blood glucose (mg/dL)
Time (h)
Vehicle
3 mg/kg CRN04777
10 mg/kg CRN04777
30 mg/kg CRN04777

Figure 3. Blood glucose was measured from the tail of male PND10 Sprague Dawley rats for up to 6 hours following a single oral dose of 3, 10, and 30 mg/kg CRN04777. A significant increase in the area under the curve of the blood glucose response from 0.5 to 6 hours post dose was observed with administration of ≥3 mg/kg CRN04777.

CRN04777 Dose Dependently Increases Blood Glucose in a Neonatal Rat Model of Glyburide-Induced Hypoglycemia

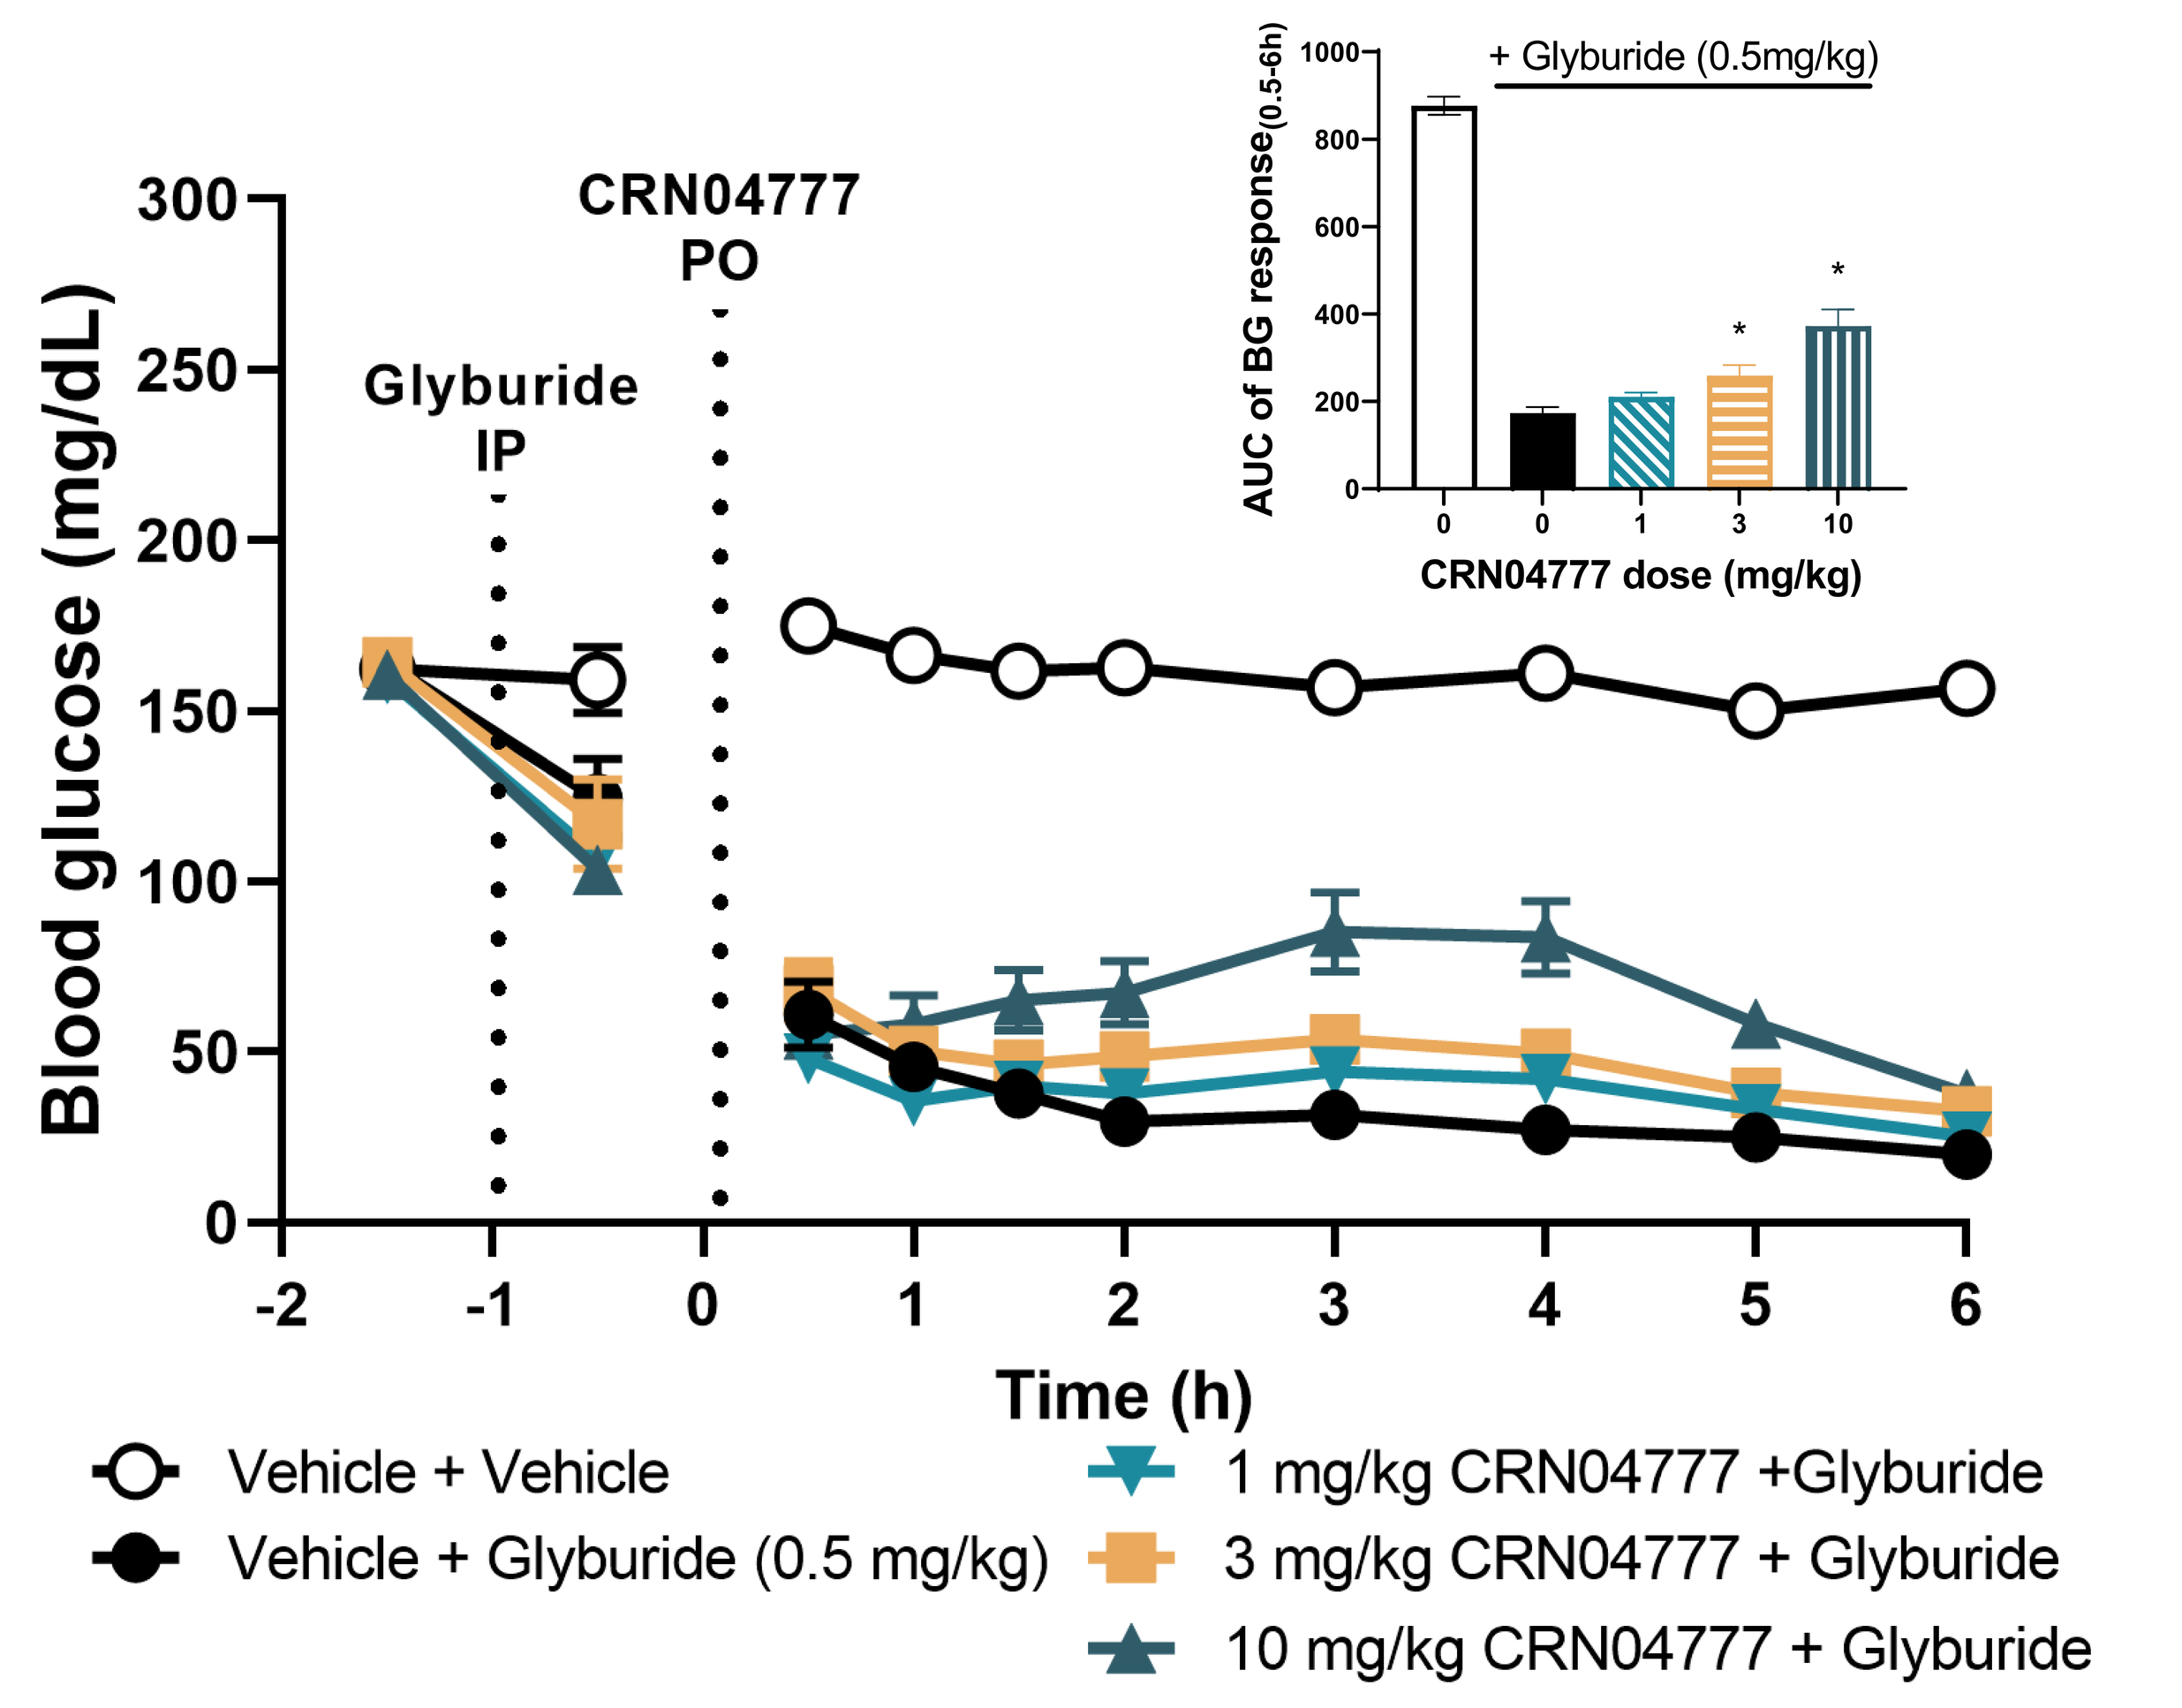
Glyburide IP
CRN04777 PO
Blood glucose (mg/dL)
Time (h)
+ Glyburide (0.5mg/kg)
AUC of BG response(0.5-6h)
CRN04777 dose (mg/kg)
Vehicle + Vehicle
Vehicle + Glyburide (0.5 mg/kg)
1 mg/kg CRN04777 +Glyburide
3 mg/kg CRN04777 + Glyburide
10 mg/kg CRN04777 + Glyburide

Figure 4. Glyburide intraperitoneally administered at 0.5 mg/kg decreased blood glucose levels to <60 mg/dL in male PND10 Sprague Dawley rats. Oral administration of ≥3 mg/kg CRN04777 one hour after glyburide significantly increased the area under the curve of the blood glucose response from 0.5 to 6 hours post CRN04777 dose.

CRN04777 Reduces Glyburide-induced Hyperinsulinemia and Raises Blood Glucose in Neonatal Rats

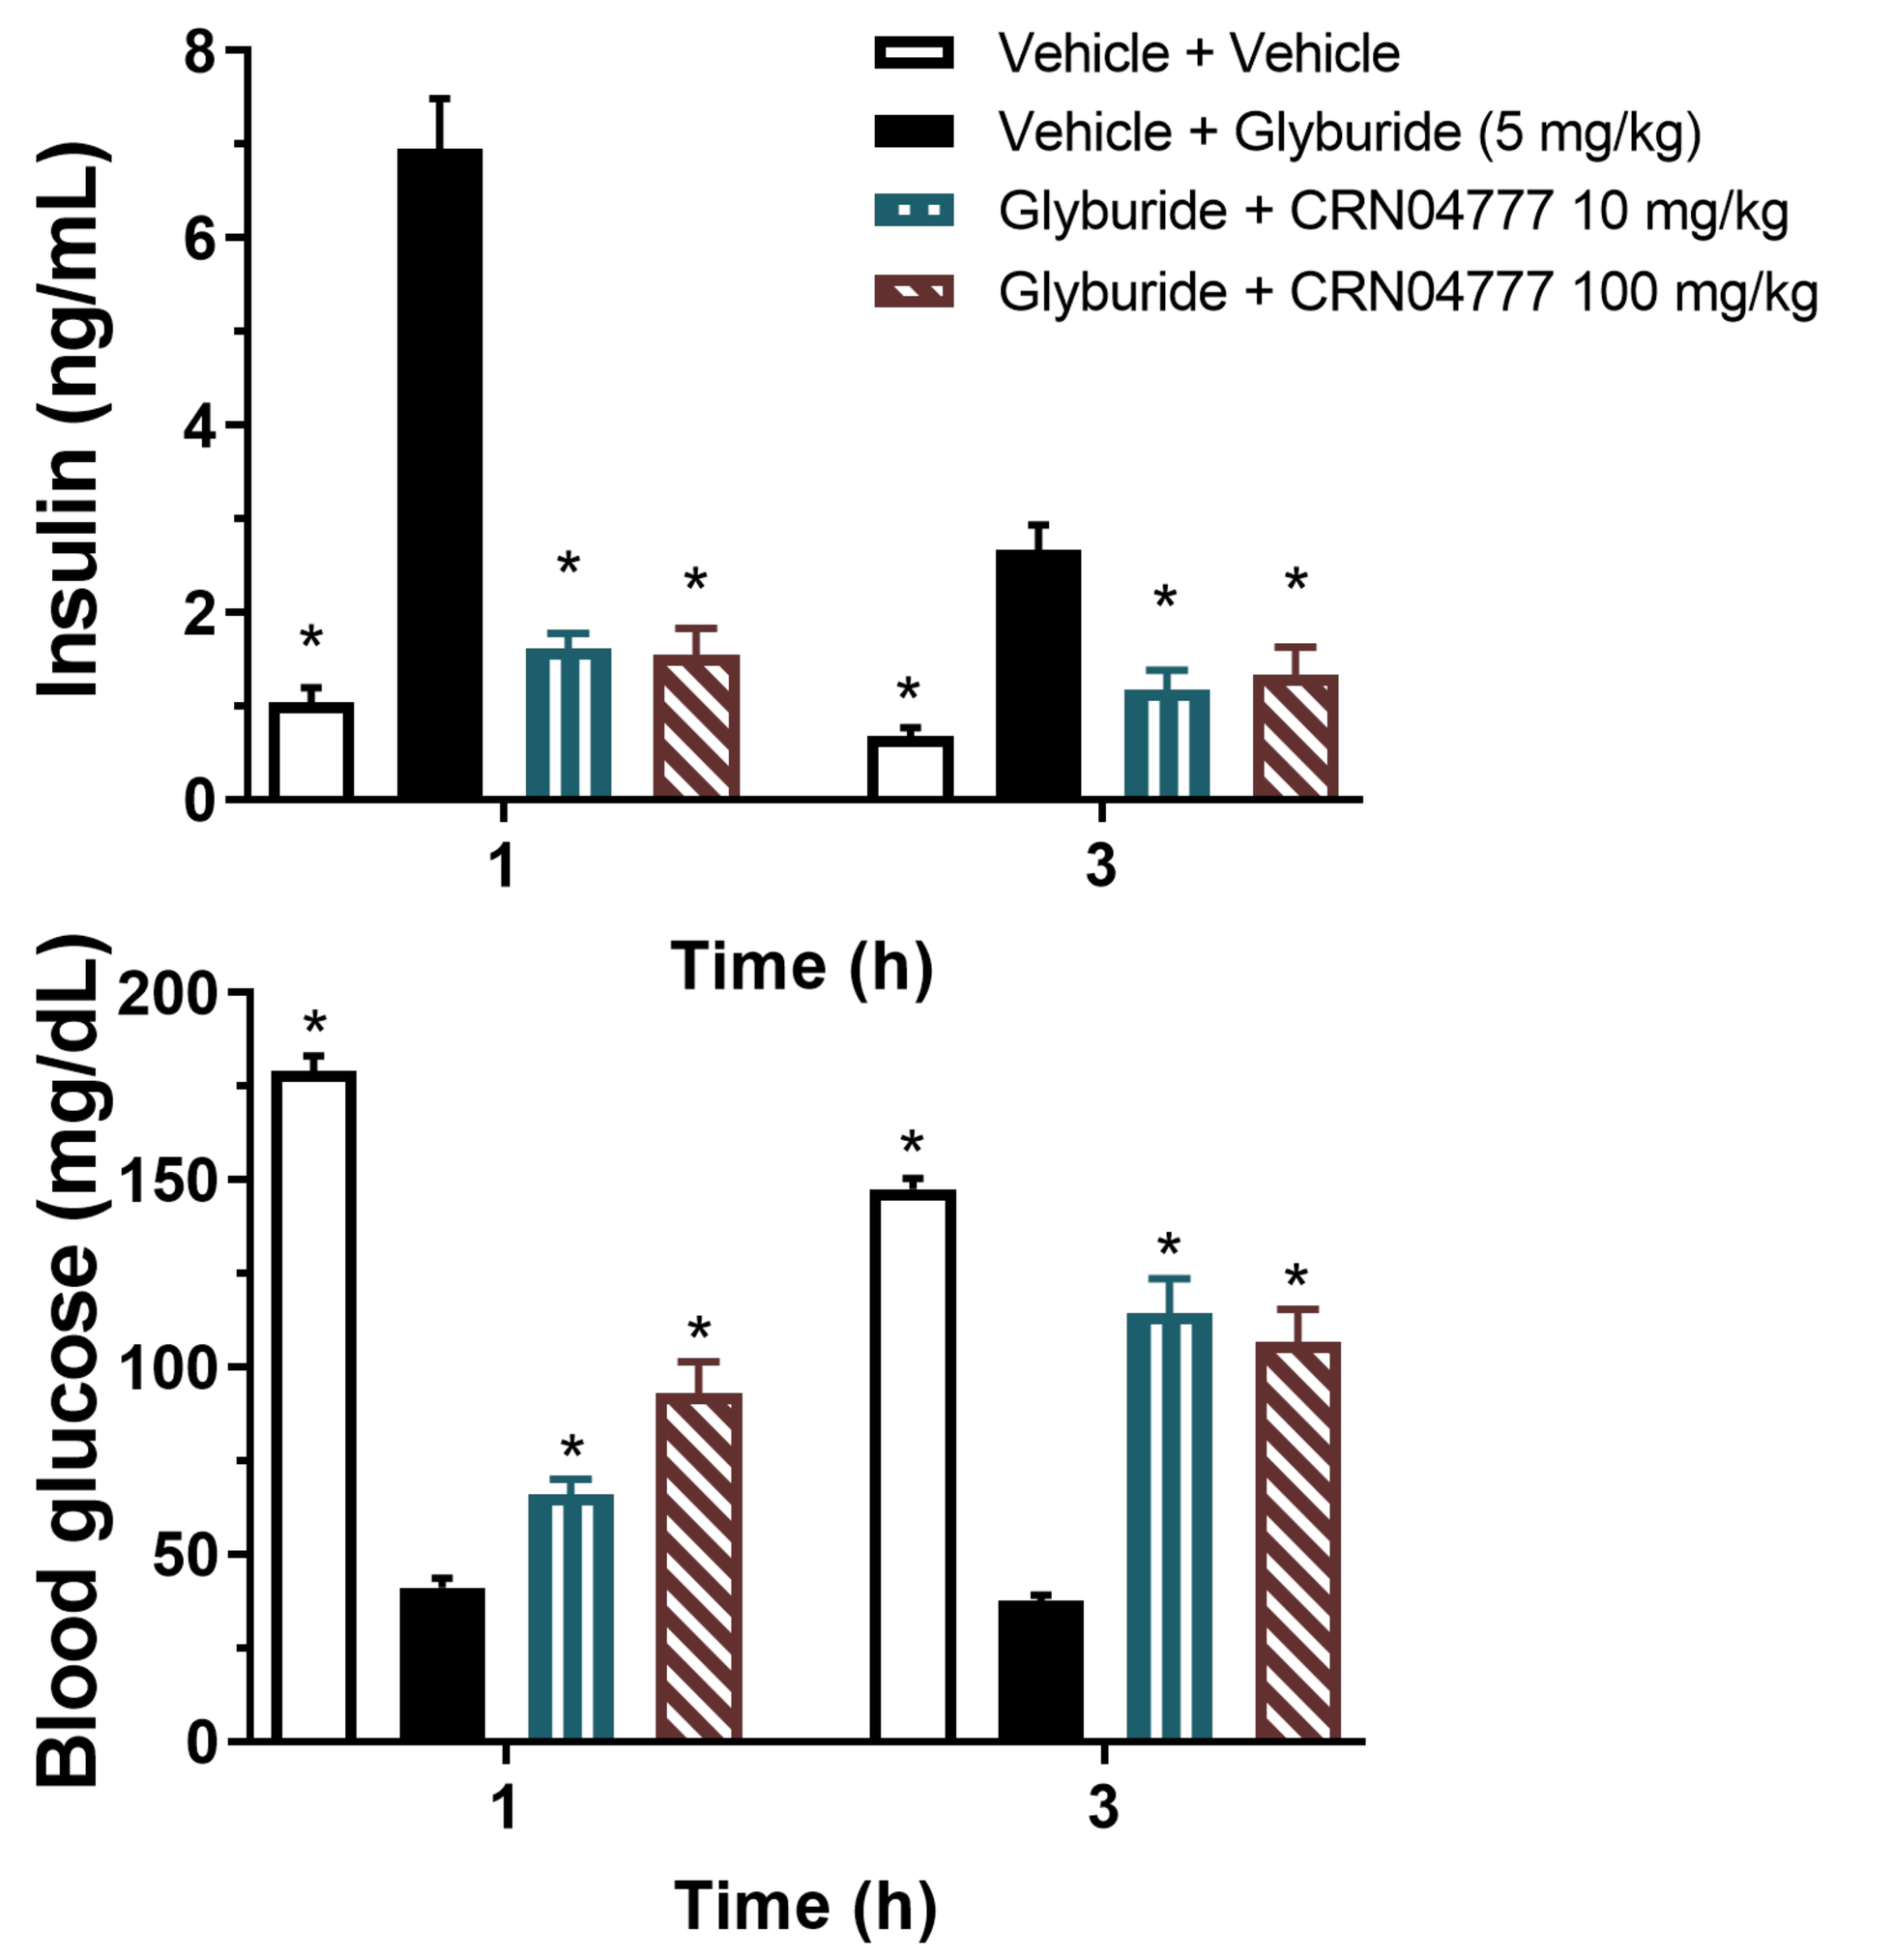
Vehicle + Vehicle
Vehicle + Glyburide (5 mg/kg)
Glyburide + CRN04777 10 mg/kg
Glyburide + CRN04777 100 mg/kg
Insulin (ng/mL)
Blood glucose (mg/dL)
Time (h)

Figure 5. Glyburide intraperitoneally administered at 5 mg/kg increased insulin secretion and decreased blood glucose in male PND10 Sprague Dawley rats. Oral administration of ≥10 mg/kg CRN04777 one hour after glyburide administration suppressed insulin and increased blood glucose for up to 3 hours post CRN04777 dose.

Effect of CRN04777 on Blood Glucose is Sustained Over Five Days of Once-Daily Oral Administration in Neonatal Rats

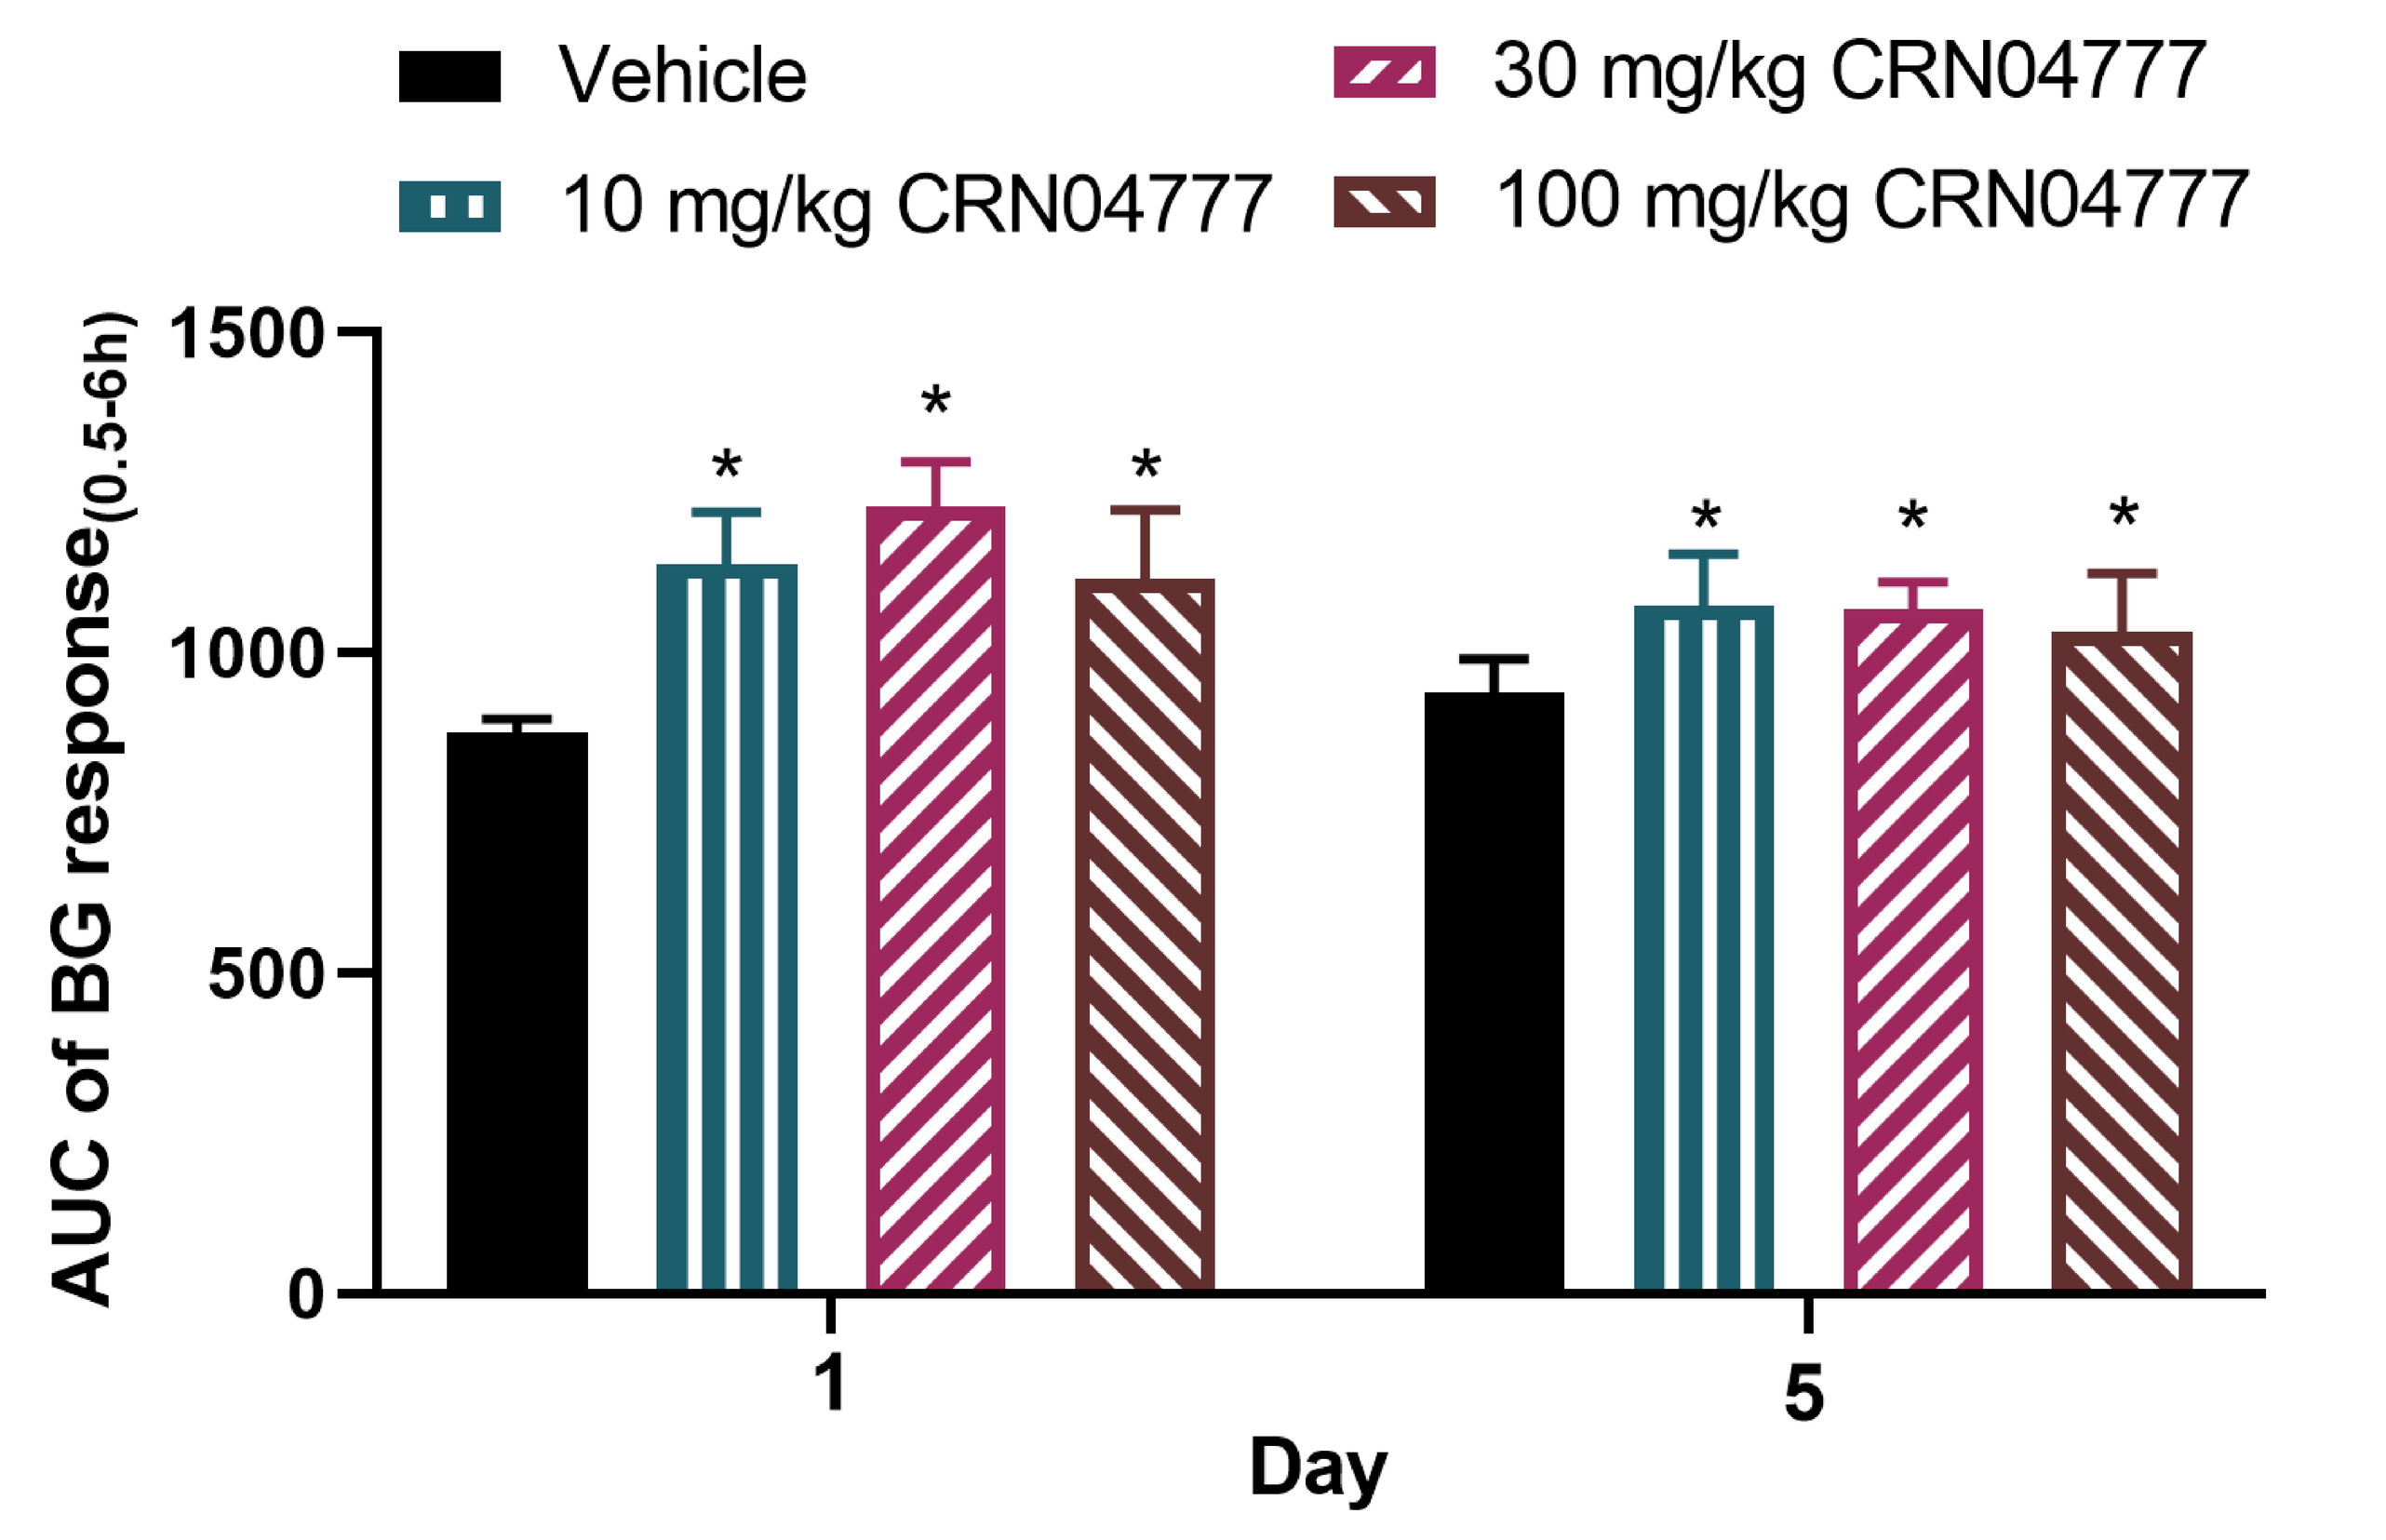
Vehicle
10 mg/kg CRN04777
30 mg/kg CRN04777
100 mg/kg CRN04777
AUC of BG response(0.5-6h)
Day

Figure 6. Blood glucose was measured from the tail in male PND7 Sprague Dawley rats for up to 6 hours on Day 1 and Day 5 of a 5-day repeat oral administration study with 10, 30 and 100 mg/kg CRN04777. A significant increase in area under the curve of the blood glucose response from 0.5 to 6 hours post dose was observed on Day 1 and Day 5 with oral administration of ≥10 mg/kg CRN04777.

Conclusions

Crinetics has discovered an orally available nonpeptide SST5 agonist, CRN04777, that:

- **is potent and selective for human SST5 receptors**
- **suppresses insulin and increases blood glucose in neonatal rats**
- **shows sustained efficacy in increasing blood glucose in neonatal rats**
- **rescues glyburide-induced hypoglycemia in neonatal rats**

Phase 1 human clinical trials with CRN04777 are currently ongoing.

Presented at: ESPE 2021
Diabetes and insulin
Melissa Fowler
P2-126